1440

Prevalence and Regional Variation in ESBLs and CRE Enterobacteriaceae (ENT) Among Hospitalized Adult Patients with ENT on a Urine Culture: A Multicenter Evaluation

Nabriva THERAPEUTICS

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

Thomas Lodise[1]; Vikas Gupta[2]; Kalvin Yu[3]; Kalpana Gupta[4]; Maureen Early[2]; Gang Ye[3]; Jennifer Schranz[2]; Steven Gelone[3]

[1]Albany College of Pharmacy and Health Sciences, Albany, NY; [2]Becton, Dickinson and Company, Franklin Lakes, NJ, USA; [3]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA; [4]Division of Infectious Diseases, Veterans Affairs Boston Healthcare System and Boston University School of Medicine, West Roxbury, Massachusetts

BACKGROUND

- The US CDC has identified extended spectrum beta-lactamase (ESBL) Enterobacteriaceae (ENT) and carbapenem resistant ENT (CRE) as significant public health threats[1].
- The World Health Organization has categorized CRE and ESBL-producing ENT as threats needing critical priority for drug development[2,3].
- Indiscriminant antibiotic use is one of the main contributors to increasing reports of drug resistant urine pathogens[4,5,6].
- Scant data are available on the actual incidence and prevalence of ESBL ENT and CRE from clinical urine samples across a large representative sample of US hospital.
- This study sought to quantify the prevalence and incidence of ESBL-producing ENT and CRE in the urine of adult hospitalized patients.

METHODS

- Isolates from all hospitalized adults with a urine culture (first urine isolate of a species per 30-day period) from 377 hospitals in 2018 were evaluated (BD Insights Research Database, Becton, Dickinson & Company).
- ESBL ENT: *E. coli* (EC), *K. pneumoniae/K. oxytoca* (KPO), and *P. mirabilis* (PM) isolates confirmed as ESBL positive by commercial laboratory panels OR with intermediate susceptibility or resistance to ceftriaxone, cefotaxime, ceftazidime, or cefepime.
- Carbapenem-resistant ENT (CRE): *E. coli, K. pneumoniae, K. oxytoca, P. mirabilis, E. aerogenes, E. cloacae, S. marcescens, C. freundi,* and *M. morganii* isolates with intermediate susceptibility or resistance to imipenem (excluded for *P. mirabilis and M. morganii*), meropenem, doripenem, or ertapenem.
- Urine isolates were classified as[7]:
 - Community-onset (CO: < 3 days of a post-inpatient admission and no previous admission within 14 days).
 - Hospital-onset (HO: ≥ 3 days post-inpatient admission or inpatient admission within 14 days of current admission).
 - Facility status:
 - Bed size: < 100, 100-300, and > 300.
 - Teaching status: teaching and non-teaching.
 - Urban or rural.

Statistical Analysis

- Prevalence (percent of non-susceptible (NS) isolates per organism tested) and rates per 1000 admissions were evaluated overall and by onset location (CO, HO). The variations in prevalence and rate by U.S. Department of Health and Human Services (HHS) geographic region were assessed using the generalized linear models (GLM) adjusting hospital characteristics (377 hospitals). The zip code territory area figures are displayed for 330 facilities.

RESULTS

- In 2018, there were 193,476 non-duplicate ENT urine isolates across 4,623,760 admissions; 63.6% were EC, 19.5% were KPO, and 8.7% were PM.
- Overall, 12.0% (95% confidence interval (CI): 11.6-12.4) were ESBL and 0.79% (95% CI: 0.7-0.9) were CRE.
 - The rates per 1000 admissions were 5.3 (95% CI: 5.1-5.5) and 0.39 (95% CI: 0.3-0.4) for ESBL and CRE, respectively **(Table 1)**.
- Among CO, 11.1% (95% CI: 10.8-11.5) were ESBL and 0.67% (95% CI: 0.6-0.8) were CRE.
 - The rates per 1000 admissions were 4.1 (95% CI: 3.9-4.2) and 0.26 (95% CI: 0.2-0.3) for ESBL and CRE ENT, respectively.
- Among HO, 15.6% (95% CI: 14.8-16.4) were ESBL and 1.4% (95% CI: 1.2-1.6) were CRE.
 - The rates per 1000 admissions were 1.3 (95% CI: 1.2-1.4) and 0.1 (95% CI: 0.1-0.2) for ESBL and CRE ENT, respectively.
- For both CO and HO, there were significant differences in the prevalence of ESBLs and rates across geographic regionals.
 - Northwest, Northeast, and Midwest regions had lower prevalence (% ESBL) relative to other areas.
- There were significant differences in CRE (prevalence (% NS) and rates per 1000 admission) across geographic regionals.
 - HO setting (P=0.118 for CRE ENT percent; P=0.057 for CRE ENT rate per 1000 admissions).

Table 1. Distribution of Urine ESBL and CRE Isolates by Geographic Location and Hospital Characteristics.

Characteristic	Hospital Demographics: Facilities	Hospital Demographics: Admissions	CRE: Mean %NS (95%CI), (n/N): CO	CRE: HO	CRE: Overall (CO + HO)	ESBL: Mean %NS (95%CI), (n/N): CO	ESBL: HO	ESBL: Overall (CO + HO)	CRE Rate/1000 Adms (95% CI): CO	CRE Rate: HO	CRE Rate: Overall (CO + HO)	ESBL Rate/1000 Adms (95% CI): CO	ESBL Rate: HO	ESBL Rate: Overall (CO + HO)
Overall	377	4,623,760	.67 (.6-.8) (1,092/151,077)	1.40 (1.2-1.6) (621/42,399)	.79 (.7-.9) (1,713/193,476)	11.12 (10.8-11.5) (16,542/140,503)	15.61 (14.8-16.4) (5,839/37,143)	11.97 (11.6-12.4) (22,381/177,646)	.26 (.2-.3)	.13 (.1-.2)	.39 (.3-.4)	4.05 (3.9-4.2)	1.31 (1.2-1.4)	5.31 (5.1-5.5)
HHS region														
Region 2 (East coast)	43 (11.4)	650,651	.96 (.7-1.2) (176/20,010)	1.65 (1.2-2.1) (118/6,229)	1.14 (.9-1.4) (294/26,239)	11.90 (10.9-12.9) (2,313/18,682)	17.04 (15.4-18.7) (981/5,513)	13.35 (12.3-14.4) (3,294/24,195)	.29 (.2-.4)	.16 (.1-.2)	.46 (.4-.6)	3.56 (3.2-3.9)	1.61 (1.4-1.9)	5.17 (4.7-5.6)
Region 3 (East)	18 (4.8)	172,815	.72 (.4-1.1) (60/6,365)	1.00 (.5-1.5) (27/1,902)	.80 (.4-1.2) (87/8,267)	"8.39 (7.0-9.8) (503/5,896)	12.00 (8.9-15.1) (204/1,643)	9.06 (7.6-10.5) (707/7,539)	.34 (.1-.5)	.11 (.1-.2)	.45 (.2-.7)	3.77 (3.1-4.4)	1.33 (1.0-1.7)	5.10 (4.2-6.0)
Region 4 (Southeast)	85 (22.5)	1,072,786	.79 (.6-1.0) (290/36,584)	1.52 (1.1-2.0) (177/10,664)	.88 (.7-1.1) (467/47,248)	11.45 (10.8-12.1) (3,986/33,938)	16.88 (15.2-18.6) (1,410/9,263)	12.48 (11.7-13.3) (5,396/43,201)	.33 (.2-.4)	.14 (.1-.2)	.47 (.4-.6)	4.83 (4.4-5.2)	1.43 (1.3-1.6)	6.19 (5.7-6.7)
Region 5 (Central north)	84 (22.3)	1,053,307	.79 (.6-1.0) (301/32,389)	1.65 (1.1-2.2) (135/9,031)	.91 (.7-1.1) (436/41,420)	9.57 (8.8-10.3) (2,990/30,010)	13.07 (11.3-14.8) (943/7,829)	10.15 (9.4-10.9) (3,933/37,839)	.28 (.2-.3)	.16 (.1-.2)	.43 (.3-.5)	3.35 (3.0-3.7)	.95 (.8-1.1)	4.23 (3.9-4.6)
Region 6 (Central south)	71 (18.8)	834,923	.53 (.4-.7) (135/27,402)	1.19 (.8-1.6) (91/7,428)	.63 (.5-.8) (226/34,830)	12.51 (11.6-13.5) (3,314/25,497)	17.49 (15.8-19.2) (1,234/6,588)	13.35 (12.4-14.3) (4,548/32,085)	.25 (.1-.4)	.11 (.1-.2)	.36 (.2-.5)	4.42 (4.0-4.8)	1.50 (1.3-1.7)	5.84 (5.3-6.4)
Region 9 (Southwest)	30 (8.0)	402,240	.64 (.5-.8) (105/15,305)	1.39 (.8-2.0) (48/3,499)	.76 (.6-1.0) (153/18,804)	15.43 (14.3-16.6) (2,405/14,416)	21.78 (18.8-24.8) (720/3,099)	16.49 (15.3-17.7) (3,125/17,515)	.23 (.2-.3)	.13 (.1-.2)	.36 (.3-.5)	6.28 (5.6-7.0)	1.82 (1.5-2.1)	8.07 (7.2-9.0)
Region 10 (Northwest)	14 (3.7)	165,753	.17 (.0-.3) (12/5,319)	1.18 (-.1-2.5) (12/1,436)	.30 (.1-.5) (24/6,755)	8.48 (6.8-10.2) (409/4,932)	13.27 (9.8-16.8) (147/1,284)	9.36 (7.8-10.9) (556/6,216)	.06 (.0-.1)	.06 (.0-.1)	.12 (.0-.2)	2.61 (2.2-3.1)	.91 (.7-1.2)	3.51 (3.0-4.0)
Region (1,7,8: New England & Midwest)	32 (8.5)	271,285	.27 (.1-.5) (13/7,703)	.92 (.0-1.8) (13/2,210)	.36 (.2-.6) (26/9,913)	8.44 (7.1-9.8) (622/7,132)	8.99 (6.6-11.4) (200/1,924)	8.47 (7.4-9.5) (822/9,056)	.07 (.0-.1)	.05 (.0-.1)	.12 (.1-.2)	2.23 (1.9-2.5)	.67 (.5-.9)	2.88 (2.5-3.3)
Bed size														
0-100	101 (26.8)	331,305	.46 (.3-.7) (59/12,328)	1.19 (.6-1.8) (24/2,325)	.53 (.4-.7) (83/14,653)	9.97 (9.1-10.9) (1,243/11,511)	15.18 (12.9-17.5) (318/2,058)	10.89 (10.0-11.8) (1,561/13,569)	.25 (.1-.4)	.11 (.1-.2)	.35 (.2-.5)	4.18 (3.8-4.6)	1.32 (1.1-1.6)	5.31 (4.8-5.8)
100-300	160 (42.4)	1,551,966	.67 (.6-.8) (395/58,854)	1.35 (1.0-1.7) (151/13,589)	.77 (.7-.9) (546/72,443)	11.31 (10.8-11.8) (6,445/54,756)	15.95 (14.9-17.0) (1,938/12,025)	12.12 (11.6-12.7) (8,383/66,781)	.27 (.2-.3)	.12 (.1-.1)	.38 (.3-.4)	4.37 (4.1-4.6)	1.28 (1.2-1.4)	5.65 (5.3-6.0)
300 over	116 (30.8)	2,740,489	.86 (.7-1.0) (638/79,895)	1.65 (1.4-1.9) (446/26,485)	1.04 (.9-1.2) (1,084/106,380)	11.89 (11.4-12.4) (8,854/74,236)	15.49 (14.6-16.4) (3,583/23,060)	12.71 (12.2-13.3) (12,437/97,296)	.26 (.2-.3)	.16 (.1-.2)	.43 (.4-.5)	3.51 (3.3-3.7)	1.33 (1.2-1.4)	4.84 (4.6-5.1)
Urban/Rural														
Rural	84 (22.3)	344,061	.53 (.3-.7) (94/14,690)	1.46 (.8-2.1) (38/3,535)	.64 (.5-.8) (132/18,225)	10.04 (9.2-10.9) (1,484/13,614)	17.59 (15.3-19.9) (486/3,149)	11.09 (10.3-11.9) (1,970/16,763)	.28 (.2-.4)	.15 (.1-.2)	.42 (.3-.6)	4.66 (4.2-5.1)	1.64 (1.4-1.9)	6.18 (5.6-6.8)
Urban	293 (77.7)	4,279,699	.72 (.6-.8) (998/136,387)	1.39 (1.2-1.6) (583/38,864)	.83 (.8-.9) (1,581/175,251)	11.44 (11.1-11.8) (15,058/126,889)	15.07 (14.3-15.8) (5,353/33,994)	12.22 (11.8-12.7) (20,411/160,883)	.26 (.2-.3)	.12 (.1-.1)	.37 (.3-.4)	3.88 (3.7-4.1)	1.21 (1.1-1.3)	"5.05 (4.8-5.3)
Teaching status														
Non-teaching	266 (70.6)	2,462,410	.60 (.5-.7) (622/88,360)	1.30 (1.0-1.6) (299/21,969)	.70 (.6-.8) (921/110,329)	11.21 (10.8-11.7) (10,164/82,355)	16.23 (15.2-17.2) (3,278/19,411)	12.09 (11.6-12.6) (13,442/101,766)	.26 (.2-.3)	.12 (.1-.2)	.38 (.3-.4)	4.39 (4.2-4.6)	1.38 (1.3-1.5)	5.69 (5.4-6.0)
Teaching	111 (29.4)	2,161,350	.87 (.7-1.0) (470/62,717)	1.65 (1.3-2.0) (322/20,430)	1.01 (.9-1.2) (792/83,147)	10.91 (10.3-11.5) (6,378/58,148)	14.15 (13.2-15.1) (2,561/17,732)	11.65 (11.1-12.2) (8,939/75,880)	.26 (.2-.3)	.14 (.1-.2)	.41 (.4-.5)	3.23 (3.0-3.5)	1.14 (1.1-1.2)	4.37 (4.1-4.6)

RESULTS (continued)

Figure 1: Geographic incidence rate (per 1000 admissions) of non-duplicate CRE urine isolates in hospitalized patients (data from 330 acute care facilities in 2018).

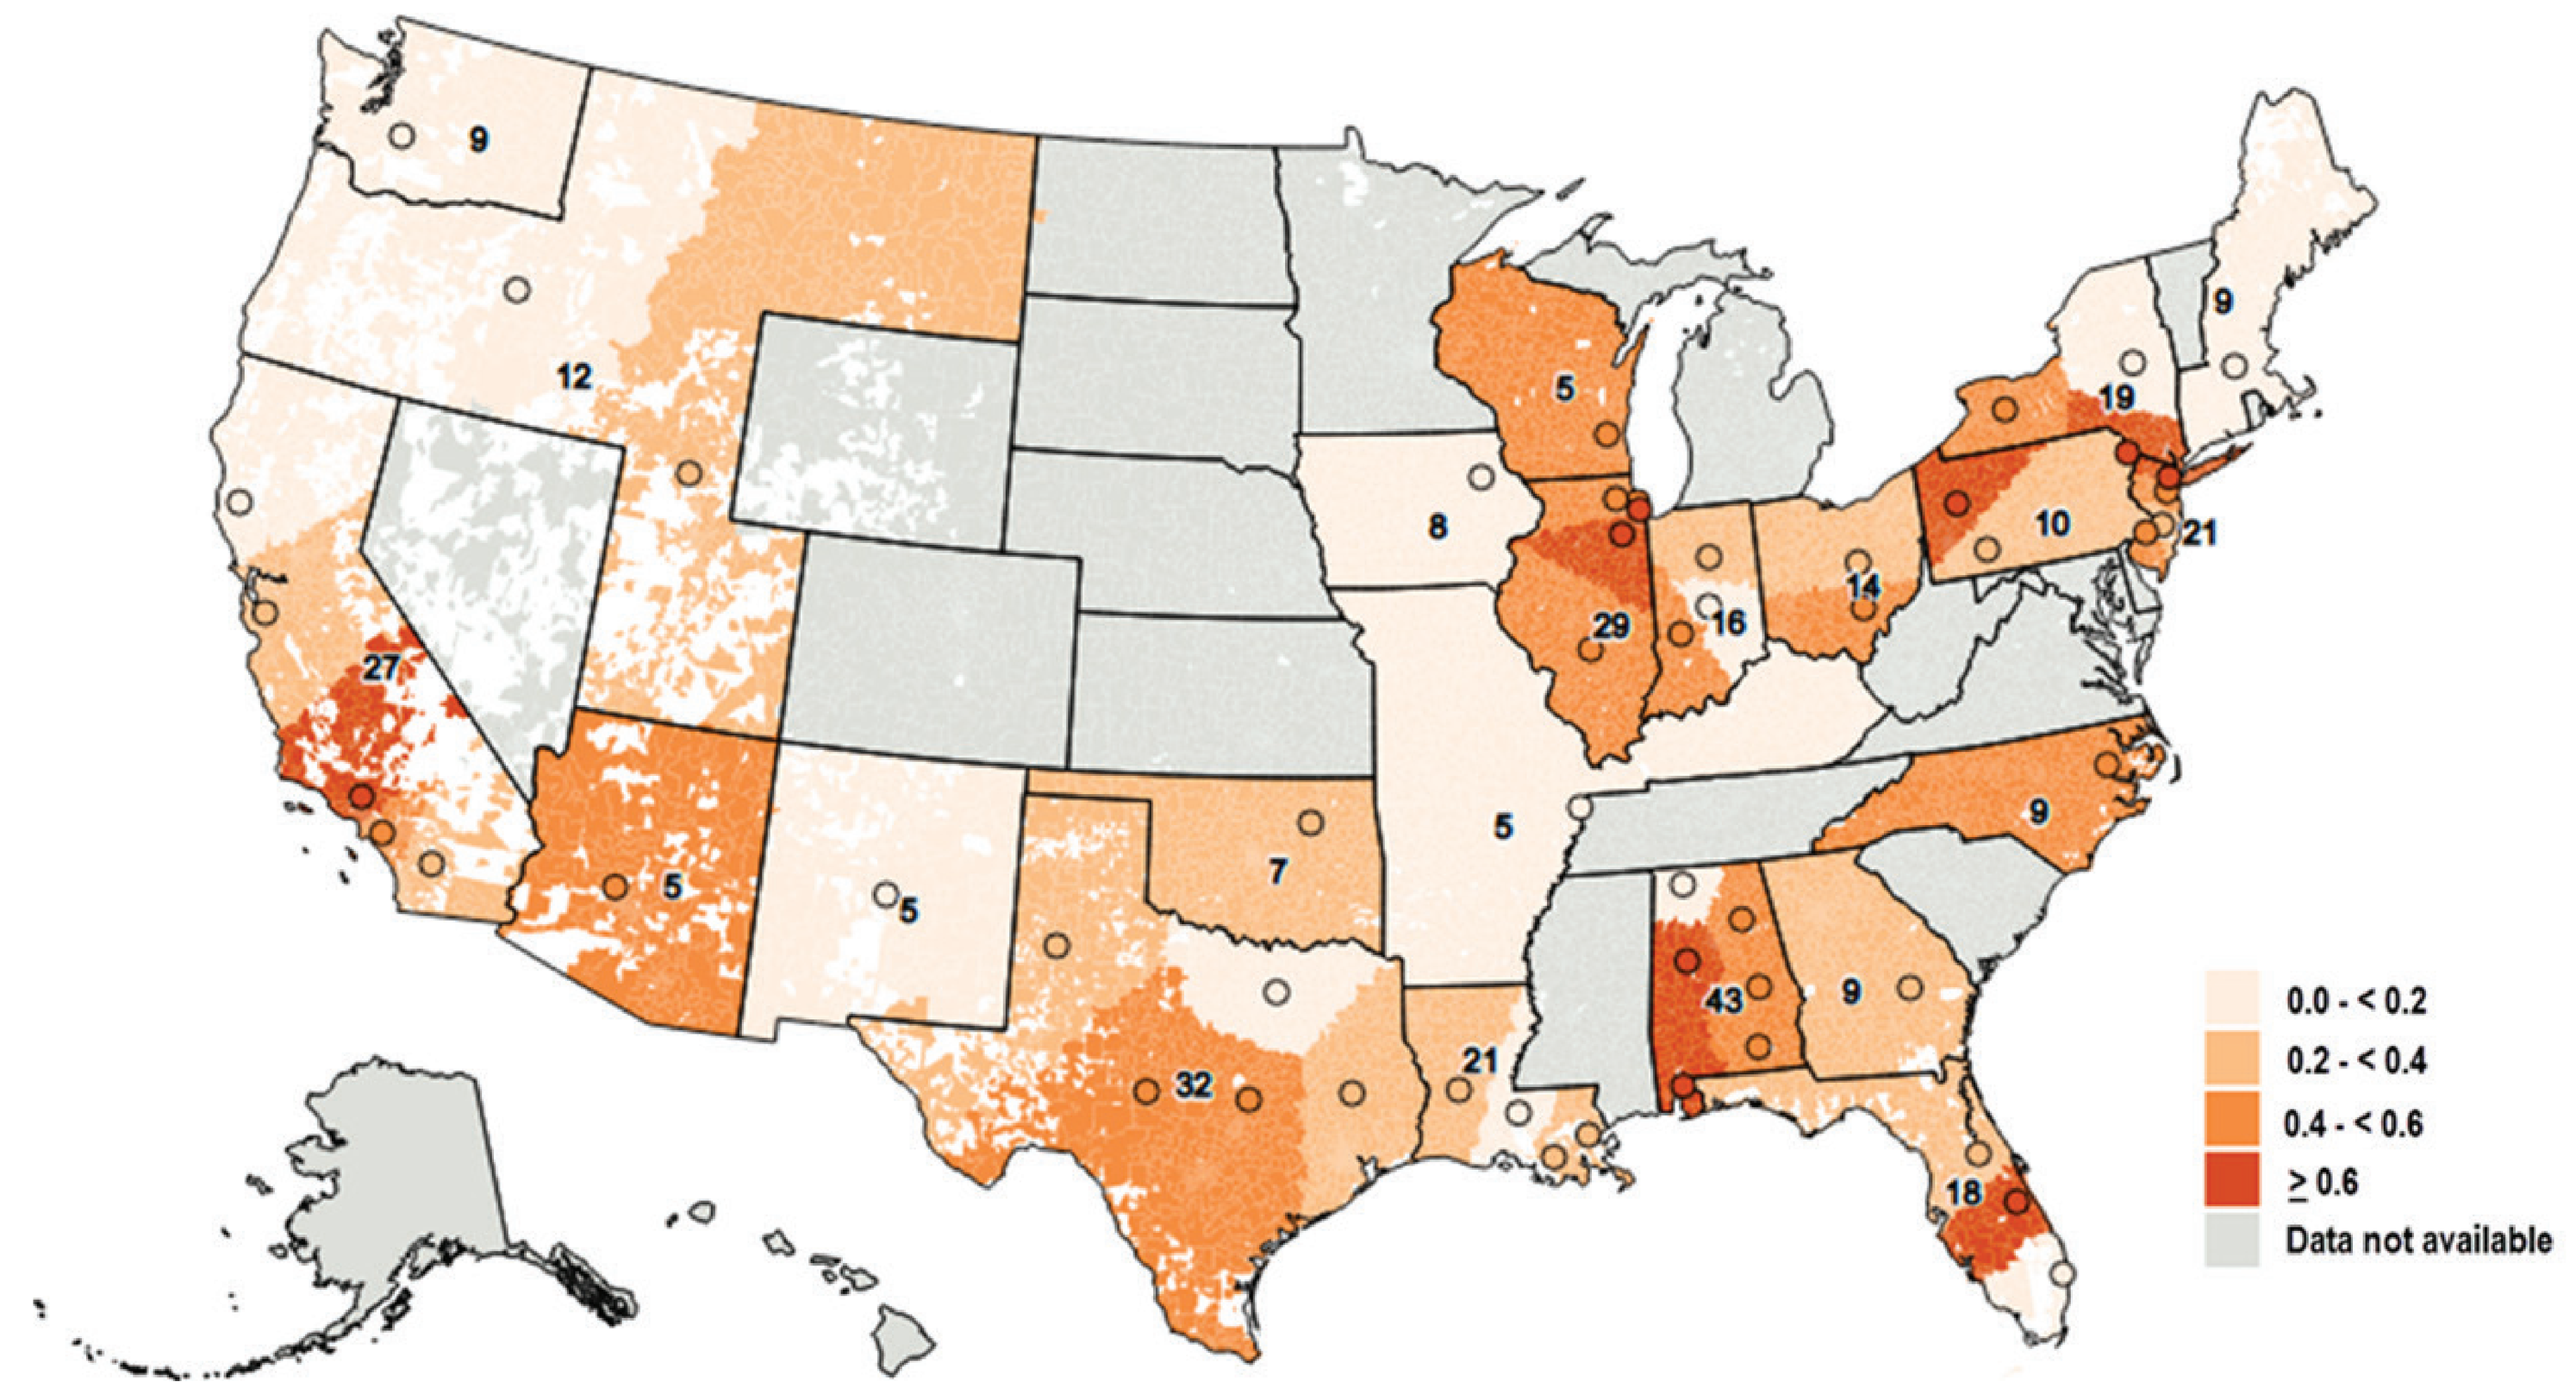

0.0 - < 0.2
0.2 - < 0.4
0.4 - < 0.6
≥ 0.6
Data not available

- Data has been aggregated into geographic clusters of five or more hospitals from two or more IDNs. Each cluster's geographic centroid is represented with shaded circles.
- Each zip code tabulation area (ZCTA) has been attributed a rate based on that ZCTA's proximity to the nearest cluster's geographic centroid, which are represented with shaded circles.
- Within each state, the number of hospitals in each cluster is distributed equally, and the total number of hospitals at the state level is labeled on the map.
- Data for contiguous states that each contain less than five hospitals has been aggregated (MT,ID,UT,OR – KY,MO,AR - ME,NH,MA,CT).

Figure 2: Geographic incidence rate (per 1000 admissions) of non-duplicate ESBL ENT urine isolates in hospitalized patients (data from 330 acute care facilities in 2018).

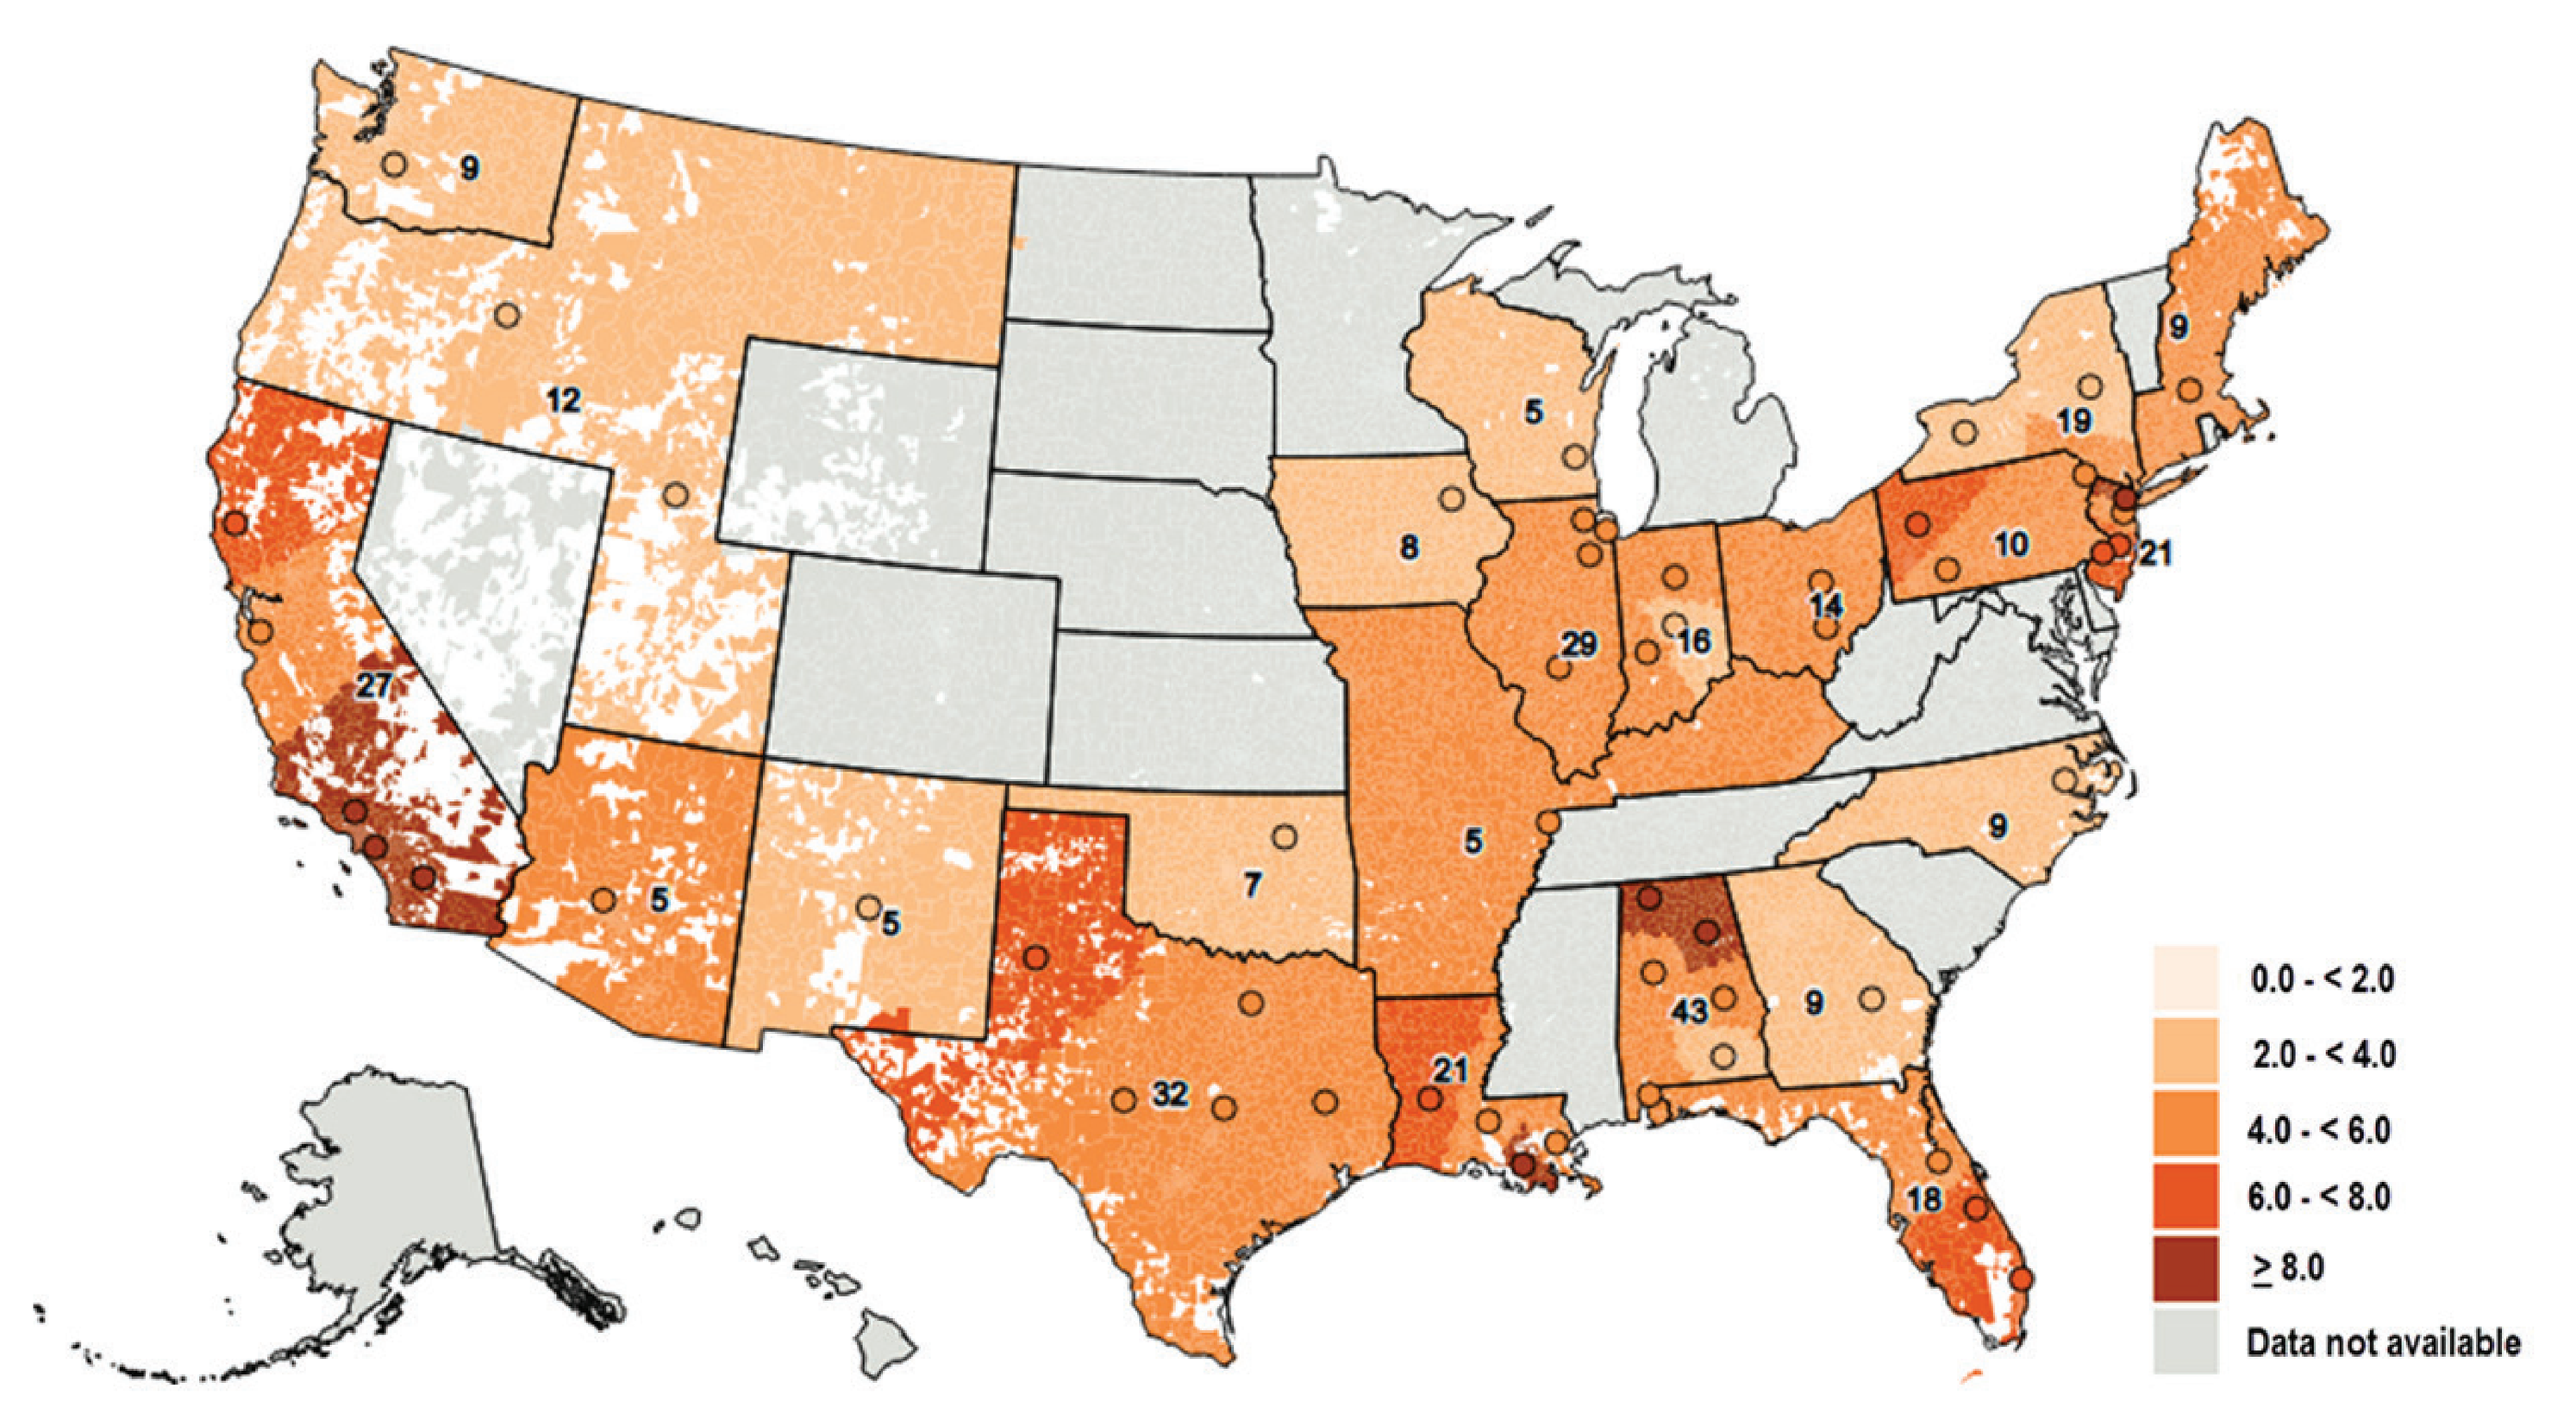

0.0 - < 2.0
2.0 - < 4.0
4.0 - < 6.0
6.0 - < 8.0
≥ 8.0
Data not available

- Data has been aggregated into geographic clusters of five or more hospitals from two or more IDNs. Each cluster's geographic centroid is represented with shaded circles.
- Each zip code tabulation area (ZCTA) has been attributed a rate based on that ZCTA's proximity to the nearest cluster's geographic centroid, which are represented with shaded circles.
- Within each state, the number of hospitals in each cluster is distributed equally, and the total number of hospitals at the state level is labeled on the map.
- Data for contiguous states that each contain less than five hospitals has been aggregated (MT,ID,UT,OR – KY,MO,AR - ME,NH,MA,CT).

Limitations

- These data were collected from the laboratory information system feeds provided by participating hospitals and relied on interpretive results reported at each facility.
- These data were collected and analyzed from the perspective of unique non-duplicate collected cultures and not from the perspective of unique patients.
- It is not clear if all included urine cultures were indicative of a urinary tract infection. The goal was to understand the volume and frequency of these organisms that were seen at the level of the hospital across a large number of geographical diverse institutions.

CONCLUSIONS

- This large national database study demonstrates that more than 1 out of every 10 urine cultures with growth of ENT that are obtained from patients admitted from the community will have an ESBL pathogen; the proportion is even higher among hospital onset urine cultures.
- This rate doubles (>20%) when combining certain regions with site of onset (Southwest and hospital onset).
- CRE ENT remains low, but also varies greatly by geographic region, with >1% along the east coast, and >1% among HO isolates regardless of geography.
- Assessing risk factors for resistance such as geographic region and site of onset is a key critical first step to optimizing empiric therapy decisions.

REFERENCES

(1) https://www.cdc.gov/drugresistance/pdf/ar-threats-2013-508.pdf
(2) World Health Organization. Global priority list of antibiotic-resistant bacteria to guide research, discover, and development of new antibiotics. 2017. http://apps.who.int/medicinedocs/documents/s23171en/s23171en.pdf. Accessed August 6, 2018.
(3) Tacconelli E, et al. Discovery, research, and development of new antibiotics: the WHO priority list of antibiotic-resistant bacteria and tuberculosis. Lancet Infect Dis. 2018; 18: 318-27.
(4) Johnson L, et al. Emergence of Fluoroquinolone Resistance in Outpatient Urinary Escherichia coli Isolates. Am J Med. 2008;121:876-884.
(5) Foxman B. The epidemiology of urinary tract infection. Nature Reviews Urology. 2010;7:653–660.
(6) Lutter SA, et al. Antibiotic Resistance Patterns in Children Hospitalized for Urinary Tract Infections. Arch Pediatr Adolesc Med. 2005;159(10):924–928.
(7) McCann E, et al. Carbapenem-Nonsusceptible Gram-Negative Pathogens in ICU and Non-ICU Settings in US Hospitals in 2017: A Multicenter Study. Open Forum Infectious Diseases, Volume 5, Issue 10, 1 October 2018, ofy241.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_Regional_Variation.

ID Week; Friday October 4, 2019; Washington, DC, USA