Sunday – CIV-177

Efficacy of Lefamulin (LEF) Versus Moxifloxacin (MOX) by Pathogen in Adults With Community-Acquired Bacterial Pneumonia (CABP): Pooled Results From the LEAP 1 and LEAP 2 Phase 3 Clinical Trials

Elizabeth Alexander,[1] Lisa Goldberg,[1] Anita F. Das,[2] Christian Sandrock,[3] Susanne Paukner,[4] Jennifer Schranz,[1] Steven P. Gelone,[1] Thomas M. File Jr[5]

[1]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA; [2]Das Consulting, Guerneville, CA, USA; [3]UC Davis School of Medicine, Sacramento, CA, USA; [4]Nabriva Therapeutics GmbH, Vienna, Austria; [5]Summa Health, Akron, OH, USA

Presented by
Robbie Stevens
Phone: 860-235-5558
Email: Robbie.Stevens@Nabriva.com

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION

- Community-acquired bacterial pneumonia (CABP) is associated with substantial morbidity, mortality, and economic burden and is among the leading causes of infection-related death in the United States[1,2]
- Increasing rates of bacterial resistance[2] and safety issues associated with fluoroquinolones have created a need for new treatment options[3,4]
- Lefamulin (LEF), a first-in-class pleuromutilin for intravenous (IV) and oral use in humans, inhibits protein synthesis by binding to the central part of the peptidyl transferase center of the 50S ribosomal subunit by forming 4 hydrogen bonds and other interactions that prevent the correct tRNA positioning in the A- and P-sites[5]
- LEF has potent in vitro activity against *Streptococcus pneumoniae*, *Haemophilus influenzae*, *Moraxella catarrhalis*, and *Staphylococcus aureus*, as well as the atypical pathogens *Mycoplasma pneumoniae*, *Chlamydophila pneumoniae*, and *Legionella pneumophila*; LEF activity is unaffected in vitro by an organism's resistance to other major antibiotic classes[6-10]
- LEF has predictable pharmacokinetics after oral and IV administration, with rapid plasma absorption[11] and comparable penetration in the epithelial lining fluid of the lung in both fed and fasted states[12]
- The favorable pharmacokinetics and spectrum of activity of LEF led to its investigation in 2 phase 3 trials in adults with CABP
 - The Lefamulin Evaluation Against Pneumonia (LEAP) 1 study evaluated the efficacy and safety of LEF as monotherapy, with an IV-to-oral switch option, compared with moxifloxacin (MOX) (± linezolid)[13]
 - The LEAP 2 study evaluated the efficacy and safety of oral LEF monotherapy compared with oral MOX monotherapy[14]
- We report efficacy outcomes by baseline pathogen in pooled LEAP 1 and LEAP 2 analyses

METHODS

Study Design

- Both studies were prospective, randomized, double-blind, double-dummy, phase 3 trials[13,14]
- Patients in LEAP 1 and LEAP 2 were enrolled at 66 centers (18 countries) and 99 centers (19 countries), respectively **(Figure 1)**
 - In LEAP 1, patients were randomized to receive LEF 150 mg IV every 12 hours (q12h) for 5–7 days or MOX 400 mg IV every 24 hours (q24h) for 7 days
 - Patients could switch to oral therapy (LEF 600 mg q12h or MOX 400 mg q24h) after 6 IV doses of study drug (~3 days) if predefined improvement criteria were met
 - In LEAP 2, patients were randomized to receive oral LEF 600 mg q12h for 5 days or oral MOX 400 mg q24h for 7 days

Figure 1. LEAP 1 and LEAP 2 Study Design

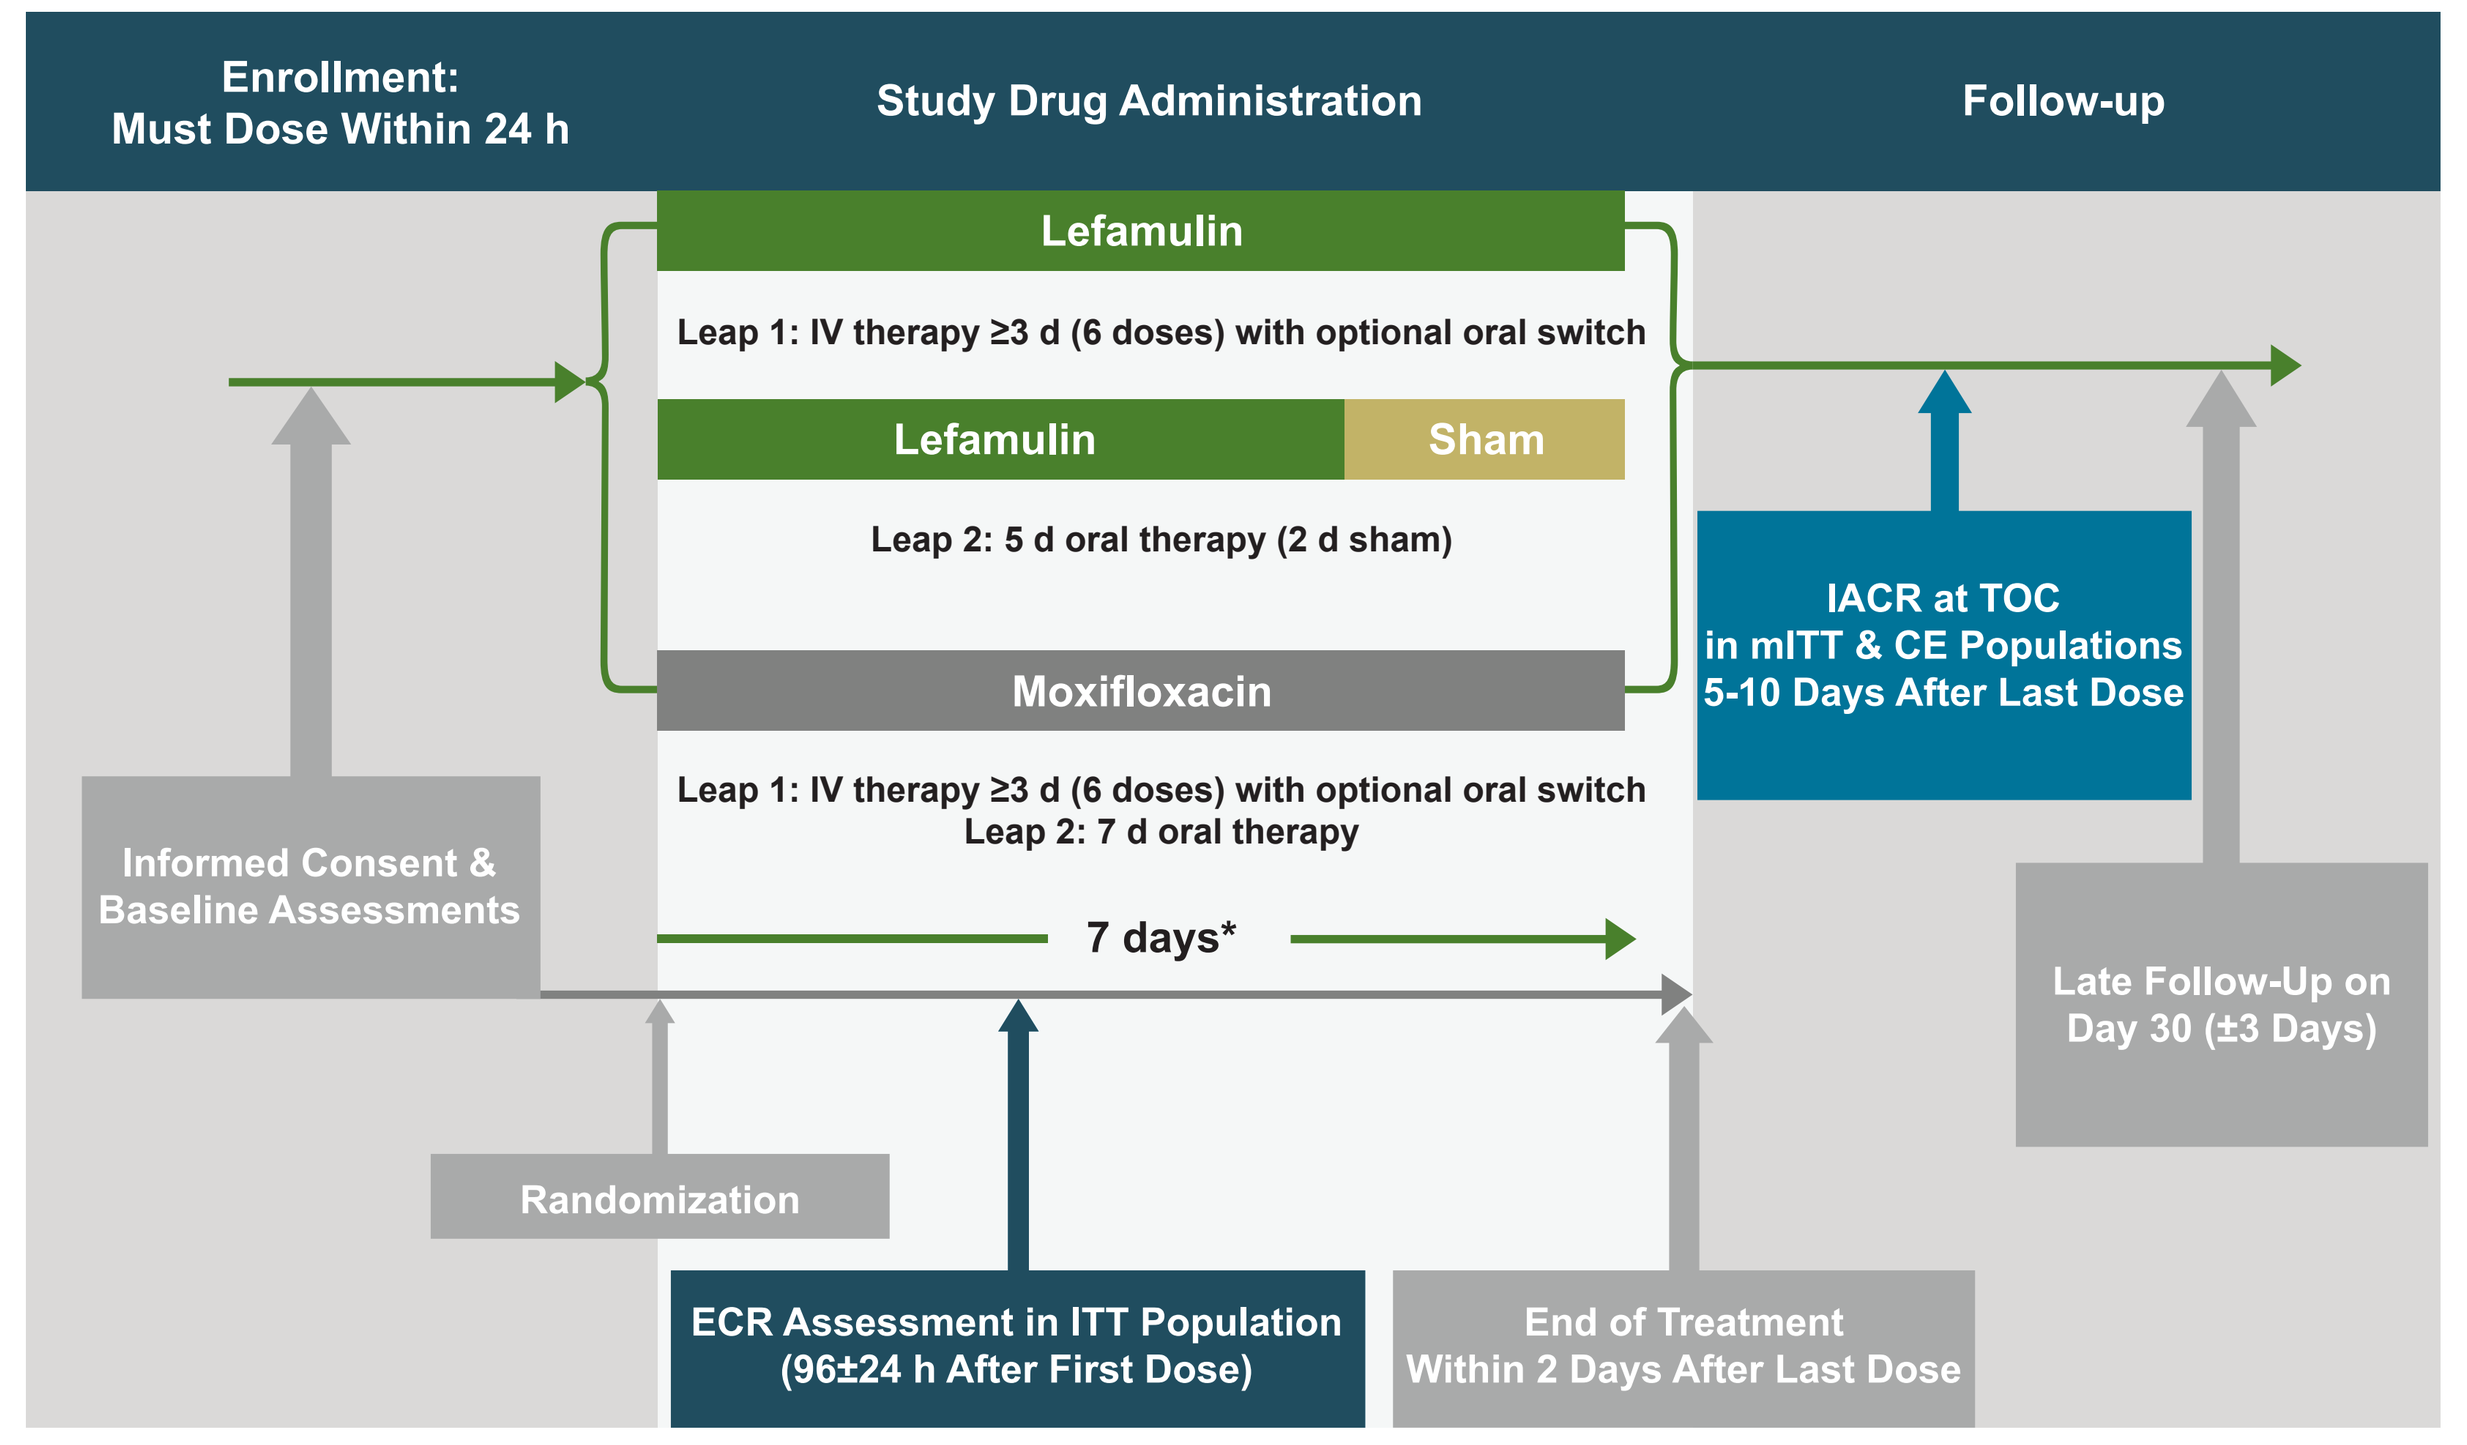
Enrollment: Must Dose Within 24 h
Study Drug Administration
Follow-up
Lefamulin
Leap 1: IV therapy ≥3 d (6 doses) with optional oral switch
Lefamulin
Sham
Leap 2: 5 d oral therapy (2 d sham)
Moxifloxacin
Leap 1: IV therapy ≥3 d (6 doses) with optional oral switch
Leap 2: 7 d oral therapy
7 days*
Informed Consent & Baseline Assessments
Randomization
ECR Assessment in ITT Population (96±24 h After First Dose)
End of Treatment Within 2 Days After Last Dose
IACR at TOC in mITT & CE Populations 5-10 Days After Last Dose
Late Follow-Up on Day 30 (±3 Days)

CABP=community-acquired bacterial pneumonia; CE=clinically evaluable (patients who met predefined specified criteria related to protocol adherence); ECR=early clinical response (patient assessed as responder if alive, showed improvement in ≥2 CABP signs and symptoms, no worsening in any CABP sign or symptom, and no receipt of a concomitant nonstudy antibiotic for the current CABP episode); IACR=investigator assessment of clinical response (patients assessed as success if alive, with signs and symptoms of CABP resolved or improved such that no additional antibacterial therapy was administered for CABP); ITT=intent to treat (all randomized patients); IV=intravenous; mITT= modified ITT (all randomized patients who received ≥1 dose of study drug); TOC=test-of-cure visit.
*In LEAP 1, if methicillin-resistant *Staphylococcus aureus* (MRSA) was suspected, linezolid or placebo was added to moxifloxacin or lefamulin therapy, respectively; if MRSA was confirmed, treatment duration was 10 days. A total of 14/275 (5.1%) MOX patients and 9/276 (3.3%) LEF patients received linezolid and linezolid placebo, respectively, because of suspected MRSA at baseline. The original protocol indicated a 5-day lefamulin treatment period but was later adjusted to 7 days to reduce medication errors and limit the burden on study sites.

Patients and Assessments

- Adults with CABP of Pneumonia Outcomes Research Team (PORT) risk class ≥III or II–IV were eligible for LEAP 1 and LEAP 2, respectively
- In both studies, the primary efficacy endpoint for the US Food and Drug Administration (FDA) was early clinical response (ECR) at 96±24 hours after first dose of study drug in the intent-to-treat (ITT) population
- The European Medicines Agency coprimary endpoints (FDA secondary endpoints) were investigator assessment of clinical response (IACR) at the test-of-cure (TOC) assessment 5–10 days after the last dose of study drug in the modified ITT and clinically evaluable populations
- In both studies, baseline pathogens were identified from specimens collected within 24 hours of the first dose of study drug
 - The microbiological intent-to-treat (microITT) population included all patients with a baseline CABP pathogen detected by ≥1 method **(Table 1)**
 - The microITT-2 population included patients with a CABP pathogen detected by a method other than real-time polymerase chain reaction (PCR)
 - Confirmatory identification and susceptibility testing of isolates, Gram staining of sputum, resistance gene determination, *S. pneumoniae* and *H. influenzae* serotyping, serology (≥4-fold increase in *L. pneumophila* antibody titer or *M. pneumoniae* or *C. pneumoniae* IgG serum antibody titer), and real-time PCR were performed by a central laboratory and by specialized laboratories (see Acknowledgments)

METHODS (continued)

Table 1. Diagnostic Modalities Used for the Identification of Baseline Pathogens of Interest

	microITT and microITT-2 Populations			microITT Population only	
Pathogen	Gram Stain and Culture*	Urinary Antigen Testing	Serological Testing	RQ- and RT-PCR from Sputum	RQ-PCR from NP Swab or OP Swab
Streptococcus pneumoniae	X†	X		X	X†
Staphylococcus aureus	X			X	
Haemophilus influenzae	X			X	
Moraxella catarrhalis	X			X	
Mycoplasma pneumoniae	X‡		X	X§	X‡
Legionella pneumophila	X‖	X	X	X§	
Chlamydophila pneumoniae	X¶		X	X§	

microITT=microbiological intent to treat; NP=nasopharyngeal; OP=oropharyngeal; PCR=polymerase chain reaction; RQ-PCR=real-time quantitative PCR; RT-PCR=real-time (qualitative) PCR.
*Specimens included blood and sputum; bronchoalveolar lavage and/or pleural fluid were cultured only if clinically indicated. Inclusion as a baseline pathogen for sputum samples also required a Gram stain with >25 polymorphonuclear cells per low power field and <10 squamous epithelial cells per low power field.
†Culture for *S. pneumoniae* was also performed on NP samples. RQ-PCR for *S. pneumoniae* was done on sputum samples and on NP swabs.
‡RT-PCR was done on OP samples. If RT-PCR was positive, OP samples were used for isolation of *M. pneumoniae* and for subsequent susceptibility testing; on some occasions, RT-PCR and culture were done in parallel.
§RT-PCR was performed for detection of *M. pneumoniae*, *L. pneumophila*, and *C. pneumoniae*.
‖Inclusion of *L. pneumophila* as a baseline pathogen did not require an appropriate morphology in the Gram stain.
¶Culture of *C. pneumoniae* by the local laboratories was allowed per protocol, but it was not cultured by any of the laboratories.

RESULTS

Figure 2. Baseline Pathogen Distribution (Pooled microITT Population [Combined Treatment Groups]; *N*=1012 Pathogens Identified*)

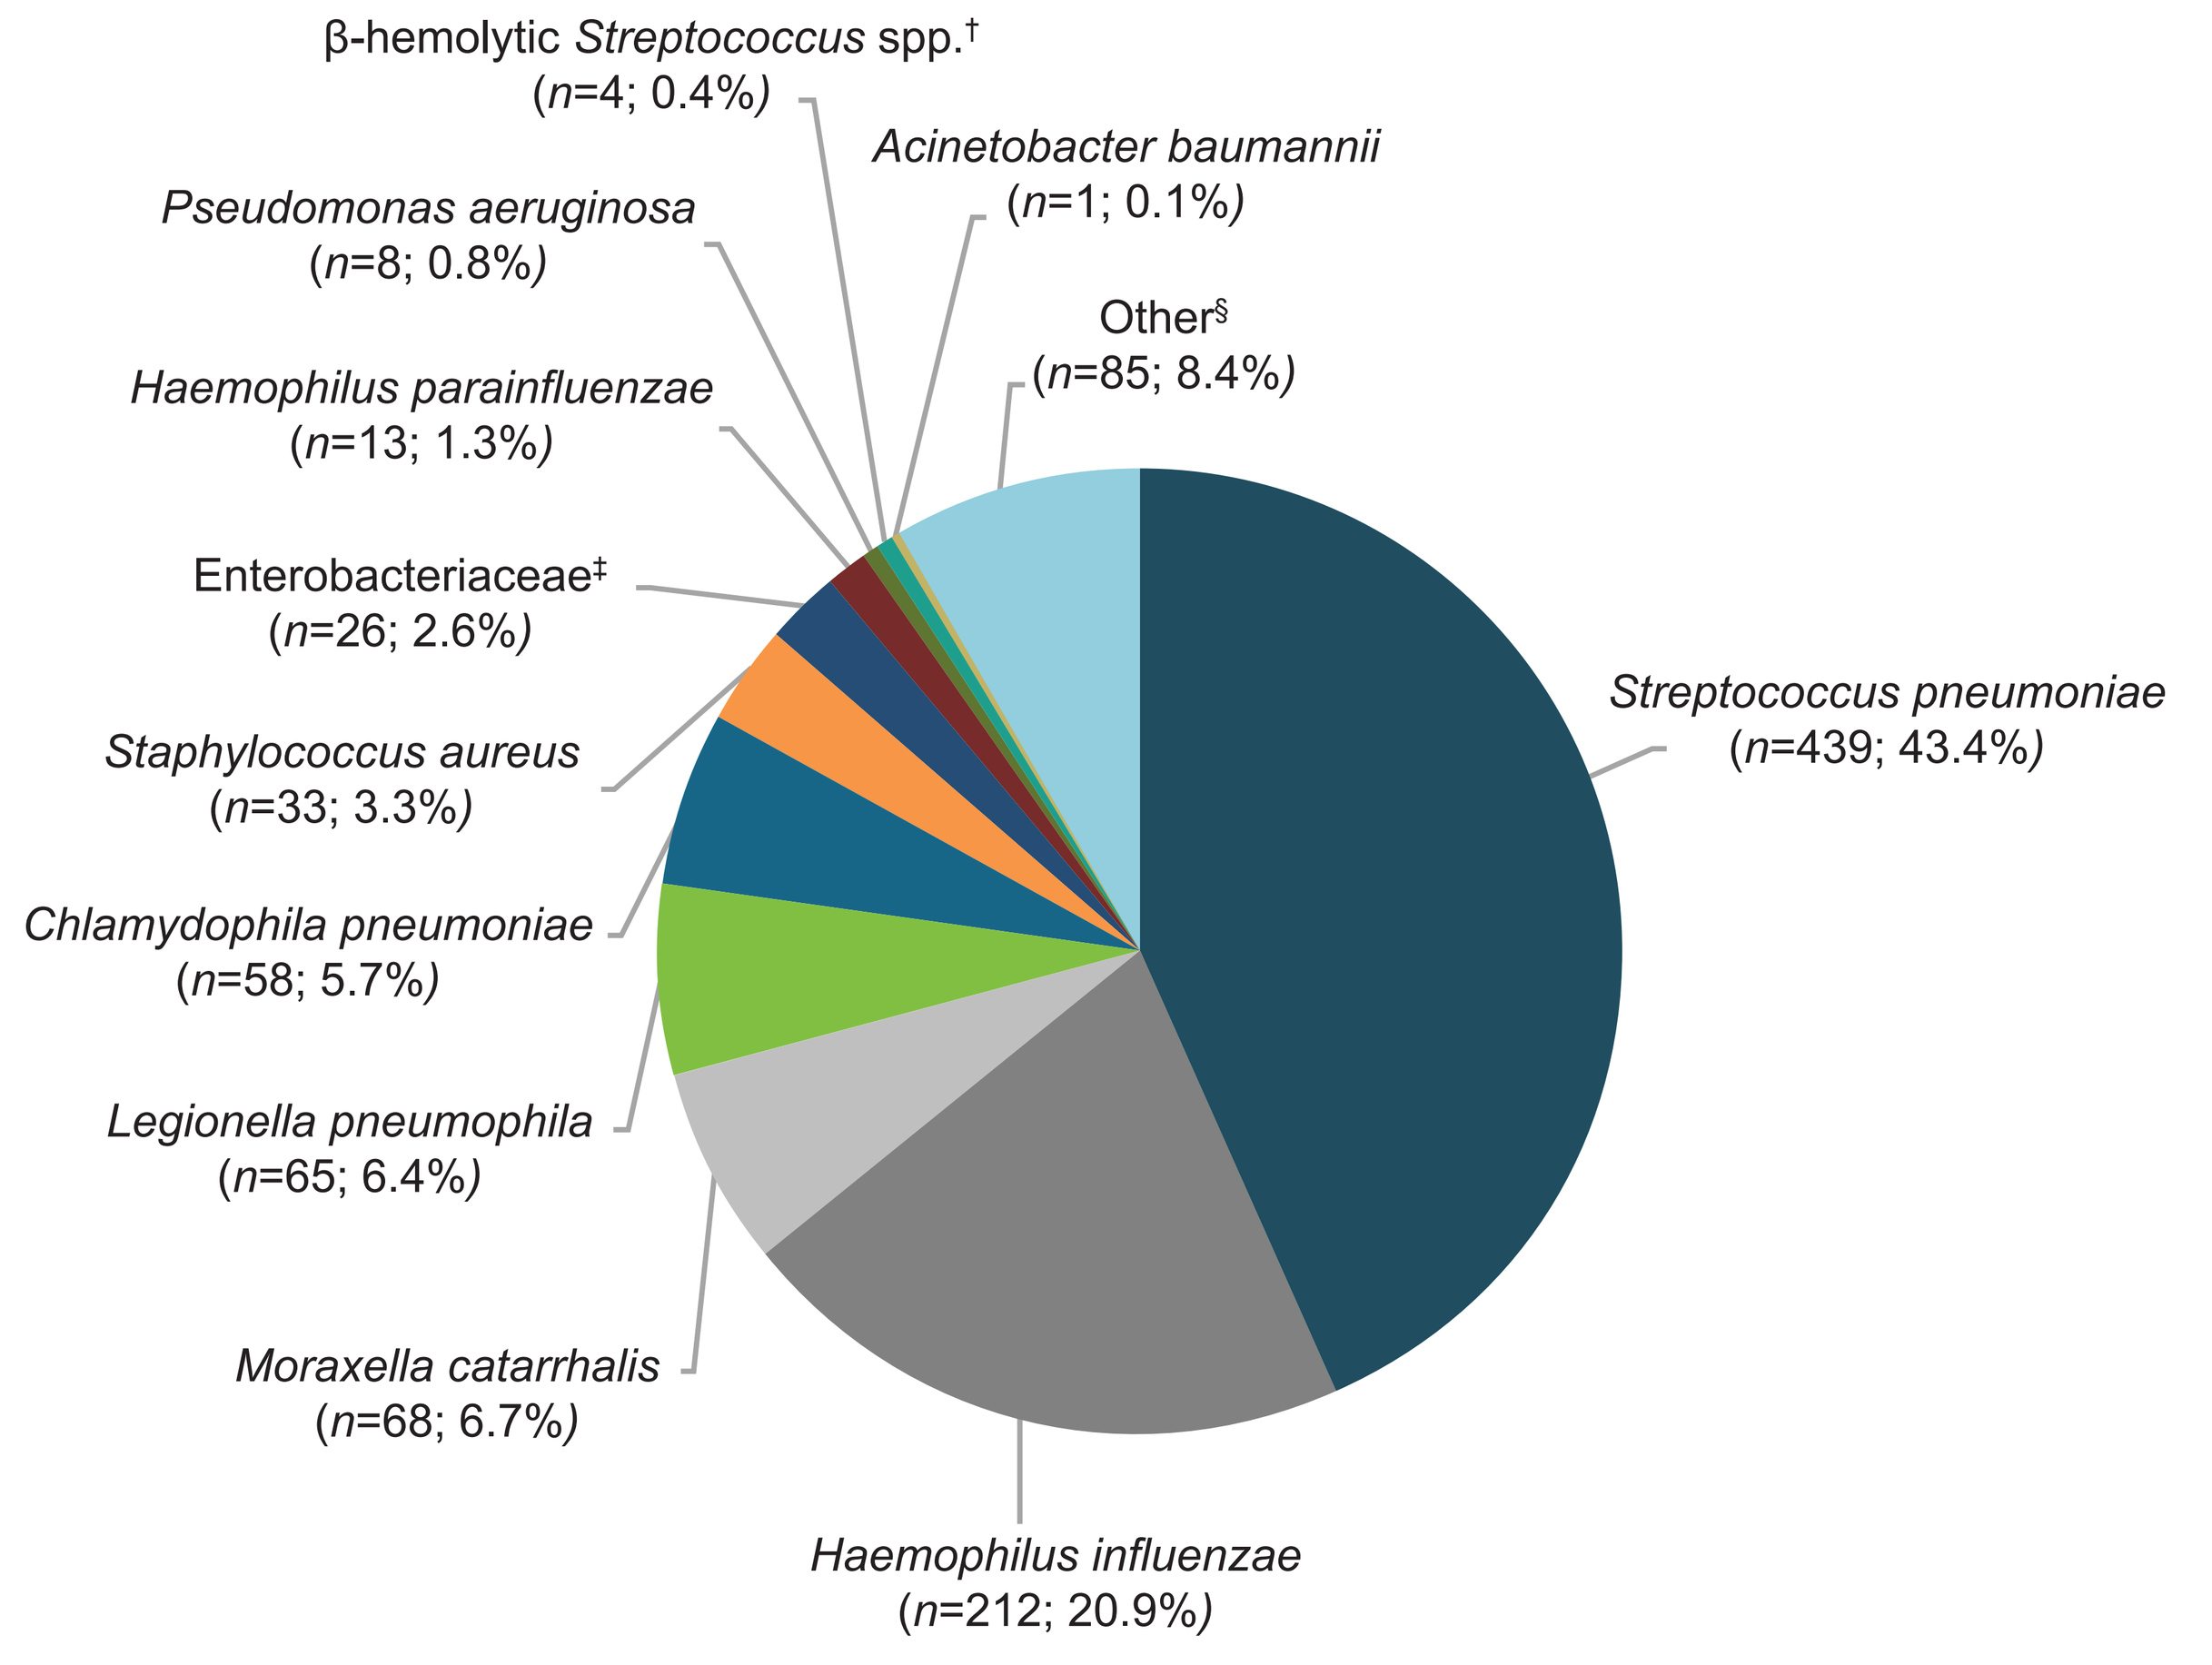
β-hemolytic *Streptococcus* spp.† (*n*=4; 0.4%)
Acinetobacter baumannii (*n*=1; 0.1%)
Pseudomonas aeruginosa (*n*=8; 0.8%)
Other§ (*n*=85; 8.4%)
Haemophilus parainfluenzae (*n*=13; 1.3%)
Enterobacteriaceae‡ (*n*=26; 2.6%)
Staphylococcus aureus (*n*=33; 3.3%)
Chlamydophila pneumoniae (*n*=58; 5.7%)
Legionella pneumophila (*n*=65; 6.4%)
Moraxella catarrhalis (*n*=68; 6.7%)
Haemophilus influenzae (*n*=212; 20.9%)
Streptococcus pneumoniae (*n*=439; 43.4%)

microITT=microbiological intent to treat; n=the number of patients with the respective baseline pathogen.
*A patient could have had >1 pathogen identified. Multiple isolates of the same species from the same patient were counted only once for each phenotype and once for the overall tabulation of the genus and species. Phenotypes were only determined for pathogens identified from cultures and with susceptibility testing results.
†Includes *Streptococcus pyogenes* and *S. agalactiae*.
‡Includes *Aeromonas caviae* complex, *Citrobacter freundii* complex, *C. koseri*, *Enterobacter aerogenes*, *E. cloacae*, *Escherichia coli*, *Klebsiella oxytoca*, *K. pneumoniae*, *K. variicola*, *Proteus mirabilis*, and *Serratia marcesens*.
§Includes *Achromobacter xylosoxidans*, *Acinetobacter calcoaceticus*, *A. calcoaceticus*/*A. baumannii* complex, *A. junii*, *A. lwoffii*, *Acinetobacter* sp., *A. ursingii*, *Burkholderia cepacia*, *Pasteurella pneumotropica*, *Pseudomonas luteola*, and *Stenotrophomonas maltophilia*.

RESULTS (continued)

Patients and Baseline Characteristics

- 709 patients randomized to LEF (*n*=364) or MOX (*n*=345) were included in the pooled microITT population
- The most commonly identified baseline pathogens are shown in **Figure 2**
- Polymicrobial infections were identified in about one third of patients in the pooled microITT population (34.6% LEF, 31.6% MOX)
- LEF and MOX showed similar in vitro activity against the most commonly isolated CABP pathogens, including drug-resistant isolates **(Table 2)**
 - Increasing LEF minimum inhibitory concentration (MIC) did not result in lower ECR response or lower IACR success rates at TOC

Table 2. Minimum Inhibitory Concentrations for Key Pathogens (Pooled microITT Population [Combined Treatment Groups])

Pathogen*	*n*	$MIC_{50/90}$, µg/mL†	
		Lefamulin	Moxifloxacin
Gram-positive bacteria			
Streptococcus pneumoniae	130	0.25/0.5	0.12/0.25
Penicillin resistant	14	0.25/0.25	0.12/0.25
Macrolide resistant‡	31	0.25/0.25	0.12/0.25
Multidrug resistant§	32	0.25/0.25	0.12/0.25
Staphylococcus aureus	24	0.12/0.25	0.06/0.5
Methicillin susceptible	21	0.12/0.25	0.06/0.06
Methicillin resistant	3	NA (0.12–0.12)	NA (0.06–>2)
Gram-negative bacteria			
Haemophilus influenzae	35‖	1/2	0.03/0.06
Moraxella catarrhalis	7	NA (0.06–0.25)	NA (0.03–0.06)
Atypical pathogens			
Mycoplasma pneumoniae	17	≤0.001/≤0.001	0.12/0.25
Legionella pneumophila	2	NA (0.5–1)	NA (0.03–0.03)

MIC=minimum inhibitory concentration; MIC_{50}=minimum inhibitory concentration required to inhibit 50% of isolates; MIC_{90}=minimum inhibitory concentration required to inhibit 90% of isolates; microITT=microbiological intent to treat; NA=not applicable because of small sample size.
*Pathogens were isolated from sputum, nasopharyngeal swab, oropharyngeal swab, blood, bronchoalveolar lavage, and/or pleural fluid via culture. A patient could have had >1 pathogen. Multiple isolates of the same species and phenotype from the same patient were counted only once, using the isolate with the highest MIC to study drug received.
†MIC_{50} and MIC_{90} values are reported for pathogens with ≥10 isolates in the relevant group. For pathogen groups with <10 isolates, the range of MIC values is provided in parentheses. Susceptibilities based on Clinical and Laboratory Standards Institute breakpoints.
‡Defined as resistant to azithromycin or erythromycin.
§Defined as resistant to ≥2 of the following: oral penicillin, moxifloxacin, ceftriaxone, clindamycin, azithromycin or erythromycin, doxycycline, or trimethoprim/sulfamethoxazole.
‖36 isolates were tested against moxifloxacin.

Efficacy

- In the pooled microITT population, ECR rates were 89.3% with LEF and 93.0% with MOX (difference, –3.7%; 95% confidence interval [CI], –7.9% to 0.5%) and IACR success rates at TOC were 83.2% with LEF and 86.7% with MOX (difference, –3.3%; 95% CI, –8.6% to 2.0%)
- In the pooled microITT-2 population, ECR rates were 90.0% with LEF and 92.8% with MOX (difference, –3.1%; 95% CI, –8.7% to 2.6%) and IACR success rates at TOC were 83.3% with LEF and 87.7% with MOX (difference, –4.6%; 95% CI, –11.5% to 2.3%)
- In both the microITT and microITT-2 populations, LEF and MOX demonstrated similar ECR responder and IACR success rates across all baseline CABP pathogens, including drug-resistant isolates **(Table 3)** and polymicrobial infections **(Table 4)**

Table 3. Responder (ECR) and Success (IACR) Rates by Baseline Pathogen

Baseline pathogen, % (*n*/*N*)*	ECR				IACR at TOC			
	Lefamulin		Moxifloxacin		Lefamulin		Moxifloxacin	
	microITT	microITT-2	microITT	microITT-2	microITT	microITT-2	microITT	microITT-2
Gram-positive bacteria								
Streptococcus pneumoniae	88.9 (192/216)	86.2 (75/87)	92.4 (206/223)	92.0 (92/100)	85.2 (184/216)	80.5 (70/87)	86.5 (193/223)	91.0 (91/100)
Penicillin resistant†	– (7/7)	– (7/7)	– (6/7)	– (6/7)	– (7/7)	– (7/7)	– (4/7)	– (4/7)
Macrolide resistant‡	92.9 (13/14)	92.9 (13/14)	82.4 (14/17)	82.4 (14/17)	92.9 (13/14)	92.9 (13/14)	82.4 (14/17)	82.4 (14/17)
Multidrug resistant§	100 (14/14)	100 (14/14)	83.3 (15/18)	83.3 (15/18)	100 (14/14)	100 (14/14)	83.3 (15/18)	83.3 (15/18)
Staphylococcus aureus	100 (23/23)	100 (19/19)	– (10/10)	– (6/6)	87.0 (20/23)	89.5 (17/19)	– (9/10)	– (5/6)
Methicillin susceptible	100 (16/16)	100 (16/16)	– (5/5)	– (5/5)	87.5 (14/16)	87.5 (14/16)	– (5/5)	– (5/5)
Methicillin resistant	– (2/2)	– (2/2)	– (1/1)	– (1/1)	– (2/2)	– (2/2)	– (0/1)	– (0/1)
Gram-negative bacteria								
Haemophilus influenzae	90.7 (97/107)	91.3 (21/23)	93.3 (98/105)	88.9 (16/18)	88.8 (95/107)	87.0 (20/23)	83.8 (88/105)	94.4 (17/18)
Moraxella catarrhalis	89.1 (41/46)	– (3/4)	100 (22/22)	– (3/3)	80.4 (37/46)	– (3/4)	100 (22/22)	– (3/3)
Haemophilus parainfluenzae	– (9/9)	– (9/9)	– (4/4)	– (4/4)	– (9/9)	– (9/9)	– (4/4)	– (4/4)
Atypical pathogens								
Mycoplasma pneumoniae	92.3 (36/39)	96.6 (28/29)	94.1 (32/34)	95.7 (22/23)	89.7 (35/39)	89.7 (26/29)	97.1 (33/34)	95.7 (22/23)
Legionella pneumophila	85.3 (29/34)	84.4 (27/32)	90.3 (28/31)	90.3 (28/31)	79.4 (27/34)	81.3 (26/32)	83.9 (26/31)	83.9 (26/31)
Chlamydophila pneumoniae	92.6 (25/27)	90.9 (20/22)	96.8 (30/31)	95.8 (23/24)	74.1 (20/27)	77.3 (17/22)	74.2 (23/31)	75.0 (18/24)

ECR=early clinical response; IACR=investigator assessment of clinical response; microITT=microbiological intent to treat; TOC=test-of-cure visit.
*microITT (lefamulin, *n*=364, moxifloxacin, *n*=345); microITT-2 (lefamulin, *n*=209, moxifloxacin, *n*=195); *n/N*=patients successfully treated/patients with a specific baseline pathogen. Percentages are reported for pathogens with ≥10 isolates in the relevant group. Multiple isolates of the same species from the same patient were counted only once for each phenotype and once for the overall tabulation of the genus and species. Phenotypes were only determined for pathogens identified from cultures and with susceptibility testing results.
†Using Clinical and Laboratory Standards Institute breakpoints.
‡Defined as resistant to azithromycin or erythromycin.
§Defined as resistant to ≥2 of the following: oral penicillin, moxifloxacin, ceftriaxone, clindamycin, azithromycin or erythromycin, doxycycline, or trimethoprim/sulfamethoxazole.

Table 4. Responder (ECR) and Success (IACR) Rates by Unique Groupings of Selected Pathogens

Baseline pathogen, % (*n*/*N*)*	ECR				IACR at TOC			
	Lefamulin		Moxifloxacin		Lefamulin		Moxifloxacin	
	microITT	microITT-2	microITT	microITT-2	microITT	microITT-2	microITT	microITT-2
Polymicrobial infections	90.5 (114/126)	82.9 (34/41)	94.5 (103/109)	90.0 (27/30)	87.3 (110/126)	80.5 (33/41)	86.2 (94/109)	93.3 (28/30)
Streptococcus pneumoniae and *Haemophilus influenzae*	96.8 (30/31)	– (2/2)	92.3 (36/39)	– (3/4)	96.8 (30/31)	– (2/2)	82.1 (32/39)	– (4/4)
Streptococcus pneumoniae and atypical pathogen†	92.9 (13/14)	– (5/5)	100 (17/17)	– (6/6)	85.7 (12/14)	– (3/5)	94.1 (16/17)	– (6/6)
Streptococcus pneumoniae and *Moraxella catarrhalis*	– (8/8)	– 0	– (4/4)	– (1/1)	– (6/8)	– 0	– (4/4)	– (1/1)
Streptococcus pneumoniae and *Staphylococcus aureus*	– (5/5)	– (3/3)	– (2/2)	– 0	– (3/5)	– (2/3)	– (1/2)	– 0
Any ≥3 pathogens	87.9 (29/33)	– (3/4)	91.7 (22/24)	– (5/6)	90.9 (30/33)	– (4/4)	83.3 (20/24)	– (5/6)

ECR=early clinical response; IACR=investigator assessment of clinical response; microITT=microbiological intent to treat; TOC=test-of-cure visit.
*microITT (lefamulin, *n*=364, moxifloxacin, *n*=345); microITT-2 (lefamulin, *n*=209, moxifloxacin, *n*=195); *n*/*N*=patients successfully treated/patients with a specific baseline pathogen. Percentages are reported for pathogens with ≥10 isolates in the relevant group.
†Atypical pathogens are defined as *Legionella pneumophila*, *Chlamydophila pneumoniae*, and *Mycoplasma pneumoniae*.

CONCLUSIONS

- In these 2 global phase 3 studies, LEF showed potent activity against the most common typical and atypical CABP pathogens, including drug-resistant strains, irrespective of whether pathogens were identified using classical detection methodologies (including culture, urinary antigen test, and serology) with or without real-time PCR
- LEF demonstrated high and similar ECR and IACR success rates to a respiratory fluoroquinolone, without the associated class safety concerns, in patients with CABP (PORT risk class II–V), regardless of baseline CABP pathogen, presence of a drug-resistant strain, or identification of a polymicrobial infection
- These results suggest that LEF may provide a valuable IV and oral monotherapy option for empiric and directed CABP treatment in adults

REFERENCES

(1) Xu J, et al. Deaths: final data for 2016. US Department of Health and Human Services, National Center for Health Statistics. Available at: https://www.cdc.gov/nchs/data/nvsr/nvsr67/nvsr67_05.pdf. Accessed April 16, 2019.
(2) Peyrani P, et al. *Expert Rev Respir Med.* 2019;13(2):139-152.
(3) US Food and Drug Administration. FDA drug safety communication: FDA updates warnings for oral and injectable fluoroquinolone antibiotics due to disabling side effects. Available at: https://www.fda.gov/Drugs/DrugSafety/ucm511530.htm. Accessed April 9, 2019.
(4) US Food and Drug Administration. FDA drug safety communication: FDA reinforces safety information about serious low blood sugar levels and mental health side effects with fluoroquinolone antibiotics; requires label changes. Available at: https://www.fda.gov/downloads/Drugs/DrugSafety/UCM612834.pdf. Accessed April 9, 2019.
(5) Eyal Z, et al. *Sci Rep.* 2016;6:39004.
(6) Mendes RE, et al. *Antimicrob Agents Chemother.* 2016;60(7):4407-4411.
(7) Waites KB, et al. *Antimicrob Agents Chemother.* 2017;61(2):e02008-16.
(8) Paukner S, et al. *Antimicrob Agents Chemother.* 2019;63(4):e02161-18.
(9) Paukner S, et al. Lefamulin activity against bacterial pathogens commonly associated with acute bacterial skin and skin structure infections (ABSSSI) collected in the 2017 global SENTRY surveillance program. Poster AAR-785. Presented at: ASM Microbe June 20-24, 2019; San Francisco, CA.
(10) Paukner S, et al. Lefamulin activity against respiratory tract pathogens collected in the 2017 global SENTRY surveillance program. Poster AAR-786. Presented at: ASM Microbe, June 20-24, 2019; San Francisco, CA.
(11) Wicha WW, et al. *J Antimicrob Chemother.* 2019;74(suppl 3):iii19-iii26.
(12) Zhang L, et al. *J Antimicrob Chemother.* 2019;74(suppl 3):iii27-iii34.
(13) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090.
(14) Alexander E, et al. Oral lefamulin is safe and effective in the treatment of adults with community-acquired bacterial pneumonia (CABP): results of Lefamulin Evaluation Against Pneumonia (LEAP 2) study. Abstract LB6. Presented at: IDWeek, October 3–7, 2018; San Francisco, CA.

Acknowledgments

We thank the personnel of Covance Central Laboratory Services (Indianapolis, IN), UAB Diagnostic Mycoplasma Laboratory (Birmingham, AL), Special Pathogens Laboratory-The Legionella Experts® (Pittsburgh, PA), Research Unit on Respiratory Pathogens at RSPH, Emory University (Atlanta, GA), and Accelero Bioanalytics (Berlin, Germany) for assistance in the performance of confirmatory and specialized testing.

Disclosures

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company. Elizabeth Alexander, Lisa Goldberg, Susanne Paukner, Jennifer Schranz, and Steven P. Gelone are employees of and have stock in Nabriva Therapeutics plc. Anita F. Das served as a consultant for Nabriva Therapeutics during the design and execution of the study and has also served as a consultant for ContraFect, Tetraphase, Paratek, Cempra, Achaogen, Zavante, UTILITY, Iterum, AntibioTx, and Wockhardt. Christian Sandrock has served as a consultant for Nabriva Therapeutics. Thomas M. File Jr has served as a consultant for Motif BioSciences, Merck, Nabriva Therapeutics, Paratek, GlaxoSmithKline, Melinta, and Shionogi.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/LEAP1:2_Pathogen.pdf

ASM Microbe 2019: June 20–24, San Francisco, CA, USA