705

Pharmacokinetics and Safety of Lefamulin After Single Intravenous Dose Administration in Subjects With Impaired Renal Function and in Those Requiring Hemodialysis

Wolfgang W. Wicha,[1] Thomas C. Marbury,[2] James A. Dowell,[3] Lori Lykens,[4] Cathie Leister,[3] James Ermer,[3] Steven P. Gelone[4]

[1]Nabriva Therapeutics GmbH, Vienna, Austria; [2]Orlando Clinical Research Center, Orlando, FL, USA;
[3]Pharmacology Development Services, LLC, Collegeville, PA, USA; [4]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA

Presented by
David Mariano, PharmD
Email: David.Mariano@Nabriva.com
Phone: 610-816-6659
Fax: 610-816-6639

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION & PURPOSE

- Lefamulin (LEF), the first pleuromutilin antibiotic for intravenous (IV) and oral treatment, was recently approved for use in adults with community-acquired bacterial pneumonia[1]
 - Approval was based on the results of the Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 phase 3 clinical studies, which demonstrated that LEF was generally well tolerated and noninferior to moxifloxacin[2,3]
- Renal comorbidities are common in patients hospitalized with pneumonia,[4] and individuals with chronic kidney disease are at increased risk of pneumonia, particularly pneumonia requiring hospitalization[5]
 - Pneumonia also is a risk factor for in-hospital mortality among incident hemodialysis patients[6]
- We investigated the pharmacokinetics (PK) and safety of LEF and its main metabolite, BC-8041, in subjects with severe renal impairment and those requiring hemodialysis
 - An abbreviated study design was used because, based on currently available data, LEF and BC-8041 do not undergo substantial renal elimination[7]

METHODS

Subjects

- Subjects were enrolled in 1 of 3 groups based on level of renal function
 - Normal: healthy controls with normal renal function (creatinine clearance ≥90 mL/min)
 - Severe: subjects with severe renal impairment (estimated glomerular filtration rate <30 mL/min/1.73 m^2) and not on hemodialysis
 - Hemodialysis: subjects with end-stage renal disease who required hemodialysis

Study Design

- Open-label, multicenter study
- Normal and Severe subjects were matched based on sex, age (±10 years), and weight (±10 kg)
- Subjects in the Normal and Severe groups received a single 1-hour IV infusion of LEF 150 mg
- Subjects in the Hemodialysis group received a single 1-hour IV infusion of LEF 150 mg in each of the 2 treatment periods, with dosing separated by ≥7 days
 - On-dialysis period: subjects started hemodialysis within 1 hour after LEF infusion
 - Off-dialysis period: subjects received LEF infusion on a non-dialysis day

Assessments

- PK analysis
 - Blood and urine samples were collected predose and over a 36-hour period postdose
 - For subjects in the Hemodialysis group, a sample of fresh (unused) dialysate was collected before the start of hemodialysis; once hemodialysis began, dialysate samples were collected at 1-hour intervals until hemodialysis completion
 - A validated liquid chromatography–tandem mass spectrometry (LC-MS/MS) method was used to quantitate concentrations of LEF and BC-8041 in plasma, urine, and dialysate (A&M Labor für Analytik und Metabolismusforschung Service GmbH, Bergheim, Germany)
 - The lower limits of quantitation for both analytes were 1.0 ng/mL for plasma and 10.0 ng/mL for urine and dialysate
- Safety assessments included treatment-emergent adverse events (TEAEs), laboratory parameters, vital signs, and electrocardiograms (ECGs)

Statistical Analysis

- PK parameters were calculated from individual concentration-time profiles using noncompartmental analysis methods in Phoenix® WinNonlin® version 6.3 Pharsight (Certara USA, Inc., Princeton, NJ, USA)
- Statistical comparisons were performed using least square geometric mean ratios (LS GMRs) (Severe/Normal, On/Off dialysis) and their 90% confidence intervals (CIs) for maximum observed plasma concentration (C_{max}), area under the plasma concentration-time curve extrapolated through infinity (AUC), systemic clearance (CL), volume of distribution based on the terminal phase (V_Z), and renal clearance (CL_R) for LEF, and C_{max} and AUC for BC-8041

RESULTS

Study Subjects

- 23 subjects enrolled in and completed the study (Normal, *n*=7; Severe, *n*=8; Hemodialysis, *n*=8); all subjects received the intended LEF dose
- While the Normal and Severe groups were well matched based on sex, age, and weight, all subjects in the Hemodialysis group were males and were younger and heavier than subjects enrolled in the other 2 groups (**Table 1**)
- In the on-dialysis period, hemodialysis was initiated within 1.67–2 hours after infusion start
 - The total hemodialysis duration was 3–4 hours across all subjects
 - Blood flow rates were 350–550 mL/min, and urea clearance was 274–400 mL/min

Table 1. Demographics and Baseline Characteristics

Parameter	Normal *n*=7	Severe *n*=8	Hemodialysis *n*=8
Age, y, mean (SD)	59 (9)	64 (9)	49 (3)
Male, *n* (%)	4 (57.1)	4 (50.0)	8 (100.0)
Race, *n* (%)			
White	5 (71.4)	4 (50.0)	2 (25.0)
Black or African American	2 (28.6)	4 (50.0)	6 (75.0)
Ethnicity, *n* (%)			
Hispanic or Latino	1 (14.3)	0	1 (12.5)
Not Hispanic or Latino	6 (85.7)	8 (100.0)	7 (87.5)
Height, cm, mean (SD)	168.4 (7.4)	165.6 (13.9)	176.1 (4.5)
Weight, kg, mean (SD)	80.2 (10.5)	80.1 (10.8)	99.8 (14.8)
BMI, kg/m^2, mean (SD)	28.2 (2.7)	29.4 (4.5)	32.2 (5.0)
BSA, m^2, mean (SD)	1.93 (0.16)	1.92 (0.18)	2.20 (0.17)
$CL_{CR,24h}$, mL/min, mean (SD)	118.9 (15.0)	28.6 (15.7)	ND
$CL_{CR,C\text{-}G}$, mL/min, mean (SD)	117.8 (23.2)	25.7 (9.6)	15.9 (8.3)
$eGFR_{MDRD}$, mL/min/1.73 m^2, mean (SD)	108.1 (29.0)	19.6 (6.6)	8.7 (5.5)

BMI=body mass index; BSA=body surface area; $CL_{CR,24h}$=creatinine clearance determined by 24-hour urine collection; $CL_{CR,C\text{-}G}$=creatinine clearance determined by Cockcroft-Gault and S_{CR}; $eGFR_{MDRD}$=estimated glomerular filtration rate determined by Modification of Diet in Renal Disease equation and S_{CR}; ND=not determined by site; S_{CR}=serum creatinine.

Pharmacokinetics

Normal Renal Function vs Severe Renal Impairment

- In both Normal and Severe subjects, mean LEF plasma concentrations peaked immediately following the 60-minute infusion, and mean BC-8041 plasma concentrations peaked within 30 minutes of the end of infusion (**Figure 1**)
 - Plasma concentrations of LEF and BC-8041 remained above lower limits of quantitation throughout the entire 36-hour sampling period for all subjects
- LEF and BC-8041 PK parameters were comparable between the Normal and Severe groups, and the majority of each LEF and BC-8041 was excreted nonrenally in both groups (**Table 2**)
- For all LEF and BC-8041 PK parameters except LEF CL_R, 90% CIs for the LS GMRs (Severe/Normal) contained 100% (**Table 3**)

On Dialysis vs Off Dialysis

- During both the On- and Off-dialysis periods, mean LEF plasma concentrations peaked immediately following the 60-minute infusion and mean BC-8041 plasma concentrations peaked within 1 hour of the end of infusion (**Figure 1**)
 - Plasma concentrations of LEF and BC-8041 remained above lower limits of quantitation throughout the entire 36-hour sampling period for all subjects
- LEF and BC-8041 PK parameters were comparable between the On- and Off-dialysis periods (**Table 2**)
- LEF and BC-8041 were not measurably filtered into dialysate
- For all LEF and BC-8041 PK parameters, 90% CIs for the LS GMRs (On/Off dialysis) contained 100% (**Table 3**)

Figure 1. Mean (SD) LEF and BC-8041 Plasma Concentrations Over Time by Renal Function Status Group

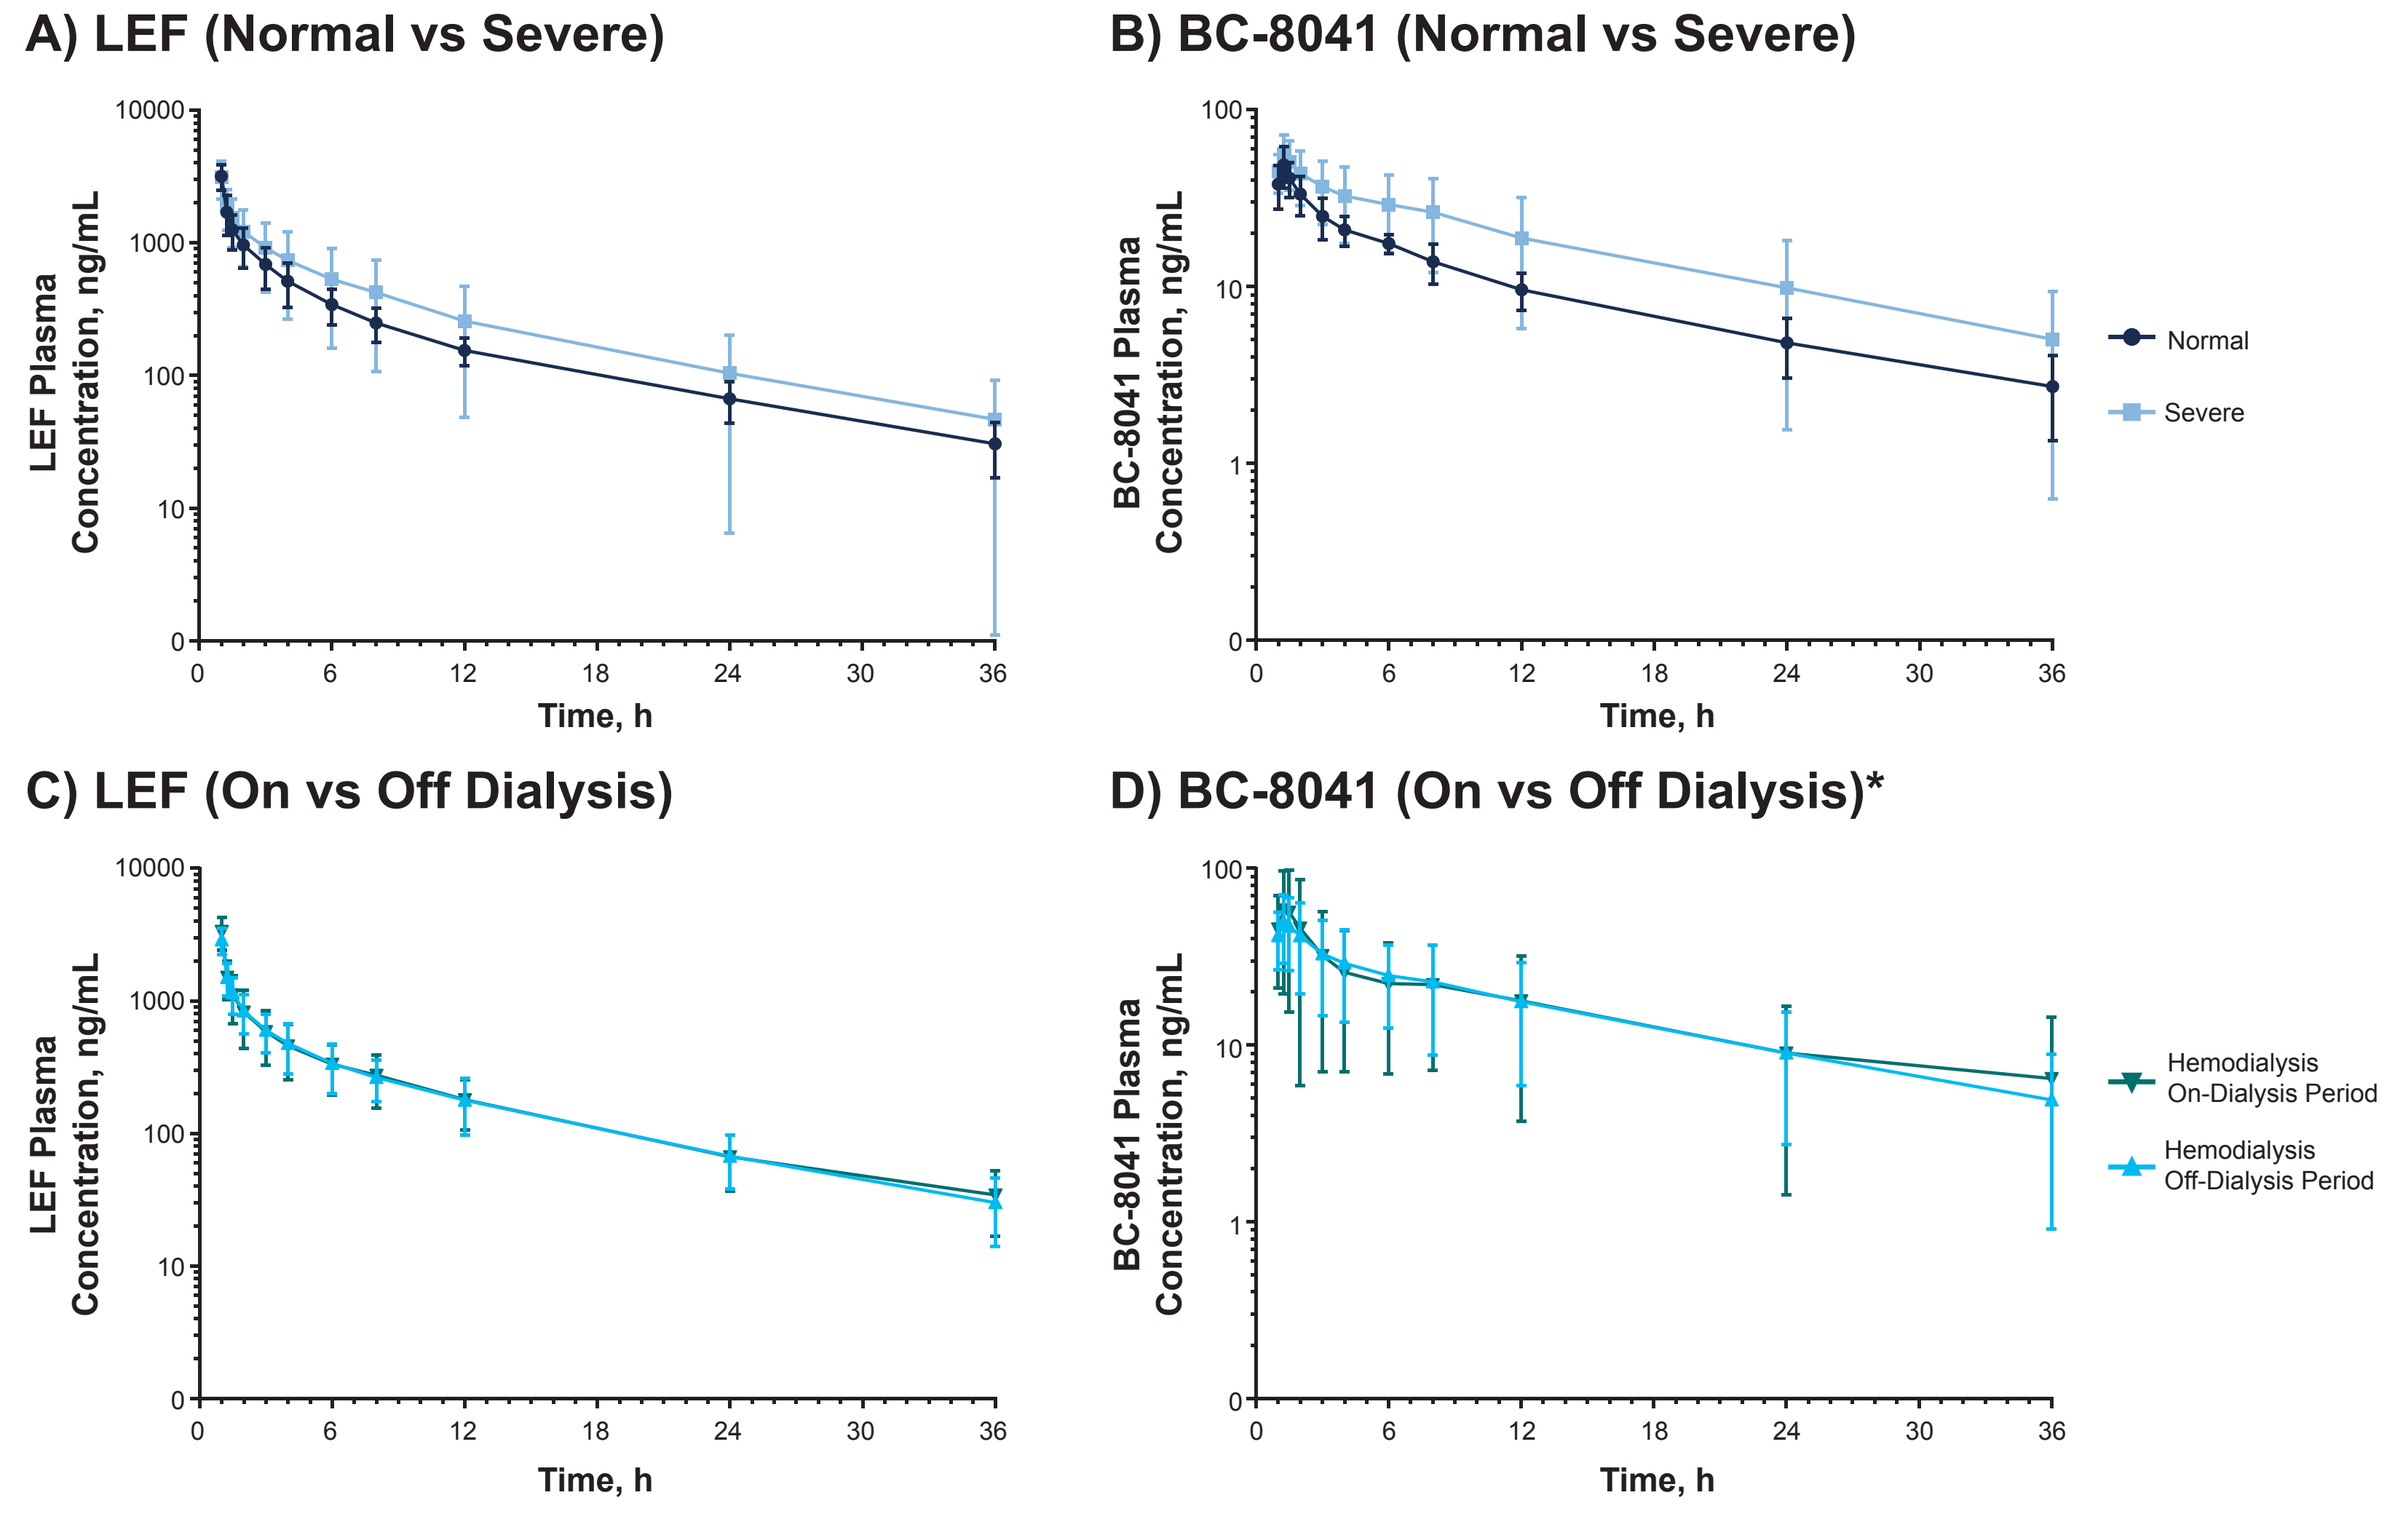
A) LEF (Normal vs Severe)
B) BC-8041 (Normal vs Severe)
C) LEF (On vs Off Dialysis)
D) BC-8041 (On vs Off Dialysis)*
LEF Plasma Concentration, ng/mL
BC-8041 Plasma Concentration, ng/mL
Time, h
Normal
Severe
Hemodialysis On-Dialysis Period
Hemodialysis Off-Dialysis Period

LEF=lefamulin.
*Downward facing error bars do not appear for some data points, since negative values cannot be graphed on a logarithmic scale.

Table 2. LEF and BC-8041 Pharmacokinetics by Renal Function Status Group

Parameter, mean (SD)	Normal *n*=7		Severe *n*=8		Off Dialysis *n*=8		On Dialysis *n*=8	
	LEF	BC-8041	LEF	BC-8041	LEF	BC-8041	LEF	BC-8041
C_{max}, ng/mL	3182 (697)	48.7 (12.8)	3138 (990)	56.1 (15.7)	2893 (653)	51.2 (21.9)	3341 (916)	60.0 (40.0)
T_{max}, h	1.0 (0.0)	1.3 (0.0)	1.1 (0.1)	1.3 (0.1)	1.0 (0.0)	1.4 (0.3)	1.0 (0.0)	1.4 (0.1)
$t_{1/2}$, h	10.1 (1.9)	13.5 (4.5)	9.4 (0.9)	11.4 (2.2)	9.1 (0.9)	12.8 (2.0)	9.3 (1.4)	15.1 (4.4)
$AUC_{0\text{-}12}$, h·ng/mL	6730 (1819)	227 (45.2)	8715 (4567)	351 (155)	6287 (1778)	315 (164)	6526 (2115)	314 (220)
AUC_t, h·ng/mL	8531 (2347)	353 (83.2)	11602 (7102)	602 (350)	8207 (2606)	551 (324)	8480.0 (2904)	560 (435)
AUC, h·ng/mL	9004 (2591)	413 (134)	12262 (7798)	695 (448)	8606 (2815)	643 (408)	8955 (3103)	734 (716)
CL, L/h	17.9 (5.4)	–	15.7 (7.2)	–	19.0 (5.6)	–	18.6 (6.4)	–
V_z, L	254 (52.2)	–	210 (94.2)	–	249 (80.7)	–	248 (88.7)	–
V_{ss}, L	154 (33.9)	–	136 (52.6)	–	164 (38.3)	–	167 (51.8)	–
A_e, mg	11.1 (5.0)	0.4 (0.2)	3.9 (1.6)	0.2 (0.1)	1.9*	0.1*	1.7*	0.1*
A_e, %	7.4 (3.3)	–	2.6 (1.1)	–	1.2*	–	1.1*	–
CL_R, L/h	1.3 (0.6)	1.1 (0.4)	0.4 (0.2)	0.3 (0.1)	0.2*	0.1*	0.1*	0.2*
CL_{NR}, L/h	16.6 (5.1)	–	15.3 (7.1)	–	18.7*	–	20.1*	–

A_e=amount excreted unchanged in urine; AUC=area under plasma concentration-time curve extrapolated through infinity; $AUC_{0\text{-}12}$=AUC from time 0 to 12 hours; AUC_t=AUC from start of infusion through to last measurable (positive) observed concentration; CL=systemic clearance; CL_{NR}=nonrenal clearance; CL_R=renal clearance; C_{max}=maximum observed plasma concentration; LEF=lefamulin; $t_{1/2}$=terminal elimination half-life; T_{max}=time of maximum observed concentration; V_{ss}= volume of distribution at steady-state (observed), estimated using mean residence time; V_Z=volume of distribution based on the terminal phase.
**n*=2 (only 2 subjects in the Hemodialysis group made urine).

Table 3. Statistical Comparisons of Pharmacokinetic Parameters by Renal Function Status Group

A. Comparison of Normal and Severe Groups

	LS Geometric Mean			
	Severe	Normal	Ratio (Severe/Normal)	90% CI
LEF				
C_{max}	2987	3126	95.6	73.4–124.4
AUC	10679	8679	123.0	82.1–184.4
CL	14.1	17.3	81.3	54.3–121.8
V_Z	190	249	76.1	53.0–109.2
CL_R	0.3	1.1	31.1	16.7–57.8
BC-8041				
C_{max}	54.0	47.4	113.9	88.2–147.2
AUC	589	397	148.3	94.5–232.6

B. Comparison of Hemodialysis Groups

	LS Geometric Mean			
	On Dialysis	Off Dialysis	Ratio (On/Off)	90% CI
LEF				
C_{max}	3235	2836	114.1	96.3–135.1
AUC	8489	8229	103.2	96.4–110.4
CL	17.7	18.2	96.9	90.6–103.8
V_Z	234	237	98.6	90.2–107.7
CL_R	–	–	–	–
BC-8041				
C_{max}	50.2	46.6	107.8	91.1–127.6
AUC	569	559	101.8	89.0–116.6

AUC=area under plasma concentration-time curve extrapolated through infinity; CL=systemic clearance; CL_R=renal clearance; C_{max}=maximum observed plasma concentration; LEF=lefamulin; LS=least squares; V_Z=volume of distribution based on the terminal phase.

Safety

- The majority of TEAEs were mild in severity and considered related to study drug (**Table 4**)
 - No severe or serious TEAEs were observed, and no TEAE resulted in study drug discontinuation
- No subjects in any renal function status group exhibited clinically significant changes in serum chemistry, hematology, or vital signs
- An increase in mean QT interval corrected according to Fridericia (QTcF) was observed in all renal function status groups
 - Within 4 hours postdose, the maximum mean increases from baseline were 8.9 and 6.6 msec in the Normal and Severe groups, respectively, 15.9 msec during the On-dialysis period, and 17.6 msec during the Off-dialysis period
 - Larger changes observed in the Hemodialysis group are consistent with previous reports and may be attributed to changes in electrolyte concentrations[8,9]
 - No subject had a postbaseline value of >480 msec or an increase from baseline of >60 msec
 - No cardiac TEAEs or TEAEs related to ECG assessments were observed

Table 4. TEAE Summary*

Category, *n* (%)	Normal *n*=7	Severe *n*=8	Hemodialysis *n*=8
Subjects with ≥1 TEAE	2 (28.6)	4 (50.0)	4 (50.0)
Mild	2 (28.6)	3 (37.5)	4 (50.0)
Moderate	0	1 (12.5)	0
Severe	0	0	0
Subjects with ≥1 drug-related TEAE	2 (28.6)	3 (37.5)	3 (37.5)
Mild	2 (28.6)	2 (25.0)	3 (37.5)
Moderate	0	1 (12.5)	0
Severe	0	0	0
TEAEs occurring in ≥1 subject†			
Application site hemorrhage	0	1 (12.5)	0
Diarrhea	0	1 (12.5)	0
Dizziness	0	0	1 (12.5)
Flatulence	0	0	1 (12.5)
Headache	1 (14.3)	1 (12.5)	0
Infusion site erythema	0	1 (12.5)	0
Infusion site induration	0	1 (12.5)	0
Infusion site pain	0	0	1 (12.5)
Infusion site phlebitis	0	1 (12.5)	0
Nausea	0	0	1 (12.5)
Rash	1 (14.3)	0	1 (12.5)
Vomiting	0	1 (12.5)	0

MedDRA=Medical Dictionary for Regulatory Activities; TEAE=treatment-emergent adverse event.
*Adverse events were coded using MedDRA Version 20.0.
†Subjects with multiple events in each system organ class and preferred term were only counted once.

CONCLUSIONS

- Results of this study demonstrate that LEF dosage adjustment is not necessary when treating subjects with severe renal impairment and that LEF can be administered without regard to hemodialysis timing
- LEF was generally well tolerated in all subjects regardless of renal function status

REFERENCES

(1) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(2) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(3) Alexander E, et al. *JAMA.* 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].
(4) Luna CM, et al. *Ann Am Thorac Soc.* 2016;13(9):1519-1526.
(5) Chou CY, et al. *Medicine (Baltimore).* 2014;93(27):e174.
(6) Bae EH, et al. *Kidney Res Clin Pract.* 2015;34(3):154-159.
(7) Nabriva Therapeutics GmbH. Data on file. Vienna, Austria. 2019.
(8) Covic A, et al. *Nephrol Dial Transplant.* 2002;17(12):2170-2177.
(9) Kim ED, et al. *BMC Nephrol.* 2019;20(1):133.

Acknowledgments

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Wolfgang W. Wicha, Lori Lykens, Steven P. Gelone, and David Mariano are employees of/stockholders in Nabriva Therapeutics plc. James A. Dowell, Cathie Leister, and James Ermer have served as consultants for Nabriva Therapeutics. Thomas C. Marbury is an employee and equity owner of Orlando Clinical Research Center.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_RenalPK

IDWeek 2019: October 2–6, Washington, DC, USA