E1006

Efficacy of Lefamulin Versus Moxifloxacin in Adults With Community-Acquired Bacterial Pneumonia: Results of the Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 Double-Blind Noninferiority Phase 3 Clinical Trials

Jennifer Schranz,[1] Lisa Goldberg,[1] Anita F. Das,[2] Elizabeth Alexander,[1] Gregory J. Moran,[3] Christian Sandrock,[4] Andrew F. Shorr,[5] Steven P. Gelone[1]

[1]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA; [2]Das Consulting, Guerneville, CA, USA; [3]Olive View-UCLA Medical Center, Sylmar, CA, USA; [4]UC Davis School of Medicine, Sacramento, CA, USA; [5]Medstar Washington Hospital Center, Washington, DC, USA

Presented by
Daniel S. Stein, MD
Email: Daniel.Stein@Nabriva.com
Phone: 610-981-2871

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

PURPOSE

- Pneumonia is among the leading causes of hospitalization and infection-related death in the United States[1-3]
- Antimicrobial surveillance programs have observed trends of generally decreasing susceptibility among bacterial isolates to antimicrobials used to treat community-acquired bacterial pneumonia (CABP), including resistance to oral penicillin, macrolides, and folate pathway inhibitors in *Streptococcus pneumoniae* and resistance to macrolides and fluoroquinolones in *Staphylococcus aureus* (particularly methicillin-resistant *S. aureus*)[4,5]
 - Increasing rates of bacterial resistance and the undesirable risks and adverse effects associated with current treatments (eg, fluoroquinolone-associated disability) are driving the need for new therapeutic options for CABP[2]
- Lefamulin (LEF), a first-in-class pleuromutilin antibiotic approved for intravenous (IV) and oral use in adults with CABP,[6] inhibits protein synthesis by binding selectively and specifically to the peptidyl transferase center of the 50S ribosomal subunit[7,8]
- LEF has demonstrated potent in vitro activity against a global collection of pathogens commonly causing CABP[4]
- LEF rapidly and predictably penetrates target sites, including plasma and the epithelial lining fluid (ELF) of the lungs
 - Unbound LEF levels are 5.7-fold higher in ELF than in plasma, making it an ideal candidate for CABP therapy[9]
 - When provided at extracellular concentrations of 1 µg/mL and 5 µg/mL, LEF exhibited ~50-fold intracellular accumulation in murine macrophages after 5h of incubation, whereas ~15–18-fold intracellular accumulation was observed with the positive control azithromycin[10]
- The favorable pharmacokinetics and spectrum of activity of LEF led to its investigation in 2 phase 3 trials in adults with CABP
 - The Lefamulin Evaluation Against Pneumonia (LEAP) 1 study evaluated the efficacy and safety of LEF as monotherapy, with an IV-to-oral switch option, compared with moxifloxacin (MOX) (± linezolid)[11]
 - The LEAP 2 study evaluated the efficacy and safety of oral LEF monotherapy compared with oral MOX monotherapy[12]
- We report overall efficacy, including analyses stratified by Pneumonia Outcomes Research Team (PORT) risk class, in pooled LEAP 1 and LEAP 2 analyses

METHODS

Study Design

- Both studies were prospective, randomized, double-blind, double-dummy, phase 3 trials **(Figure 1)**[11,12]
- Patients in LEAP 1 and LEAP 2 were enrolled at 66 centers (18 countries) and 99 centers (19 countries), respectively
 - In LEAP 1, patients were randomized to receive LEF 150 mg IV every 12 hours (q12h) for 5–7 days or MOX 400 mg IV every 24 hours (q24h) for 7 days
 - Patients could switch to oral therapy (LEF 600 mg q12h or MOX 400 mg q24h) after 6 IV doses of study drug (approximately 3 days) if predefined improvement criteria were met
 - In LEAP 2, patients were randomized to receive oral LEF 600 mg q12h for 5 days or oral MOX 400 mg q24h for 7 days

Figure 1. LEAP 1 and LEAP 2 Study Design

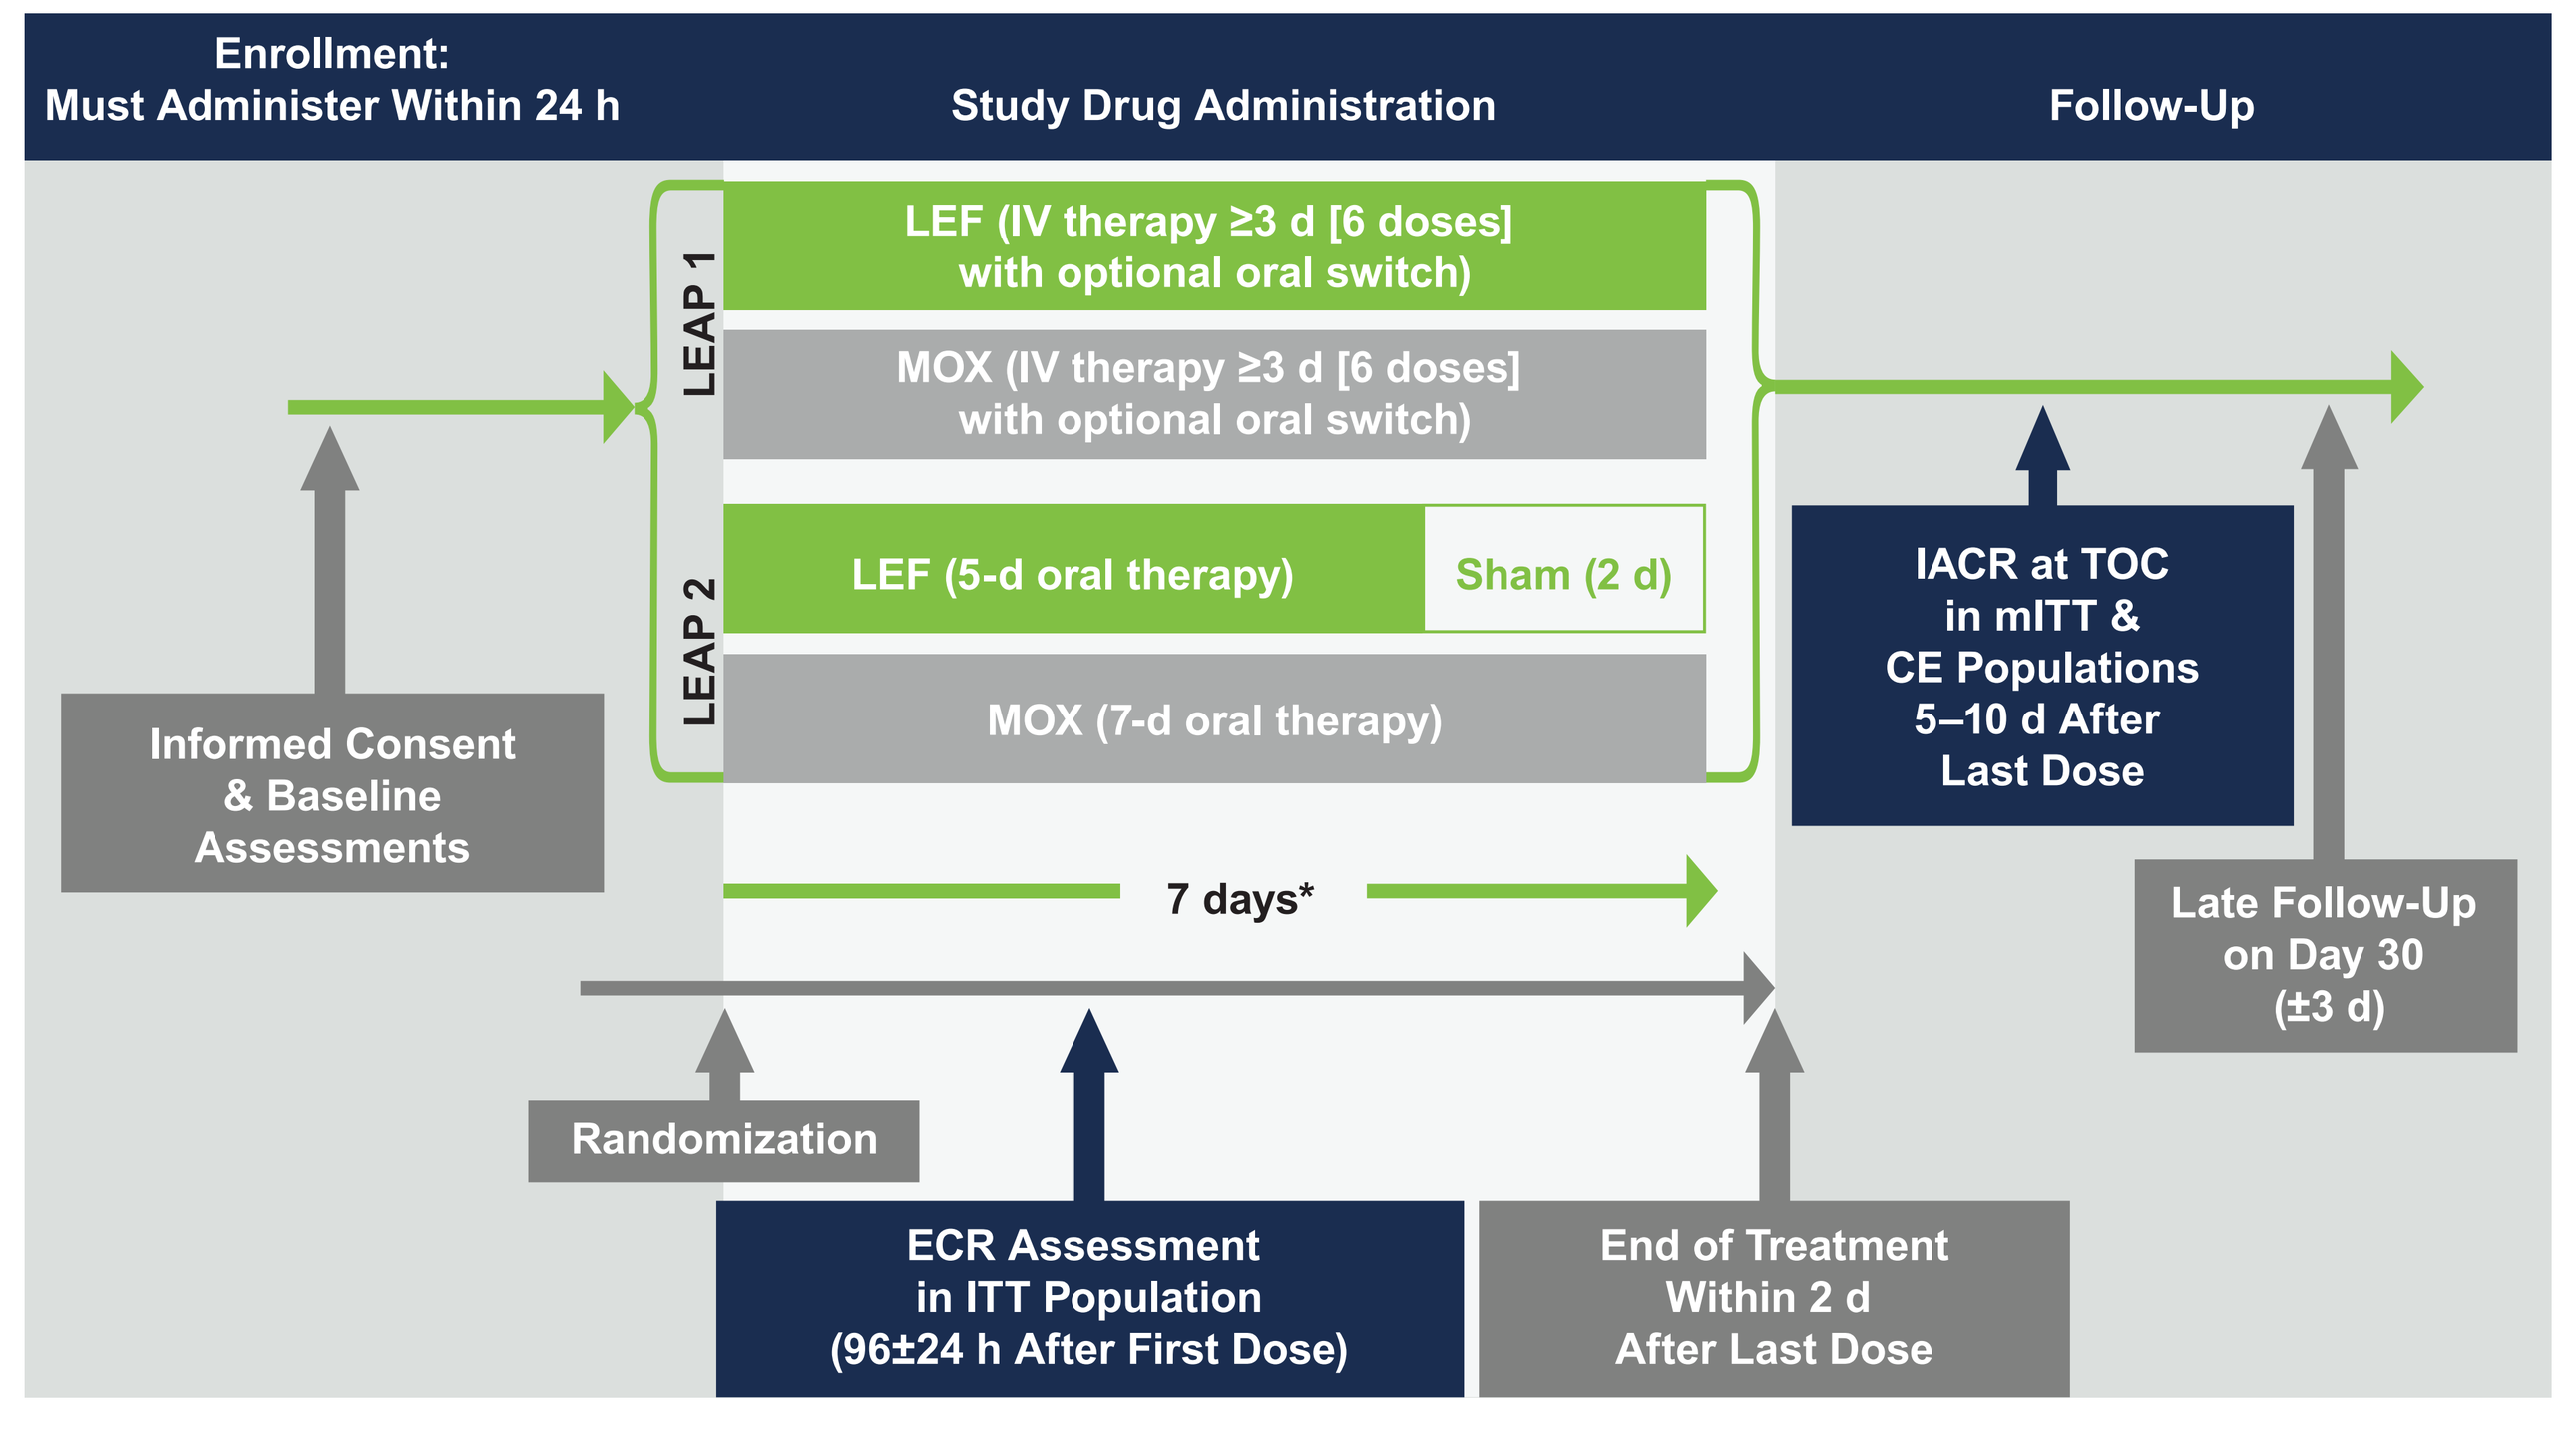

Enrollment:
Must Administer Within 24 h
Study Drug Administration
Follow-Up
LEAP 1
LEF (IV therapy ≥3 d [6 doses] with optional oral switch)
MOX (IV therapy ≥3 d [6 doses] with optional oral switch)
LEAP 2
LEF (5-d oral therapy)
Sham (2 d)
MOX (7-d oral therapy)
Informed Consent & Baseline Assessments
7 days*
IACR at TOC in mITT & CE Populations 5–10 d After Last Dose
Late Follow-Up on Day 30 (±3 d)
Randomization
ECR Assessment in ITT Population (96±24 h After First Dose)
End of Treatment Within 2 d After Last Dose

CABP=community-acquired bacterial pneumonia; CE=clinically evaluable (patients who met predefined specified criteria related to protocol adherence); ECR=early clinical response (patient assessed as responder if alive, showed improvement in ≥2 CABP signs and symptoms, no worsening in any CABP sign or symptom, and no receipt of a concomitant nonstudy antibiotic for the current CABP episode); IACR=investigator assessment of clinical response (patients assessed as success if alive, with signs and symptoms of CABP resolved or improved such that no additional antibacterial therapy was administered for CABP); ITT=intent to treat (all randomized patients); IV=intravenous; LEAP=Lefamulin Evaluation Against Pneumonia; LEF=lefamulin; mITT=modified ITT (all randomized patients who received any amount of study drug); MOX=moxifloxacin; TOC=test-of-cure visit.
*In LEAP 1, the original protocol indicated a LEF treatment period of 5 days (but 10 days in patients with CABP due to *Legionella pneumophila* or methicillin-resistant *Staphylococcus aureus* [MRSA] or in patients with *Streptococcus pneumoniae* and bacteremia); however, this was later adjusted to 7 days (except in cases of confirmed MRSA, which continued to receive 10 days of treatment) to reduce medication errors and limit the burden on study sites.[11] If MRSA was suspected, linezolid or linezolid placebo was added to MOX or LEF therapy, respectively. A total of 14/275 (5.1%) patients randomized to MOX and 9/276 (3.3%) patients randomized to LEF received linezolid and linezolid placebo, respectively, because of suspected MRSA at baseline.

Patients and Assessments

- Patients ≥18 years old with CABP of PORT risk class III–V or II–IV were eligible for LEAP 1 and LEAP 2, respectively
 - In both studies, the primary efficacy endpoint for the US Food and Drug Administration (FDA) was early clinical response (ECR) at 96±24 hours after first dose of study drug in the intent-to-treat (ITT) population
 - The European Medicines Agency coprimary endpoints (FDA secondary endpoints) were investigator assessment of clinical response (IACR) at the test-of-cure (TOC) assessment 5–10 days after the last dose of study drug in the modified ITT (mITT) and clinically evaluable (CE) populations (see **Figure 1** footnote for study population definitions)
 - Pooled analyses used a 10% noninferiority margin

RESULTS

Patients

- 1289 patients randomized to LEF (*n*=646) and MOX (*n*=643) were included in the pooled ITT population
- In this pooled analysis, patient demographics and disease characteristics were generally well balanced between treatment groups **(Table 1)**
 - Overall, patients in the pooled analysis were predominantly male (55.6%) and white (79.3%), with a mean (SD) age of 58.7 (16.1) years
 - Approximately 51% of patients had impaired renal function, and approximately 71% had a PORT risk class of ≥III

Table 1. Demographics and Baseline Characteristics (Pooled ITT Population)

	LEF (*n*=646)	MOX (*n*=643)
Age, y, mean (SD)	58.9 (16.5)	58.5 (15.7)
Men, *n* (%)	377 (58.4)	340 (52.9)
White, *n* (%)	513 (79.4)	509 (79.2)
BMI, kg/m², mean (SD)	26.5 (5.8)	26.4 (6.0)
Renal status,* *n* (%)		
Normal function	311 (48.1)	312 (48.5)
Mild impairment	201 (31.1)	192 (29.9)
Moderate impairment	125 (19.3)	132 (20.5)
Severe impairment	7 (1.1)	6 (0.9)
Missing	2 (0.3)	1 (0.2)
Prior antibiotic use,† *n* (%)	147 (22.8)	145 (22.6)
Smoking history, *n* (%)	284 (44.0)	242 (37.6)
Hypertension history, *n* (%)	248 (38.4)	253 (39.3)
Diabetes mellitus history, *n* (%)	80 (12.4)	88 (13.7)
Arrhythmia history, *n* (%)	43 (6.7)	30 (4.7)
PORT risk class,‡ *n* (%)		
I/II	184 (28.5)	192 (29.9)
III	341 (52.8)	334 (51.9)
IV/V	121 (18.7)	117 (18.2)
CURB-65 score,§ *n* (%)		
0–2	610 (94.4)	604 (93.9)
3–5	36 (5.6)	39 (6.1)
Minor ATS severity criteria,‖ *n* (%)	85 (13.2)	85 (13.2)
Modified ATS severity criteria,¶ *n* (%)	53 (8.2)	57 (8.9)
SIRS,^ *n* (%)	621 (96.1)	609 (94.7)
Baseline liver enzyme elevation,# *n* (%)	119 (18.4)	144 (22.4)
Bacteremia, *n* (%)	13 (2.0)	12 (1.9)

ALT=alanine aminotransferase; AST=aspartate aminotransferase; ATS=American Thoracic Society; BMI=body mass index; BUN=blood urea nitrogen; CrCl=creatinine clearance; eCRF=electronic case report form; ITT=intent to treat; LEF=lefamulin; MOX=moxifloxacin; PORT=Pneumonia Outcomes Research Team; SIRS=Systemic Inflammatory Response Syndrome; ULN=upper limit of normal; WBC=white blood cell (count).
*National Kidney Foundation categories of renal impairment[13] based on baseline central laboratory serum creatinine. When baseline central laboratory serum creatinine was not available, local serum creatinine results were used. Renal impairment categories are: normal (CrCl ≥90 mL/min), mild (CrCl of 60 to <90 mL/min), moderate (CrCl of 30 to <60 mL/min), and severe (CrCl <30 mL/min). †Patients received a single dose of short-acting systemic antibacterial medication within 72 hours before randomization; randomization was stratified and capped such that no more than 25% of the total ITT population met these criteria. ‡PORT risk class was calculated programmatically using data obtained at the site and reported in the eCRF and was not always consistent with the site-reported PORT risk class used for enrollment/stratification; as a result, 3 patients with PORT risk class I (LEF, *n*=1; MOX, *n*=2) were enrolled. §Defined as confusion of new onset, BUN >19 mg/dL, respiratory rate ≥30 breaths/min, systolic blood pressure <90 mm Hg or diastolic blood pressure ≤60 mm Hg, and age ≥65 years. ‖Defined as presence of ≥3 of the following 9 criteria at baseline: respiratory rate ≥30 breaths/min, O_2 saturation <90% or PaO_2 <60 mm Hg, BUN ≥20 mg/dL, WBC <4000 cells/mm³, confusion, multilobar infiltrates, platelets <100,000 cells/mm³, temperature <36°C, or systolic blood pressure <90 mm Hg.[14] ¶Defined as presence of ≥3 of the following 6 criteria at baseline: respiratory rate ≥30 breaths/min, SpO_2/FiO_2 <274 where $SpO_2/FiO_2 = 64+0.84\ (PaO_2/FiO_2)$, BUN ≥20 mg/dL, confusion, age ≥65 years, or multilobar infiltrates.[15] ^Defined as having ≥2 of the following 4 criteria at baseline: temperature <36°C or >38°C; heart rate >90 bpm; respiratory rate >20 breaths/min; and WBC <4000 cells/mm³, WBC >12,000 cells/mm³, or immature polymorphonuclear neutrophils >10%. #Defined as AST or ALT >ULN.

Early Clinical Response and Investigator Assessment of Clinical Response

- In the pooled ITT population, LEF was noninferior (10% margin) to MOX for ECR **(Figure 2)**
- Similarly, LEF was noninferior (10% margin) to MOX for IACR success **(Figure 2)**, with rates at TOC in the mITT and CE populations that were high and similar for both LEF and MOX

Clinical Efficacy by PORT Risk Class

- LEF demonstrated high efficacy across all PORT-defined severities of CABP **(Table 2)**

Clinical Efficacy by Subpopulations

- Overall, LEF demonstrated high ECR and IACR success rates across most baseline demographic characteristics and CABP severity indices **(Figure 3)**; similar results were seen at TOC in the pooled CE population (data not shown)

Other Findings From LEAP 1 and LEAP 2 Pooled Analyses

- Please refer to Poster E1053 for safety and tolerability results and Poster E1142 for efficacy and safety results in patients with atypical respiratory pathogens

Figure 2. Pooled Early Clinical Responder and Investigator Assessment of Clinical Response Success Rates

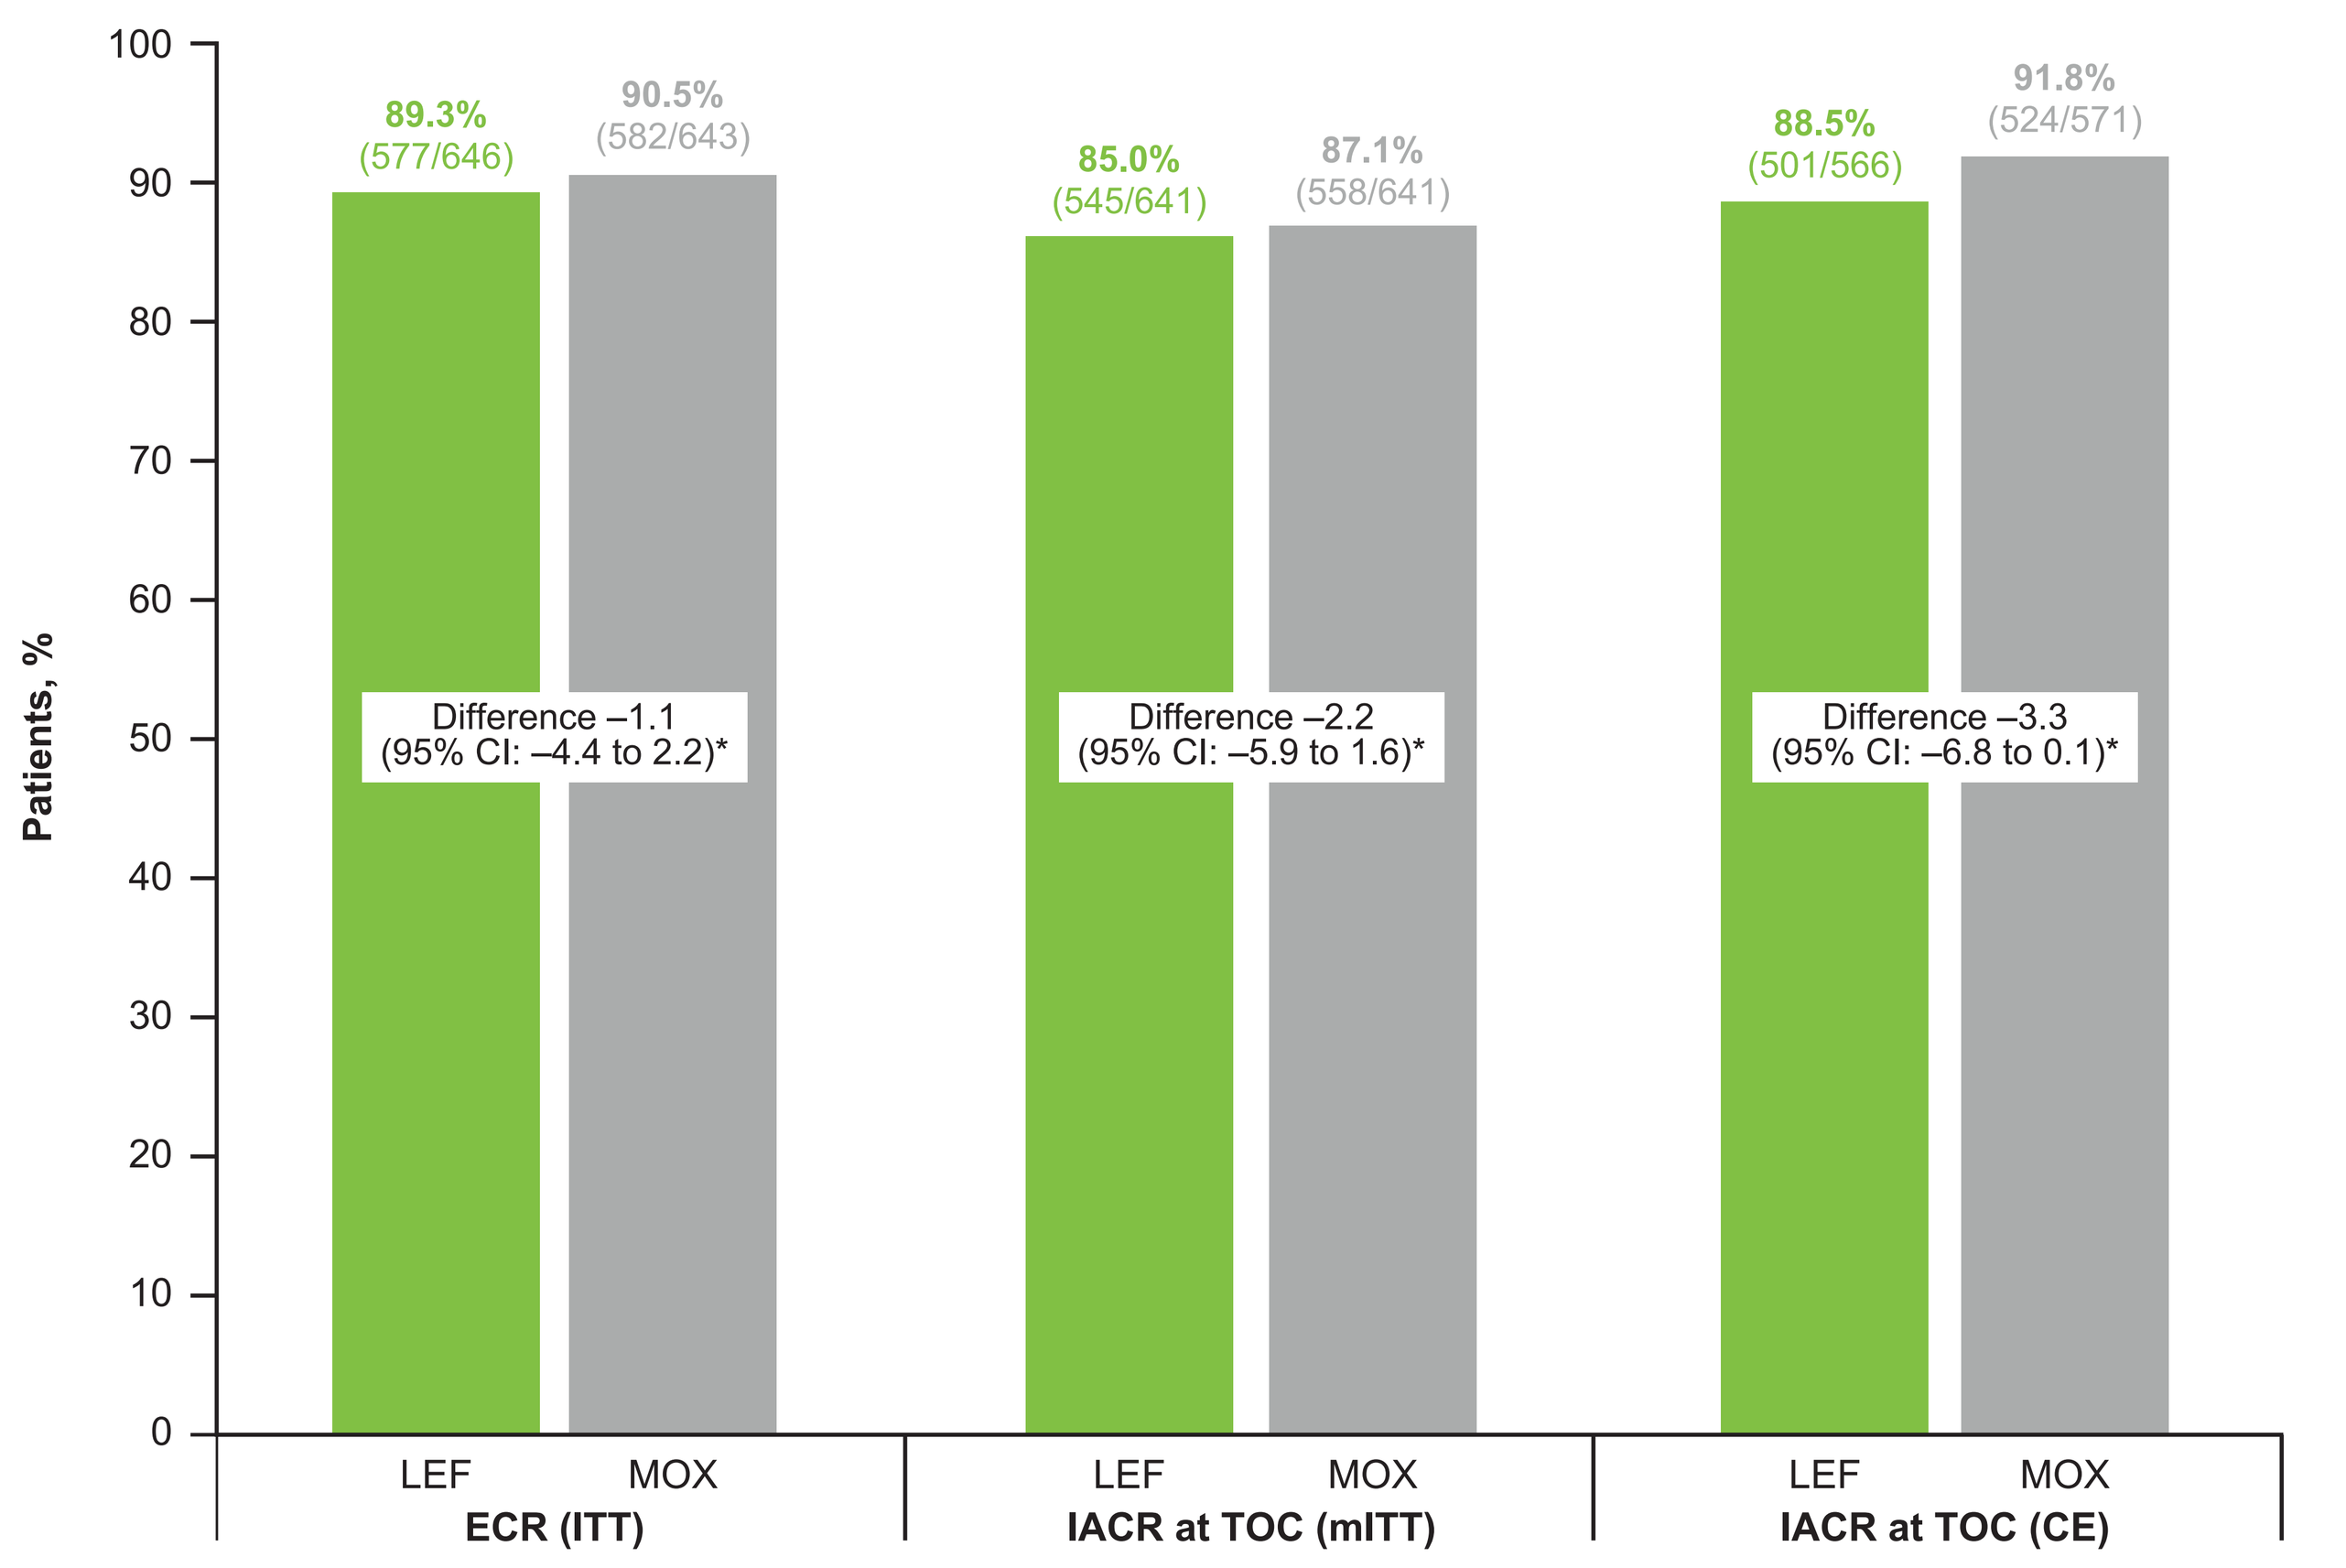

Patients, %
100
90
80
70
60
50
40
30
20
10
0
89.3% (577/646)
90.5% (582/643)
Difference –1.1 (95% CI: –4.4 to 2.2)*
85.0% (545/641)
87.1% (558/641)
Difference –2.2 (95% CI: –5.9 to 1.6)*
88.5% (501/566)
91.8% (524/571)
Difference –3.3 (95% CI: –6.8 to 0.1)*
LEF MOX
ECR (ITT)
LEF MOX
IACR at TOC (mITT)
LEF MOX
IACR at TOC (CE)

CE=clinically evaluable; ECR=early clinical response; IACR=investigator assessment of clinical response; ITT=intent to treat; LEF=lefamulin; mITT=modified ITT; MOX=moxifloxacin; TOC=test-of-cure visit.
*Computed using the method of Miettinen and Nurminen, adjusted for study (ECR and IACR) and receipt of a prior single-dose short-acting antibiotic (IACR only), with the inverse variance of the effect size as the stratum weights.

Table 2. Pooled Response by PORT Risk Class

Outcome (Analysis Population)	PORT Risk Class,* *n/N* (%)	LEF	MOX	Treatment Difference (95% CI)
ECR (ITT)	I/II†	169/184 (91.8)	179/192 (93.2)	–1.4 (–7.2 to 4.5)‡
	III	307/341 (90.0)	307/334 (91.9)	–1.9 (–6.4 to 2.5)§
	IV/V	101/121 (83.5)	96/117 (82.1)	1.5 (–8.2 to 11.3)§
IACR at TOC (mITT)	I/II†	158/184 (85.9)	176/192 (91.7)	–5.8 (–12.7 to 1.1)‡
	III	294/337 (87.2)	286/333 (85.9)	1.5 (–3.6 to 6.6)§
	IV/V	93/120 (77.5)	96/116 (82.8)	–5.3 (–15.6 to 5.0)§
IACR at TOC (CE)	I/II†	142/162 (87.7)	164/172 (95.3)	–7.7 (–14.3 to –1.1)‡
	III	279/307 (90.9)	271/300 (90.3)	0.6 (–4.1 to 5.3)§
	IV/V	80/97 (82.5)	89/99 (89.9)	–7.3 (–17.4 to 2.4)§

CE=clinically evaluable; ECR=early clinical response; eCRF=electronic case report form; IACR=investigator assessment of clinical response; ITT=intent to treat; LEF=lefamulin; mITT=modified ITT; MOX=moxifloxacin; PORT=Pneumonia Outcomes Research Team; TOC=test-of-cure visit.
*PORT risk class was calculated programmatically using data obtained at the site and reported in the eCRF and was not always consistent with the site-reported PORT risk class used for enrollment/stratification. †In LEAP 2, the ITT and mITT populations included 3 patients with PORT risk class I (LEF, *n*=1; MOX, *n*=2). ‡Absolute treatment difference (LEF minus MOX). CI was computed using a continuity-corrected Z-test. §Weighted treatment difference and CI were computed using the method of Miettinen and Nurminen, adjusted for study, with the inverse variance of the effect size as the stratum weights.

Figure 3. Pooled (A) Early Clinical Response and (B) Investigator Assessment of Clinical Response by Baseline Variables

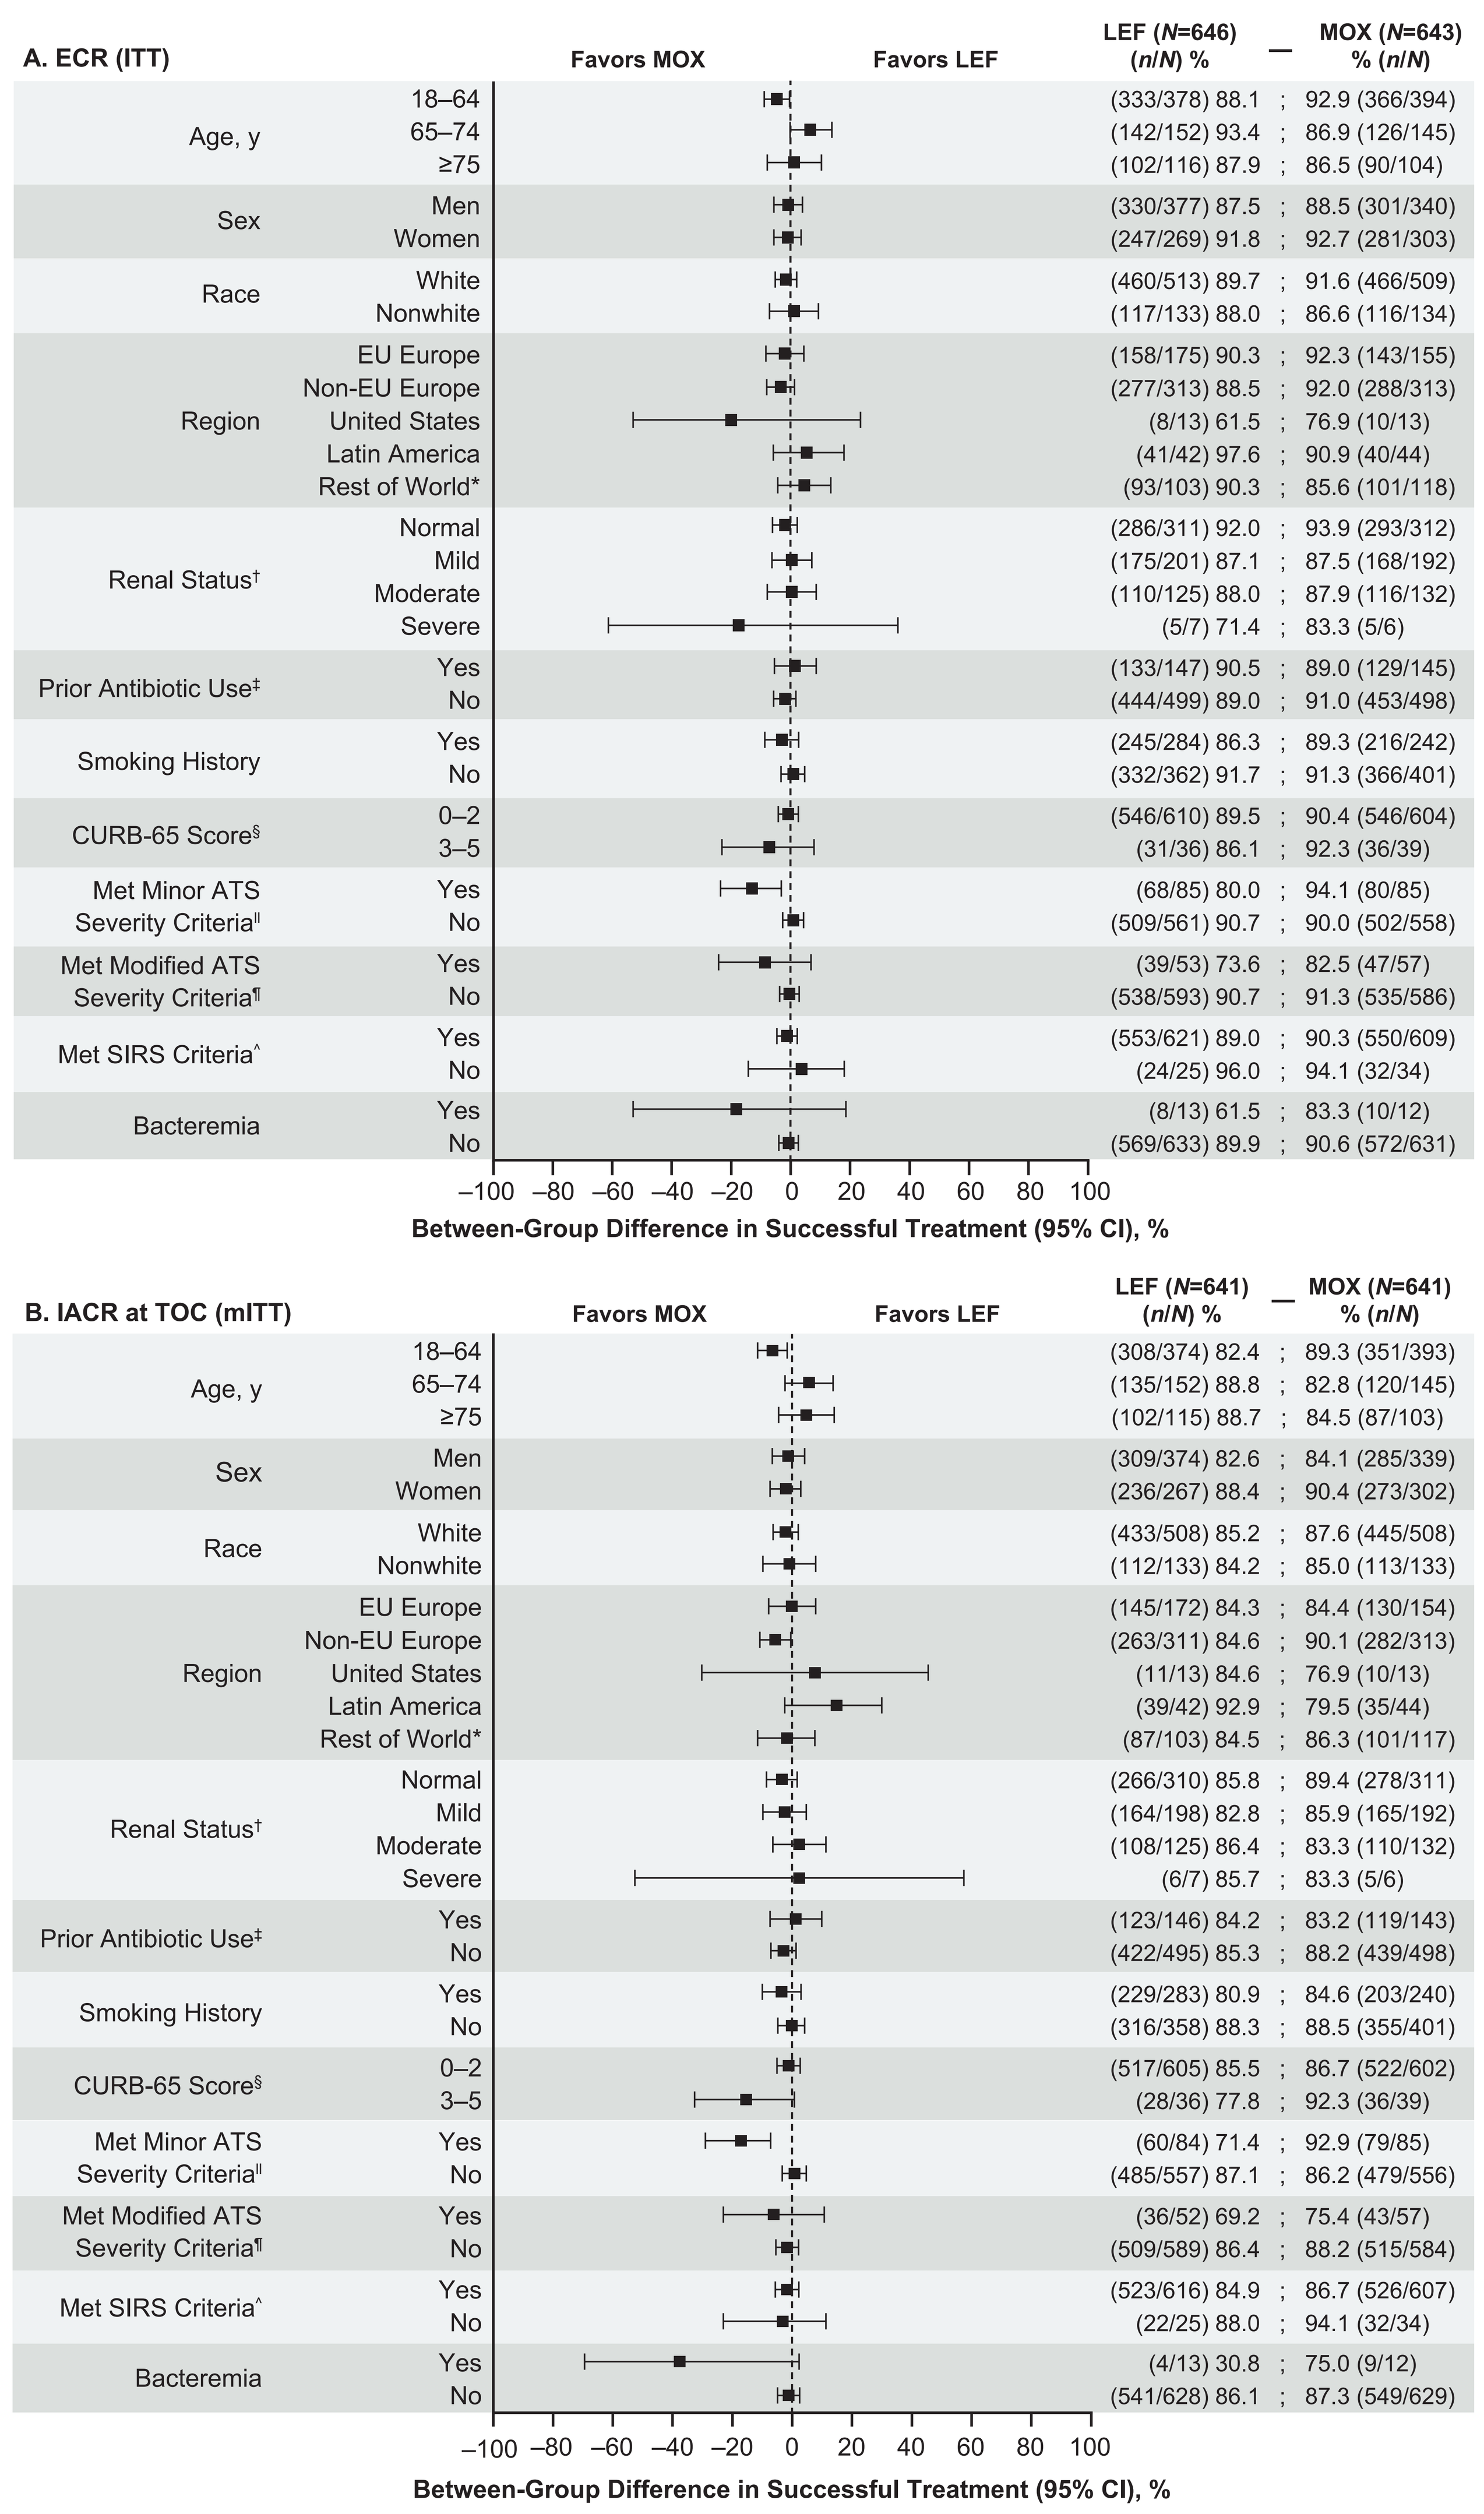

A. ECR (ITT)
Favors MOX
Favors LEF
LEF (N=646) (n/N) % — MOX (N=643) % (n/N)
Age, y 18–64 (333/378) 88.1 ; 92.9 (366/394)
65–74 (142/152) 93.4 ; 86.9 (126/145)
≥75 (102/116) 87.9 ; 86.5 (90/104)
Sex Men (330/377) 87.5 ; 88.5 (301/340)
Women (247/269) 91.8 ; 92.7 (281/303)
Race White (460/513) 89.7 ; 91.6 (466/509)
Nonwhite (117/133) 88.0 ; 86.6 (116/134)
Region EU Europe (158/175) 90.3 ; 92.3 (143/155)
Non-EU Europe (277/313) 88.5 ; 92.0 (288/313)
United States (8/13) 61.5 ; 76.9 (10/13)
Latin America (41/42) 97.6 ; 90.9 (40/44)
Rest of World* (93/103) 90.3 ; 85.6 (101/118)
Renal Status† Normal (286/311) 92.0 ; 93.9 (293/312)
Mild (175/201) 87.1 ; 87.5 (168/192)
Moderate (110/125) 88.0 ; 87.9 (116/132)
Severe (5/7) 71.4 ; 83.3 (5/6)
Prior Antibiotic Use‡ Yes (133/147) 90.5 ; 89.0 (129/145)
No (444/499) 89.0 ; 91.0 (453/498)
Smoking History Yes (245/284) 86.3 ; 89.3 (216/242)
No (332/362) 91.7 ; 91.3 (366/401)
CURB-65 Score§ 0–2 (546/610) 89.5 ; 90.4 (546/604)
3–5 (31/36) 86.1 ; 92.3 (36/39)
Met Minor ATS Severity Criteria‖ Yes (68/85) 80.0 ; 94.1 (80/85)
No (509/561) 90.7 ; 90.0 (502/558)
Met Modified ATS Severity Criteria¶ Yes (39/53) 73.6 ; 82.5 (47/57)
No (538/593) 90.7 ; 91.3 (535/586)
Met SIRS Criteria^ Yes (553/621) 89.0 ; 90.3 (550/609)
No (24/25) 96.0 ; 94.1 (32/34)
Bacteremia Yes (8/13) 61.5 ; 83.3 (10/12)
No (569/633) 89.9 ; 90.6 (572/631)
–100 –80 –60 –40 –20 0 20 40 60 80 100
Between-Group Difference in Successful Treatment (95% CI), %
B. IACR at TOC (mITT)
Favors MOX
Favors LEF
LEF (N=641) (n/N) % — MOX (N=641) % (n/N)
Age, y 18–64 (308/374) 82.4 ; 89.3 (351/393)
65–74 (135/152) 88.8 ; 82.8 (120/145)
≥75 (102/115) 88.7 ; 84.5 (87/103)
Sex Men (309/374) 82.6 ; 84.1 (285/339)
Women (236/267) 88.4 ; 90.4 (273/302)
Race White (433/508) 85.2 ; 87.6 (445/508)
Nonwhite (112/133) 84.2 ; 85.0 (113/133)
Region EU Europe (145/172) 84.3 ; 84.4 (130/154)
Non-EU Europe (263/311) 84.6 ; 90.1 (282/313)
United States (11/13) 84.6 ; 76.9 (10/13)
Latin America (39/42) 92.9 ; 79.5 (35/44)
Rest of World* (87/103) 84.5 ; 86.3 (101/117)
Renal Status† Normal (266/310) 85.8 ; 89.4 (278/311)
Mild (164/198) 82.8 ; 85.9 (165/192)
Moderate (108/125) 86.4 ; 83.3 (110/132)
Severe (6/7) 85.7 ; 83.3 (5/6)
Prior Antibiotic Use‡ Yes (123/146) 84.2 ; 83.2 (119/143)
No (422/495) 85.3 ; 88.2 (439/498)
Smoking History Yes (229/283) 80.9 ; 84.6 (203/240)
No (316/358) 88.3 ; 88.5 (355/401)
CURB-65 Score§ 0–2 (517/605) 85.5 ; 86.7 (522/602)
3–5 (28/36) 77.8 ; 92.3 (36/39)
Met Minor ATS Severity Criteria‖ Yes (60/84) 71.4 ; 92.9 (79/85)
No (485/557) 87.1 ; 86.2 (479/556)
Met Modified ATS Severity Criteria¶ Yes (36/52) 69.2 ; 75.4 (43/57)
No (509/589) 86.4 ; 88.2 (515/584)
Met SIRS Criteria^ Yes (523/616) 84.9 ; 86.7 (526/607)
No (22/25) 88.0 ; 94.1 (32/34)
Bacteremia Yes (4/13) 30.8 ; 75.0 (9/12)
No (541/628) 86.1 ; 87.3 (549/629)
–100 –80 –60 –40 –20 0 20 40 60 80 100
Between-Group Difference in Successful Treatment (95% CI), %

ATS=American Thoracic Society; BUN=blood urea nitrogen; CrCl=creatinine clearance; ECR=early clinical response; EU=European Union; IACR=investigator assessment of clinical response; ITT=intent to treat; LEF=lefamulin; mITT=modified ITT; MOX=moxifloxacin; SIRS=Systemic Inflammatory Response Syndrome; WBC=white blood cell (count).
Weighted treatment difference and CI were computed using the method of Miettinen and Nurminen, adjusted for study, with the inverse variance of the effect size as the stratum weights.
*Philippines, South Africa, South Korea, Taiwan, and Thailand. †National Kidney Foundation categories of renal impairment[13] based on baseline central laboratory serum creatinine. When baseline central laboratory serum creatinine was not available, local serum creatinine results were used. Renal impairment categories are: normal (CrCl ≥90 mL/min), mild (CrCl of 60 to <90 mL/min), moderate (CrCl of 30 to <60 mL/min), and severe (CrCl <30 mL/min). ‡Patients received a single dose of short-acting antibacterial medication within 72 hours before randomization; randomization was stratified and capped such that no more than 25% of the total ITT population met these criteria. §Defined as confusion of new onset, BUN >19 mg/dL, respiratory rate ≥30 breaths/min, systolic blood pressure <90 mm Hg or diastolic blood pressure ≤60 mm Hg, and age ≥65 years. ‖Defined as presence of ≥3 of the following 9 criteria at baseline: respiratory rate ≥30 breaths/min, O_2 saturation <90% or PaO_2 <60 mm Hg, BUN ≥20 mg/dL, WBC <4000 cells/mm³, confusion, multilobar infiltrates, platelets <100,000 cells/mm³, temperature <36°C, or systolic blood pressure <90 mm Hg.[14] ¶Defined as presence of ≥3 of the following 6 criteria at baseline: respiratory rate ≥30 breaths/min, SpO_2/FiO_2 <274 where $SpO_2/FiO_2 = 64+0.84\ (PaO_2/FiO_2)$, BUN ≥20 mg/dL, confusion, age ≥65 years, or multilobar infiltrates.[15] ^Defined as having ≥2 of the following 4 criteria at baseline: temperature <36°C or >38°C; heart rate >90 bpm; respiratory rate >20 breaths/min; and WBC <4000 cells/mm³, WBC >12,000 cells/mm³, or immature polymorphonuclear neutrophils >10%.

CONCLUSIONS AND CLINICAL IMPLICATIONS

- LEF demonstrated high ECR and IACR rates and was found to be noninferior for both endpoints to standard of care comparator MOX
- Response rates remained high across pneumonia severities as assessed by PORT risk class and baseline variables
- LEF was generally safe and well tolerated regardless of the route of administration (IV only, IV-to-oral, oral only)
- LEF may provide a valuable IV and oral monotherapy alternative to fluoroquinolones or macrolides for empiric treatment of CABP in adults

REFERENCES

(1) Xu J, et al. Deaths: final data for 2016. US Department of Health and Human Services, Centers for Disease Control and Prevention, National Center for Health Statistics. Available at: https://www.cdc.gov/nchs/data/nvsr/nvsr67/nvsr67_05.pdf. Accessed August 26, 2019.
(2) Peyrani P, et al. *Expert Rev Respir Med.* 2019;13(2):139-152.
(3) McDermott KW, et al. Trends in hospital inpatient stays in the United States, 2005-2014: statistical brief #225. *Healthcare Cost and Utilization Project (HCUP) Statistical Briefs.* Rockville, MD: Agency for Healthcare Research and Quality; 2017.
(4) Paukner S, et al. *Antimicrob Agents Chemother.* 2019;63(4):e02161-18.
(5) Aliberti S and Kaye KS. *Postgrad Med.* 2013;125(6):31-42.
(6) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(7) Schlünzen F, et al. *Mol Microbiol.* 2004;54(5):1287-1294.
(8) Eyal Z, et al. *Sci Rep.* 2016;6:39004.
(9) Zeitlinger M, et al. *J Antimicrob Chemother.* 2016;71(4):1022-1026.
(10) Wicha WW, et al. *J Antimicrob Chemother.* 2019;74(suppl 3):iii11-iii18.
(11) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(12) Alexander E, et al. *JAMA.* 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].
(13) Cockcroft DW and Gault MH. *Nephron.* 1976;16(1):31-41.
(14) Mandell LA, et al. *Clin Infect Dis.* 2007;44(suppl 2):S27-S72.
(15) Li HY, et al. *Medicine (Baltimore).* 2015;94(36):e1474.

Acknowledgments & Disclosures

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Jennifer Schranz, Lisa Goldberg, Elizabeth Alexander, and Steven P. Gelone are employees of/stockholders in Nabriva Therapeutics plc. Anita F. Das has served as a consultant for Achaogen, AntibioTx, Boston Pharmaceuticals, Cempra, ContraFect, IterumTx, Nabriva Therapeutics, Paratek, Tetraphase, Theravance, UTILITY, Wockhardt, and Zavante. Christian Sandrock has served as a consultant for Allergan and Nabriva Therapeutics, received grants from the National Institutes of Health and the Health Resources & Services Administration, and received nonfinancial support from the State of California. Gregory J. Moran has received grants from ContraFect and Nabriva Therapeutics. Andrew F. Shorr has served as a consultant for or received grant support from Achaogen, Melinta, Merck, Nabriva Therapeutics, Paratek, and Tetraphase.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/CHEST2019_LEAP1_2_Efficacy

CHEST Annual Meeting 2019: October 19–23, New Orleans, LA, USA