02146

Evaluation of Pleuromutilins Resistance Mechanisms Among Surveillance Clinical Isolates: Results from the Worldwide Surveillance Program for Lefamulin in 2019

Nabriva
THERAPEUTICS

Nabriva Therapeutics
Vienna, Austria
and Fort Washington, PA, USA
www.nabriva.com

RE Mendes[1], C Hubler[1], TB Doyle[1], SP Gelone[2], S Paukner[3], M Castanheira[1], HS Sader[1]

[1]JMI Laboratories, North Liberty, Iowa USA; [2]Nabriva Therapeutics Inc., Fort Washington, PA USA; [3]Nabriva Therapeutics, GmbH, Vienna, Austria

INTRODUCTION

- Lefamulin (Xenleta™) is a first in human pleuromutilin antibiotic approved in the US, Europe, and Canada for the treatment of community acquired bacterial pneumonia (CABP).
 - Both intravenous and oral formulations are available for the treatment of CABP in adults.
 - Lefamulin has a targeted spectrum of activity against key pathogens including typical Gram-positive, fastidious Gram-negative and atypical organisms, including strains resistant to standard-of-care therapies.
 - Lefamulin is a first-in-class, semi-synthetic pleuromutilin that inhibits bacterial protein synthesis by binding to the 50S ribosomal subunit at the A- and P- sites in the peptidyl transferase center (PTC) via an "induced-fit" mechanism, which prohibits the correct positioning of the tRNA.
- The *in vitro* activity and emergence of resistance to lefamulin are being monitored against a global collection of Gram-positive and fastidious Gram-negative organisms through the SENTRY Antimicrobial Surveillance Program.
- This study characterized possible resistance mechanisms among surveillance isolates with elevated lefamulin MICs collected globally in 2019.

MATERIALS AND METHODS

Bacterial isolates

- A total of 3,975 *Staphylococcus aureus*, coagulase-negative staphylococci (CoNS), *Streptococcus pneumoniae*, β-haemolytic streptococci, viridans group streptococci, *Moraxella catarrhalis*, and *Haemophilus* spp. were included as part of the surveillance study for 2019 (Table 1).
 - A total of 36 (0.8%) isolates met the MIC screening criteria based on the US FDA and EUCAST breakpoints or tentative epidemiological cut-off (ECOFF) values.
 - *S. aureus*: ≥0.5 mg/L, with an FDA susceptible breakpoint ≤0.25 mg/L
 - CoNS and β-haemolytic *Streptococcus* spp.: ≥0.5 mg/L
 - *S. pneumoniae*: ≥1 mg/L, with an FDA susceptible breakpoint ≤0.5 mg/L
 - *H. influenzae*: ≥4 mg/L, with an FDA susceptible breakpoint ≤2 mg/L
- Bacterial isolate identification was confirmed by standard algorithms and supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany) and genome sequencing.

Antimicrobial susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines using frozen-form broth microdilution panels containing CAMHB; supplemented with 2.5-5% lysed horse blood for streptococci; HTM was used for *H. influenzae*.
- Quality assurance was performed by concurrently testing the CLSI-recommended quality control reference strains.

Characterization of resistance mechanisms by next-generation sequencing

- Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific™ KingFisher™ Flex Magnetic Particle Processor (Cleveland, OH, USA).
- DNA libraries were prepared using the Nextera™ library construction protocol (Illumina, San Diego, CA, USA) according to manufacturer instructions.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.9.0.
- Additional sequences of intrinsic genes associated with the pleuromutilin binding site, including 23S rRNA (PTC), *rplC* (L3), *rplD* (L4), and *rplV* (L22), were evaluated against a susceptible reference strain of the corresponding species.
- All intrinsic 23S rRNA target genes or ribosomal protein amino acid sequences were considered wild type if 100.0% homology with the respective reference sequences was displayed.

Transcriptional levels of AcrA

- *Haemophilus* spp. were subjected to sequencing analysis of AcrR and the quantification of AcrAB-TolC expression.
 - The total mRNA was extracted and purified using the RNeasy Mini Kit in the QIAcube workstation according to manufacturer instructions and treatment with RNAse-free DNase (Promega, Madison, Wisconsin).
- The quantification of mRNA and sample quality were assessed using the RNA 6000 Nano kit from Agilent on the Agilent 2100 Bioanalyzer, according to manufacturer instructions.
- The transcription levels of *acrA* were determined by relative quantification using real-time PCR (qRT-PCR) assays in the StepOne Plus instrument (Life Technologies, Foster City, California).
- Transcription levels of *acrA* were measured by the quantification of the target gene mRNA using a normalized expression analysis method with a housekeeping reference gene (*gyrA*).

Multilocus sequence typing

- Multilocus sequence typing (MLST) was performed by extracting the previously defined set of 7 housekeeping gene fragments (~500 bp) and comparing with allelic variants on the MLST website.

RESULTS

- Among 873 *S. aureus* included in the 2019 lefamulin surveillance program, 2 (0.2%) isolates had lefamulin MIC values of 1 or >16 mg/L and were selected for this study (Table 1).
 - These 2 non-susceptible isolates harboured *vga*(A) or *lsa*(E)
 - 871 (99.8%) *S. aureus* isolates showed lefamulin MICs of 0.015-0.25 mg/L ($MIC_{50/90}$ of 0.06/0.12 mg/L) and are susceptible based on FDA and EUCAST breakpoints (S ≤0.25 mg/L).
- A total of 24 of 329 (7.3%) CoNS, spanning 4 total species with lefamulin MIC values of 0.5–≥32 mg/L, were selected for genetic characterization.
 - Most CoNS isolates carried *vga* gene variants (19/24; 79.2%) and had lefamulin MIC results of 0.5–≥16 mg/L).
 - Two *S. epidermidis* isolates from Mexico carried *cfr* (2/24; lefamulin MIC, 8 mg/L). These 2 isolates were from 2 patients hospitalized in the same medical centre and exhibited the same MLST.
 - One *S. epidermidis* from Germany carried a G2576T mutation (1/24; lefamulin MIC of 0.5 mg/L).
 - One *S. haemolyticus* and 1 *S. saprophyticus* with a lefamulin MIC of 0.5–1 mg/L did not show any mechanisms previously associated with lefamulin resistance and MICs represent the upper end of the MIC wildtype distributions.
- Three *S. pneumoniae* were non-susceptible to lefamulin (MIC, 1 mg/L), but these isolates did not show any clear mechanisms implicated with pleuromutilin resistance and MIC values represent the upper end of the MIC wildtype distributions (Tables 1 and 4)
- A single *Streptococcus agalactiae* (0.3% of β-haemolytic streptococci) with an elevated lefamulin MIC (32 mg/L) was detected in this study (collected in Italy).
 - This strain carried *lsa*(E) (Tables 1 and 4)
- A total of 6 (1.2%) *H. influenzae* showed lefamulin nonsusceptible MICs of 4–8 mg/L.
 - All but 1 isolate (1129277) showed derepression of *acrA* and *acrB* expression due to a premature stop codon in *acrR*.
 - Additionally, these isolates had either an 23S rRNA (A2058G or A2059G) or a L4/L22 alteration, except for isolate 1126213 that had an *acrR* with a premature stop codon only (Table 5).

Table 1. Lefamulin MIC distributions obtained during the 2019 surveillance program

Organism/organism group (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (mg/L) of: [a]														MIC50	MIC90
	≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>32		
Staphylococcus aureus (873)	0 0.0	3 0.3	46 5.6	490 61.7	312 97.4	20 99.7	0 99.7	1 99.9	0 99.9	0 99.9	0 99.9	0 99.9		1 100.0	0.06	0.12
Coagulase-negative staphylococci (329)	6 1.8	10 4.9	120 41.3	132 81.5	35 92.1	2 92.7	4 93.9	7 96.0	4 97.3	1 97.6	2 98.2	3 99.1		3 100.0	0.06	0.12
Streptococcus pneumoniae (1,384)	3 0.2	20 1.7	131 11.1	510 48.0	571 89.2	143 99.6	3 99.8	3 100.0							0.12	0.25
β-haemolytic streptococci (367)			253 68.9	94 94.6	17 99.2	2 99.7	0 99.7	0 99.7	0 99.7	0 99.7	0 99.7	0 99.7	1 100.0		≤0.03	0.06
Haemophilus influenzae (490)	2 0.4	5 1.4	1 1.6	5 2.7	18 6.3	73 21.2	214 64.9	123 90.0	43 98.8	3 99.4	3 100.0				0.5	1
Moraxella catarrhalis (300)	6 2.0	5 3.7	5 5.3	177 64.3	103 98.7	4 100.0									0.06	0.12

[a] Shading represents MICs above the breakpoints or ECOFF values for the respective species or groups of species. *S. bovis* group display higher and broader MIC distributions for lefamulin and were not included in the study.

Table 2. Molecular epidemiology and resistance mechanism results for *S. aureus* isolates

Collection no.	MLST	Country	MIC[a] (mg/L)	Resistance mechanisms: Gene			Ribosomal mutations[b]			
				cfr	*lsa*(E)	*vga*(A)	23S rRNA	L3	L4	L22
1099706	9	Mexico	>16	—	+	—	WT	WT	V118D	WT
1127385	9	Brazil	1	—	—	+	WT	WT	V118D	WT

MLST, multilocus sequence typing; WT, wild type
[a] MIC, minimal inhibitory concentration.
[b] The 23S rRNA mutational analysis was performed on nucleotide sequences. Mutations outside of PTC were observed but were considered polymorphisms and suppressed. The 23S rRNA sequence was designated as WT. Protein sequences that were analysed for annotating were L3, L4, and L22.

Table 3. Molecular epidemiology and resistance mechanisms results for *Staphylococcus* spp. other than *S. aureus*

Collection no.	Organism	MLST	Country	MIC[a] (mg/L)	Resistance mechanisms: Gene	Ribosomal mutations[b]			
						23S rRNA	L3	L4	L22
1117096	*S. capitis*	NA	Australia	0.5	*vga*(A)	WT	T83A	WT	WT
1118328	*S. capitis*	NA	Australia	4	*vga*(A)	WT	T83A	I78N	WT
1074687	*S. capitis*	23	Mexico	8	*vga*(A)	WT	S158Y,D159Y	WT	WT
1090848	*S. epidermidis*	23	Mexico	8	cfr	WT	S158Y,D159Y	WT	WT
1090853	*S. epidermidis*	23	Mexico	8	cfr	WT	S158Y,D159Y	WT	WT
1091111	*S. epidermidis*	16	USA	1	*vga*(A)	WT	WT	WT	WT
1092976	*S. epidermidis*	2	Germany	0.5	G2576T	WT	H146R	G71_R72insG	WT
1101658	*S. epidermidis*	87	England	16	*vga*(A)	WT	WT	WT	WT
1101683	*S. epidermidis*	35	England	16	*vga*(A)	WT	WT	WT	WT
1104351	*S. epidermidis*	25	Spain	>16	*vga*(A)	WT	WT	WT	WT
1107456	*S. epidermidis*	5	France	1	*vga*(A)	WT	WT	WT	WT
1112499	*S. epidermidis*	87	Wales	1	*vga*(A)	WT	WT	WT	WT
1112518	*S. epidermidis*	142	Wales	2	*vga*(A)	WT	WT	WT	WT
1116902	*S. epidermidis*	87	Australia	1	*vga*(A)	WT	WT	WT	WT
1123315	*S. epidermidis*	87	Belgium	1	*vga*(A)	WT	WT	WT	WT
1123338	*S. epidermidis*	87	Belgium	0.5	*vga*(A)	WT	WT	WT	WT
1123389	*S. epidermidis*	5	Belgium	2	*vga*(A)	WT	WT	WT	WT
1126561	*S. epidermidis*	59	Argentina	2	*vga*(A)	WT	WT	WT	WT
1126563	*S. epidermidis*	59	Argentina	2	*vga*(A)	WT	WT	WT	WT
1126635	*S. epidermidis*	87	Argentina	1	*vga*(A)	WT	WT	WT	WT
1099446	*S. haemolyticus*	29	Italy	16	*vga*(A)LC	WT	T82A,G137A	V130G,I142V	WT
1103323	*S. haemolyticus*	3	USA	0.5	—	WT	T82A,G137A	V130G,I142V	WT
1107535	*S. haemolyticus*	29	France	>16	*vga*(A)LC	WT	T82A,G137A	V130G,I142V	WT
1117552	*S. haemolyticus*	29	USA	>16	*vga*(A)LC	WT	T82A,G137A	V130G,I142V	WT
1125669	*S. saprophyticus*	NA	Taiwan	1	—	WT	I35V	WT	WT

MLST, multilocus sequence typing; NA, MLST method and database non-existent; WT, wild type
[a] MIC, minimal inhibitory concentration
[b] The 23S rRNA mutational analysis was performed on nucleotide sequences. Mutations outside of PTC were observed but were considered polymorphisms and suppressed. The 23S rRNA sequence was designated as WT. Protein sequences that were analysed for annotating were L3, L4, and L22.

Table 4. Molecular epidemiology results and resistance mechanisms obtained for *Streptococcus* spp.

Collection no.	Organism	Country	MIC[a] (mg/L)	Resistance mechanisms: Gene	Ribosomal mutations[b]			
					23S rRNA	L3	L4	L22
1108216	*S. pneumoniae*	USA	1	—	A2061G	WT	N20S	WT
1122375	*S. pneumoniae*	Wales	1	—	A2060G	WT	WT	WT
1129127	*S. pneumoniae*	USA	1	—	A2061G	WT	N20S	WT
1122688	*S. agalactiae*	Italy	32	*lsa*(E)	WT	WT	H45R	WT

WT, wild type
[a] MIC, minimal inhibitory concentration.
[b] The 23S rRNA mutational analysis was performed on nucleotide sequences. Additional mutations outside of PTC were observed but were considered polymorphisms and suppressed. Protein sequences that were analysed for annotating were L3, L4, and L22.

Table 5. Molecular epidemiology results and resistance mechanisms obtained for *H. influenzae*

Collection no.	Country	MIC[a] (mg/L)	Resistance mechanisms: AcrA	AcrR	Ribosomal mutations[b]			
					23S rRNA	L3	L4	L22
1091435	USA	8	1.29323	L102X	A2059G	WT	WT	WT
1114745	USA	8	25.2846	E144X	A2058G	WT	WT	WT
1126213	Vietnam	4	NA	Q57X	WT	WT	WT	WT
1126852	USA	4	55.2783	E144X	WT	WT	K57N	G91D
1129277	Chile	4	4.72255	L31H,Y45D	WT	WT	R60H	R88_G91del
1129385	Brazil	8	72.7533	L105X	WT	WT	G65D,T82I	G91D

WT, wild type; NA, not available.
[a] MIC, minimal inhibitory concentration
[b] The 23S rRNA mutational analysis was performed on nucleotide sequences. Protein sequences that were analysed for annotating were L3, L4, and L22.

CONCLUSIONS

- Gram-positive and fastidious Gram-negative isolates causing human infections with elevated (non-susceptible) lefamulin MIC results remained rare (0.9%).
- Lefamulin resistance mechanisms identified in *S. aureus*, CoNS, and streptococcal isolates were *vga*(A) and *lsa*(E), which have been described to mediate ribosomal protection.
- Target site alterations, such as mutations in the large ribosomal proteins or 23S rRNA or methylation of A2503 by the Cfr methyltransferase, were less common.
 - The *cfr* gene was detected in only 2 *S. epidermidis* surveillance isolates.
- Longitudinal surveillance studies will continue to monitor the emergence of resistance and *in vitro* stability of lefamulin activity among Gram-positive and fastidious Gram-negative clinical isolates.

Acknowledgements

This study was supported by Nabriva Therapeutics. JMI Laboratories received compensation fees for services related to preparing this poster.

References

(1) Alexander E, Goldberg L, Das AF, Moran GJ, Sandrock C, Gasink LB, Spera P, Sweeney C, Paukner S, Wicha WW, Gelone SP, Schranz J. (2019). *JAMA* 322: 1661-1671.
(2) Clinical and Laboratory Standards Institute (2019). M100Ed29E. Wayne, PA, USA.
(3) Clinical and Laboratory Standards Institute (2018). M07Ed11E. Approved Standard - Eleventh edition. Wayne, PA, USA.
(4) File TM, Goldberg L, Das A, Sweeney C, Saviski J, Gelone SP, Seltzer E, Paukner S, Wicha WW, Talbot GH, Gasink LB (2019). *Clin Infect Dis.* 69: 1856-1867.
(5) Hot C, Berthet N, Chesneau O (2014). *Antimicrob Agents and Chemother.* 58: 3335-3341.
(6) Mendes RE, Paukner S, Doyle TB, Gelone SP, Flamm RK, Sader HS (2019). *Antimicrob Agents Chemother.* 63: e02158-18.
(7) Paukner S, Riedl R (2017). *Cold Spring Harb Perspect Med.* 7:a027110.

Contact Information

Rodrigo E. Mendes, Ph.D.
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

Scan this QR code with your electronic device to receive a PDF file of the poster or visit https://www.jmilabs.com/data/posters/ECCMID2021_PleuromutilinsResistanceMechanisms.pdf.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit https://nabriva.com/news

ECCMID 2021, July 9–12, 2021