2233

Efficacy and Symptom Resolution by Visit in Adults With Community-Acquired Bacterial Pneumonia Treated With Lefamulin or Moxifloxacin: Pooled Analysis of Lefamulin Evaluation Against Pneumonia (LEAP) 1 and LEAP 2 Study Results

Jennifer Schranz,[1] Anita F. Das,[2] Elizabeth Alexander,[1] Gregory J. Moran,[3] Christian Sandrock,[4] Thomas M. File Jr,[5] Steven P. Gelone[1]

[1]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA; [2]Das Consulting, Guerneville, CA, USA; [3]Olive View-UCLA Medical Center, Los Angeles, CA, USA; [4]UC Davis School of Medicine, Sacramento, CA, USA; [5]Summa Health, Akron, OH, USA

Presented by
Elizabeth Alexander, MD, MSc
Email: Elizabeth.Alexander@Nabriva.com
Phone: 610-981-2840
Fax: 610-816-6639

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION & PURPOSE

- Pneumonia is associated with substantial morbidity and mortality and is among the leading causes of hospitalizations and infection-related deaths in the United States[1,2]
- Increasing bacterial resistance and growing safety concerns associated with some of the most frequently used community-acquired bacterial pneumonia (CABP) therapies has created a need for new safe and effective treatment options[1,3,4]
- Lefamulin (LEF), a first-in-class pleuromutilin antibiotic approved for intravenous (IV) and oral use in adults with CABP,[5] selectively inhibits bacterial protein synthesis by binding to the 50S ribosomal subunit at the A- and P-sites in the peptidyl transferase center[6,7]
- In patients with CABP, LEF demonstrated noninferiority to moxifloxacin (MOX) in the IV-to-oral switch Lefamulin Evaluation Against Pneumonia (LEAP) 1 phase 3 study,[8] and in the LEAP 2 oral-only phase 3 study[9]
- The objective of this analysis was to examine efficacy outcomes by study visit using pooled data from the LEAP 1 and LEAP 2 clinical trials

METHODS

Study Design

- Both studies were prospective, randomized, double-blind, double-dummy, phase 3 trials **(Figure 1)**[8,9]
 - In LEAP 1, patients were randomized to receive LEF 150 mg IV every 12 hours (q12h) for 5–7 days or MOX 400 mg IV every 24 hours (q24h) for 7 days
 - Patients could switch to oral therapy (LEF 600 mg q12h or MOX 400 mg q24h) after 6 IV doses of study drug (~3 days) if predefined improvement criteria were met
 - In LEAP 2, patients were randomized to receive oral LEF 600 mg q12h for 5 days or oral MOX 400 mg q24h for 7 days

Figure 1. LEAP 1 and LEAP 2 Study Design

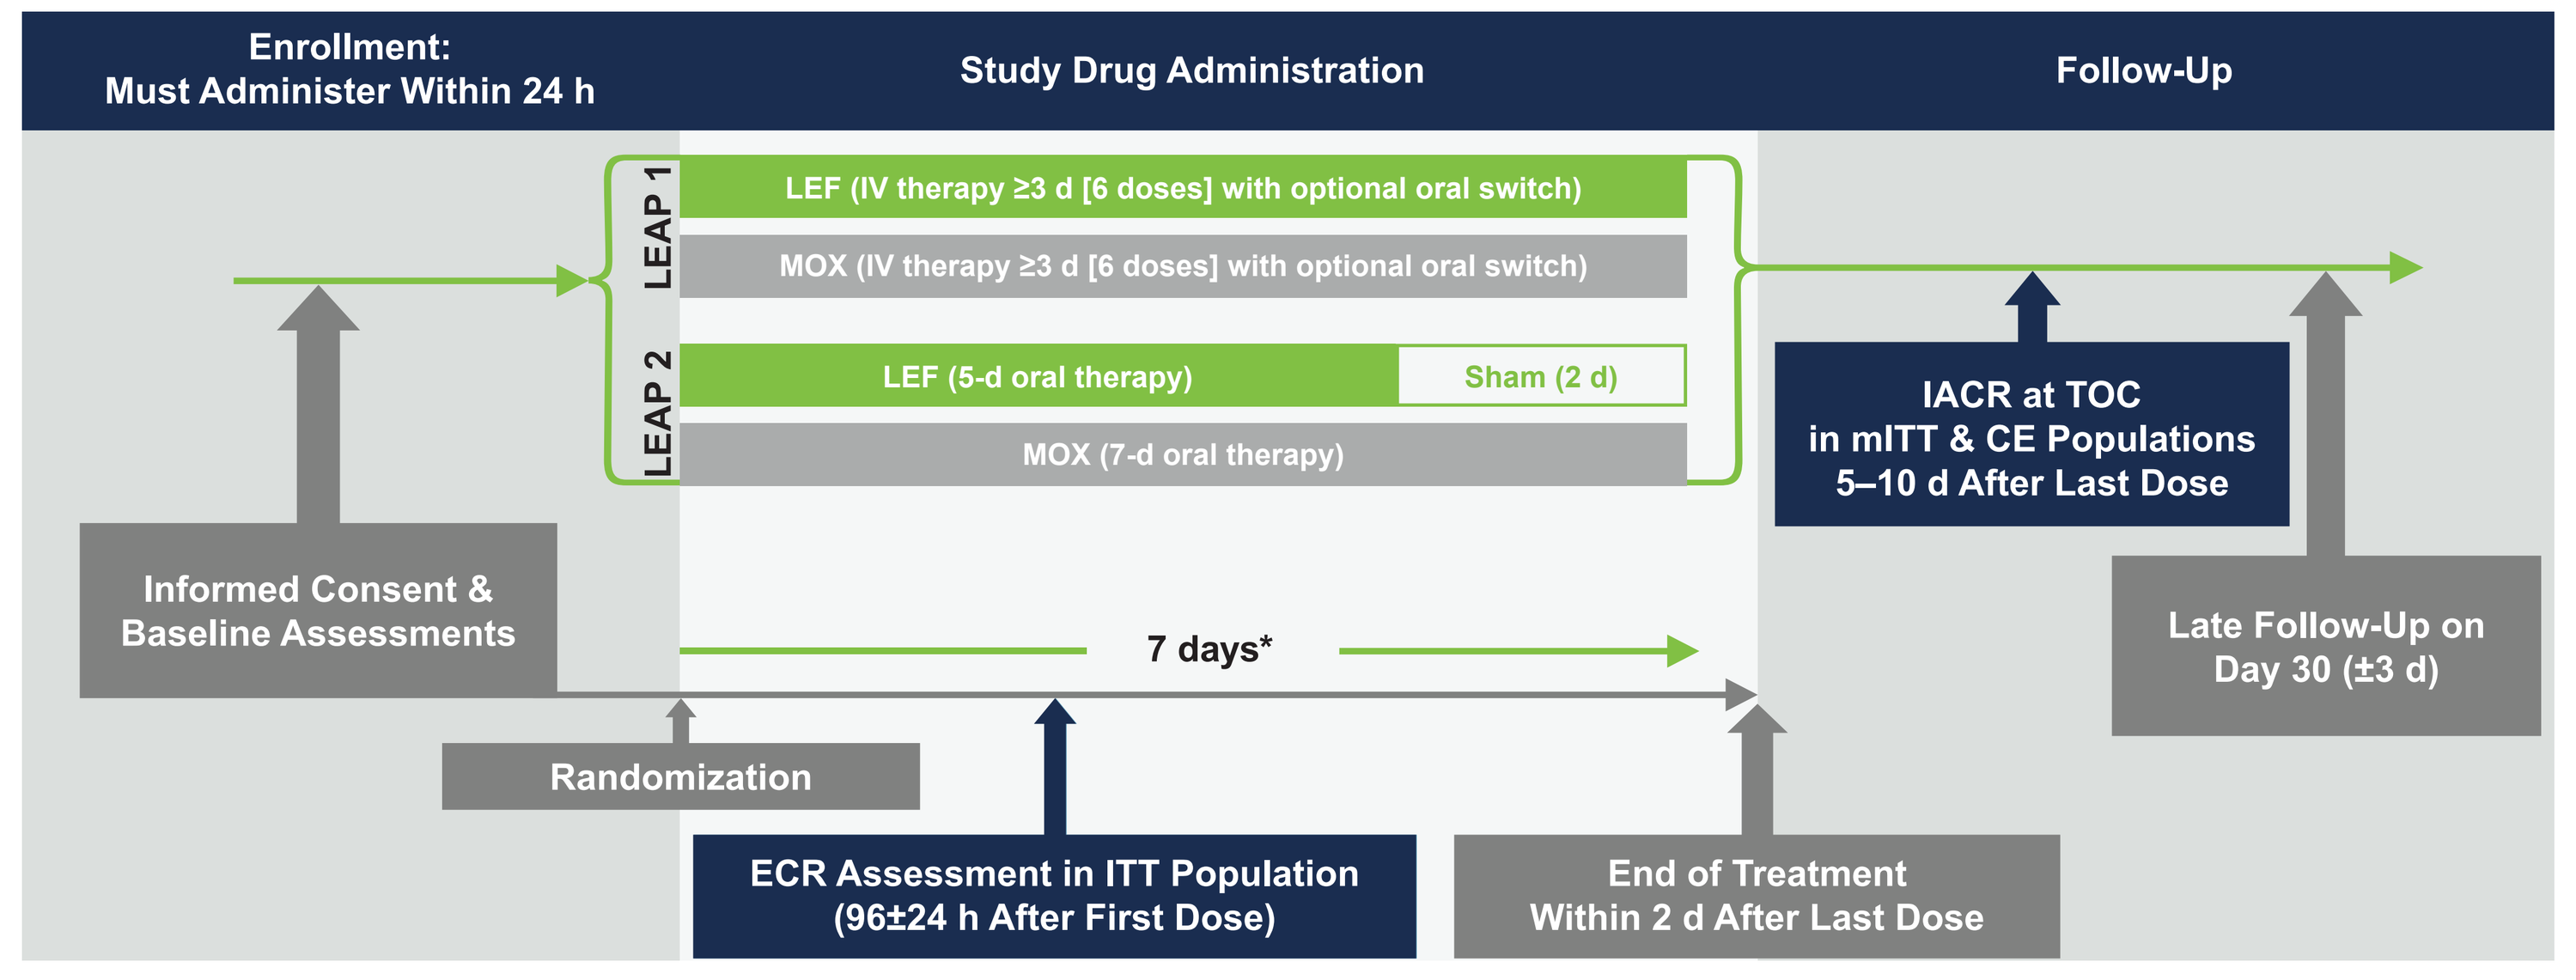

Enrollment: Must Administer Within 24 h
Study Drug Administration
Follow-Up
LEAP 1
LEF (IV therapy ≥3 d [6 doses] with optional oral switch)
MOX (IV therapy ≥3 d [6 doses] with optional oral switch)
LEAP 2
LEF (5-d oral therapy)
Sham (2 d)
MOX (7-d oral therapy)
IACR at TOC in mITT & CE Populations 5–10 d After Last Dose
Informed Consent & Baseline Assessments
7 days*
Late Follow-Up on Day 30 (±3 d)
Randomization
ECR Assessment in ITT Population (96±24 h After First Dose)
End of Treatment Within 2 d After Last Dose

CABP=community-acquired bacterial pneumonia; CE=clinically evaluable (patients who met predefined specified criteria related to protocol adherence); ECR=early clinical response (patient assessed as responder if alive, showed improvement in ≥2 CABP signs and symptoms, no worsening in any CABP sign or symptom, and no receipt of a concomitant nonstudy antibiotic for the current CABP episode); IACR=investigator assessment of clinical response (patients assessed as success if alive, with signs and symptoms of CABP resolved or improved such that no additional antibacterial therapy was administered for CABP); ITT=intent to treat (all randomized patients); IV=intravenous; LEAP=Lefamulin Evaluation Against Pneumonia; LEF=lefamulin; mITT=modified ITT (all randomized patients who received any amount of study drug); MOX=moxifloxacin; TOC=test of cure visit.

*In LEAP 1, the original protocol indicated a LEF treatment period of 5 days (but 10 days in patients with CABP due to *Legionella pneumophila* or methicillin-resistant *Staphylococcus aureus* [MRSA] or in patients with *Streptococcus pneumoniae* and bacteremia); however, this was later adjusted to 7 days (except in cases of confirmed MRSA, which continued to receive 10 days of treatment) to reduce medication errors and limit the burden on study sites.[8] If MRSA was suspected, linezolid or linezolid placebo was added to MOX or LEF therapy, respectively. A total of 14/275 (5.1%) patients randomized to MOX and 9/276 (3.3%) patients randomized to LEF received linezolid and linezolid placebo, respectively, because of suspected MRSA at baseline.

Patients and Assessments

- Patients ≥18 years old with CABP of Pneumonia Outcomes Research Team (PORT) risk class III–V or II–IV were eligible for LEAP 1 and LEAP 2, respectively
- Patients were included if they had the following signs and symptoms as well as radiographically-diagnosed pneumonia:
 - Acute onset of ≥3 signs/symptoms: dyspnea, new or increased cough, purulent sputum production, or chest pain due to pneumonia
 - ≥2 vital sign abnormalities: hypotension, tachycardia, or tachypnea
 - ≥1 other clinical sign or laboratory finding of CABP: hypoxemia, auscultatory/percussion findings consistent with pneumonia, white blood cell (WBC) count >10,000 cells/mm³ or <4500 cells/mm³ or >15% immature neutrophils (bands) regardless of total WBC count
- In both studies, the primary efficacy endpoint for the US Food and Drug Administration (FDA) was early clinical response (ECR) at 96±24 hours after first dose of study drug in the intent-to-treat (ITT) population
 - In this pooled analysis, ECR criteria were applied to each study visit through late follow-up (LFU; days 27–34) in the ITT population

METHODS (continued)

- The European Medicines Agency coprimary endpoints (FDA secondary endpoints) were investigator assessment of clinical response (IACR) at the test-of-cure (TOC) assessment 5–10 days after the last dose of study drug in the modified ITT (mITT) and clinically evaluable (CE) populations (see **Figure 1** footnote for study population definitions)
 - In this pooled analysis, IACR was examined at end of treatment (EOT; within 2 d after last dose), TOC, and LFU in the mITT and CE populations
- CABP clinical signs and symptoms (dyspnea, cough, production of purulent sputum, chest pain) were assessed at baseline and by study visit through LFU

RESULTS

Demographics and Baseline Characteristics

- 1289 patients were randomized to LEF (*n*=646) and MOX (*n*=643) and included in the ITT population
- In both individual LEAP studies and the pooled analysis, patient demographics and disease characteristics were generally well balanced between treatment groups **(Table 1)**
 - Overall, patients in the pooled analysis were predominantly male (55.6%) and white (79.3%), with a mean (SD) age of 58.7 (16.1)
 - Approximately 51% of patients had impaired renal function and approximately 71% had a PORT risk class of ≥III

Table 1. Demographics and Baseline Characteristics (Pooled ITT Population)

Parameter	LEF (*n*=646)	MOX (*n*=643)	Overall (*N*=1289)
Age, y, mean (SD)	58.9 (16.5)	58.5 (15.7)	58.7 (16.1)
Male, *n* (%)	377 (58.4)	340 (52.9)	717 (55.6)
White, *n* (%)	513 (79.4)	509 (79.2)	1022 (79.3)
Renal status,* *n* (%)			
Normal function	311 (48.1)	312 (48.5)	623 (48.3)
Mild impairment	201 (31.1)	192 (29.9)	393 (30.5)
Moderate impairment	125 (19.3)	132 (20.5)	257 (19.9)
Severe impairment	7 (1.1)	6 (0.9)	13 (1.0)
History of COPD/asthma, *n* (%)	119 (18.4)	113 (17.6)	232 (18.0)
Prior antibiotic use,† *n* (%)	147 (22.8)	145 (22.6)	292 (22.7)
Smoking history, *n* (%)	284 (44.0)	242 (37.6)	526 (40.8)
PORT risk class,‡ *n* (%)			
I/II	184 (28.5)	192 (29.9)	376 (29.2)
III	341 (52.8)	334 (51.9)	675 (52.4)
IV/V	121 (18.7)	117 (18.2)	238 (18.5)
CURB-65 score,§ *n* (%)			
0–2	610 (94.4)	604 (93.9)	1214 (94.2)
3–5	36 (5.6)	39 (6.1)	75 (5.8)
Minor ATS severity criteria,‖ *n* (%)	85 (13.2)	85 (13.2)	170 (13.2)
Modified ATS severity criteria,¶ *n* (%)	53 (8.2)	57 (8.9)	110 (8.5)
SIRS,^ *n* (%)	621 (96.1)	609 (94.7)	1230 (95.4)
Bacteremia, *n* (%)	13 (2.0)	12 (1.9)	25 (1.9)
CABP clinical signs and symptoms, *n* (%)			
Dyspnea	629 (97.4)	625 (97.2)	1254 (97.3)
Cough	645 (99.8)	643 (100)	1288 (99.9)
Purulent sputum production	568 (87.9)	576 (89.6)	1144 (88.8)
Chest pain	551 (85.3)	550 (85.5)	1101 (85.4)

ATS=American Thoracic Society; BMI=body mass index; BUN=blood urea nitrogen; CABP=community-acquired bacterial pneumonia; COPD=chronic obstructive pulmonary disease; CrCl=creatinine clearance; eCRF=electronic case report form; ITT=intent to treat; LEF=lefamulin; MOX=moxifloxacin; PORT=Pneumonia Outcomes Research Team; SIRS=Systemic Inflammatory Response Syndrome; WBC=white blood cell (count).

*National Kidney Foundation categories of renal impairment[10] based on baseline central laboratory serum creatinine. When baseline central laboratory serum creatinine was not available, local serum creatinine results were used. Renal impairment categories are: normal [CrCl ≥90 mL/min], mild [CrCl of 60 to <90 mL/min], moderate [CrCl of 30 to <60 mL/min], and severe [CrCl <30 mL/min].

†Patients received a single dose of short-acting systemic antibacterial medication within 72 hours before randomization; randomization was stratified and capped such that no more than 25% of the total ITT population met these criteria.

‡PORT risk class was calculated programmatically using data obtained at the site and reported in the eCRF and was not always consistent with the site-reported PORT risk class used for enrollment/stratification.

§Defined as confusion of new onset, BUN >19 mg/dL, respiratory rate ≥30 breaths/min, systolic blood pressure <90 mm Hg or diastolic blood pressure ≤60 mm Hg, and age ≥65 years.

‖Defined as presence of ≥3 of the following 9 criteria at baseline: respiratory rate ≥30 breaths/min, O_2 saturation <90% or PaO_2 <60 mm Hg, BUN ≥20 mg/dL, WBC <4000 cells/mm³, confusion, multilobar infiltrates, platelets <100,000 cells/mm³, temperature <36°C, or systolic blood pressure <90 mm Hg.[11]

¶Defined as presence of ≥3 of the following 6 criteria at baseline: respiratory rate ≥30 breaths/min, SpO_2/FiO_2 <274 where SpO_2/FiO_2 = 64+0.84 (PaO_2/FiO_2), BUN ≥20 mg/dL, confusion, age ≥65 years, or multilobar infiltrates.[12]

^Defined as having ≥2 of the following 4 criteria at baseline: temperature <36°C or >38°C; heart rate >90 bpm; respiratory rate >20 breaths/min; and WBC <4000 cells/mm³, WBC >12,000 cells/mm³, or immature polymorphonuclear neutrophils >10%.

RESULTS (continued)

Early Clinical Response

- The majority of patients in the LEF and MOX treatment groups met ECR criteria at Day 3, with >80% of patients in both groups meeting ECR criteria by Day 4; further increases through Day 7 and sustained efficacy through LFU were observed **(Figure 2)**

Figure 2. Patients Meeting Criteria for Early Clinical Response by Visit (Pooled ITT Population)

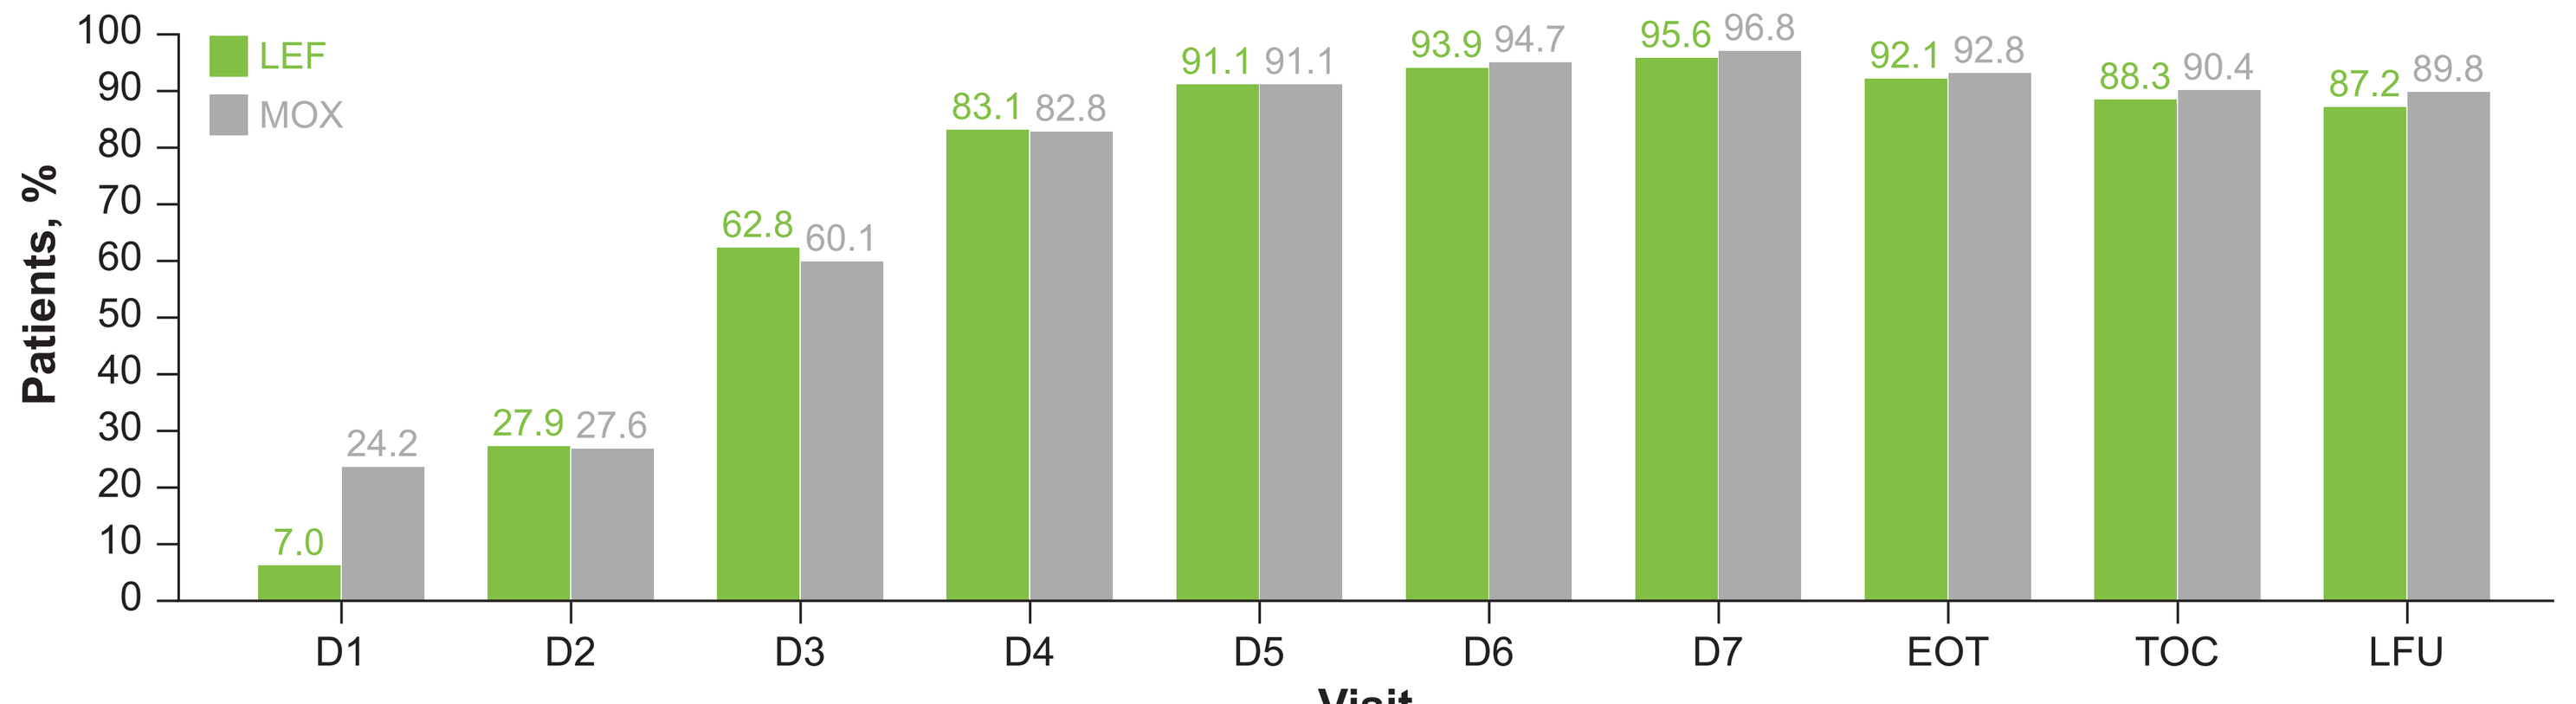

LEF
MOX
Patients, %
7.0 24.2
27.9 27.6
62.8 60.1
83.1 82.8
91.1 91.1
93.9 94.7
95.6 96.8
92.1 92.8
88.3 90.4
87.2 89.8
D1 D2 D3 D4 D5 D6 D7 EOT TOC LFU
Visit

Number of patients evaluated

	D1	D2	D3	D4	D5	D6	D7	EOT	TOC	LFU
LEF, *n/N*=	3/43	176/630	393/626	516/621	564/619	567/604	576/601	579/629	552/625	537/616
MOX, *n/N*=	8/33	175/634	379/631	515/622	561/616	577/609	578/597	582/627	562/622	555/618

EOT=end of treatment; ITT=intent to treat; LEF=lefamulin: LFU=late follow-up; MOX=moxifloxacin; TOC=test of cure.

Investigator Assessment of Clinical Response

- IACR success rates at TOC, as well as at EOT and LFU, in the mITT and CE populations were high (>83%) and similar between LEF and MOX treatment groups (**Figures 3A** and **3B**)

Figure 3. Pooled Investigator Assessment of Clinical Response Success Rates

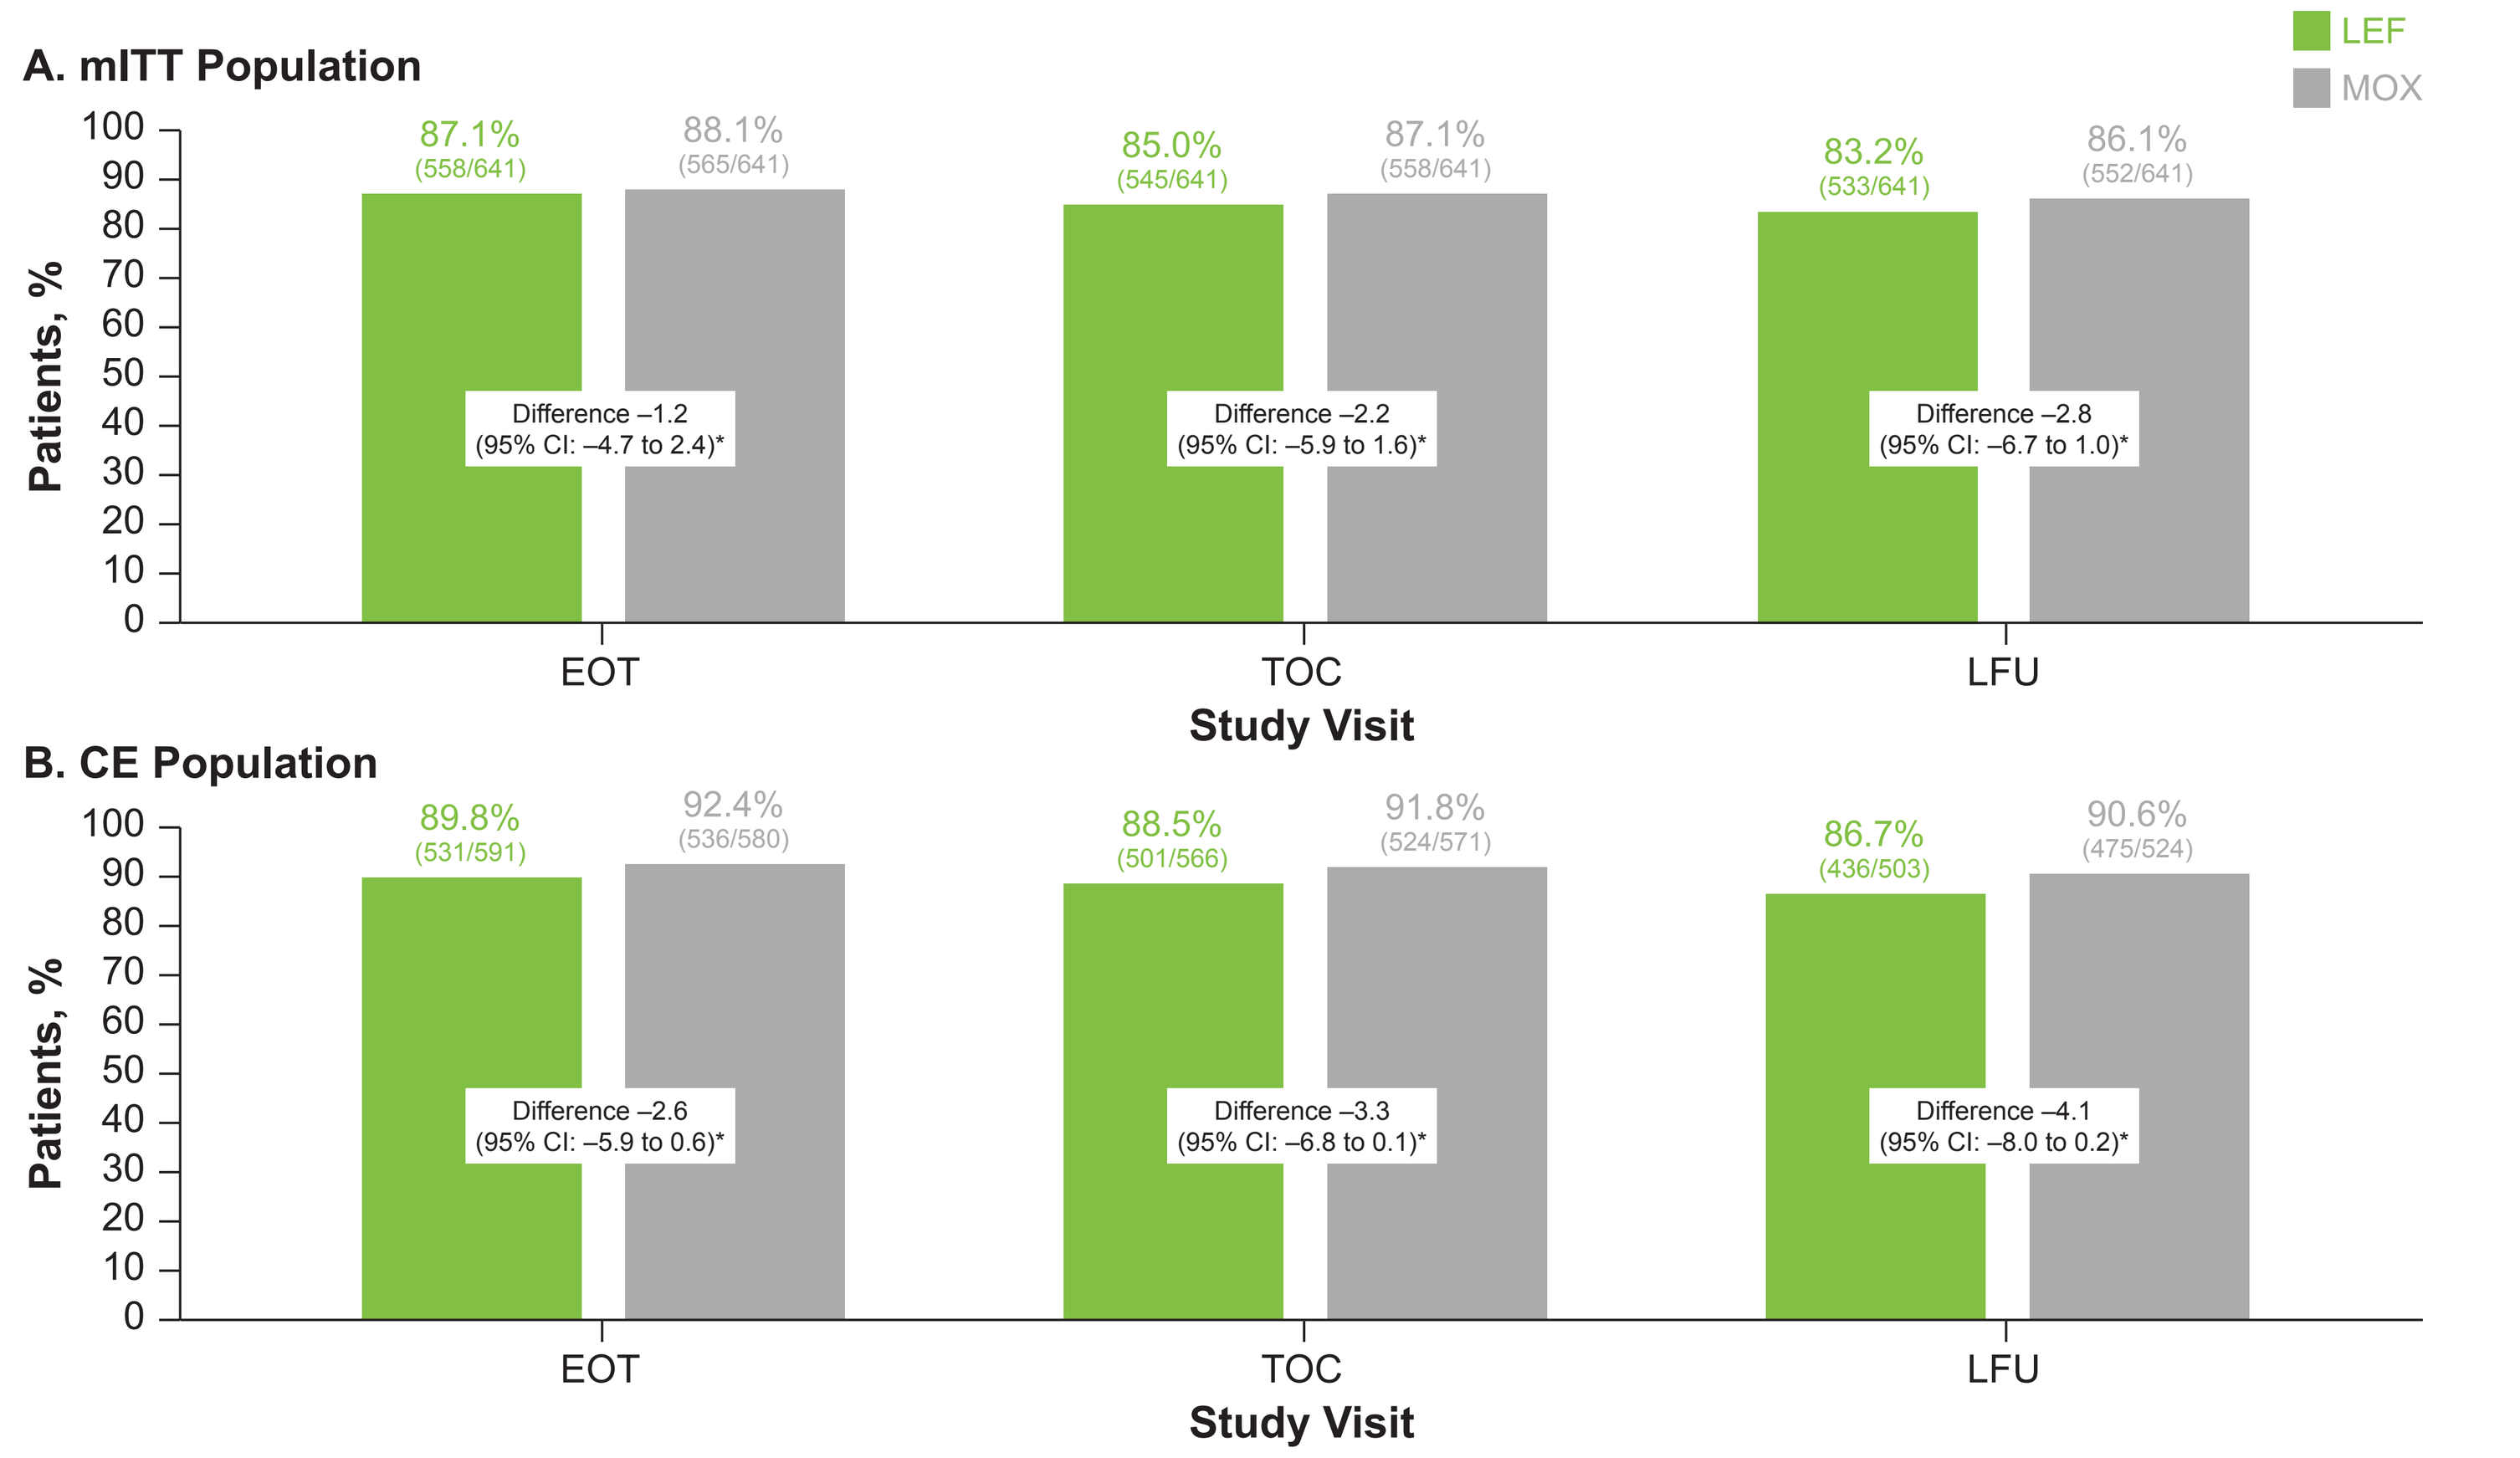

LEF
MOX
A. mITT Population
Patients, %
87.1% (558/641)
88.1% (565/641)
Difference –1.2 (95% CI: –4.7 to 2.4)*
85.0% (545/641)
87.1% (558/641)
Difference –2.2 (95% CI: –5.9 to 1.6)*
83.2% (533/641)
86.1% (552/641)
Difference –2.8 (95% CI: –6.7 to 1.0)*
EOT TOC LFU
Study Visit
B. CE Population
89.8% (531/591)
92.4% (536/580)
Difference –2.6 (95% CI: –5.9 to 0.6)*
88.5% (501/566)
91.8% (524/571)
Difference –3.3 (95% CI: –6.8 to 0.1)*
86.7% (436/503)
90.6% (475/524)
Difference –4.1 (95% CI: –8.0 to 0.2)*
EOT TOC LFU
Study Visit

CE=clinically evaluable; CI=confidence interval; EOT=end of treatment; LEF=lefamulin; LFU=late follow-up; mITT=modified intent to treat; MOX=moxifloxacin; TOC=test of cure.

*Computed using the method of Miettinen and Nurminen, adjusted for study (all analyses) and receipt of a prior single-dose short-acting antibiotic (analyses at TOC only), with the inverse variance of the effect size as the stratum weights.

Symptom Resolution

- As was seen with ECR rates by visit, the proportions of ITT patients with resolution of all baseline signs and symptoms of CABP increased by visit in both treatment groups **(Figure 4)**
- Most patients did not achieve complete sign and symptom resolution until TOC or later **(Figure 4)**, which is reflective of the known lag between antimicrobial treatment and complete symptom resolution, with fever generally being the first and cough the last CABP symptom to resolve[13]

Figure 4. Patients With Resolution of All Baseline Signs/Symptoms* of CABP by Visit (Pooled ITT Population)

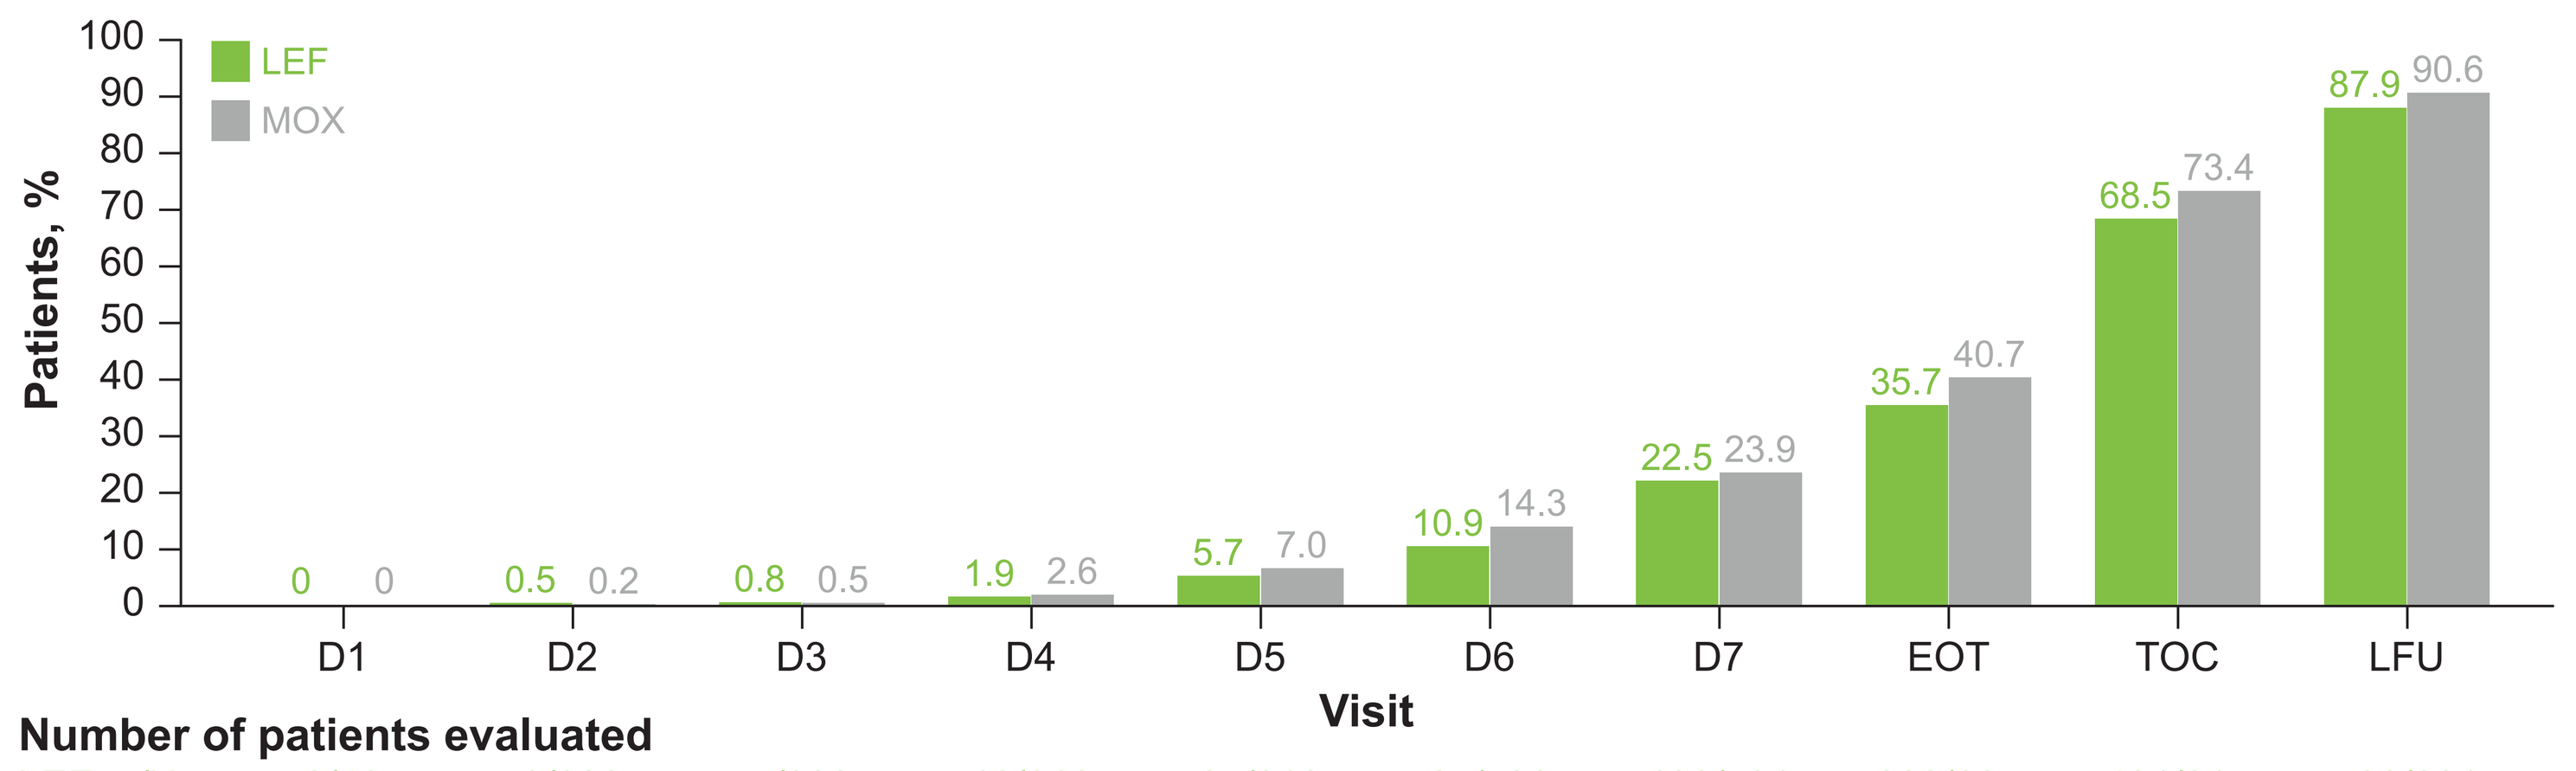

LEF
MOX
Patients, %
0 0
0.5 0.2
0.8 0.5
1.9 2.6
5.7 7.0
10.9 14.3
22.5 23.9
35.7 40.7
68.5 73.4
87.9 90.6
D1 D2 D3 D4 D5 D6 D7 EOT TOC LFU
Visit

Number of patients evaluated

	D1	D2	D3	D4	D5	D6	D7	EOT	TOC	LFU
LEF, *n/N*=	0/42	3/628	5/623	12/616	35/612	65/596	133/591	223/625	421/615	528/601
MOX, *n/N*=	0/32	1/633	3/630	16/616	43/611	86/603	141/590	254/624	453/617	550/607

EOT=end of treatment; ITT=intent to treat; LEF=lefamulin; LFU=late follow-up; MOX=moxifloxacin; TOC=test of cure.
*Includes cough, dyspnea, purulent sputum production, and chest pain.

CONCLUSIONS

- In this pooled analysis of 2 pivotal CABP studies (LEAP 1 and LEAP 2), efficacy results (ECR, IACR, and symptom resolution) were similar in the LEF and MOX groups, with fairly rapid achievement and maintenance of clinical response (ECR and symptom resolution) through the LFU visit in the majority of patients with CABP
- Consistent with published literature,[13-15] complete resolution of all CABP clinical signs and symptoms lags noticeably behind early clinical response and cessation of an appropriate course of antimicrobial therapy, which may be attributed to the inflammatory nature of CABP and the exacerbation of preexisting conditions (eg, asthma, chronic obstructive pulmonary disease) or disability associated with smoking
- The high percentage of patients at LFU with resolution of baseline symptoms suggests that symptom resolution was sustained
- LEF may provide a valuable IV and oral monotherapy alternative to fluoroquinolones or macrolides for empiric treatment of CABP in adults

REFERENCES

(1) Peyrani P, et al. *Expert Rev Respir Med.* 2019;13(2):139-152.
(2) Xu J, et al. Deaths: Final Data for 2016. US Department of Health and Human Services, Centers for Disease Control and Prevention, National Center for Health Statistics. Available at: https://www.cdc.gov/nchs/data/nvsr/nvsr67/nvsr67_05.pdf. Accessed July 17, 2019.
(3) US Food and Drug Administration. FDA drug safety communication: FDA updates warnings for oral and injectable fluoroquinolone antibiotics due to disabling side effects. Available at: https://www.fda.gov/Drugs/DrugSafety/ucm511530.htm. Accessed June 23, 2019.
(4) US Food and Drug Administration. FDA drug safety communication: FDA reinforces safety information about serious low blood sugar levels and mental health side effects with fluoroquinolone antibiotics; requires label changes. Available at: https://www.fda.gov/downloads/Drugs/DrugSafety/UCM612834.pdf. Accessed June 23, 2019.
(5) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(6) Eyal Z, et al. *Sci Rep.* 2016;6:39004.
(7) Schlünzen F, et al. *Mol Microbiol.* 2004;54(5):1287-1294.
(8) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(9) Alexander E, et al. *JAMA*. 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].
(10) Cockcroft DW and Gault MH. *Nephron.* 1976;16(1):31-41.
(11) Mandell LA, et al. *Clin Infect Dis.* 2007;44(suppl 2):S27-S72.
(12) Li HY, et al. *Medicine (Baltimore).* 2015;94(36):e1474.
(13) Wyrwich KW, et al. *Patient Relat Outcome Meas.* 2015;6:215-223.
(14) Metlay JP, et al. *Respir Med.* 1998;92(9):1137-1142.
(15) Wootton DG, et al. *Eur Respir J.* 2017;49(6):1602170.

Acknowledgments

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Jennifer Schranz, Elizabeth Alexander, and Steven P. Gelone are employees of/stockholders in Nabriva Therapeutics plc. Anita F. Das has served as a consultant for Achaogen, AntibioTx, Boston Pharmaceuticals, Cempra, ContraFect, IterumTx, Nabriva Therapeutics, Paratek, Tetraphase, Theravance, UTILITY, Wockhardt, and Zavante. Gregory J. Moran has received grants from ContraFect and Nabriva Therapeutics. Christian Sandrock has served as a consultant for Allergan and Nabriva Therapeutics, received grants from the National Institutes of Health and the Health Resources & Services Administration, and received nonfinancial support from the State of California. Thomas M. File Jr has served as a consultant for bioMérieux, Curetis, Melinta, Merck, Motif BioSciences, Nabriva Therapeutics, Paratek, Pfizer, and Shionogi Inc. and received grants from Nabriva Therapeutics.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_LEAP1_2_ECR_by-visit

IDWeek 2019: October 2–6, Washington, DC, USA