722

Pharmacokinetics and Safety of Lefamulin After Single Intravenous Dose Administration in Subjects With Impaired Hepatic Function

Nabriva THERAPEUTICS

Wolfgang W. Wicha,[1] Thomas C. Marbury,[2] James A. Dowell,[3] Lori Lykens,[4] Cathie Leister,[3] James Ermer,[3] Steven P. Gelone[4]

[1]Nabriva Therapeutics GmbH, Vienna, Austria; [2]Orlando Clinical Research Center, Orlando, FL, USA;
[3]Pharmacology Development Services, LLC, Collegeville, PA, USA; [4]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA

Presented by
Wolfgang W. Wicha, MSc
Email: Wolfgang.Wicha@Nabriva.com
Phone: +43 1 664 851 5864
Fax: +1 610-816-6639

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION & PURPOSE

- Lefamulin (LEF), the first pleuromutilin antibiotic for intravenous (IV) and oral treatment, was recently approved for use in adults with community-acquired bacterial pneumonia (CABP)[1]
 - Approval was based on the results of the Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 phase 3 clinical studies, which demonstrated that LEF was generally well tolerated and noninferior to moxifloxacin[2,3]
 - Preclinical studies have indicated that the liver is the major route of elimination for LEF[4]
 - LEF is metabolized by CYP450 phase I reactions to hydroxylated metabolites, with the monohydroxy BC-8041 being the primary metabolite[4]
- Pneumonia is among the leading causes of hospitalization and infection-related death in the United States[5-7]; novel therapeutic options are needed to address increasing rates of bacterial resistance and risks associated with current CABP treatments[6]
 - Patients with chronic liver disease (CLD) are at substantially increased risk of pneumonia,[8] and those with both pneumonia and CLD experience higher mortality[8-11] and healthcare costs[8,11] than patients with pneumonia alone
 - Liver disease may affect LEF pharmacokinetics (PK), highlighting the need for PK studies in subjects with impaired hepatic function
- We investigated the PK and safety of LEF and its main metabolite, BC-8041, in subjects with hepatic impairment

METHODS

Subjects

- Subjects were enrolled in 1 of 3 groups based on level of hepatic function
 - Normal: healthy controls with no liver cirrhosis and normal hepatic function
 - Moderate: subjects with liver cirrhosis and moderate hepatic impairment as classified by their Child-Pugh score (Class B, 7–9 points)
 - Severe: subjects with liver cirrhosis and severe hepatic impairment as classified by their Child-Pugh score (Class C, ≥10 points)

Study Design

- Open-label, multicenter study
- Moderate and Severe subjects were matched to Normal subjects based on sex, age (±10 years), and weight (±10 kg)
- All subjects received a single 1-hour IV infusion of LEF 150 mg

Assessments

- PK analysis
 - Blood and urine samples were collected predose and over a 48-hour period postdose
 - A validated liquid chromatography–tandem mass spectrometry (LC-MS/MS) method was used to quantitate concentrations of LEF and BC-8041 in plasma and urine (A&M Labor für Analytik und Metabolismusforschung Service GmbH, Bergheim, Germany)
 - The lower limits of quantitation were 1.0 ng/mL for plasma and 10.0 ng/mL for urine
 - Plasma protein binding (PPB) was determined using a fit-for-purpose validated LC-MS/MS method
- Safety assessments included treatment-emergent adverse events (TEAEs), laboratory parameters, vital signs, and electrocardiograms

Statistical Analysis

- PK parameters were calculated from individual concentration-time profiles using noncompartmental analysis methods in Phoenix® WinNonlin® version 6.3, Pharsight (Certara USA, Inc., Princeton, NJ, USA)
- Statistical comparisons were performed using least square geometric mean ratios (LS GMRs) (Moderate/Normal, Severe/Normal) and their 90% confidence intervals (CIs) for maximum observed plasma concentration (C_{max}), area under the plasma concentration-time curve extrapolated through infinity (AUC), systemic clearance (CL), volume of distribution based on the terminal phase (V_Z), and renal clearance (CL_R) for LEF, and C_{max} and AUC for BC-8041

RESULTS

Study Subjects

- 27 subjects enrolled in and completed the study (Normal, *n*=11; Moderate, *n*=8; Severe, *n*=8); all subjects received the intended LEF dose
- All groups were well matched based on sex, age, and weight **(Table 1)**
 - Most subjects with hepatic impairment (15/16 [94%]) presented at screening with ascites described as “slight” for most subjects in the Moderate group and as “moderate” for most subjects in the Severe group
 - Child-Pugh scores ranged from 7–9 points in the Moderate group and from 10–12 points in the Severe group
 - Subjects in the Moderate or Severe groups had mean laboratory values at baseline that reflected CLD

Table 1. Demographics and Baseline Characteristics

Parameter	Normal *n*=11	Moderate *n*=8	Severe *n*=8
Age, y, mean (SD)	57 (7)	59 (4)	59 (6)
Male, *n* (%)	9 (81.8)	5 (62.5)	7 (87.5)
Race, *n* (%)			
White	9 (81.8)	5 (62.5)	8 (100.0)
Black or African American	2 (18.2)	1 (12.5)	0
Other	0	2 (25.0)	0
Ethnicity, *n* (%)			
Hispanic or Latino	1 (9.1)	2 (25.0)	3 (37.5)
Not Hispanic or Latino	10 (90.9)	6 (75.0)	5 (62.5)
Height, cm, mean (SD)	176.6 (8.0)	173.7 (9.0)	173.2 (9.7)
Weight, kg, mean (SD)	90.4 (17.4)	91.7 (16.8)	86.9 (16.9)
BMI, kg/m², mean (SD)	28.8 (3.7)	30.2 (4.1)	29.0 (5.1)
BSA, m², mean (SD)	2.10 (0.24)	2.10 (0.24)	2.04 (0.22)
Ascites, *n* (%)			
Absent	–	1 (12.5)	0
Slight	–	5 (62.5)	1 (12.5)
Moderate	–	2 (25.0)	7 (87.5)
Child-Pugh score, *n* (%)			
7	–	3 (37.5)	0
8	–	3 (37.5)	0
9	–	2 (25.0)	0
10	–	0	4 (50.0)
11	–	0	3 (37.5)
12	–	0	1 (12.5)
Serum albumin, g/dL, mean (SD)	4.3 (0.5)	3.8 (0.6)	3.3 (0.3)
Prothrombin time, s, mean (SD)	11.3 (0.7)	13.9 (1.4)	15.5 (1.2)
Total bilirubin, mg/dL, mean (SD)	0.6 (0.1)	1.5 (0.5)	2.7 (1.0)

BMI=body mass index; BSA=body surface area.

Pharmacokinetics

- LEF and BC-8041 plasma concentrations were comparable across hepatic function status groups through the first 12 hours after the start of infusion, followed by slightly slower rates of elimination for subjects with hepatic impairment in the later phases of elimination **(Figure 1)**
 - Mean LEF plasma concentrations peaked within 30 minutes following the 60-minute infusion, and mean BC-8041 plasma concentrations peaked within 60 minutes of the end of infusion
 - LEF plasma concentrations remained above lower limits of quantitation throughout the entire 48-hour sampling period for all subjects
- Overall, LEF and BC-8041 exposures were similar across hepatic function status groups **(Table 2)**
 - LEF C_{max} decreased slightly with hepatic impairment and appeared to be related to the presence and degree of ascites
 - LEF AUC increased slightly (<20%) in subjects with severe hepatic impairment vs normal hepatic function
- The majority of LEF and BC-8041 was excreted nonrenally in all subjects **(Table 2)**
- For LEF and BC-8041 PK parameters **(Table 3)**, 90% CIs for the LS GMRs contained 100% except for LEF C_{max} (Moderate vs Normal, Severe vs Normal), V_z (Severe vs Normal), and CL_R (Moderate vs Normal, Severe vs Normal) and BC-8041 C_{max} (Severe vs Normal) and AUC (Severe vs Normal)
- A decrease in LEF PPB was observed in subjects with impaired vs normal hepatic function **(Table 4)**

Figure 1. Mean (SD) LEF and BC-8041 Plasma Concentrations Over Time by Hepatic Function Status Group

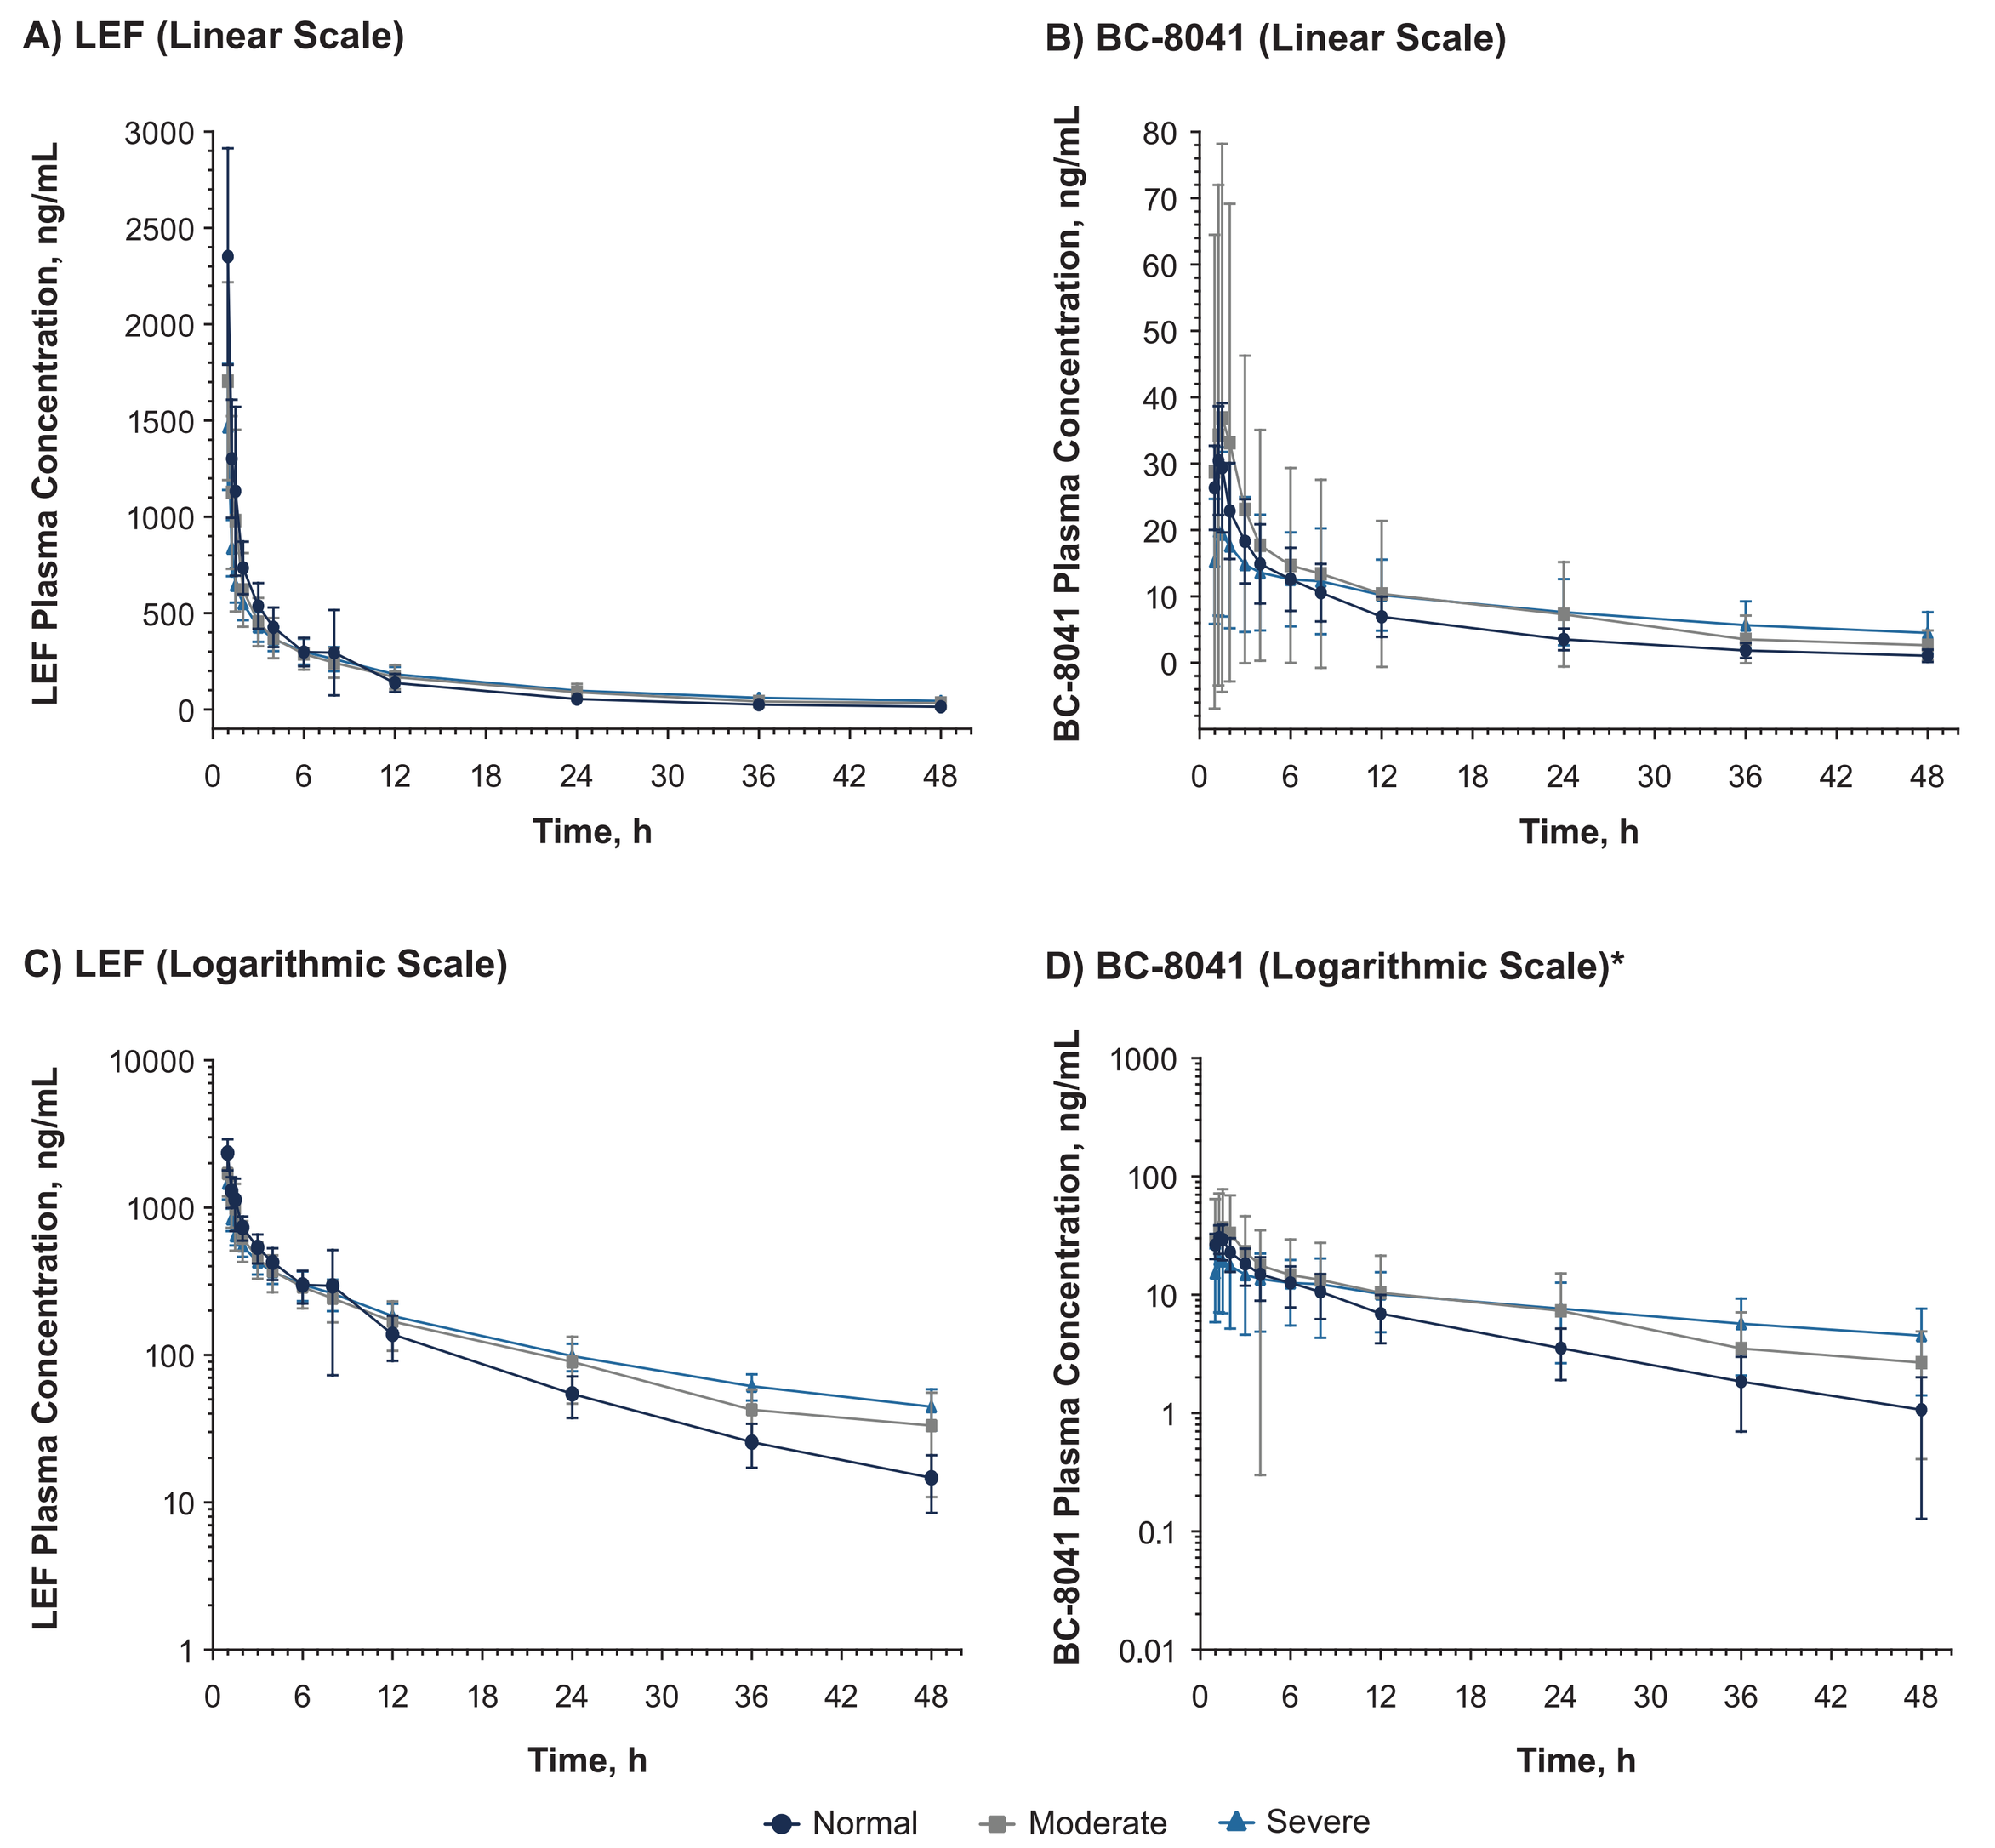

A) LEF (Linear Scale)
B) BC-8041 (Linear Scale)
C) LEF (Logarithmic Scale)
D) BC-8041 (Logarithmic Scale)*
LEF Plasma Concentration, ng/mL
BC-8041 Plasma Concentration, ng/mL
Time, h
Normal
Moderate
Severe

LEF=lefamulin.
*Downward facing error bars do not appear for some data points, since negative values cannot be graphed on a logarithmic scale.

Table 2. LEF and BC-8041 Pharmacokinetics by Hepatic Function Status Group

Parameter, mean (SD)	Normal *n*=11		Moderate *n*=8		Severe *n*=8	
	LEF	BC-8041	LEF	BC-8041	LEF	BC-8041
C_{max}, ng/mL	2463 (403)	33.3 (9.7)	1746 (524)	37.9 (41.2)	1468 (328)	20.4 (12.3)
T_{max}, h	1.0 (0.2)	1.3 (0.1)	1.1 (0.2)	1.5 (0.3)	1.0 (0.0)	1.4 (0.1)
$t_{1/2}$, h	11.5 (1.8)	14.4 (4.5)	13.6 (3.1)	24.4 (20.0)	17.5 (3.4)	33.8 (14.8)
AUC_{0-12}, h·ng/mL	5599 (1112)	163 (55.1)	4781 (1081)	203 (212)	4496 (766)	152 (93.5)
AUC_t, h·ng/mL	7368 (1503)	269 (106)	7538 (1971)	412 (431)	7764 (1308)	401 (244)
AUC, h·ng/mL	7615 (1554)	303 (116)	8233 (2286)	499 (463)	8938 (1640)	647 (441)
CL, L/h	20.5 (4.5)	–	19.6 (6.0)	–	17.4 (3.8)	–
V_z, L	343 (94.1)	–	377 (121)	–	429 (68.9)	–
V_{ss}, L	198 (39.3)	–	291 (107)	–	353 (61.9)	–
A_e, mg	9.7 (2.5)	0.3 (0.1)	21.0 (6.5)	0.7 (0.4)	24.5 (6.9)	1.0 (0.6)
A_e, %	6.5 (1.7)	–	14.0 (4.3)	–	16.3 (4.6)	–
CL_R, L/h	1.3 (0.4)	1.2 (0.4)	2.7 (1.2)	1.6 (0.5)	2.8 (0.8)	1.6 (0.6)
CL_{NR}, L/h	19.2 (4.1)	–	16.9 (5.5)	–	14.6 (3.4)	–

A_e=amount excreted unchanged in urine; AUC=area under plasma concentration-time curve extrapolated through infinity; AUC_{0-12}=AUC from time 0 to 12 hours; AUC_t=AUC from start of infusion through to last measurable (positive) observed concentration; CL=systemic clearance; CL_{NR}=nonrenal clearance; CL_R=renal clearance; C_{max}=maximum observed plasma concentration; LEF=lefamulin; $t_{1/2}$=terminal elimination half-life; T_{max}=time of maximum observed concentration; V_{ss}=volume of distribution at steady-state (observed), estimated using mean residence time; V_Z=volume of distribution based on the terminal phase.

Table 3. Statistical Comparisons of Pharmacokinetic Parameters by Hepatic Function Status Group

	LS Geometric Mean			Ratio (90% CI)	
	Normal	Moderate	Severe	Moderate vs Normal	Severe vs Normal
LEF					
C_{max}	2434	1683	1438	69.1 (58.1–82.3)	59.1 (49.6–70.4)
AUC	7467	7945	8793	106.4 (88.4–128.1)	117.8 (97.8–141.8)
CL	20.1	18.9	17.1	94.0 (78.1–113.2)	84.9 (70.5–102.3)
V_Z	331	361	423	109.4 (88.3–135.4)	128.1 (103.4–158.5)
CL_R	1.3	2.5	2.7	202.3 (150.1–272.7)	215.2 (159.6–290.1)
BC-8041					
C_{max}	32.0	24.0	17.7	75.2 (44.5–127.2)	55.5 (32.8–93.8)
AUC	278	397	536	142.5 (90.3–224.9)	192.4 (121.9–303.5)

AUC=area under plasma concentration-time curve extrapolated through infinity; CL=systemic clearance; CL_R=renal clearance; C_{max}=maximum observed plasma concentration; LEF=lefamulin; LS=least squares; V_Z=volume of distribution based on the terminal phase.

Table 4. LEF PPB by Hepatic Function Status Group

PPB, % (SD)	Normal *n*=11	Moderate *n*=8	Severe *n*=8
1 h	94.8 (1.3)	89.2 (3.2)	86.5 (3.3)
3 h	97.0 (0.6)	91.8 (2.9)	89.6 (2.2)
8 h	97.1 (0.6)	92.8 (2.9)	90.8 (2.8)

LEF=lefamulin; PPB=plasma protein binding.

Safety

- Of the few TEAEs reported across hepatic function status groups, most were mild in severity **(Table 5)**
 - No severe or serious TEAEs were observed, and no TEAEs resulted in study drug discontinuation
- No subjects in any hepatic function status group exhibited clinically significant changes in serum chemistry, hematology, or vital signs, and no subject met Hy’s law criteria
- An increase in mean QT interval corrected according to Fridericia (QTcF) was observed in all hepatic function status groups
 - Within 4 hours postdose, the maximum mean increases from baseline were 12.4, 19.2, and 14.1 msec in the Normal, Moderate, and Severe groups, respectively
 - One subject in the Moderate group had a postbaseline value of >500 msec; no subject had an increase from baseline of >60 msec

Table 5. TEAE Summary*

Category, *n* (%)	Normal *n*=11	Moderate *n*=8	Severe *n*=8
Subjects with ≥1 TEAE	2 (18.2)	2 (25.0)	1 (12.5)
Mild	1 (9.1)	1 (12.5)	1 (12.5)
Moderate	1 (9.1)	1 (12.5)	0
Severe	0	0	0
Subjects with ≥1 drug-related TEAE	1 (9.1)	1 (12.5)	1 (12.5)
Mild	0	1 (12.5)	1 (12.5)
Moderate	1 (9.1)	0	0
Severe	0	0	0
TEAEs occurring in ≥1 subject†			
Constipation	1 (9.1)	0	0
Diarrhea	1 (9.1)	0	1 (12.5)
Electrocardiogram QT prolonged	0	1 (12.5)	0
Headache	1 (9.1)	0	1 (12.5)
Joint injury	0	1 (12.5)	0
Nausea	0	0	1 (12.5)
Pyrexia	1 (9.1)	0	0

MedDRA=Medical Dictionary for Regulatory Activities; TEAE=treatment-emergent adverse event.
*Adverse events were coded using MedDRA Version 20.0.
†Subjects with multiple events in each system organ class and preferred term were only counted once.

CONCLUSIONS

- LEF was generally well tolerated in all subjects regardless of hepatic function status
- Differences in LEF PK across the hepatic function groups were small relative to the overall variability, and changes appear to be compensated by increases in CL_R and decreases in PPB
 - The clinical significance of these changes with respect to efficacy is low, as LEF efficacy has been shown to correlate with AUC,[12] which was consistent across the hepatic function groups
- Dosage adjustment is required for IV LEF when treating subjects with severe hepatic impairment but not mild or moderate hepatic impairment
 - Oral LEF has not been studied in subjects with hepatic impairment and, based on available data, is not recommended for subjects with moderate or severe hepatic impairment

REFERENCES

(1) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(2) File TM Jr, et al. *Clin Infect Dis*. 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(3) Alexander E, et al. *JAMA*. 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].
(4) Nabriva Therapeutics GmbH. Data on file. Vienna, Austria. 2019.
(5) Xu J, et al. Deaths: final data for 2016. US Department of Health and Human Services, Centers for Disease Control and Prevention, National Center for Health Statistics. Available at: https://www.cdc.gov/nchs/data/nvsr/nvsr67/nvsr67_05.pdf. Accessed August 26, 2019. Peyrani P, et al. *Expert Rev Respir Med*. 2019;13(2):139-152.
(6) McDermott KW, et al. Trends in hospital inpatient stays in the United States, 2005-2014: statistical brief #225. *Healthcare Cost and Utilization Project (HCUP) Statistical Briefs*. Rockville, MD: Agency for Healthcare Research and Quality; 2017.
(7) Htun ZM and Gul M. *Chest*. 2018;154(4):958A.
(8) Hung TH, et al. *BMC Gastroenterol*. 2013;13:25.
(9) Xu L, et al. *Respir Res*. 2018;19(1):242.
(10) Charatcharoenwitthaya P, et al. *Medicine (Baltimore)*. 2017;96(32):e7782.
(11) Wicha WW, et al. *J Antimicrob Chemother*. 2019;74(suppl 3):iii5-iii10.

Acknowledgments

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Wolfgang W. Wicha, Lori Lykens, and Steven P. Gelone are employees of/stockholders in Nabriva Therapeutics plc. James A. Dowell, Cathie Leister, and James Ermer have served as consultants for Nabriva Therapeutics. Thomas C. Marbury is an employee and equity owner of Orlando Clinical Research Center.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_HepaticPK

IDWeek 2019: October 2–6, Washington, DC, USA