Saturday – AAR-786

Lefamulin Activity Against Respiratory Tract Pathogens Collected in the 2017 Global SENTRY Antimicrobial Surveillance Program

Susanne Paukner,[1] Steven P. Gelone,[2] S. J. Ryan Arends,[3] Helio S. Sader[3]

[1]Nabriva Therapeutics GmbH, Vienna, Austria; [2]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA; [3]JMI Laboratories, North Liberty, IA, USA

Presented by
Susanne Paukner
Phone: +43 1 74093-1224
Email: Susanne.Paukner@Nabriva.com

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION

- Community-acquired bacterial pneumonia (CABP) is a common infection-related cause of hospitalization and/or death in older adults in Europe, Latin America, Asia-Pacific, and North America[1-5]
- Annual CABP care costs an estimated €10.1 billion[1] in Europe and >$17 billion in the United States[5]
- Increasing rates of antibiotic resistance among CABP-causing pathogens and safety concerns around available antibiotics drive a worldwide need for new CABP treatment options[6]
- Lefamulin (LEF) is a first-in-class pleuromutilin antibiotic in phase 3 clinical development for intravenous (IV) and oral treatment of CABP[7,8]
 - LEF selectively inhibits bacterial protein synthesis by binding to the 50S ribosomal subunit at the A- and P-sites in the peptidyl transferase center[7,8] **(Figure 1)**
- LEF was noninferior to moxifloxacin in two phase 3 trials (one IV-to-oral switch study and one oral-only study) in adults with CABP[7,9]
- The objective of this study was to evaluate the in vitro activity of LEF and comparators against a contemporary global set of respiratory tract pathogens

Figure 1. (A) Structure of Lefamulin and (B) Lefamulin in the Peptidyl Transferase Center Overlapping With A- and P-site tRNA

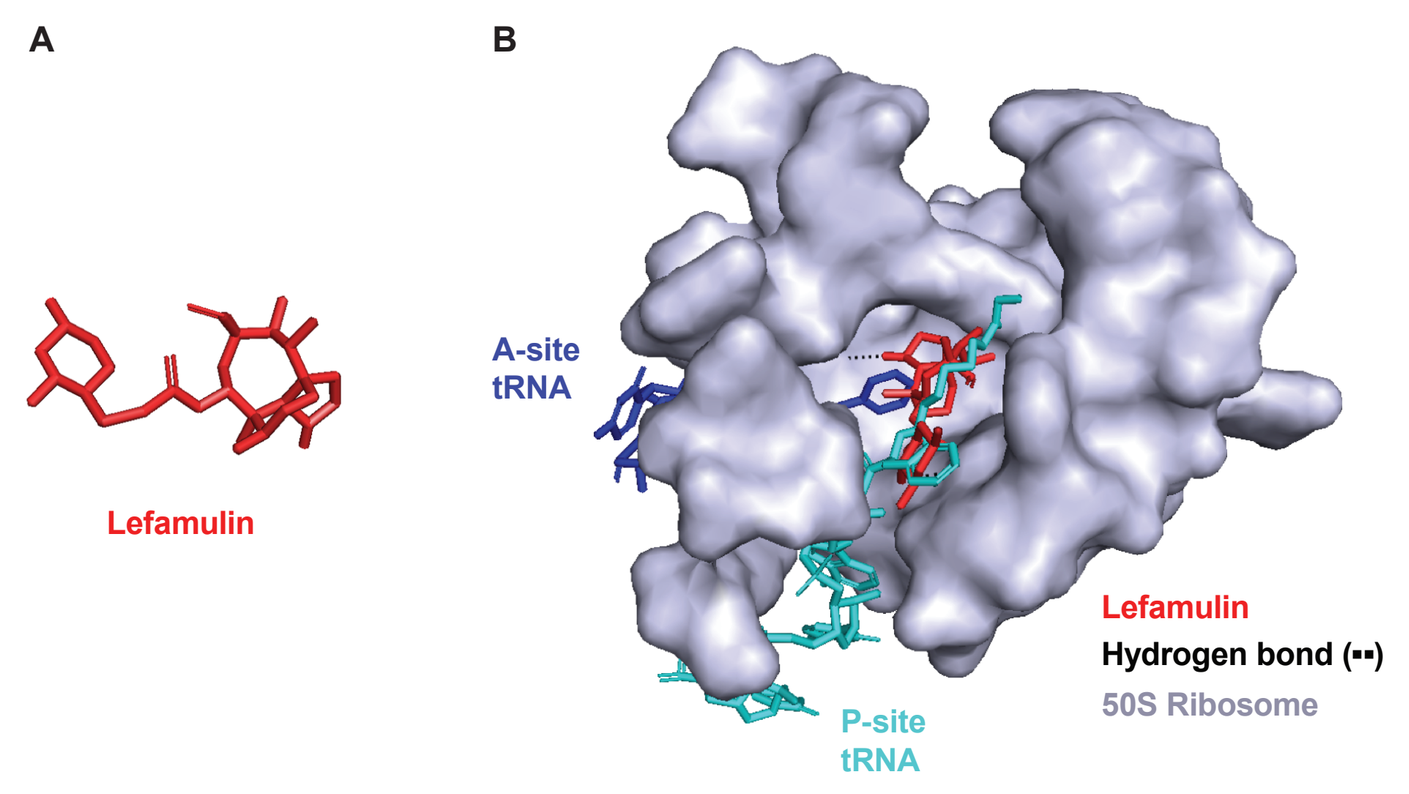
A
B
Lefamulin
A-site tRNA
Lefamulin
Hydrogen bond (••)
50S Ribosome
P-site tRNA

METHODS

- As part of the 2017 global SENTRY Antimicrobial Surveillance Program, 3190 unique isolates (1 per patient) were collected from patients with community-acquired respiratory tract infections (77.2%) or hospitalized patients with pneumonia (22.8%)
- Isolates were collected from 109 sites in 33 countries
 - 40.3% of isolates were collected from the United States, 41.1% from Europe, 10.3% from Asia Pacific, and 8.2% from Latin America
- Minimum inhibitory concentration (MIC) for LEF and comparators was determined using Clinical and Laboratory Standards Institute (CLSI) broth microdilution[10]; susceptibility was evaluated using the CLSI (2019) breakpoints[11]

RESULTS

- LEF showed potent antibacterial activity against all tested CABP pathogens and its activity was not affected by resistance to other antibiotic classes **(Table 1)**

Streptococcus pneumoniae

- *S. pneumoniae* isolates showed resistance to macrolides (35.8% resistant to erythromycin), penicillin (14.4%), trimethoprim-sulfamethoxazole (18.4%), and tetracycline (24.9%), whereas they were largely susceptible (≥84%) to the tested cephalosporins and fluoroquinolones **(Table 2)**
- LEF inhibited 99.5% of *S. pneumoniae* isolates at ≤0.25 µg/mL, with $MIC_{50/90}$ values of 0.06/0.12 µg/mL for multidrug-resistant and penicillin-resistant subsets and 0.12/0.25 µg/mL for the macrolide-resistant subset **(Table 3)**

Staphylococcus aureus

- *S. aureus* isolates overall, and particularly methicillin-resistant *S. aureus* (MRSA) strains, were resistant to macrolides (40.2% and 70.2%, respectively, resistant to erythromycin) and fluoroquinolones (26.4% and 67.5%, respectively, resistant to moxifloxacin; **Table 4**)
- LEF showed potent activity against *S. aureus* as well as MRSA, with $MIC_{50/90}$ values of 0.06/0.12 µg/mL for both **(Table 4)**

Haemophilus influenzae

- *H. influenzae* isolates were largely susceptible to all antimicrobial agents except ampicillin (27.6% resistant) and trimethoprim-sulfamethoxazole (33.1% resistant; **Table 5**)
- LEF inhibited *H. influenzae* isolates **(Table 5)**, including the β-lactamase–positive subset (*n*=144; data not shown), with $MIC_{50/90}$ values of 0.5/2 µg/mL for both

Haemophilus parainfluenzae

- *H. parainfluenzae* isolates were largely susceptible to all antimicrobial agents except ampicillin (15.2% resistant), clarithromycin (8.7% resistant), and trimethoprim-sulfamethoxazole (16.3% resistant; **Table 5**)
- LEF inhibited *H. parainfluenzae* isolates at $MIC_{50/90}$ values of 1/2 µg/mL **(Table 5)**

Moraxella catarrhalis

- *M. catarrhalis* isolates included a substantial proportion of β-lactamase producers (96.9%) and were largely susceptible to all tested antimicrobial agents **(Table 5)**
- LEF inhibited *M. catarrhalis* isolates at concentrations of ≤0.12 µg/mL ($MIC_{50/90}$ values of 0.06/0.06 µg/mL; **Table 5**)

β-hemolytic and viridans group streptococci

- LEF showed potent activity against both β-hemolytic and viridans group streptococci, with $MIC_{50/90}$ values of 0.03/0.03 µg/mL and 0.06/2 µg/mL, respectively **(Table 1)**
- β-hemolytic and viridans group streptococcal isolates showed resistance to clindamycin (11.1% and 23.1%, respectively), erythromycin (33.3% and 46.2%, respectively), and levofloxacin (5.6% and 7.7%, respectively; data not shown)

Table 1. Activity of Lefamulin and Comparators Against CABP Pathogens

		$MIC_{50/90}$, µg/mL					
Organism	*n*	Lefamulin	Amoxicillin-clavulanic acid	Azithromycin	Ceftriaxone	Moxifloxacin	Tigecycline
S. pneumoniae	1862	0.12/0.25	≤0.03/2	0.06/>4	0.03/1	0.12/0.25	0.06/0.06
Penicillin resistant*	268	0.06/0.12	2/>4	>4/>4	1/2	0.12/0.25	0.06/0.12
Macrolide resistant†	667	0.12/0.25	0.5/>4	>4/>4	0.5/2	0.12/0.25	0.06/0.06
Multidrug resistant‡	162	0.06/0.12	4/>4	>4/>4	1/2	0.12/0.25	0.06/0.12
S. aureus	428	0.06/0.12	–	0.5/>32	0.25/1§	≤0.06/>4	0.12/0.12
MRSA	151	0.06/0.12	–	>32/>32	1/2§	2/>4	0.12/0.25
H. influenzae	550	0.5/2	1/2	1/2	0.004/0.015	0.03/0.06	0.25/0.5
H. parainfluenzae	92	1/2	0.5/1	0.5/2	0.004/0.06	0.06/>2	0.25/0.5
M. catarrhalis	227	0.06/0.06	≤0.25/≤0.25	0.015/0.03	0.25/1	0.06/0.06	0.06/0.06
β-hemolytic *Streptococcus* spp.¶	18	0.03/0.03	–	0.03/8‖	0.06/0.06	0.12/0.25	0.06/0.06
Viridans group *Streptococcus* spp.^	13	0.06/2	–	0.03/>16‖	0.12/0.5	0.12/0.25	0.06/0.12

CABP=community-acquired bacterial pneumonia; CLSI=Clinical and Laboratory Standards Institute; MIC=minimum inhibitory concentration; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; MRSA=methicillin-resistant *S. aureus*.
*Applying oral CLSI breakpoint of ≥2 µg/mL. †Using erythromycin breakpoint. ‡Resistant to oral penicillin, erythromycin, and tetracycline. §Ceftaroline tested instead of ceftriaxone. ¶Organisms include: *Streptococcus pyogenes* (*n*=8) and *S. agalactiae* (*n*=10). ‖Erythromycin tested instead of azithromycin. ^Organisms include: *Streptococcus anginosus* (*n*=1), *S. anginosus* group (*n*=1), *S. gallolyticus* (*n*=2), *S. mitis* group (*n*=7), *S. parasanguinis* (*n*=1), *S. salivarius/vestibularis* (*n*=1).

Table 2. Activity of Lefamulin and Comparators Against *S. pneumoniae*

	µg/mL			CLSI*		
Antimicrobial Agent	MIC_{50}	MIC_{90}	Range	%S	%I	%R
***S. pneumoniae* (*n*=1862)**						
Lefamulin	**0.12**	**0.25**	**≤0.008–1**	**[100.0]†**		
Amoxicillin-clavulanic acid	≤0.03	2	≤0.03–>4	93.1	2.2	4.8
Azithromycin	0.06	>4	≤0.03–>4	63.7	0.6	35.7
Ceftaroline	≤0.008	0.12	≤0.008–>1	99.9	–	–
Ceftriaxone	0.03	1	≤0.015–>2	83.8‡ 94.9§	11.2 4.1	5.1 1.0
Clindamycin	≤0.25	>2	≤0.25–>2	80.8	0.4	18.8
Erythromycin	0.03	>16	≤0.015–>16	63.7	0.4	35.8
Levofloxacin	1	2	0.25–>4	98.0	0.3	1.7
Moxifloxacin	0.12	0.25	≤0.03–>4	98.4	1.1	0.4
Penicillin	0.03	2	≤0.008–>4	64.9¶ 64.9‖ 94.0^	20.7 – 5.3	14.4 35.1 0.6
Tetracycline	0.5	>4	0.06–>4	74.8	0.3	24.9
Tigecycline	0.06	0.06	0.015–0.25	93.7#	–	–
Trimethoprim-sulfamethoxazole	0.25	>4	≤0.12–>4	71.4	10.2	18.4

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC=minimum inhibitory concentration; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; R=resistant; S=susceptible.
*Criteria as published by CLSI 2019. †Percentage inhibited at proposed lefamulin breakpoint of ≤1 µg/mL for *S. pneumoniae* shown for reference. ‡Using meningitis breakpoints. §Using nonmeningitis breakpoints. ¶Using oral breakpoints. ‖Using parenteral, meningitis breakpoints. ^Using parenteral, nonmeningitis breakpoints. #US Food and Drug Administration breakpoints accessed February 2018.

Table 3. Activity of Lefamulin and Comparators Against Drug-Resistant *S. pneumoniae*

	µg/mL			CLSI*		
Antimicrobial Agent	MIC_{50}	MIC_{90}	Range	%S	%I	%R
Penicillin-resistant† *S. pneumoniae* (*n*=268)						
Lefamulin	**0.06**	**0.12**	**0.015–0.5**	**[100.0]‡**		
Amoxicillin-clavulanic acid	2	>4	1–>4	51.7	15.0	33.3
Azithromycin	>4	>4	≤0.03–>4	15.7	0.7	83.6
Ceftaroline	0.12	0.25	0.03–>1	99.3	–	–
Ceftriaxone	1	2	0.25–>2	5.2§ 66.0¶	60.8 27.6	34.0 6.3
Clindamycin	>2	>2	≤0.25–>2	46.3	0.7	53.0
Erythromycin	>16	>16	≤0.015–>16	15.7	0.0	84.3
Levofloxacin	1	2	0.5–>4	96.6	0.0	3.4
Moxifloxacin	0.12	0.25	0.06–>4	97.4	1.1	1.5
Penicillin	2	4	2–>4	0.0‖ 0.0^ 58.6#	0.0 – 36.9	100.0 100.0 4.5
Tetracycline	>4	>4	0.25–>4	38.4	0.0	61.6
Tigecycline	0.06	0.12	0.015–0.12	89.6**	–	–
Trimethoprim-sulfamethoxazole	>4	>4	≤0.12–>4	24.3	11.2	64.6
Macrolide-resistant†† *S. pneumoniae* (*n*=667)						
Lefamulin	**0.12**	**0.25**	**0.015–1**	**[100.0]‡**		
Amoxicillin-clavulanic acid	0.5	>4	≤0.03–>4	83.8	5.6	10.7
Azithromycin	>4	>4	≤0.03–>4	0.6	0.9	98.5
Ceftaroline	0.06	0.12	≤0.008–>1	99.7	–	–
Ceftriaxone	0.5	2	≤0.015–>2	62.0§ 86.2¶	24.2 11.3	13.8 2.6
Clindamycin	>2	>2	≤0.25–>2	46.5	1.2	52.3
Erythromycin	>16	>16	1–>16	0.0	0.0	100.0
Levofloxacin	1	2	0.5–>4	97.8	0.3	1.9
Moxifloxacin	0.12	0.25	≤0.03–>4	98.4	0.9	0.7
Penicillin	0.5	4	≤0.008–>4	28.0‖ 28.0^ 85.5#	38.1 – 12.7	33.9 72.0 1.8
Tetracycline	>4	>4	0.12–>4	39.7	0.1	60.1
Tigecycline	0.06	0.06	0.015–0.25	92.4**	–	–
Trimethoprim-sulfamethoxazole	1	>4	≤0.12–>4	49.5	17.3	33.2
Multidrug-resistant‡‡ *S. pneumoniae* (*n*=162)						
Lefamulin	**0.06**	**0.12**	**0.015–0.25**	**[100.0]‡**		
Amoxicillin-clavulanic acid	4	>4	1–>4	42.9	18.6	38.5
Azithromycin	>4	>4	1–>4	0.0	1.2	98.8
Ceftaroline	0.12	0.25	0.06–0.5	100.0	–	–
Ceftriaxone	1	2	0.5–>2	4.3§ 50.6¶	46.3 41.4	49.4 8.0
Clindamycin	>2	>2	≤0.25–>2	21.0	1.2	77.8
Erythromycin	>16	>16	1–>16	0.0	0.0	100.0
Levofloxacin	1	2	0.5–>4	95.7	0.0	4.3
Moxifloxacin	0.12	0.25	0.06–>4	96.9	1.9	1.2
Penicillin	4	4	2–>4	0.0‖ 0.0^ 45.1#	0.0 – 48.8	100.0 100.0 6.2
Tetracycline	>4	>4	>4–>4	0.0	0.0	100.0
Tigecycline	0.06	0.12	0.015–0.12	85.8**	–	–
Trimethoprim-sulfamethoxazole	>4	>4	≤0.12–>4	18.5	13.6	67.9

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC=minimum inhibitory concentration; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; R=resistant; S=susceptible.
*2019 CLSI criteria. †Penicillin MIC ≥2 µg/mL using the oral breakpoint. ‡Percentages inhibited at proposed lefamulin breakpoint of ≤1 µg/mL for *S. pneumoniae* shown for reference. §Using meningitis breakpoint. ¶Using nonmeningitis breakpoint. ‖Using oral breakpoint. ^Using parenteral, meningitis breakpoints. #Using parenteral, nonmeningitis breakpoints. **US Food and Drug Administration breakpoints accessed February 2018. ††Using erythromycin breakpoint. ‡‡Resistant to oral penicillin, erythromycin, and tetracycline.

Table 4. Activity of Lefamulin and Comparators Against *S. aureus*

	µg/mL			CLSI*		
Antimicrobial Agent	MIC_{50}	MIC_{90}	Range	%S	%I	%R
***S. aureus* (*n*=428)**						
Lefamulin	**0.06**	**0.12**	**0.015–1**	**[99.5]†**		
Azithromycin	0.5	>32	0.06–>32	56.8	0.0	43.2
Ceftaroline	0.25	1	≤0.06–4	92.5‡	7.5	0.0
Clindamycin	0.06	>2	≤0.03–>2	81.3	0.0	18.7
Doxycycline	0.12	1	≤0.06–>8	95.1	3.0	1.9
Erythromycin	0.25	>8	≤0.06–>8	56.3	3.5	40.2
Gentamicin	≤1	>8	≤1–>8	88.3	0.7	11.0
Levofloxacin	0.25	>4	0.12–>4	69.9	0.2	29.9
Linezolid	1	2	0.25–4	100.0	–	0.0
Moxifloxacin	≤0.06	>4	≤0.06–>4	70.3	3.3	26.4
Oxacillin	0.5	>2	0.25–>2	64.7	–	35.3
Tigecycline	0.12	0.12	≤0.015–0.5	100.0§	–	–
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5–>16	97.9	–	2.1
Vancomycin	1	1	0.5–2	100.0	0.0	0.0
MRSA (*n*=151)						
Lefamulin	**0.06**	**0.12**	**0.03–0.25**	**[98.6]†**		
Azithromycin	>32	>32	0.12–>32	26.5	0.0	73.5
Ceftaroline	1	2	0.25–4	78.8‡	21.2	0.0
Clindamycin	0.06	>2	≤0.03–>2	53.6	0.0	46.4
Doxycycline	0.12	8	≤0.06–>8	86.8	8.6	4.6
Erythromycin	>8	>8	≤0.06–>8	26.5	3.3	70.2
Gentamicin	≤1	>8	≤1–>8	76.8	2.0	21.2
Levofloxacin	>4	>4	0.12–>4	24.5	0.0	75.5
Linezolid	1	2	0.5–2	100.0	–	0.0
Moxifloxacin	2	>4	≤0.06–>4	25.2	7.3	67.5
Oxacillin	>2	>2	>2–>2	0.0	–	100.0
Tigecycline	0.12	0.25	0.03–0.5	100.0§	–	–
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5–>16	95.4	–	4.6
Vancomycin	1	1	0.5–2	100.0	0.0	0.0

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC=minimum inhibitory concentration; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; MRSA=methicillin-resistant *S. aureus*; R=resistant; S=susceptible.
*2019 CLSI criteria. †Percentages inhibited at proposed lefamulin breakpoint of ≤0.5 µg/mL for *S. aureus* shown for reference. ‡Intermediate interpreted as susceptible-dose dependent. §US Food and Drug Administration breakpoints accessed February 2018.

Table 5. Activity of Lefamulin and Comparators Against *H. influenzae*, *H. parainfluenzae*, and *M. catarrhalis*

	µg/mL			CLSI*		
Antimicrobial Agent	MIC_{50}	MIC_{90}	Range	%S	%I	%R
***H. influenzae* (*n*=550)**						
Lefamulin	**0.5**	**2**	**0.015–4**	**[100.0]†**		
Amoxicillin-clavulanic acid	1	2	≤0.06–16	98.9	–	1.1
Ampicillin	0.5	>8	≤0.12–>8	67.3	5.1	27.6
Azithromycin	1	2	≤0.12–>8	99.1	–	–
Cefepime	0.12	0.25	≤0.015–>2	99.8	–	–
Ceftriaxone	0.004	0.015	≤0.002–0.25	100.0	–	–
Ciprofloxacin	0.015	0.015	0.008–>1	98.9	–	–
Clarithromycin	8	16	≤0.12–>16	80.5	17.6	1.8
Moxifloxacin	0.03	0.06	≤0.008–>2	99.1	–	–
Tetracycline	0.5	1	0.12–>8	98.4	0.2	1.5
Tigecycline	0.25	0.5	0.03–2	86.2‡	–	–
Trimethoprim-sulfamethoxazole	0.12	>4	≤0.06–>4	62.9	4.0	33.1
***H. parainfluenzae* (*n*=92)**						
Lefamulin	**1**	**2**	**≤0.008–8**	**[100.0]†**		
Amoxicillin-clavulanic acid	0.5	1	≤0.06–8	98.9	–	1.1
Ampicillin	0.5	>8	≤0.12–>8	81.5	3.3	15.2
Azithromycin	0.5	2	≤0.12–>8	95.7	–	–
Cefepime	0.06	2	≤0.015–>2	96.7	–	–
Ceftriaxone	0.004	0.06	≤0.002–>2	98.9	–	–
Ciprofloxacin	0.015	>1	≤0.004–>1	88.0	–	–
Clarithromycin	8	16	0.5–>16	68.5	22.8	8.7
Moxifloxacin	0.06	>2	≤0.008–>2	85.7	–	–
Tetracycline	0.5	1	0.25–>8	92.4	0.0	7.6
Tigecycline	0.25	0.5	0.06–1	–	–	–
Trimethoprim-sulfamethoxazole	≤0.06	4	≤0.06–>4	79.3	4.3	16.3
***M. catarrhalis* (*n*=227)**						
Lefamulin	**0.06**	**0.06**	**≤0.008–0.12**	**[100.0]†**		
Amoxicillin-clavulanic acid	≤0.25	≤0.25	≤0.25–2	100.0	–	0.0
Azithromycin	0.015	0.03	≤0.004–0.06	100.0	–	–
Ceftriaxone	0.25	1	≤0.002–>2	99.6	–	–
Clarithromycin	≤0.12	≤0.12	≤0.12–0.5	100.0	–	–
Moxifloxacin	0.06	0.06	0.015–0.12	–	–	–
Tetracycline	0.25	0.5	≤0.06–>8	99.1	0.0	0.9
Tigecycline	0.06	0.06	≤0.015–0.12	–	–	–
Trimethoprim-sulfamethoxazole	0.25	0.5	≤0.06–2	94.7	5.3	0.0

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC=minimum inhibitory concentration; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; R=resistant; S=susceptible.
*2019 CLSI criteria. †Percentages inhibited at proposed lefamulin breakpoints of ≤4 µg/mL for *H. influenzae*, ≤8 µg/mL for *H. parainfluenzae*, and ≤0.5 µg/mL for *M. catarrhalis* shown for reference. ‡US Food and Drug Administration breakpoints accessed February 2018.

CONCLUSIONS

- LEF demonstrated potent in vitro activity against this contemporary (2017) collection of community-acquired bacterial pathogens collected worldwide from patients with respiratory tract infections and hospitalized patients with pneumonia compared with the most potent comparator agents used to treat CABP
- LEF activity was unaffected by resistance to other antibiotic classes, including macrolides, fluoroquinolones, and tetracyclines
- These in vitro data suggest that LEF may offer an important empiric monotherapy treatment option for CABP, in particular, where resistance to antimicrobials commonly used for CABP is high

REFERENCES

(1) Welte T, et al. *Thorax*. 2012;67(1):71-79.
(2) Buzzo AR, et al. *Int J Infect Dis*. 2013;17(9):e673-677.
(3) Song JH, et al. *Int J Antimicrob Agents*. 2011;38(2):108-117.
(4) Jain S, et al. *N Engl J Med*. 2015;373(5):415-427.
(5) File TM and Marrie TJ. *Postgrad Med*. 2010;122(2):130-141.
(6) Peyrani P, et al. *Expert Rev Respir Med*. 2019;13(2):139-152.
(7) File TM Jr, et al. *Clin Infect Dis*. 2019; doi: 10.1093/cid/ciz090.
(8) Eyal Z, et al. *Sci Rep*. 2016;6:39004.
(9) Alexander E, et al. Oral lefamulin is safe and effective in the treatment of adults with community-acquired bacterial pneumonia (CABP): results of Lefamulin Evaluation Against Pneumonia (LEAP 2) study. Abstract LB6. Presented at: IDWeek, October 3–7, 2018; San Francisco, CA.
(10) Clinical and Laboratory Standards Institute. Performance standards for antimicrobial susceptibility testing: 28th informational supplement. Wayne, PA: Clinical and Laboratory Standards Institute; 2018. M100Ed28E.
(11) Clinical and Laboratory Standards Institute. Performance standards for antimicrobial susceptibility testing, 29th edition. Wayne, PA: Clinical and Laboratory Standards Institute; 2019. M100Ed29.

Acknowledgments

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Susanne Paukner and Steven P. Gelone are employees/stockholders of Nabriva Therapeutics plc. S.J. Ryan Arends and Helio S. Sader are employees of JMI Laboratories, which was contracted by Nabriva Therapeutics to conduct these analyses.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/SENTRY_Global

ASM Microbe 2019: June 20–24, San Francisco, CA, USA