Pharmacokinetics-Pharmacodynamics (PK-PD) Target Attainment Analyses to Support ZTI-01 (Fosfomycin for Injection) Dose Selection for Patients with Complicated Urinary Tract Infections (cUTI)

S. M. Bhavnani[1], M. Trang[1], C. M. Rubino[1], A. J. Lepak[2], D. R. Andes[2], R. K. Flamm[3], K. Sweeney[4], E. J. Ellis-Grosse[4], P. G. Ambrose[1]

[1]Inst. for Clinical Pharmacodynamics Inc., Schenectady, NY

[2]Wisconsin Univ., Madison, WI

[3]JMI Lab., N. Liberty, IA

[4]Zavante Therapeutics, Inc., San Diego, CA

Background: ZTI-01, fosfomycin for injection, has *in vitro* activity against Gram -positive and -negative organisms, including carbapenem-resistant Enterobacteriaceae. ZTI-01 is currently in Phase 2/3 development for the treatment of patients with cUTI. PK-PD target attainment analyses were undertaken to provide support for the ZTI-01 dosing regimen to treat patients with cUTI.

Methods: Using parameter estimates from a population PK model (3-compartment model with zero-order input and 1st-order elimination), total-drug plasma concentration-time profiles were generated for 6000 simulated patients with varying creatinine clearance (CLcr, mL/min/1.73m^2). Simulated patients received ZTI-01 according to CLcr: 6 g IV q8h for > 50 mL/min/1.73 m^2, 4 g IV q8h for >40 to 50 mL/min/1.73 m^2, 6 g IV loading followed 3 g IV q8h for >30 to 40 mL/min/1.73 m^2, and 6 g IV load followed by 5 g IV q24h for >10 to 30 mL/min/1.73 m^2. Day 1 AUC_{0-24} was calculated. Percent probabilities of PK–PD target attainment by MIC and overall (i.e., weighted over the MIC distribution for 1091 Enterobacteriaceae isolates from the USA) were determined using median total-drug plasma AUC:MIC ratio targets associated with net bacterial stasis and a 1-$\log_{10}$ CFU reduction from baseline at 24 h from a neutropenic murine- thigh infection model for Enterobacteriaceae (19.1 and 41.6, respectively).

Results: Percent probabilities of attaining PK-PD targets associated with net bacterial stasis (Figure 1) and a 1 $\log_{10}$ CFU reduction were ≥98.3% at a MIC=64 mg/L and ≥97.3% at a MIC=32 mg/L, respectively, across renal function groups. These MIC values were 1 to 2 dilutions higher than the MIC_{90} of 16 mg/L for Enterobacteriaceae. Overall percent probabilities of PK-PD target attainment for each target were 97.8 and 95.5%, respectively.

Conclusions: Together with clinical outcome data, these data will provide support for ZTI-01 dosing recommendations and fosfomycin susceptibility breakpoints for Enterobacteriaceae.

Figure 1. Percent probabilities of PK-PD target attainment by MIC based on the AUC:MIC ratio target associated with net bacterial stasis among simulated patients by renal function group after administration of ZTI-01 6 g q8h-based dosing regimens, overlaid on the fosfomycin MIC distribution for Enterobacteriaceae

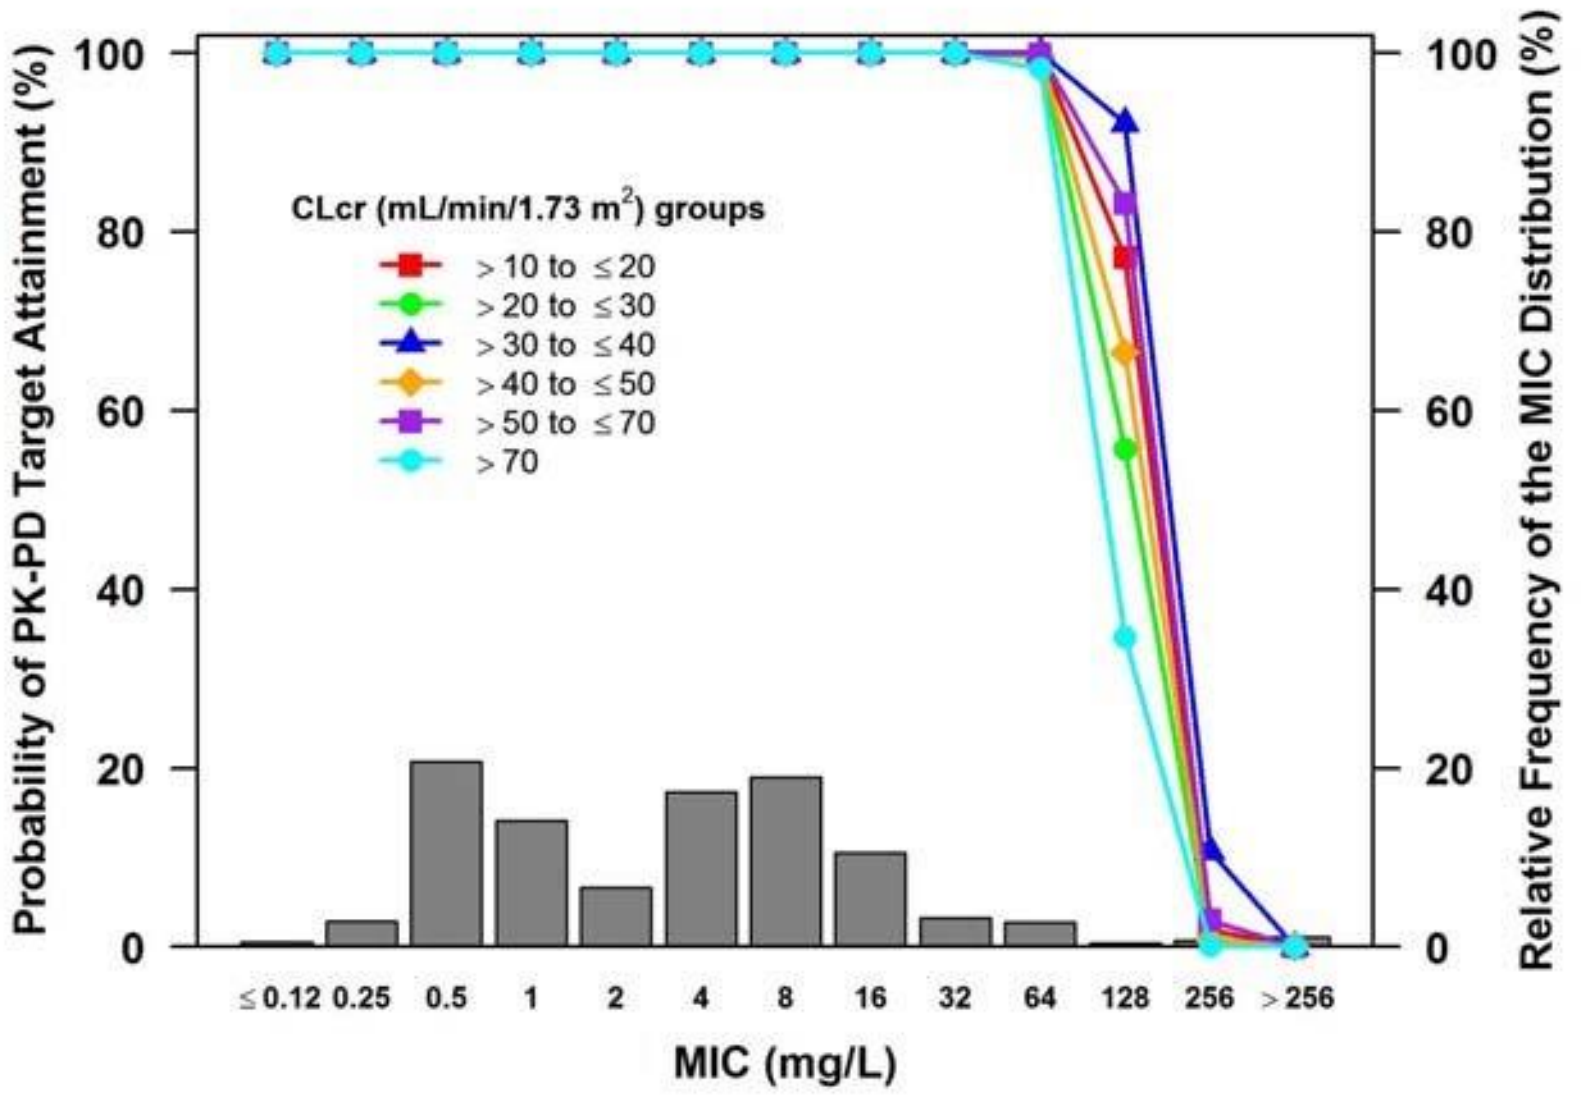
Probability of PK-PD Target Attainment (%)
Relative Frequency of the MIC Distribution (%)
CLcr (mL/min/1.73 m2) groups
> 10 to ≤ 20
> 20 to ≤ 30
> 30 to ≤ 40
> 40 to ≤ 50
> 50 to ≤ 70
> 70
0
20
40
60
80
100
≤ 0.12 0.25 0.5 1 2 4 8 16 32 64 128 256 > 256
MIC (mg/L)

Keywords: fosfomycin, pharmacokinetics/pharmacodynamics, ZTI-01

Sub-track: AAID03 Antimicrobial pharmacokinetics, pharmacodynamics and general pharmacology