703

In Vitro Activity of Lefamulin Against Bacterial Pathogens Causing Community-Acquired Bacterial Pneumonia: SENTRY Surveillance 2017–2018 Results From the United States

Susanne Paukner,[1] S. J. Ryan Arends,[2] Steven P. Gelone,[3] Helio S. Sader[2]

[1]Nabriva Therapeutics GmbH, Vienna, Austria; [2]JMI Laboratories, North Liberty, IA, USA; [3]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA

Presented by
Helio S. Sader, MD, PhD
Email: Helio-Sader@jmilabs.com
Phone: 319-665-3370
Fax: 319-665-3371

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION & PURPOSE

- Pneumonia is one of the leading causes of infection-related death in the United States (US)[1] and is associated with substantial morbidity, mortality, and economic burden[2]
- *Streptococcus pneumoniae, Staphylococcus aureus, Haemophilus influenzae, Moraxella catarrhalis,* and the atypical pathogens *Mycoplasma pneumoniae, Chlamydophila pneumoniae,* and *Legionella pneumophila,* are among the most common pathogens that cause community-acquired bacterial pneumonia (CABP) and are usually treated with macrolides, β-lactams, or fluoroquinolones[2,3]
 - Surveillance programs have observed trends of generally decreasing antimicrobial susceptibility for *S. pneumoniae* strains in North America, including 64.1% and 56.1% susceptibility to penicillin (using oral breakpoints) and erythromycin, respectively[4]
 - Methicillin-resistant *S. aureus* (MRSA) has emerged as an important CABP-causing pathogen because of its disproportionate frequency of infecting young, otherwise healthy patients[5]; in the US, methicillin-resistance rates range from 40.7%–54.7%, depending on the region[6]
 - Increasing rates of antimicrobial resistance, combined with increasing safety concerns associated with fluoroquinolones,[7,8] have created a need for new safe and effective treatment options[2]
- Lefamulin (LEF) is the first antimicrobial in the pleuromutilin class approved for intravenous (IV) and oral administration in adults with CABP.[9] LEF selectively inhibits bacterial protein synthesis by binding to the 50S ribosomal subunit at the A- and P-sites in the peptidyl transferase center[10,11] **(Figure 1)**
- In patients with CABP, LEF demonstrated noninferiority to moxifloxacin in the IV-to-oral switch Lefamulin Evaluation Against Pneumonia (LEAP) 1 phase 3 study,[12] and in the LEAP 2 oral-only phase 3 study[13]
- The objective of this analysis was to investigate the in vitro activity of LEF and comparators against a contemporary set of CABP-causing pathogens collected in the US in 2017 and 2018

Figure 1. (A) Structure of Lefamulin and (B) Lefamulin in the Peptidyl Transferase Center of the Large Ribosomal Subunit

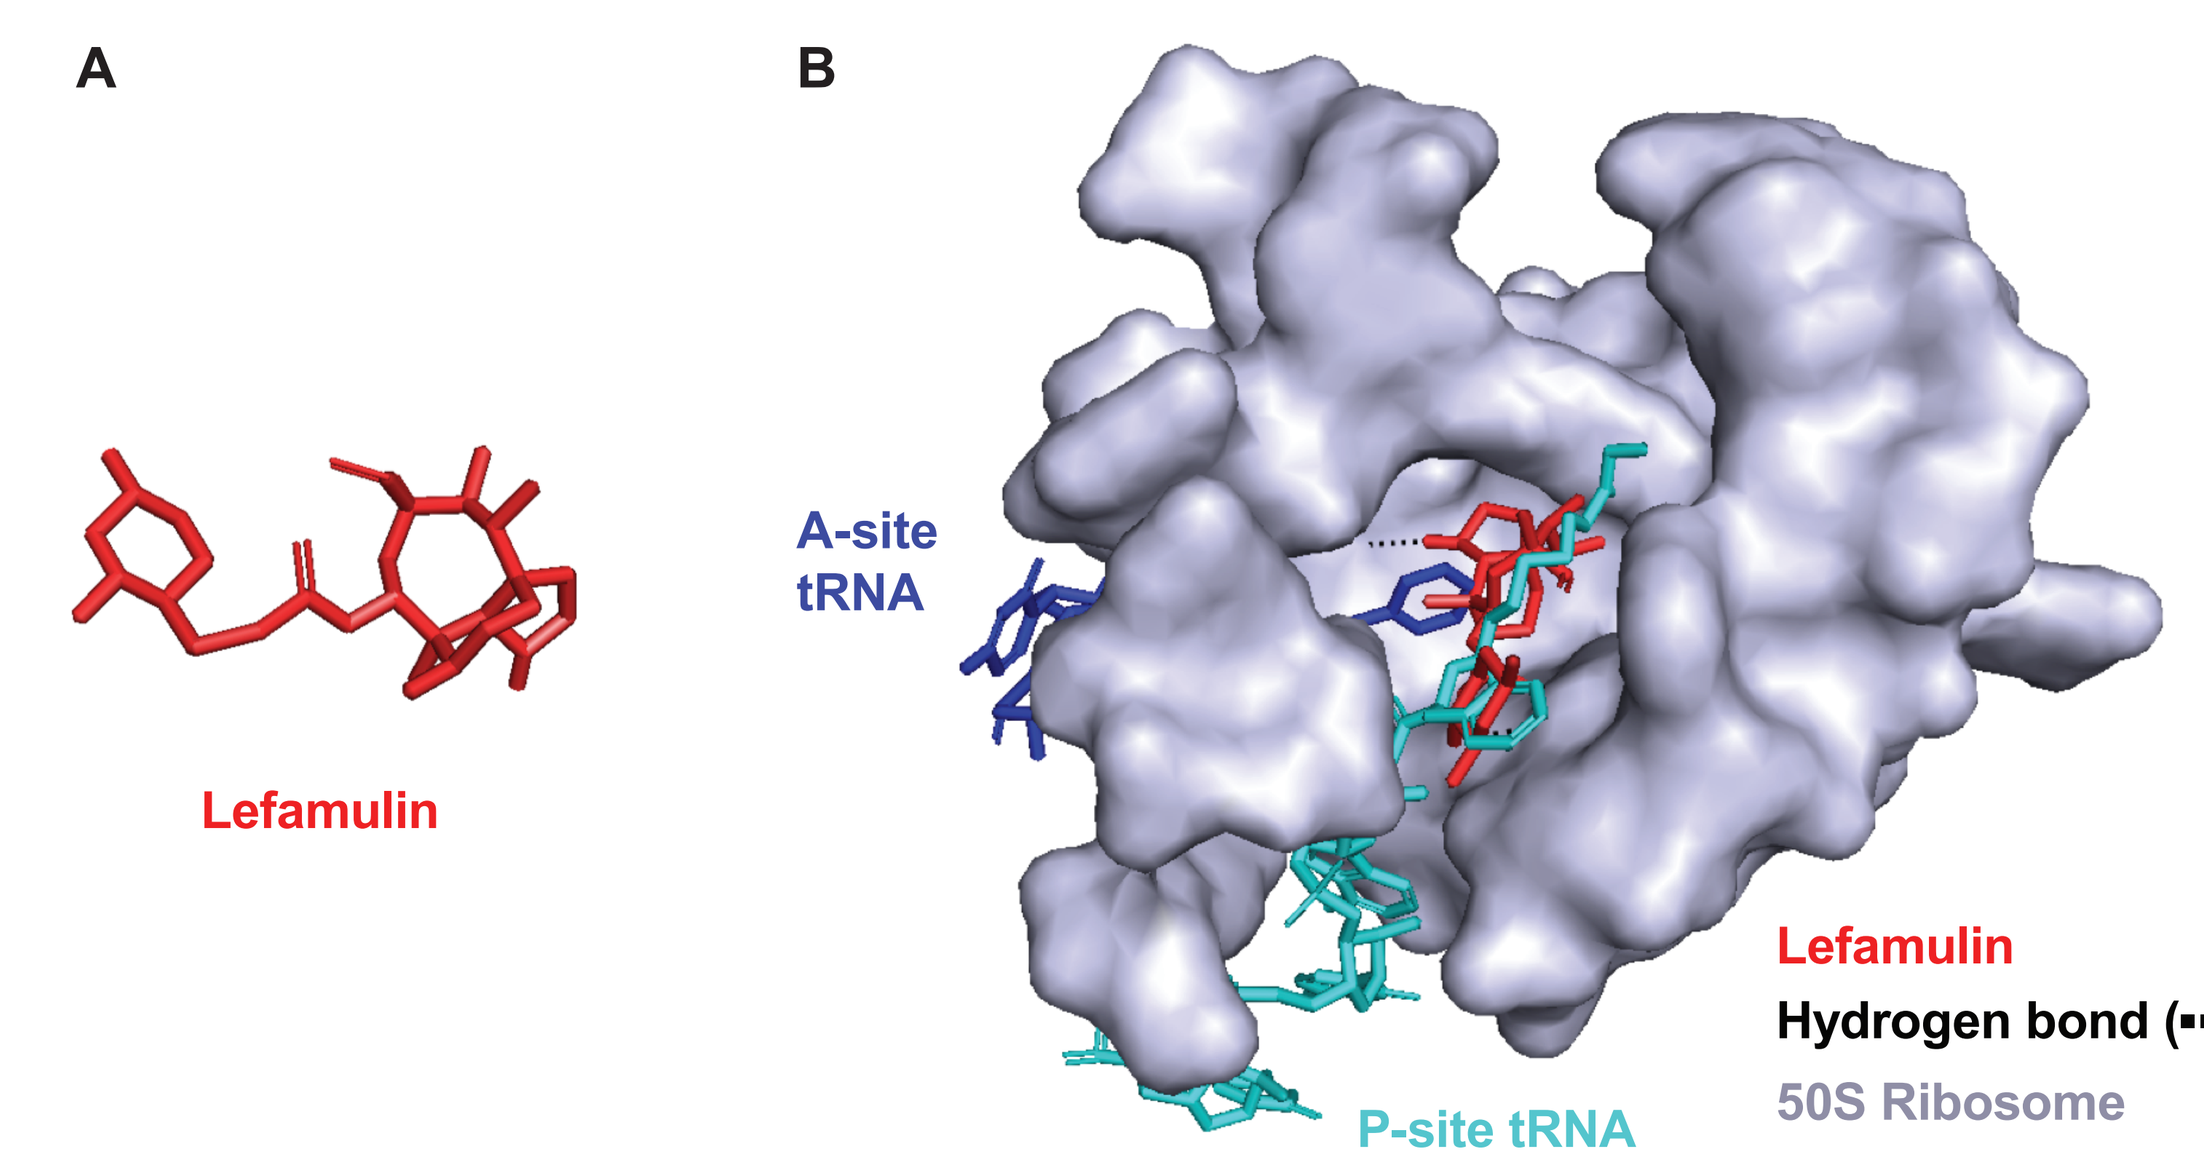

A
B
Lefamulin
A-site tRNA
P-site tRNA
Lefamulin
Hydrogen bond (••)
50S Ribosome

METHODS

- As part of the SENTRY Antimicrobial Surveillance Program, 2299 unique isolates (1 per patient) were collected from 34 US medical centers in 2017 and 2018 from patients with community-acquired respiratory tract infections (1812/2299 [78.8%]) and hospitalized patients with pneumonia (487/2299 [21.2%])
- Minimum inhibitory concentration (MIC) for LEF and comparators was determined using Clinical and Laboratory Standards Institute (CLSI) broth microdilution[14]; susceptibility was evaluated using the CLSI (2019) breakpoints[15]

RESULTS

- LEF demonstrated potent antibacterial activity against all tested CABP pathogens, and its activity was unaffected by resistance to other antibiotic classes

S. pneumoniae

- *S. pneumoniae* isolates showed considerable resistance to macrolides (45.6%), penicillin (36.8%), and tetracycline (20.4%), whereas they were largely susceptible (>85%) to the tested cephalosporins and fluoroquinolones **(Table 1)**
- LEF was highly active against *S. pneumoniae*, with a minimum concentration at which 50% or 90% of the isolates were inhibited ($MIC_{50/90}$) of 0.12/0.25 µg/mL (range ≤0.008–2 µg/mL; **Table 1**)
- LEF was effective against all tested resistant subsets ($MIC_{50/90}$ of 0.12/0.25 µg/mL), with 100% of penicillin- and tetracycline-resistant isolates and 99.5% of macrolide-resistant isolates inhibited at ≤0.5 µg/mL **(Table 2)**

S. aureus

- *S. aureus* isolates overall, and particularly MRSA and fluoroquinolone-resistant strains, were commonly resistant to macrolides; 81.2% of MRSA and 80.4% of fluoroquinolone-resistant strains were resistant to erythromycin (**Table 3**)
- LEF demonstrated potent activity against *S. aureus* isolates, including methicillin-resistant, macrolide-resistant (75.0% MRSA) and fluoroquinolone-resistant (87.6% MRSA) subsets ($MIC_{50/90}$ of 0.06/0.12 µg/mL for each; **Table 3**)

H. influenzae

- *H. influenzae* isolates were largely susceptible to all comparators except ampicillin (31.4% resistant) and trimethoprim-sulfamethoxazole (35.3% resistant; **Table 4**)
- LEF demonstrated activity against *H. influenzae* ($MIC_{50/90}$ of 0.5/2 µg/mL; **Table 4**), with 99.2% of isolates inhibited at ≤2 µg/mL

M. catarrhalis

- *M. catarrhalis* isolates included a large proportion (97.6%) of β-lactamase producers and were susceptible (96.4%–100%) to all comparators, including amoxicillin-clavulanic acid (**Table 4**)
- LEF inhibited all *M. catarrhalis* isolates at concentrations of ≤0.5 µg/mL ($MIC_{50/90}$ of 0.06/0.12 µg/mL; **Table 4**)

β-hemolytic streptococci

- LEF effectively inhibited β-hemolytic streptococci (*n*=14), with $MIC_{50/90}$ values of 0.03/0.06 µg/mL (data not shown)

Table 1. Activity of Lefamulin and Comparators Against *S. pneumoniae*

Antimicrobial Agent	(µg/mL) MIC50	MIC90	Range	CLSI* %S	%I	%R
***S. pneumoniae* (n=1441)**						
Lefamulin	**0.12**	**0.25**	**≤0.008–2**	**99.8†**	**–**	**–**
Amoxicillin-clavulanic acid‡	≤0.03	2	≤0.03–>4	95.1	1.9	3.0
Azithromycin‡	0.12	>4	≤0.03–>4	53.0	1.4	45.6
Ceftaroline	≤0.008	0.12	≤0.008–0.5	100.0	–	–
Ceftriaxone§	0.03	1	≤0.015–>2	86.0‖	11.0	2.9
				97.1¶	2.5	0.4
Clindamycin	≤0.25	>2	≤0.25–>2	85.2	0.4	14.4
Erythromycin	0.06	>16	≤0.015–>16	53.9	0.6	45.6
Levofloxacin	1	1	0.25–>4	99.2	0.1	0.7
Moxifloxacin	0.12	0.25	≤0.03–4	99.4	0.5	0.1
Penicillin	0.03	2	≤0.008–>4	63.2^	26.0	10.8
				63.2#	–	36.8
				96.3**	3.1	0.7
Tetracycline‡	0.5	>4	0.06–>4	79.6	0.1	20.4
Trimethoprim-sulfamethoxazole	0.25	>4	≤0.12–>4	73.6	11.7	14.8

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; R=resistant; S=susceptible.
*2019 CLSI criteria. †2019 FDA susceptibility breakpoint of ≤0.5 µg/mL applied. ‡*n*=1439. §*n*=1440. ‖Using meningitis breakpoints. ¶Using nonmeningitis breakpoints. ^Using oral breakpoints. #Using parenteral, meningitis breakpoints. **Using parenteral, nonmeningitis breakpoints.

Table 2. Activity of Lefamulin and Comparators Against Drug-Resistant *S. pneumoniae*

Antimicrobial Agent	(µg/mL) MIC50	MIC90	Range	CLSI* %S	%I	%R
Penicillin-resistant† *S. pneumoniae* (n=156)						
Lefamulin	**0.12**	**0.25**	**0.015–0.25**	**100.0‡**	**–**	**–**
Amoxicillin-clavulanic acid	2	>4	1–>4	55.1	17.3	27.6
Azithromycin	>4	>4	0.06–>4	5.8	0.0	94.2
Ceftaroline	0.12	0.25	0.06–0.5	100.0	–	–
Ceftriaxone	1	2	0.5–>2	5.8§	67.9	26.3
				73.7‖	22.4	3.8
Clindamycin	≤0.25	>2	≤0.25–>2	57.7	0.6	41.7
Erythromycin	16	>16	0.03–>16	5.8	0.0	94.2
Levofloxacin	1	1	0.5–>4	98.7	0.0	1.3
Moxifloxacin	0.12	0.25	0.06–2	99.4	0.6	0.0
Penicillin	2	4	2–>4	0.0¶	0.0	100.0
				0.0^	–	100.0
				65.4#	28.2	6.4
Tetracycline	1	>4	0.12–>4	50.0	0.0	50.0
Trimethoprim-sulfamethoxazole	>4	>4	≤0.12–>4	26.3	5.1	68.6
Macrolide-resistant *S. pneumoniae* (n=657)**						
Lefamulin	**0.12**	**0.25**	**0.015–2**	**99.5‡**	**–**	**–**
Amoxicillin-clavulanic acid	0.25	4	≤0.03–>4	89.5	4.0	6.5
Azithromycin	>4	>4	0.06–>4	0.2	1.4	98.5
Ceftaroline	0.06	0.12	≤0.008–0.5	100.0	–	–
Ceftriaxone	0.25	1	≤0.015–>2	71.5§	22.1	6.4
				93.6‖	5.5	0.9
Clindamycin	≤0.25	>2	≤0.25–>2	67.9	0.8	31.4
Erythromycin	8	>16	1–>16	0.0	0.0	100.0
Levofloxacin	1	1	0.5–>4	99.2	0.2	0.6
Moxifloxacin	0.12	0.25	≤0.03–2	99.5	0.5	0.0
Penicillin	0.25	2	≤0.008–>4	33.0¶	44.6	22.4
				33.0^	–	67.0
				91.8#	6.7	1.5
Tetracycline	0.5	>4	0.06–>4	60.0	0.0	40.0
Trimethoprim-sulfamethoxazole	0.5	>4	≤0.12–>4	53.9	18.9	27.2
Tetracycline-resistant *S. pneumoniae* (n=293)						
Lefamulin	**0.12**	**0.25**	**0.015–0.5**	**100.0‡**	**–**	**–**
Amoxicillin-clavulanic acid	0.25	>4	≤0.03–>4	79.9	6.5	13.7
Azithromycin††	>4	>4	≤0.03–>4	6.8	2.1	91.1
Ceftaroline	0.06	0.12	≤0.008–0.5	100.0	–	–
Ceftriaxone	0.25	2	≤0.015–>2	71.3§	15.7	13.0
				87.0‖	11.6	1.4
Clindamycin	>2	>2	≤0.25–>2	33.1	1.4	65.5
Erythromycin	>16	>16	≤0.015–>16	9.2	1.0	89.8
Levofloxacin	1	2	0.5–>4	99.0	0.3	0.7
Moxifloxacin	0.12	0.25	0.06–2	99.7	0.3	0.0
Penicillin	0.25	4	≤0.008–>4	21.2¶	52.2	26.6
				21.2^	–	78.8
				83.6#	13.3	3.1
Tetracycline	>4	>4	4–>4	0.0	0.0	100.0
Trimethoprim-sulfamethoxazole	1	>4	≤0.12–>4	39.6	24.9	35.5

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC=minimum inhibitory concentration; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; R=resistant; S=susceptible.
*2019 CLSI criteria. †Penicillin MIC ≥2 µg/mL for oral breakpoint. ‡2019 FDA susceptibility breakpoint of ≤0.5 µg/mL applied. §Using meningitis breakpoints. ‖Using nonmeningitis breakpoints. ¶Using oral breakpoints. ^Using parenteral, meningitis breakpoints. #Using parenteral, nonmeningitis breakpoints. **Using erythromycin breakpoints. ††*n*=292.

Table 3. Activity of Lefamulin and Comparators Against *S. aureus*

Antimicrobial Agent	(µg/mL) MIC50	MIC90	Range	CLSI* %S	%I	%R
***S. aureus* (n=297)**						
Lefamulin	**0.06**	**0.12**	**0.03–0.25**	**100.0†**	**–**	**–**
Azithromycin	32	>32	0.12–>32	46.5	0.3	53.2
Ceftaroline	0.25	1	≤0.06–2	94.3‡	5.7	0.0
Clindamycin	0.06	>2	≤0.03–>2	79.5	0.0	20.5
Doxycycline	0.12	0.5	≤0.06–>8	98.0	1.7	0.3
Erythromycin	4	>8	≤0.06–>8	46.5	5.1	48.5
Gentamicin	≤1	≤1	≤1–>8	96.6	0.3	3.0
Levofloxacin	0.25	>4	0.12–>4	62.0	0.0	38.0
Linezolid	1	2	0.25–4	100.0	–	0.0
Moxifloxacin	≤0.06	>4	≤0.06–>4	62.0	5.4	32.7
Oxacillin	1	>2	0.12–>2	55.2	–	44.8
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5–>16	98.7	–	1.3
Vancomycin	1	1	0.25–2	100.0	0.0	0.0
MRSA (n=133)						
Lefamulin	**0.06**	**0.12**	**0.03–0.25**	**100.0†**	**–**	**–**
Azithromycin	>32	>32	0.12–>32	14.3	0.0	85.7
Ceftaroline	1	2	0.25–2	87.2‡	12.8	0.0
Clindamycin	0.06	>2	≤0.03–>2	58.6	0.0	41.4
Doxycycline	0.12	1	≤0.06–8	97.0	3.0	0.0
Erythromycin	>8	>8	≤0.06–>8	14.3	4.5	81.2
Gentamicin	≤1	≤1	≤1–>8	94.7	0.0	5.3
Levofloxacin	>4	>4	0.12–>4	26.3	0.0	73.7
Linezolid	1	2	0.25–2	100.0	–	0.0
Moxifloxacin	2	>4	≤0.06–>4	26.3	9.8	63.9
Oxacillin	>2	>2	>2–>2	0.0	–	100.0
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5–>16	97.0	–	3.0
Vancomycin	1	1	0.5–2	100.0	0.0	0.0
Macrolide-resistant§ *S. aureus* (n=144)						
Lefamulin	**0.06**	**0.12**	**0.03–0.25**	**100.0†**	**–**	**–**
Azithromycin	>32	>32	8–>32	0.0	0.0	100.0
Ceftaroline	0.5	2	≤0.06–2	88.2‡	11.8	0.0
Clindamycin	0.06	>2	≤0.03–>2	57.6	0.0	42.4
Doxycycline	0.12	1	≤0.06–>8	96.5	2.8	0.7
Erythromycin	>8	>8	8–>8	0.0	0.0	100.0
Gentamicin	≤1	≤1	≤1–>8	94.4	0.0	5.6
Levofloxacin	>4	>4	0.12–>4	35.4	0.0	64.6
Linezolid	1	2	0.25–2	100.0	–	0.0
Moxifloxacin	2	>4	≤0.06–>4	35.4	10.4	54.2
Oxacillin	>2	>2	0.25–>2	25.0	–	75.0
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5–>16	98.6	–	1.4
Vancomycin	1	1	0.5–2	100.0	0.0	0.0
Fluoroquinolone-resistant‖ *S. aureus* (n=97)						
Lefamulin	**0.06**	**0.12**	**0.03–0.25**	**100.0†**	**–**	**–**
Azithromycin	>32	>32	0.25–>32	14.4	1.0	84.5
Ceftaroline	1	2	0.12–2	82.5‡	17.5	0.0
Clindamycin	>2	>2	≤0.03–>2	47.4	0.0	52.6
Doxycycline	0.12	1	≤0.06–8	96.9	3.1	0.0
Erythromycin	>8	>8	≤0.06–>8	14.4	5.2	80.4
Gentamicin	≤1	≤1	≤1–>8	92.8	0.0	7.2
Levofloxacin	>4	>4	4–>4	0.0	0.0	100.0
Linezolid	1	2	0.5–2	100.0	–	0.0
Moxifloxacin	>4	>4	2–>4	0.0	0.0	100.0
Oxacillin	>2	>2	0.25–>2	12.4	–	87.6
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5–>16	95.9	–	4.1
Vancomycin	1	1	0.5–2	100.0	0.0	0.0

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; MRSA=methicillin-resistant *S. aureus*; MSSA=methicillin-susceptible *S. aureus*; R=resistant; S=susceptible.
*2019 CLSI criteria. †2019 FDA susceptibility breakpoint for MSSA of ≤0.25 µg/mL applied. ‡Intermediate interpreted as susceptible-dose dependent. §Using erythromycin breakpoints. ‖Using moxifloxacin breakpoints.

Table 4. Activity of Lefamulin and Comparators Against *H. influenzae* and *M. catarrhalis*

Antimicrobial Agent	(µg/mL) MIC50	MIC90	Range	CLSI* %S	%I	%R
***H. influenzae* (n=382)**						
Lefamulin	**0.5**	**2**	**0.015–8**	**99.2†**	**–**	**–**
Amoxicillin-clavulanic acid	0.5	2	≤0.06–>8	99.2	–	0.8
Ampicillin	0.5	>8	≤0.12–>8	65.4	3.1	31.4
Azithromycin	1	2	≤0.12–>8	98.4	–	–
Cefepime	0.12	0.25	≤0.015–>2	99.7	–	–
Ceftriaxone	0.004	0.015	≤0.002–0.25	100.0	–	–
Ciprofloxacin	0.015	0.015	0.008–>1	98.7	–	–
Clarithromycin	8	16	≤0.12–>16	80.9	16.2	2.9
Moxifloxacin	0.03	0.06	0.015–>2	99.0	–	–
Tetracycline	0.5	1	0.12–>8	99.0	0.0	1.0
Trimethoprim-sulfamethoxazole	0.12	>4	≤0.06–>4	62.0	2.6	35.3
***M. catarrhalis* (n=165)**						
Lefamulin	**0.06**	**0.12**	**≤0.008–0.12**	**–**	**–**	**–**
Amoxicillin-clavulanic acid	≤0.25	≤0.25	≤0.25–0.5	100.0	–	0.0
Azithromycin	≤0.03	≤0.03	≤0.03–0.06	100.0	–	–
Ceftriaxone	0.25	1	0.004–2	100.0	–	–
Clarithromycin	≤0.12	≤0.12	≤0.12–0.5	100.0	–	–
Moxifloxacin	0.06	0.06	0.015–0.12	–	–	–
Tetracycline	0.25	0.5	0.12–>8	99.4	0.0	0.6
Trimethoprim-sulfamethoxazole	0.12	0.25	≤0.06–2	96.4	3.6	0.0

CLSI=Clinical and Laboratory Standards Institute; I=intermediate; MIC_{50}=minimum concentration at which 50% of isolates were inhibited; MIC_{90}=minimum concentration at which 90% of isolates were inhibited; R=resistant; S=susceptible.
*2019 CLSI criteria. †2019 FDA susceptibility breakpoint of ≤2 µg/mL applied.

CONCLUSIONS

- LEF demonstrated potent in vitro activity against this contemporary (2017–2018) set of pathogens collected in the US from patients with respiratory tract infections and hospitalized patients with pneumonia
- LEF activity was comparable with the most common antimicrobial agents used to treat CABP and was unaffected by resistance to other antibiotic classes, including macrolides, fluoroquinolones, β-lactams, and tetracyclines
- These in vitro data, as well as the high efficacy in CABP patients from phase 3 clinical trials (LEAP 1 and 2),[12,13] suggest that LEF may offer an important empiric monotherapy treatment option for CABP, particularly where resistance to antimicrobial agents commonly used to treat CABP is high

REFERENCES

(1) Xu J, et al. Deaths: Final Data for 2016. US Department of Health and Human Services, Centers for Disease Control and Prevention, National Center for Health Statistics. Available at: https://www.cdc.gov/nchs/data/nvsr/nvsr67/nvsr67_05.pdf. Accessed July 17, 2019.
(2) Peyrani P, et al. *Expert Rev Respir Med.* 2019;13(2):139-152.
(3) Pfaller MA, et al. *Clin Infect Dis.* 2012;55(suppl 3):S187-193.
(4) Sader HS, et al. *Open Forum Infect Dis.* 2019;6(suppl 1):S14-S23.
(5) Wunderink RG. *Infect Dis Clin North Am.* 2013;27(1):177-188.
(6) Diekema DJ, et al. *Open Forum Infect Dis.* 2019;6(suppl 1):S47-S53.
(7) US Food and Drug Administration. FDA drug safety communication: FDA updates warnings for oral and injectable fluoroquinolone antibiotics due to disabling side effects. Available at: https://www.fda.gov/Drugs/DrugSafety/ucm511530.htm. Accessed April 9, 2019.
(8) US Food and Drug Administration. FDA drug safety communication: FDA reinforces safety information about serious low blood sugar levels and mental health side effects with fluoroquinolone antibiotics; requires label changes. Available at: https://www.fda.gov/downloads/Drugs/DrugSafety/UCM612834.pdf. Accessed April 9, 2019.
(9) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(10) Eyal Z, et al. *Sci Rep.* 2016;6:39004.
(11) Schlünzen F, et al. *Mol Microbiol.* 2004;54(5):1287-1294.
(12) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(13) Alexander E, et al. *JAMA.* 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].
(14) Clinical and Laboratory Standards Institute. Performance standards for antimicrobial susceptibility testing: 28th informational supplement. Wayne, PA 2018. M100Ed28E.
(15) Clinical and Laboratory Standards Institute. Performance standards for antimicrobial susceptibility testing, 29th edition. Wayne, PA 2019. M100Ed29.

Acknowledgments

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Susanne Paukner and Steven P. Gelone are employees of/stockholders in Nabriva Therapeutics plc. S. J. Ryan Arends and Helio S. Sader are employees of JMI Laboratories, which was contracted by Nabriva Therapeutics to conduct these analyses.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_SENTRY2017-18_USA

IDWeek 2019: October 2–6, Washington, DC, USA