699

Hepatobiliary Safety in Adults With Community-Acquired Bacterial Pneumonia Treated With Lefamulin or Moxifloxacin: Pooled Analysis of Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 Study Results

James H. Lewis,[1] Anita F. Das,[2] Daniel S. Stein,[3] Steven P. Gelone,[3] Jennifer Schranz[3]

[1]Georgetown University Medical Center, Washington, DC, USA; [2]Das Consulting, Guerneville, CA, USA; [3]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA

Presented by
Jennifer Schranz, MD
Email: Jennifer.Schranz@Nabriva.com
Phone: 610-585-6294
Fax: 610-816-6639

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION & PURPOSE

- Lefamulin (LEF) is the first pleuromutilin antibiotic approved for intravenous (IV) and oral use in adults with community-acquired bacterial pneumonia (CABP)[1]
- LEF inhibits protein synthesis by binding to the central part of the peptidyl transferase center of the 50S ribosomal subunit, forming 4 hydrogen bonds and other interactions that prevent the correct transfer RNA positioning in the A- and P-sites[2,3]
- LEF displays potent in vitro activity against pathogens that commonly cause CABP, and its activity is unaffected in vitro by an organism's resistance to other major antimicrobial classes[4,5]
- Most antibiotic classes (eg, cephalosporins, fluoroquinolones, macrolides, tetracyclines) have hepatic enzyme elevation and other hepatic adverse events listed in their package inserts[6-9]
- Extensive nonclinical evaluations of LEF have suggested that substantial hepatotoxicity as a direct result of LEF is unlikely[10]
- However, because potential liver injury has been a concern with multiple antibiotic classes, we assessed hepatobiliary safety in adults with CABP treated with LEF or moxifloxacin (MOX), a standard-of-care fluoroquinolone, in a pooled analysis of the Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 clinical trial data[11,12]

METHODS

Study Design and Patients

- Both studies were prospective, randomized, double-blind, double-dummy, phase 3 trials[11,12]
 - In LEAP 1, patients with Pneumonia Outcomes Research Team (PORT) risk classes III–V were randomized to receive LEF 150 mg IV every 12 hours (q12h) for 5–7 days or MOX 400 mg IV every 24 hours (q24h) for 7 days
 - Patients could switch to oral therapy (LEF 600 mg q12h or MOX 400 mg q24h) after 6 IV doses of study drug (approximately 3 days) if predefined improvement criteria were met
 - In LEAP 2, patients with PORT risk classes II–IV were randomized to receive oral LEF 600 mg q12h for 5 days or oral MOX 400 mg q24h for 7 days
 - Refer to **Poster 664** for further details on study design for the LEAP 1 and LEAP 2 trials
- Patients with evidence of significant hepatic disease were excluded from both studies, including:
 - Known acute hepatitis, including active viral hepatitis
 - Cirrhosis
 - Aspartate aminotransferase (AST) or alanine aminotransferase (ALT) >5× upper limit of normal (ULN)
 - Total bilirubin >3× ULN (unless Gilbert's disease)
 - AST or ALT >3× ULN and total bilirubin >2× ULN
 - Manifestation of end-stage liver disease (eg, ascites, hepatic encephalopathy)

Hepatobiliary Safety Assessments

- Hepatobiliary safety assessments were based on central laboratory evaluation (Covance Central Laboratory Services, Indianapolis, IN, USA; Geneva, Switzerland; Singapore), treatment-emergent adverse events (TEAEs), and expert consultant adjudication
- Blood samples for assessment of clinical chemistry parameters were collected at baseline, on Day 4, at end of treatment, and at test of cure in both studies
- To focus on cases with more probable liver injury, patients with Grade 2 or higher ALT values were evaluated using the Common Terminology Criteria for Adverse Events (CTCAE) version 5.0[13]
- The pattern of hepatic injury was assessed by calculating the R-value, which was defined as the ratio of the (maximum postbaseline ALT value divided by ULN) divided by the (maximum postbaseline alkaline phosphatase [ALP] value divided by ULN). The ALP value was taken from the same day as the maximum ALT value. If ALP was not available, then the ALP assessment closest to the day of the maximum ALT value was used
- Case narratives of patients with liver-related TEAEs or AST/ALT increases >3× ULN were reviewed by an independent external expert

RESULTS

Patients

- The intent-to-treat population (*N*=1289) comprised all randomized patients (LEF, *n*=646; MOX, *n*=643), and the safety analysis set (*n*=1282) comprised all randomized and treated patients (LEF, *n*=641; MOX, *n*=641)
- Patient demographics and baseline disease characteristics were generally well balanced between treatment groups (**Table 1**)
 - In both treatment groups, few patients had known hepatic disease
- Approximately 20% of patients enrolled in the CABP studies presented with elevated ALT or AST values at baseline

RESULTS (continued)

Hepatobiliary Safety Analyses

- The postbaseline distribution of ALT, AST, ALP, and total bilirubin values was generally similar for both treatment groups (**Table 2**)
 - Mean maximum increases from baseline were somewhat higher in the LEF group compared with the MOX group: ALT (19.0 U/L vs 10.8 U/L), AST (10.6 U/L vs 5.9 U/L), and ALP (13.2 U/L vs 8.5 U/L)
 - The mean maximum increase from baseline in total bilirubin was similar in the LEF (1.6 U/L) and MOX (1.0 U/L) groups
- Similar and small percentages of patients in both treatment groups had hepatobiliary laboratory values of interest (**Figure 1**)
 - The percentages of patients with postbaseline ALT >3× ULN were 5.5% for LEF and 5.4% for MOX
- In both treatment groups, patients with elevated transaminases at baseline were more likely to have postbaseline elevations >3× ULN, but the majority remained below 5× ULN (**Table 3**)
- Among patients with postbaseline ALT >5× ULN, peak increases were generally seen in the first week after the first LEF dose and declined to within/near normal levels by late follow-up (Day 28); for MOX, time to peak ALT was less consistent (**Figure 2**)
- No LEF patient and 1 MOX patient met laboratory criteria for Hy's Law (ie, any postbaseline ALT or AST value >3× ULN, any postbaseline total bilirubin value >2× ULN, and any postbaseline ALP value ≤2× ULN)
- 20 LEF-treated patients had a CTCAE Grade 2 or higher ALT value
 - The hepatic injury biochemical pattern as classified by R-values was: hepatocellular for 10 patients (50%), cholestatic for 2 patients (10%), and mixed for 8 patients (40%), with no apparent sex, age, or ethnic predominance
- TEAEs in the hepatobiliary disorders system organ class were reported in 6 LEF patients (0.9%) and 6 MOX patients (0.9%), with similar levels seen in patients with elevated baseline transaminases (**Table 4**)
- In terms of severity, all patients identified in this hepatobiliary safety analysis were asymptomatic with no hypersensitivity features (eg, fever, rash, eosinophilia), and the transaminase time course indicated reversible injury (within 2–4 weeks) with no development of chronic injury

Table 1. Demographics and Baseline Disease Characteristics (Pooled ITT Population)

Parameter	LEF (*n*=646)	MOX (*n*=643)
Age, y, mean (SD)	58.9 (16.5)	58.5 (15.7)
Male, *n* (%)	377 (58.4)	340 (52.9)
BMI, kg/m², mean (SD)	26.5 (5.8)	26.4 (6.0)
PORT risk class,* *n* (%)		
I/II	184 (28.5)	192 (29.9)
III	341 (52.8)	334 (51.9)
IV/V	121 (18.7)	117 (18.2)
Hepatobiliary disorders, *n* (%)		
Cholelithiasis	9 (1.4)	3 (0.5)
Hepatic steatosis	8 (1.2)	7 (1.1)
Cholecystitis, chronic	7 (1.1)	7 (1.1)
Chronic hepatitis	3 (0.5)	0
Liver disorder	2 (0.3)	1 (0.2)
Hepatic function, abnormal	1 (0.2)	1 (0.2)
Cholecystitis	1 (0.2)	0
Nonalcoholic fatty liver	1 (0.2)	0
Postcholecystectomy syndrome	1 (0.2)	0
Hepatic cyst	0	1 (0.2)

BMI=body mass index; ITT=intent to treat; LEAP=Lefamulin Evaluation Against Pneumonia; LEF=lefamulin; MOX=moxifloxacin; PORT=Pneumonia Outcomes Research Team.
*PORT risk class was calculated programmatically using data obtained at the site and reported in the electronic case report form and was not always consistent with the site-reported PORT risk class used for enrollment/stratification; as a result, 3 patients with PORT risk class I (LEF, *n*=1; MOX, *n*=2) were enrolled in LEAP 2.

Table 2. Change From Baseline in Hepatobiliary Parameters (Pooled Safety Analysis Set)

Parameter* Time Point	LEF (*n*=641)	MOX (*n*=641)
Baseline ALT value,† U/L	25.3 (20.0) *n*=588	29.3 (55.8) *n*=594
Change from baseline ALT,‡ U/L		
Day 4/5	9.9 (43.1)	4.6 (23.4)
EOT	8.5 (33.2)	2.4 (60.0)
TOC	3.3 (27.0)	–3.2 (58.5)
Baseline AST value,† U/L	27.1 (21.2) *n*=568	27.3 (20.7) *n*=582
Change from baseline AST,‡ U/L		
Day 4/5	3.1 (28.2)	0.6 (23.1)
EOT	–0.8 (22.9)	–2.0 (23.7)
TOC	–2.4 (23.6)	–4.3 (21.3)
Baseline ALP value,† U/L	86.5 (55.6) *n*=621	86.0 (51.5) *n*=625
Change from baseline ALP,‡ U/L		
Day 4/5	1.8 (38.6)	–2.7 (27.5)
EOT	1.6 (39.9)	–3.5 (30.2)
TOC	–1.1 (38.7)	–2.5 (36.4)
Baseline total bilirubin value,† µmol/L	9.4 (5.7) *n*=590	9.3 (6.1) *n*=596
Change from baseline total bilirubin,‡ µmol/L		
Day 4/5	–1.8 (4.8)	–1.9 (5.1)
EOT	–1.5 (5.4)	–1.9 (5.7)
TOC	–0.7 (6.2)	–1.4 (5.8)

ALP=alkaline phosphatase; ALT=alanine aminotransferase; AST=aspartate aminotransferase; EOT=end of treatment; LEF=lefamulin; MOX=moxifloxacin; TOC=test of cure.
*All values are means (standard deviations).
†Last assessment before the first dose of study drug.
‡Includes all patients with values for the specified laboratory parameter at both baseline and the specified time point.

Figure 1. Percentages of Patients With Postbaseline Hepatobiliary Values of Interest* (Pooled Safety Analysis Set)

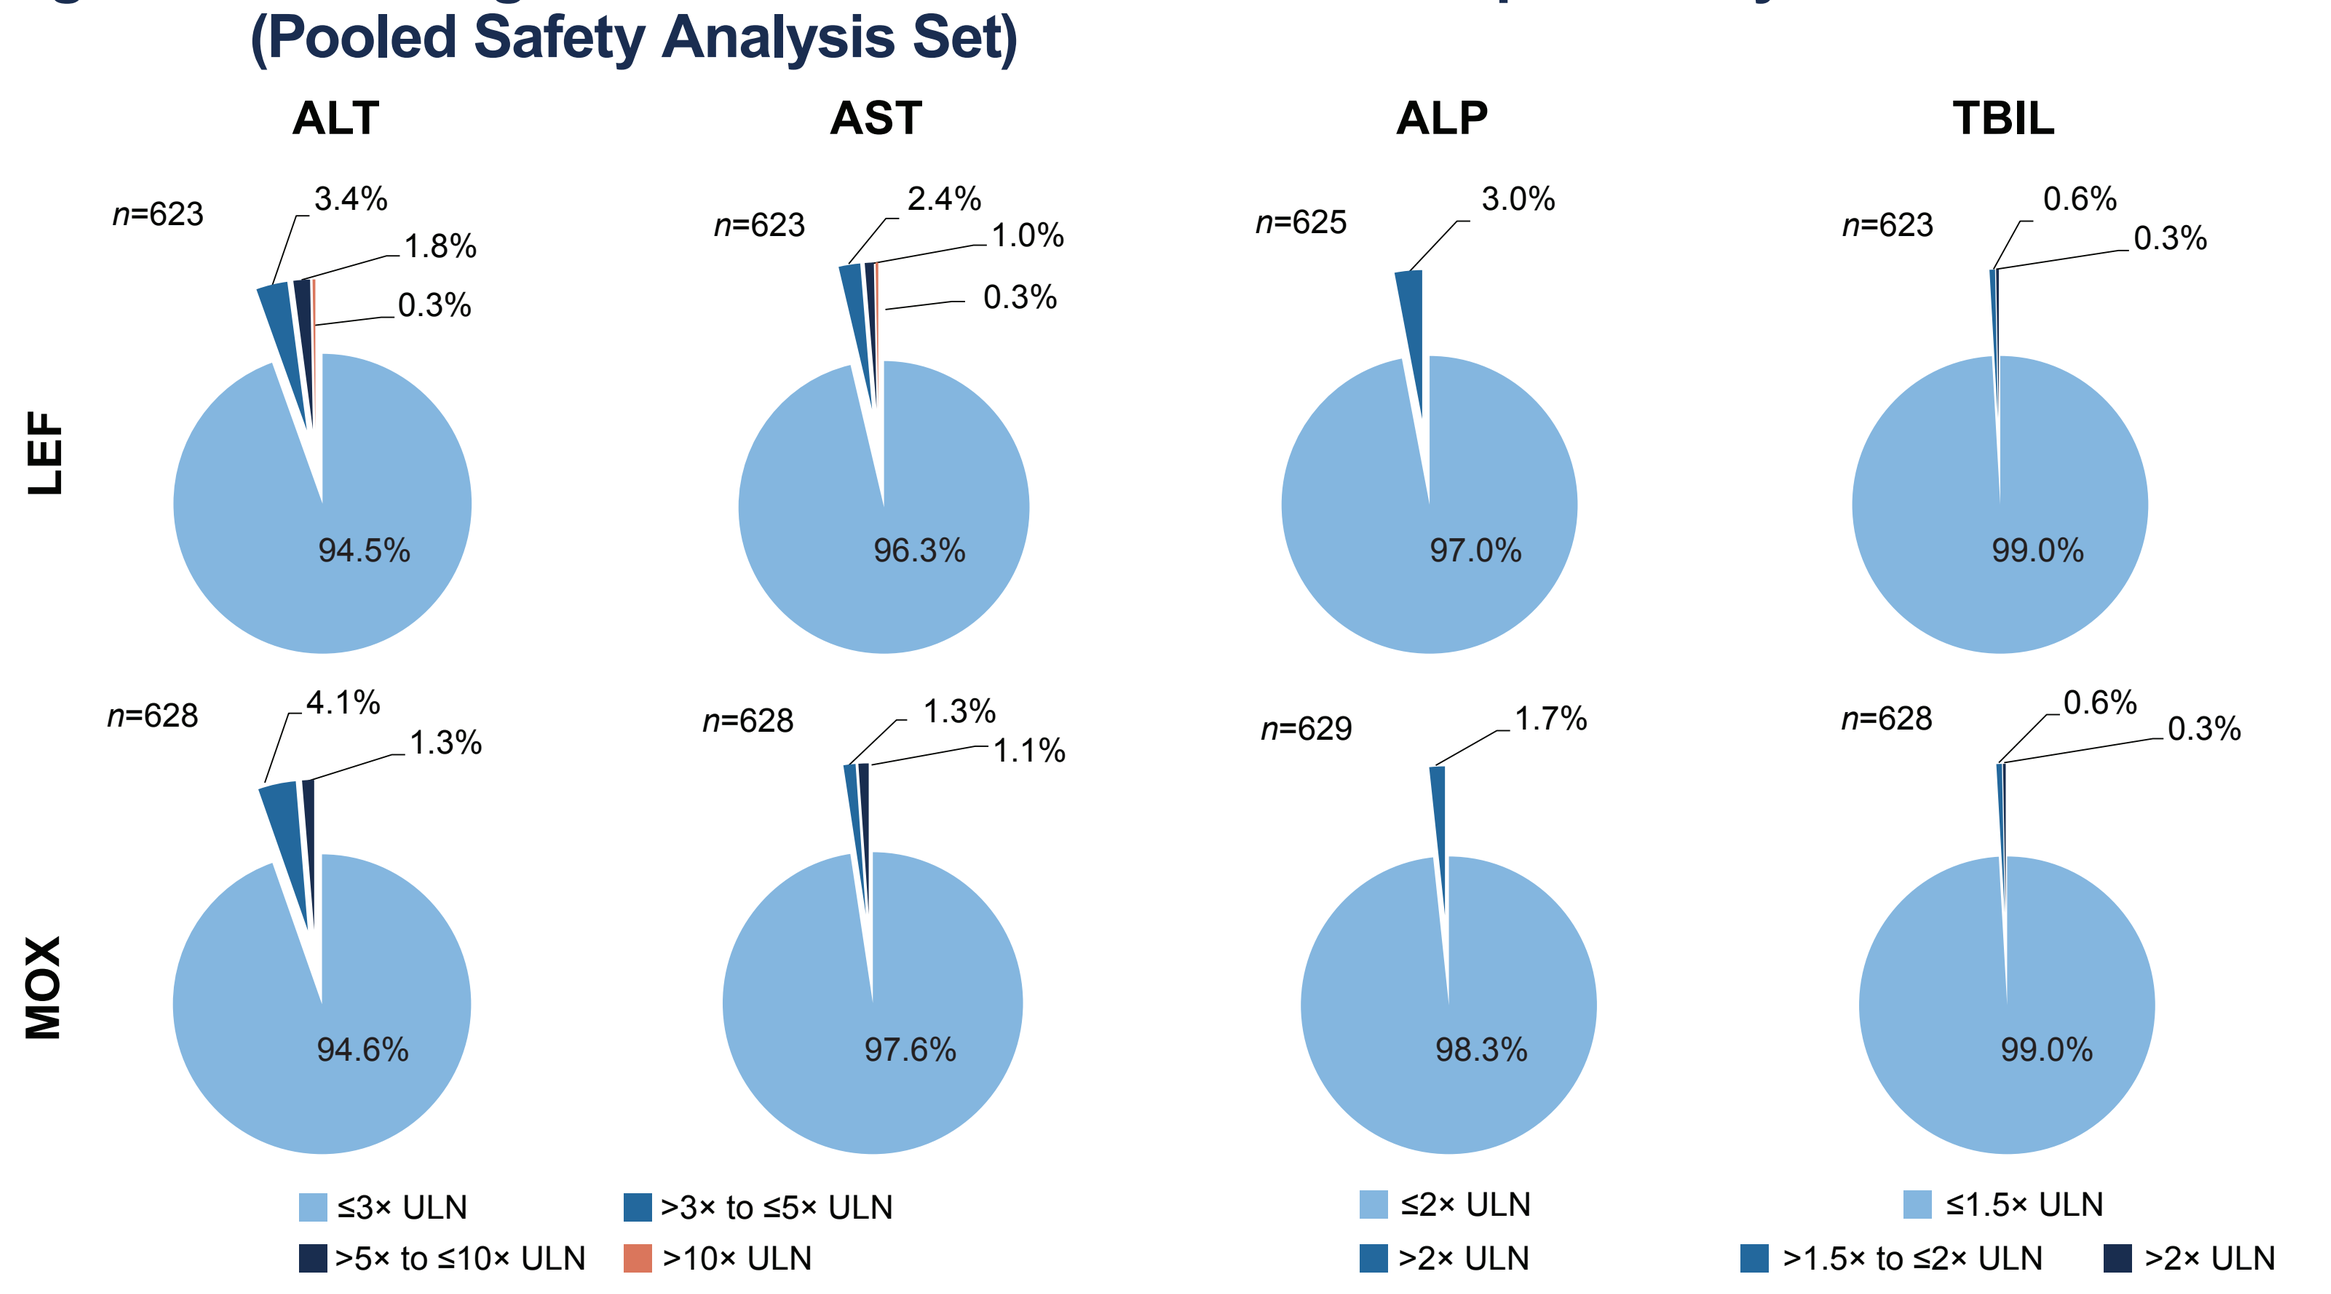
ALT
AST
ALP
TBIL
LEF
MOX
n=623 3.4% 1.8% 0.3% 94.5%
n=623 2.4% 1.0% 0.3% 96.3%
n=625 3.0% 97.0%
n=623 0.6% 0.3% 99.0%
n=628 4.1% 1.3% 94.6%
n=628 1.3% 1.1% 97.6%
n=629 1.7% 98.3%
n=628 0.6% 0.3% 99.0%
≤3× ULN
>3× to ≤5× ULN
>5× to ≤10× ULN
>10× ULN
≤2× ULN
>2× ULN
≤1.5× ULN
>1.5× to ≤2× ULN
>2× ULN

ALP=alkaline phosphatase; ALT=alanine aminotransferase; AST=aspartate aminotransferase; EOT=end of treatment; LEF=lefamulin; LFU=late follow-up; MOX=moxifloxacin; TBIL=total bilirubin; TOC=test of cure; ULN=upper limit of normal.
*In LEAP 1, 1 LEF patient who did not have a baseline ALT value had an ALT value >10× ULN at EOT on Day 8 (396 U/L; normal range, 6–35 U/L); the elevated value returned to the normal range at LFU on Day 27 (15 U/L). Also in LEAP 1, 1 LEF patient who did not have a baseline AST value had an AST value >10× ULN on Day 4 (428 U/L; normal range, 11–36 U/L); the elevated value returned to the normal range at TOC on Day 12 (35 U/L). In LEAP 2, 1 LEF patient who had a baseline ALT value of 24 U/L (normal range, 6–43 U/L) had an ALT value >10× ULN on Day 5 (595 U/L); the elevated value returned to the normal range at LFU on Day 21 (43 U/L). Also in LEAP 2, 1 LEF patient who had a baseline AST value of 40 U/L (normal range, 11–36 U/L) had an AST value >10× ULN on Day 4 (791 U/L); the elevated value returned to the normal range at EOT on Day 8 (36 U/L) and at subsequent visits.

Table 3. Postbaseline ALT and AST Values by Baseline Liver Enzyme Status (Pooled Safety Analysis Set)

Postbaseline Threshold	Baseline Status	Postbaseline ALT LEF (*n*=641)	Postbaseline ALT MOX (*n*=641)	Postbaseline AST LEF (*n*=641)	Postbaseline AST MOX (*n*=641)
>3× ULN	Normal ALT and AST	15/457 (3.3%)	9/437 (2.1%)	8/435 (1.8%)	3/432 (0.7%)
	ALT or AST > ULN	15/114 (13.2%)	23/142 (16.2%)	12/114 (10.5%)	10/142 (7.0%)
>5× ULN	Normal ALT and AST	7/457 (1.5%)	1/437 (0.2%)	5/435 (1.1%)	1/432 (0.2%)
	ALT or AST > ULN	4/114 (3.5%)	7/142 (4.9%)	2/114 (1.8%)	5/142 (3.5%)

ALT=alanine aminotransferase; AST=aspartate aminotransferase; LEF=lefamulin; MOX=moxifloxacin; ULN=upper limit of normal.

Figure 2. Individual ALT Values for Patients With Postbaseline ALT >5× ULN (Pooled Safety Analysis Set)

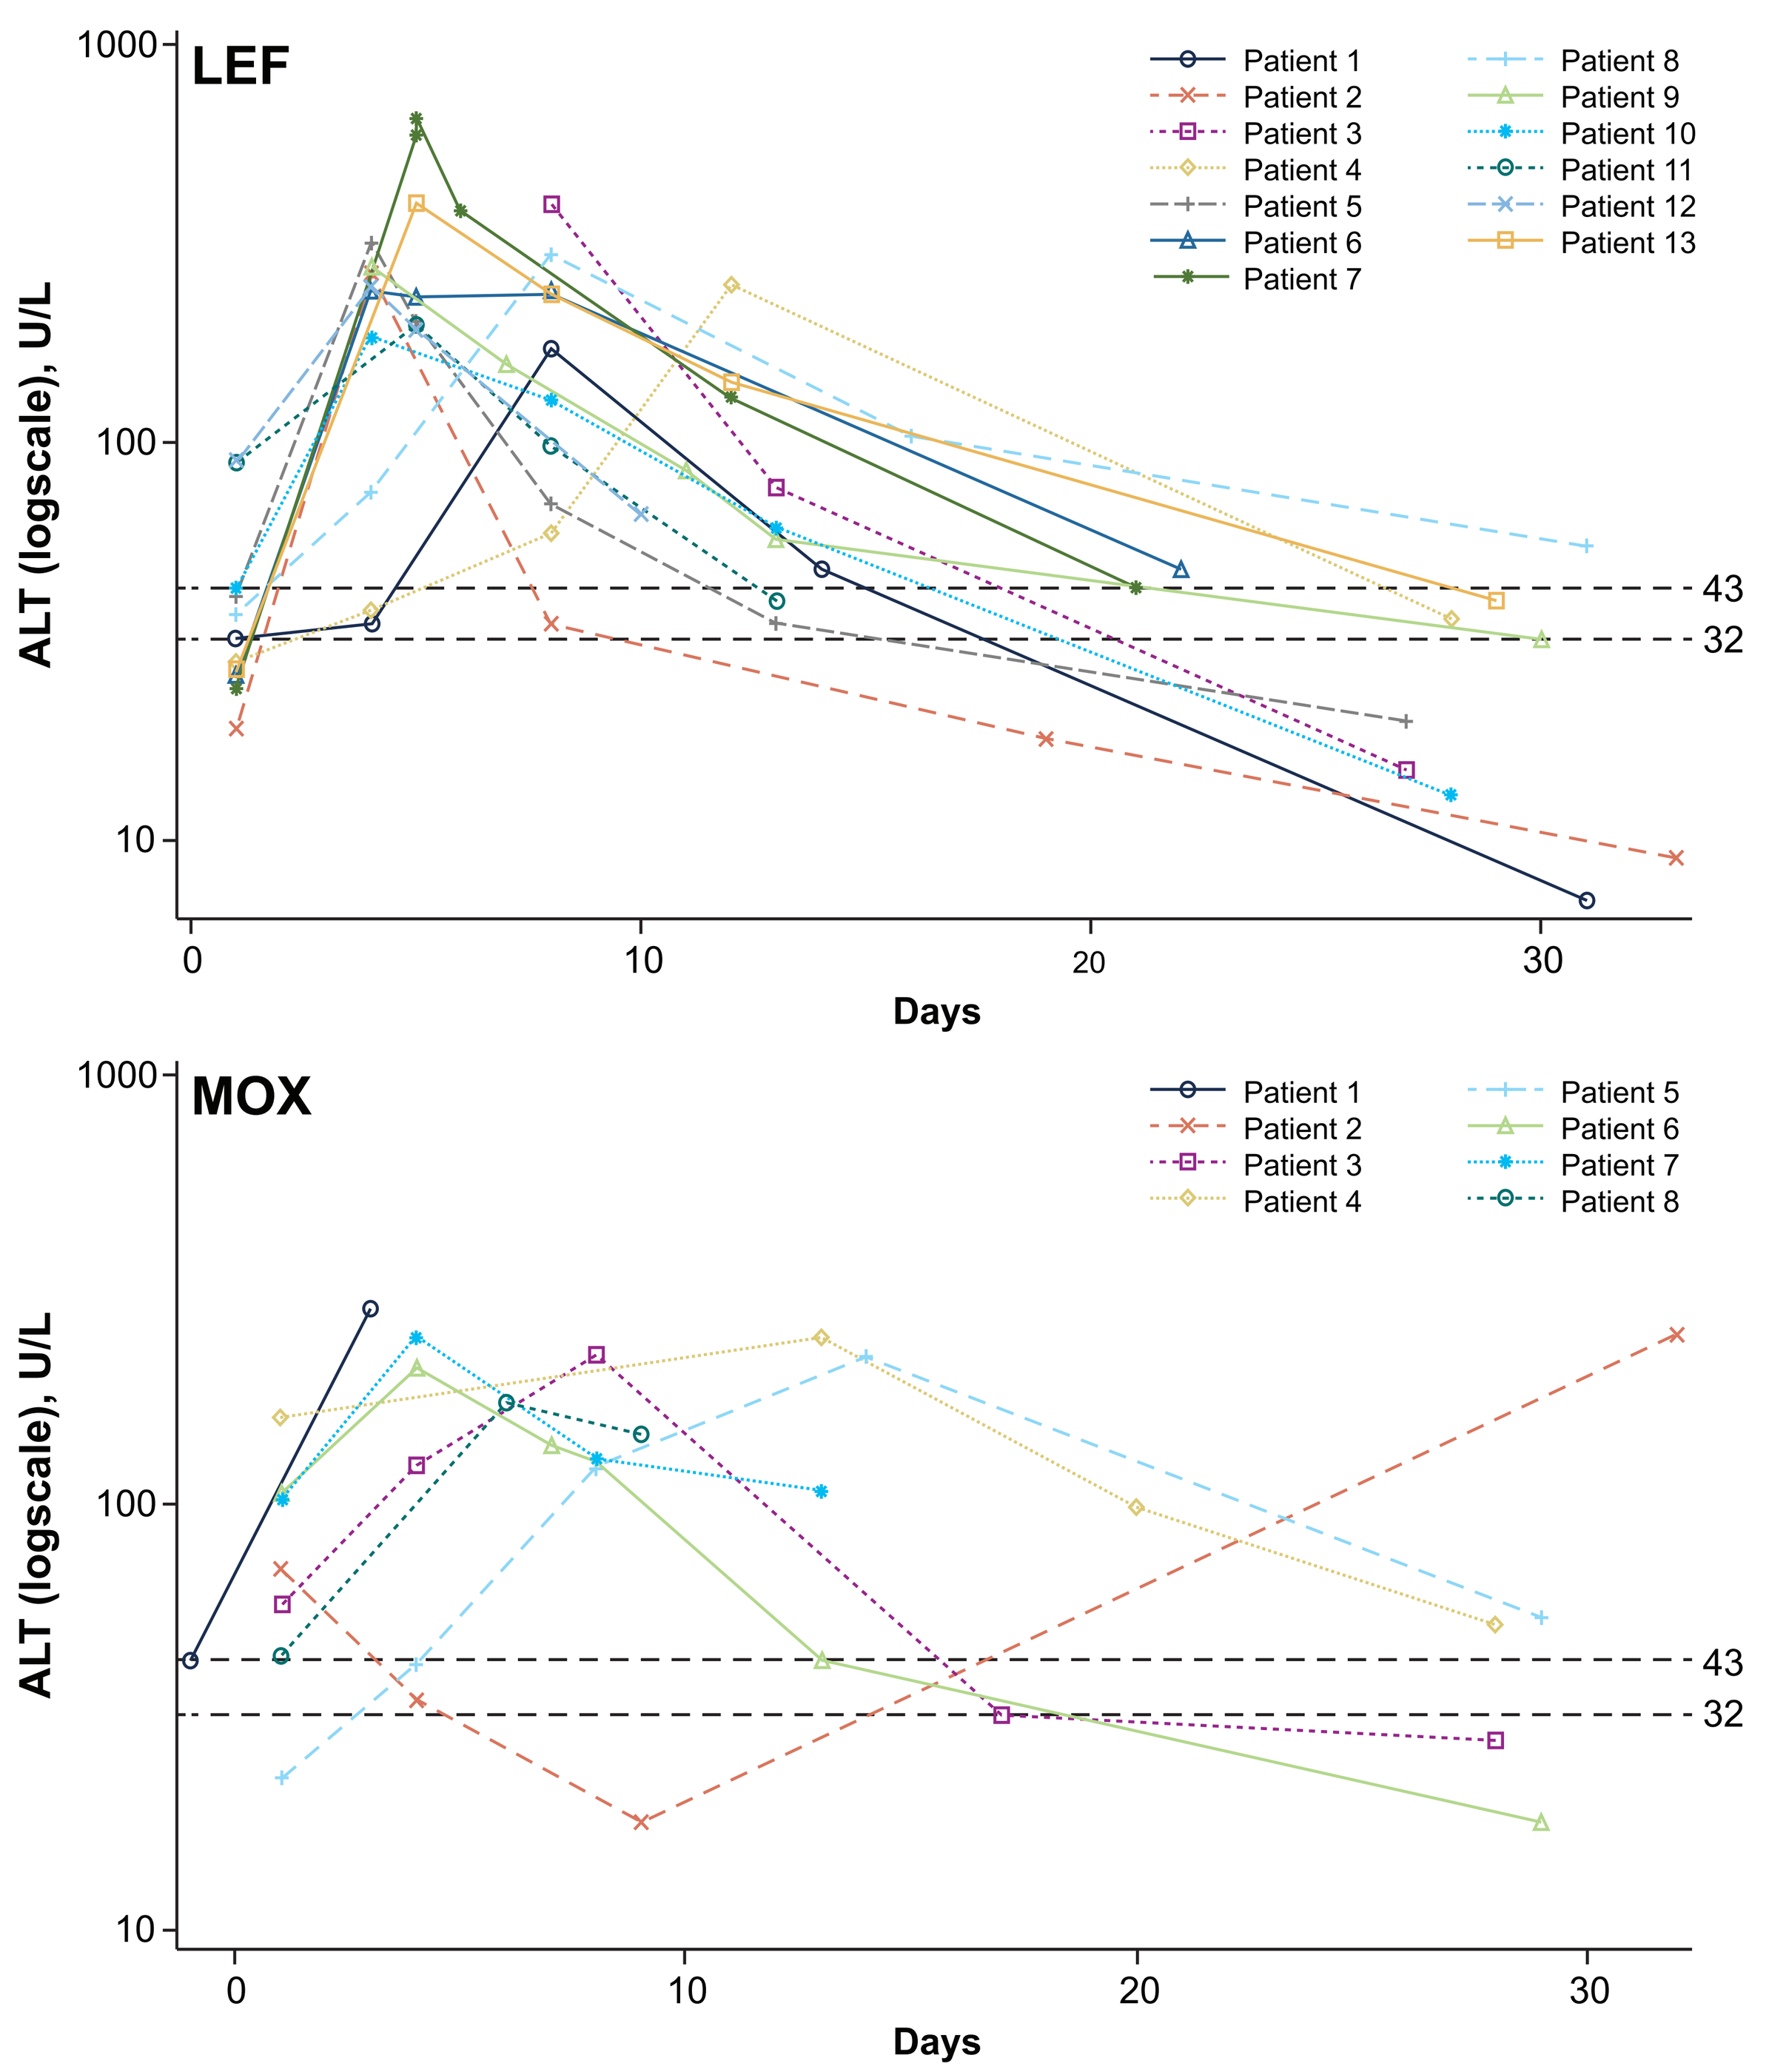
LEF
MOX
ALT (logscale), U/L
1000
100
10
0
10
20
30
Days
43
32
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5
Patient 6
Patient 7
Patient 8
Patient 9
Patient 10
Patient 11
Patient 12
Patient 13

ALT=alanine aminotransferase; LEF=lefamulin; MOX=moxifloxacin; ULN=upper limit of normal.
Horizontal dashed lines represent the range of the central laboratory ULN, which varied by patient, depending on age and sex.

Table 4. Hepatobiliary Disorder TEAEs (Pooled Safety Analysis Set)

System Organ Class Preferred Term	LEF (*n*=641)	MOX (*n*=641)
Hepatobiliary disorders	6 (0.9)*	6 (0.9)*
Cholecystitis, chronic	1 (0.2)	1 (0.2)
Cholelithiasis	1 (0.2)	1 (0.2)
Hepatic steatosis	1 (0.2)	1 (0.2)
Cholecystitis	1 (0.2)	0
Drug-induced liver injury	1 (0.2)	0
Hepatic cyst	1 (0.2)	0
Hepatitis, toxic	1 (0.2)	0
Liver disorder	1 (0.2)	0
Steatohepatitis	1 (0.2)	0
Cholecystitis, acute	0	1 (0.2)
Hypertransaminasemia	0	1 (0.2)
Nonalcoholic fatty liver	0	1 (0.2)

LEF=lefamulin; MOX=moxifloxacin; TEAE=treatment-emergent adverse event.
*Although a patient may have had ≥2 TEAEs, a patient was counted only once within a system organ class category and within a preferred term category.

CONCLUSIONS

- Low incidences of hepatic enzyme elevations and TEAEs were observed in these 2 pivotal phase 3 trials of adults with CABP, with no apparent differences between LEF and MOX
- Expert review of probable related cases suggested a clinical signature of rapid and reversible onset in patients who were asymptomatic, with no evidence of hypersensitivity and a general hepatocellular pattern
- These results suggest a favorable benefit-risk profile for LEF, given its high efficacy in phase 3 trials and low incidence of reversible, nonsevere transaminase elevation
- IV or oral LEF may offer an important empiric systemic treatment option for patients with CABP

REFERENCES

(1) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(2) Eyal Z, et al. *Sci Rep.* 2016;6:39004.
(3) Schlünzen F, et al. *Mol Microbiol.* 2004;54(5):1287-1294.
(4) Waites KB, et al. *Antimicrob Agents Chemother.* 2017;61(2):e02008-16.
(5) Paukner S, et al. *Antimicrob Agents Chemother.* 2019;63(4):e02161-18.
(6) Avelox® (moxifloxacin hydrochloride). Full Prescribing Information, Bayer HealthCare Pharmaceuticals Inc., Whippany, NJ, 2016.
(7) Teflaro® (ceftaroline fosamil). Full Prescribing Information, Allergan USA, Madison, NJ, 2018.
(8) Zithromax® (azithromycin dihydrate). Full Prescribing Information, Pfizer, New York, NY, 2019.
(9) Nuzyra® (omadacycline). Full Prescribing Information, Paratek Pharmaceuticals, Inc., Boston, MA, 2019.
(10) Nabriva Therapeutics GmbH. Data on file. Vienna, Austria. 2019.
(11) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(12) Alexander E, et al. *JAMA.* 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].
(13) National Cancer Institute. Common terminology criteria for adverse events (CTCAE) v5.0. National Institutes of Health. Available at: https://ctep.cancer.gov/protocolDevelopment/electronic_applications/ctc.htm#ctc_50. Accessed June 12, 2019.

Acknowledgments

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Daniel S. Stein, Steven P. Gelone, and Jennifer Schranz are employees of/stockholders in Nabriva Therapeutics plc. James H. Lewis served as a consultant for Nabriva Therapeutics to perform the hepatobiliary safety analyses described here. Anita F. Das served as a consultant for Achaogen, AntibioTx, Boston Pharmaceuticals, Cempra, ContraFect, IterumTx, Nabriva Therapeutics, Paratek, Tetraphase, Theravance, UTILITY, Wockhardt, and Zavante.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_LEAP1_2_Hepatobiliary

IDWeek 2019: October 2–6, Washington, DC, USA