P912

ECCMID 2010

Disk Diffusion and MIC Quality Control Ranges for BC-3205 and BC-3781, Novel Pleuromutilin Class Antimicrobials

J.E. Ross[1], S.D. Putnam[1], R. Novak[2], S. Paukner[2], Z. Ivezic-Schoenfeld[2], H.S. Sader[1], R.N. Jones[1]

[1] JMI Laboratories, North Liberty, Iowa, USA [2] Nabriva Therapeutics AG, Vienna, Austria

Nabriva Therapeutics AG
Leberstrasse 20
1110 Vienna, Austria
www.nabriva.com

Amended Abstract

Objectives: To establish the disk diffusion (DD) and MIC quality control (QC) ranges for BC-3205 and BC-3781, novel semi-synthetic pleuromutilin derivatives in the early stage clinical development for oral treatment of skin and skin structure infections (SSSI).

Methods: These QC studies for the 20 µg BC-3205 and 20 µg BC-3781 disk and broth microdilution method follow the CLSI M02-A10 (2009), M07-A8 (2009) and M23-A3 (2008) document using eight laboratories, two lots of BC-3205 disks, two lots of BC-3781 disks, and three or more different medium lots. The results are presented as proposed QC ranges for four ATCC strains: *S. aureus* ATCC 25923, *H. influenzae* ATCC 49247, *S. pneumoniae* ATCC 49619 and *S. aureus* ATCC 29213 (MIC only). DD and MIC QC ranges for BC-3205 and BC-3781 were established per a CLSI M23-A3 study design. Ten replicates with each of 3 QC strains produced ≥1,440 zone diameters for two disk lots of each pleuromutilin (20 µg disk) provided by the Mast Group. Clindamycin (CL; 2 µg disk), azithromycin (AZ; 15 µg disk) and linezolid (LZ; 30 µg disk) were utilized as control agents for DD, while retapamulin, AZ and levofloxacin were the control agents for MIC testing.

Results: Proposed QC ranges for BC-3205 are listed in the Table. No significant differences (>1 mm) were noted between media or disk lots when testing BC-3205, BC-3781 or control agents. *S. aureus* ATCC 25923 produced larger variations between laboratories with the most extreme laboratory modes having a 7 mm difference. One laboratory submitted significantly larger zone diameters for *H. influenzae* ATCC 49247 and was excluded from evaluation leaving seven laboratories for a valid CLSI QC study. MIC values for *H. influenzae* 49247 showed trailing endpoints of at least one dilution step. One laboratory was excluded from analysis due to outlier values. A 7 mm zone diameter is proposed for *S. pneumoniae* ATCC 49619 which includes 99.7 % of all results. Excluding one aberrant laboratory from the MIC testing analysis produced all seven participant results within the proposed 0.06–0.25 mg/l range. All but one MIC result for the control agents were within expected ranges, when applicable. The control disks (LZ, AZ, CL) provided a valid internal control for the study, with 97.7 to 100.0 % of zones within CLSI published QC ranges.

Conclusions: An acceptable QC range was established for the four QC organisms that will guide clinical and reference laboratories involved in clinical trials and facilitate the regulatory review process of BC-3205 and BC-3781.

QC organism	MIC/Disk diffusion zone diameters for BC-3205: Proposed range [mg/l / mm]	% in range
***S. aureus* ATCC 29213/25923**	**0.03–0.25 / 25–33**	**100.0 / 95.4**
***H. influenzae* ATCC 49247**	**1–4 / 18–24**	**81.8 / 91.6**
Excludes one participant	**1–4 / 18–24**	**87.6[a] / 100.0[a]**
***S. pneumoniae* ATCC 49619**	**0.06–0.25 / 20–26**	**100.0[a] / 99.7**

[a] One laboratory was excluded from evaluation, *H. influenzae* QC range for MICs was not acceptable.

Introduction

Two new pleuromutilin class compounds from Nabriva Therapeutics AG (Vienna, Austria), BC-3205 and BC-3781, are in clinical development for intravenous and/or oral treatment of skin and skin structure infections (SSSI) and community acquired pneumonia (CAP). Both compounds exhibit excellent activity against clinical pathogens identified in SSSI and CAP, including methicillin-resistant *Staphylococcus aureus* (MRSA). The activity for both Nabriva agents is not adversely influenced by resistance to methicillin among staphylococci or vancomycin among enterococci. The establishment of these disk diffusion (DD) and broth microdilution (BMD) quality control (QC) ranges will guide clinical and reference laboratories involved in clinical trials and facilitate the regulatory review process worldwide.

BMD and DD QC studies of BC-3205 and BC-3781 were performed following the Clinical Laboratory Standards Institute (CLSI) M23-A3 (2008) guideline document using eight laboratories, different manufacturers of media and three antimicrobial control agents. The results are presented as proposed QC ranges in mg/l concentrations and zone diameters measured in mm for four American Type Culture Collection (ATCC) strains: *S. aureus* ATCC 29213 (BMD only), *S. aureus* ATCC 25923 (DD only), *Haemophilus influenzae* ATCC 49247, and *Streptococcus pneumoniae* ATCC 49619.

Materials and Methods

A total of eight laboratories were recruited to provide data for this QC investigation. For the BMD study, four cation-adjusted Mueller-Hinton (MH) broth media included lots produced by Difco Laboratories (Detroit, MI), Becton-Dickinson (BD; Sparks, MD), and Oxoid (Hampshire, United Kingdom [UK]). Four cation-adjusted MH broth lots supplemented with 2-5 % lysed horse blood were also supplied by Difco, BD and Oxoid. BC-3205 and BC-3781 were provided by Nabriva Therapeutics; azithromycin, clindamycin and linezolid were acquired from Sigma-Aldrich (St. Louis, Missouri, USA). Panels were prepared by a certified GMP source (TREK Diagnostics Cleveland, Ohio, USA). Appropriate inoculum concentrations were established by performing colony counts from the broth microdilution trays which were subcultured onto drug-free agar plates.

Three lots of agar medium were used for the disk diffusion (DD) study from BD and Remel (Lenexa, KS, USA). The BC-3205 and BC-3781 disks (20 µg) were provided by Mast Group (Merseyside, UK; BC-3205 lots #257105 and #257106; BC-3781 lots #257108 and #257109). Internal QC was established using azithromycin (15 µg), clindamycin (2 µg) and linezolid (30 µg) disks obtained from BD. Ten replicates of each of the three ATCC strains produced at least 1,440 zone diameters for each investigational drug.

Results

- Colony counts for MIC testing averaged between 1.8×10^5 to 8.6×10^5 CFU/ml for *S. aureus* ATCC 29213, 1.1×10^5 to 8.6×10^5 CFU/ml for *S. pneumoniae* ATCC 49619 and 1.0×10^5 to 1.2×10^6 CFU/ml for *H. influenzae* ATCC 49247.
- Figure 1 shows a consistent bimodal MIC distribution (0.06 and 0.12 mg/l) for *S. aureus* ATCC 29213 for BC-3205 where a four dilution QC range is needed (0.03–0.25 mg/l). Figure 2 shows BC-3781 with a clear mode at 0.12 mg/l and a three dilution range of 0.03–0.12 mg/l was proposed.
- Figures 3 and 4 show *H. influenzae* ATCC 49247 results for BC-3205 and BC-3781, both with a three dilution QC range. The BC-3205 range of 1 to 4 mg/l includes only 87.6 % of results from seven laboratories (one laboratory was excluded as an outlier; Table 1). The range for BC-3781 of 0.5 to 2 only included 94.3 % of all results excluding one laboratory. For both agents, a QC range that included at least 95 % of MIC values could not be achieved.
- *S. pneumoniae* ATCC 49619 (data not shown) has a proposed MIC QC range of 0.06-0.25 for BC-3205 which includes 100.0 % of results (excluding one outlier laboratory). BC-3781 has a proposed range of 0.06–0.5 with 98.6 % results included (excluding one laboratory).

Table 1. Inter- and intra-laboratory comparisons of the BC-3205 zone diameter results versus *Haemophilus influenzae* ATCC 49247 for an eight medical center protocol meeting the study design guidelines found in CLSI M23-A3 (2008)

Zone diameter [mm]	Disk lot[b] A	Disk lot[b] B	Occurrences by lot[c] A	Occurrences by lot[c] B	Occurrences by lot[c] C	Laboratory code (occurrences): A	B	C	D	E	F	G	H	Total
13														
14	1	2	3						3					3
15	11	13	12	12					24					24
16	8	5	5	8					13					13
17	0	0	0	0					0					0
18	2	1	2	1		1	1		0			1		3[a]
19	31	28	30	28	1	0	6	38	0		10	5		59[a]
20	62	48	38	44	28	12	24	22	4		25	20	3	110[a]
21	49	48	33	30	34	30	9		6		22	20	10	97[a]
22	28	47	13	16	46	11	9		6		2	9	38	75[a]
23	48	26	22	18	48	6	11		4	52	1	5	9	88[a]
24		8	2	3	3					8				8[a]
25														
Total	240	240	160	160	160	60	60	60	60	60	60	60	60	480
Median	21	21	20	21	22	21	20	19	16	23	20	21	22	21
Geomean	20.4	20.6	19.9	19.9	21.7	21.1	20.8	19.4	17.1	23.1	20.3	20.7	21.9	20.5
Range	10	11	11	10	6	6	6	2	10	2	5	6	4	11

[a] 91.6 % of qualified results are in proposed QC range of 18–24 mm. 100.0 % of qualified results are in the proposed QC range of 18–24 mm when Laboratory D is removed as an outlier.

Table 2. Summary of disk diffusion (DD) and MIC proposed QC ranges for BC-3205 and BC-3781

QC Organism	Proposed DD range in mm (% included in range) BC-3205	BC-3781	Proposed MIC range in mg/l (% included in range) BC-3205	BC-3781
S. aureus ATCC 25923	25–33 (95.4)	25–33 (97.3)	0.03–0.25 (100.0)	0.06–0.25 (100.0)
H. influenzae ATCC 49247	18–24 (100.0)	22–28 (99.8)	1–4 (87.6)	0.5–2 (94.3)
S. pneumoniae ATCC 49619	20–26 (99.2)	20–26 (96.0)	0.06–0.25 (100.0)	0.06–0.5 (98.6)

Figure 1. BC-3205 MIC distribution for *S. aureus* ATCC 29213

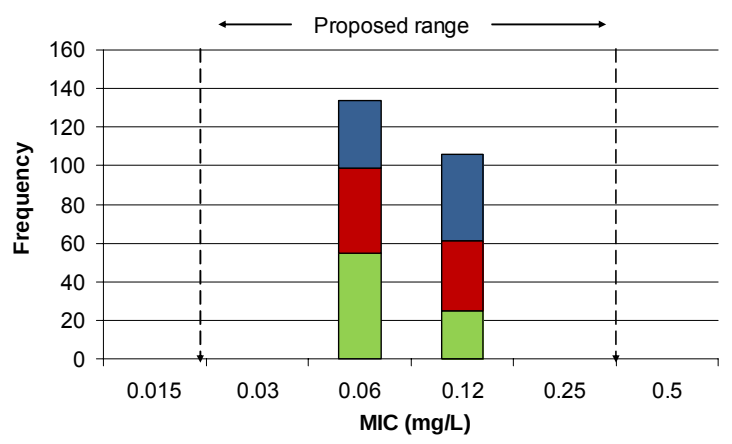

Proposed range
Frequency
MIC (mg/L)
0.015
0.03
0.06
0.12
0.25
0.5

Figure 2. BC-3781 MIC distribution for *S. aureus* ATCC 29213

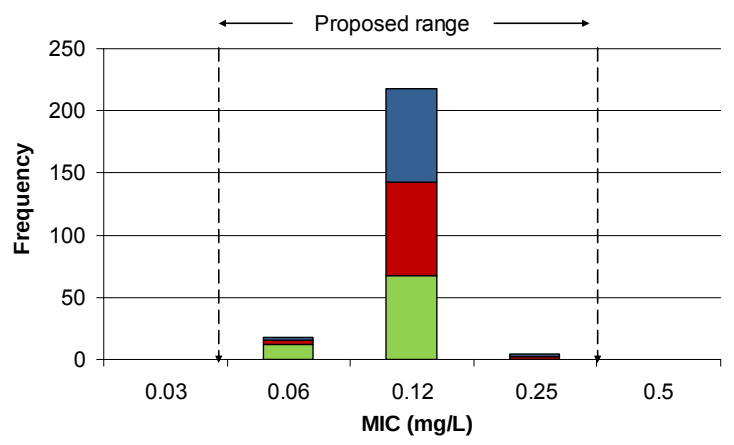

Proposed range
Frequency
MIC (mg/L)
0.03
0.06
0.12
0.25
0.5

Figure 3. BC-3205 MIC distribution for *H. influenzae* ATCC 49247

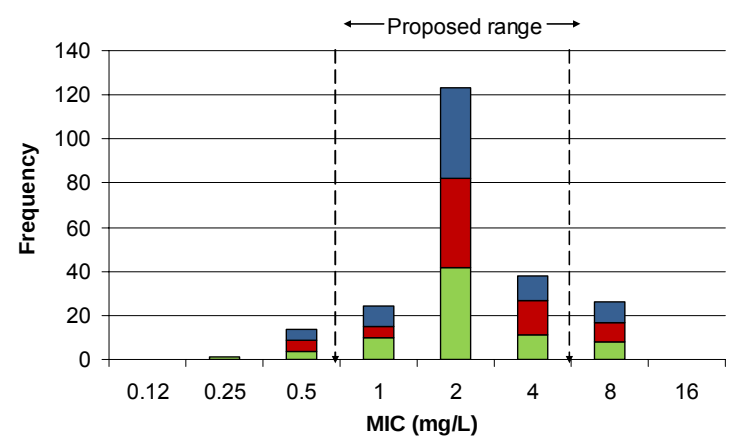

Proposed range
Frequency
MIC (mg/L)
0.12
0.25
0.5
1
2
4
8
16

Figure 4. BC-3781 MIC distribution for *H. influenzae* ATCC 49247

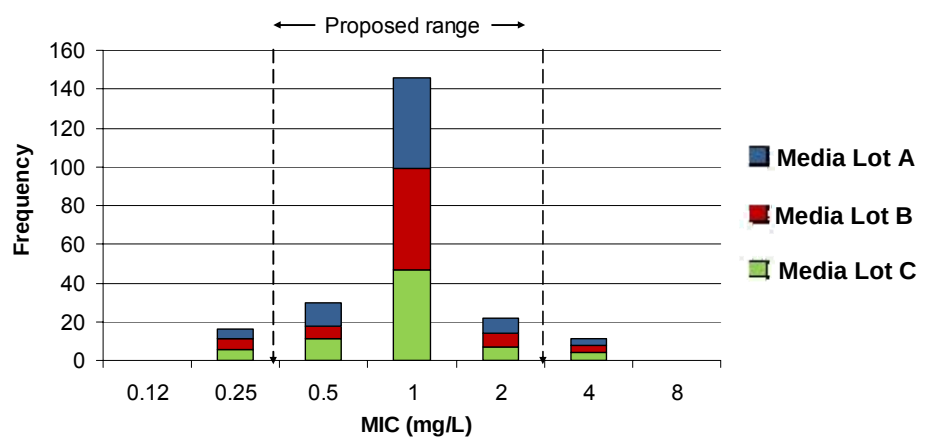

Proposed range
Frequency
MIC (mg/L)
0.12
0.25
0.5
1
2
4
8
Media Lot A
Media Lot B
Media Lot C

- The proposed DD QC ranges for BC-3205 against *S. aureus* ATCC 25923 (data not shown) needed a nine mm range of 25–33 mm to include at least 95 % of all zones (95.4 %). Very similar results were found for BC-3781 showing a range of 25–33 mm (97.3 %; Table 2).
- The DD QC ranges results for BC-3205 and *H. influenzae* ATCC 49247 are shown in Table 1 where a range of 18–24 mm is proposed. When the outlier laboratory is excluded from analysis, 100.0 % of the results fall within this QC range. For BC-3781, a range of 22–28 mm (99.8 %) is proposed with the same laboratory excluded.
- *S. pneumoniae* ATCC 49619 DD ranges for BC-3205 and BC-3781 are both proposed as 20–26 mm (99.2 % and 96.0 %, respectively; Table 2).
- No significant medium or disk lot variations were noted. Control agents tested provided acceptable internal controls, with each antimicrobial having ≥97 % of all values within the CLSI published range.

Conclusions

- The proposed QC ranges for DD and BMD methods showed that BC-3205 and BC-3781 have generally good inter- and intra-laboratory reproducibility for the commonly utilized control isolates, *S. aureus* ATCC 29213 and 25923, *H. influenzae* ATCC 49247 (DD only) and *S. pneumoniae* ATCC 49619 (see Table 2).
- These studies established CLSI method QC ranges that can be utilized to support accurate testing for susceptibility of BC-3205 and BC-3781 during clinical trials and continued product development.

Acknowledgements

The QC study sites and primary investigators were: The Cleveland Clinic Foundation, Cleveland, OH (G. Hall, Ph.D.); University of Texas Medical Center, Houston, TX (A. Wanger, Ph.D.); Massachusetts General Hospital, Boston, MA (M. J. Ferraro, M.D.); JMI Laboratories, North Liberty, IA (H. S. Sader, M.D., Ph.D.); Robert Wood Johnson Medical School, New Brunswick, NJ (M. Weinstein, M.D.); TREK Diagnostics, Cleveland, OH (C. Knapp, M.S.); University of Alberta, Edmonton, Canada (R. Rennie, Ph.D.); Duke University Medical Center, Durham, NC (L. B. Reller, M.D.), Wheaton Franciscan Laboratory, Wauwatosa, WI (E. Munson, Ph.D.) and the University of Washington Medical Center, Seattle, WA (S. Swanzy, B.S., M.T. [ASCP]).

Selected References

1. Clinical and Laboratory Standards Institute (2009). *M02-A10. Performance standards for antimicrobial disk susceptibility tests; approved standard - tenth edition*. Wayne, PA: CLSI.
2. Clinical and Laboratory Standards Institute (2009). *M07-A8. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard - eighth edition* Wayne, PA: CLSI.
3. Clinical and Laboratory Standards Institute (2008). *M23-A3. Development of in vitro susceptibility testing criteria and quality control parameters - third edition*. Wayne, PA: CLSI.
4. Clinical and Laboratory Standards Institute (2010). *M100-S20. Performance standards for antimicrobial susceptibility testing. 20th informational supplement* Wayne, PA: CLSI.