2245

Oral 5-Day Lefamulin for Outpatient Management of Pneumonia Outcomes Research Team Risk Class III/IV Community-Acquired Bacterial Pneumonia: Post Hoc Analysis of the Lefamulin Evaluation Against Pneumonia (LEAP) 2 Phase 3 Study

Jennifer Schranz,[1] Elizabeth Alexander,[1] David Fitts,[1] David Mariano,[1] Andrew Meads,[1] Christian Sandrock,[2] Gregory J. Moran,[3] Steven P. Gelone[1]

[1]Nabriva Therapeutics US, Inc., King of Prussia, PA, USA; [2]UC Davis School of Medicine, Sacramento, CA, USA; [3]Olive View-UCLA Medical Center, Los Angeles, CA, USA

Presented by
David Mariano, PharmD
Email: David.Mariano@Nabriva.com
Phone: 610-816-6659
Fax: 610-816-6639

Nabriva Therapeutics
Dublin, Ireland
www.nabriva.com

INTRODUCTION & PURPOSE

- Site-of-care decisions (eg, admission vs outpatient) in community-acquired bacterial pneumonia (CABP) management can be challenging for healthcare providers[1]
- Clinical prediction rules, such as the Pneumonia Outcomes Research Team (PORT) risk class and CURB-65 score (confusion of new onset, blood urea nitrogen >19 mg/dL, respiratory rate ≥30 breaths/min, systolic blood pressure <90 mm Hg or diastolic blood pressure ≤60 mm Hg, and age ≥65 years), were developed to classify patients with community-acquired pneumonia based on the risk of short-term mortality[2,3]
 - These tools are also used to inform hospital admission decisions, but their application can vary based on a physician's subjective assessment of an individual patient (eg, patients with PORT risk class III and/or CURB-65 score 2 can be admitted for a short hospitalization or treated as outpatients)[1-3]
- The ability to initiate appropriate empiric therapy and prevent unnecessary admission of patients with CABP could have significant economic benefits and infection control implications (eg, decreased chance of nosocomial infections and decreased exposure to second- and third-line antimicrobials)[4-6]
- Lefamulin (LEF), a first-in-class systemic pleuromutilin antibiotic approved for intravenous (IV) and oral use in adults with CABP,[7] was shown to be noninferior to moxifloxacin (MOX) based on standard early and posttreatment clinical response endpoints in 2 phase 3 clinical trials (Lefamulin Evaluation Against Pneumonia [LEAP] 1 and LEAP 2)[8,9]
- Here we describe a post hoc analysis of adults with CABP, including patients classified as PORT risk class III or IV and patients with CURB-65 scores of 2 or 3, who were managed as outpatients in the double-blind, noninferiority, phase 3, oral-only LEAP 2 study

METHODS

Study Design

- LEAP 2 was a multicenter, randomized, double-blind, double-dummy, active-controlled study that compared the efficacy and safety of oral LEF 600 mg every 12 hours for 5 days vs oral MOX 400 mg every 24 hours for 7 days in adults with PORT risk class II–IV **(Figure 1)**
- Hospitalization was not a study requirement, and site of care (eg, admission vs outpatient) was at the investigator's discretion

Figure 1. Study Design

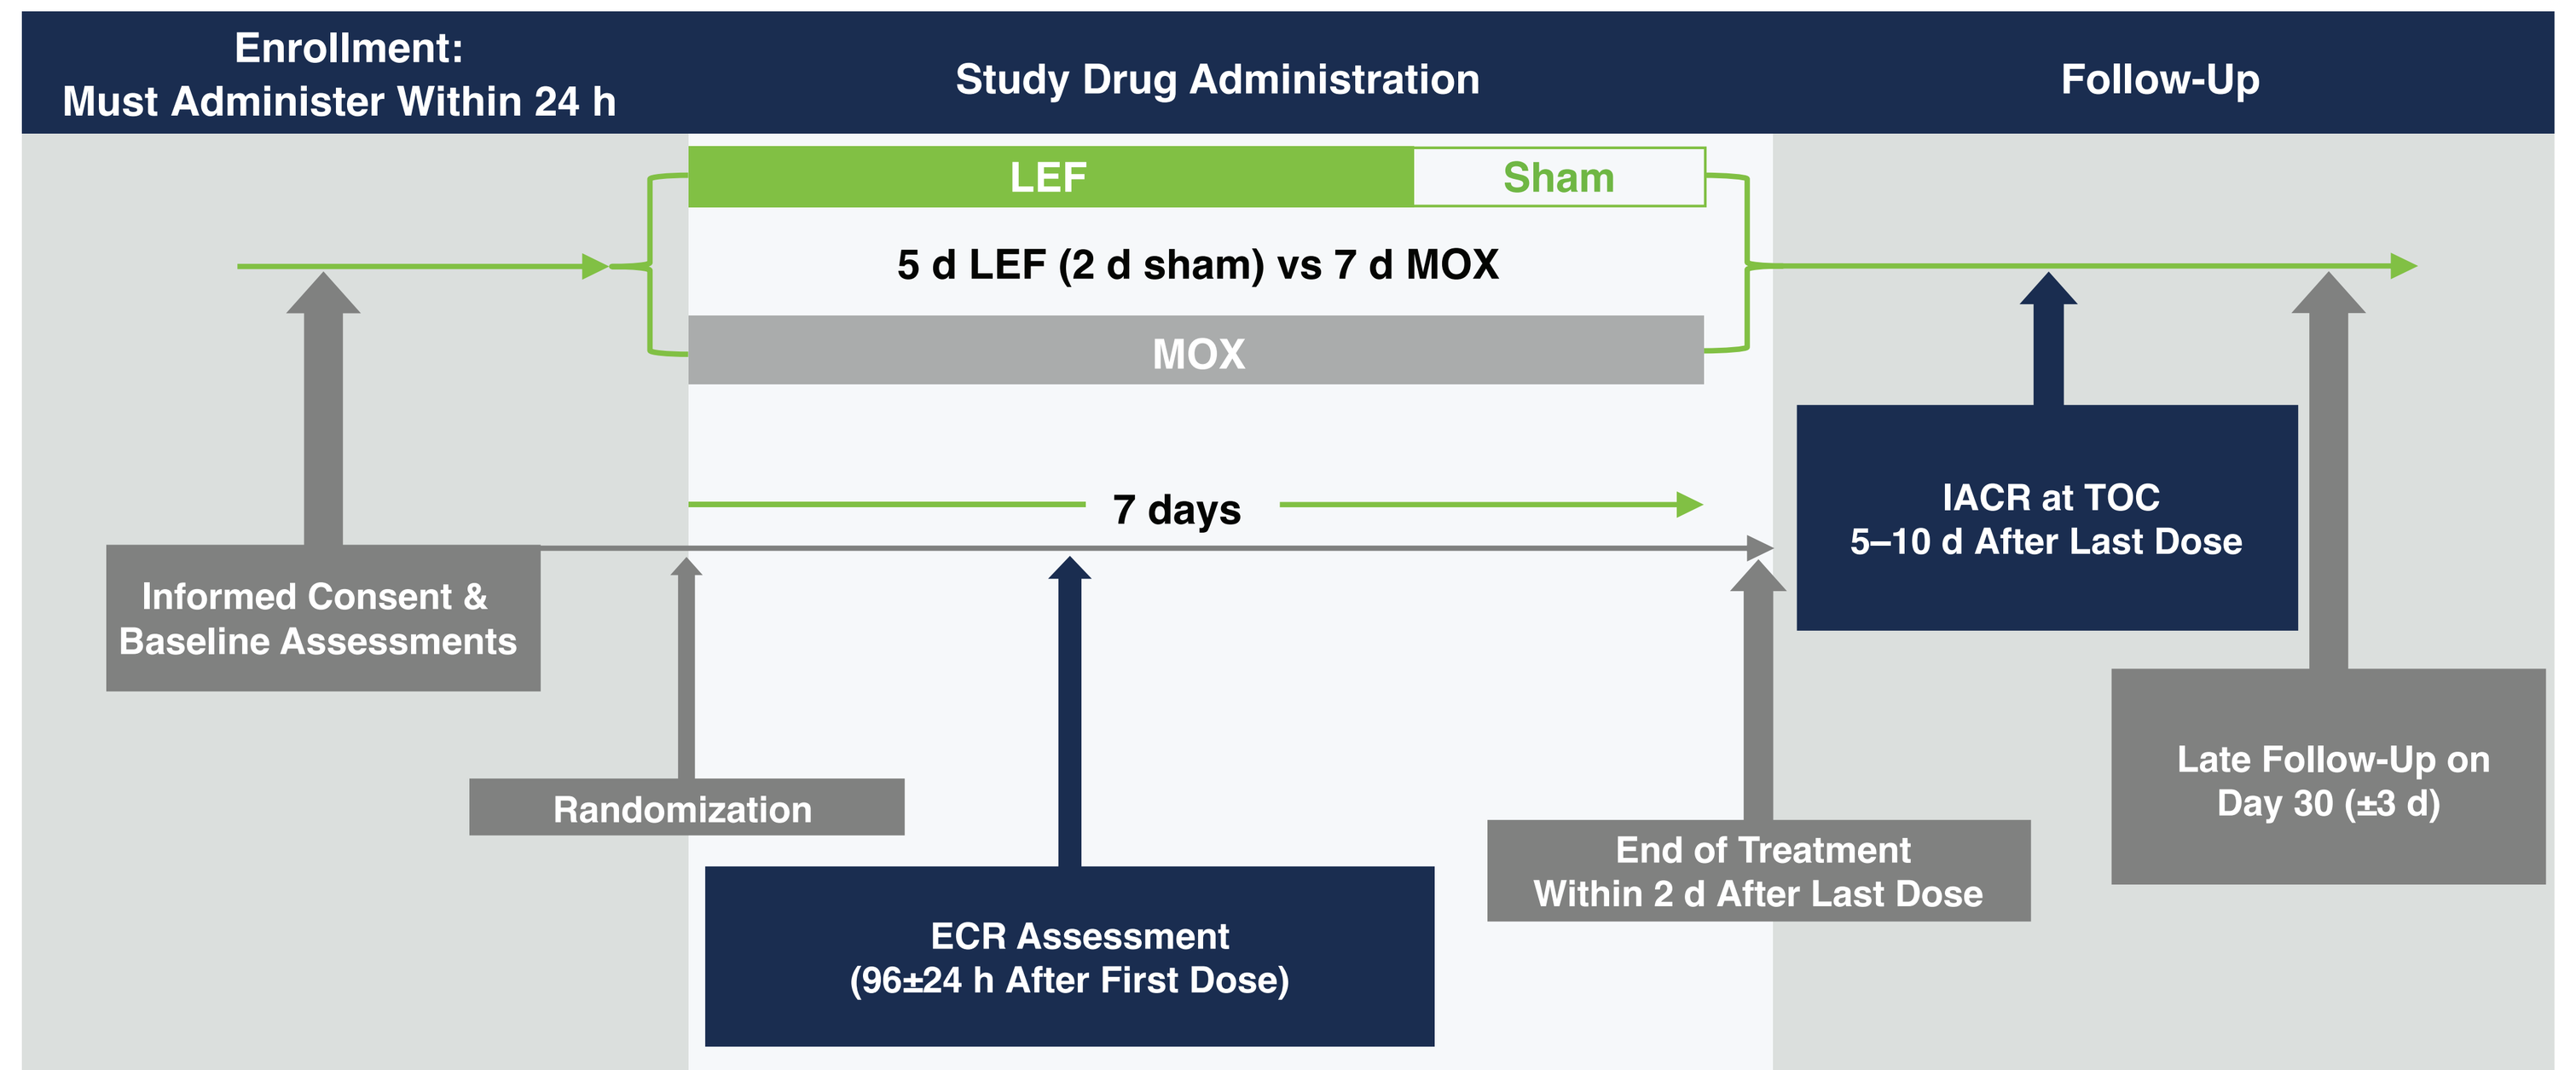
Enrollment:
Must Administer Within 24 h
Study Drug Administration
Follow-Up
LEF
Sham
5 d LEF (2 d sham) vs 7 d MOX
MOX
7 days
IACR at TOC
5–10 d After Last Dose
Informed Consent &
Baseline Assessments
Randomization
ECR Assessment
(96±24 h After First Dose)
End of Treatment
Within 2 d After Last Dose
Late Follow-Up on
Day 30 (±3 d)

ECR=early clinical response; IACR=investigator assessment of clinical response; LEF=lefamulin; MOX=moxifloxacin; TOC=test of cure.

Patients

- Key inclusion criteria: ≥18 years old with PORT risk class II–IV (≥50% of patients were required to have PORT risk class III or IV) radiographically documented pneumonia; acute illness for ≤7 days with ≥3 CABP symptoms (dyspnea, cough, sputum production, or chest pain); ≥2 vital sign abnormalities (fever or hypothermia, hypotension, tachycardia, or tachypnea); and ≥1 other clinical sign or laboratory finding of CABP (eg, hypoxemia)
- Key exclusion criteria: >1 dose of a short-acting oral or IV antibacterial for CABP within 72 hours before randomization; hospitalized for ≥2 days within 90 days before onset of symptoms; confirmed or suspected methicillin-resistant *Staphylococcus aureus*; at risk for major cardiac events or dysfunction; evidence of significant hepatic, hematologic, or immunologic disease; or severe renal impairment (estimated creatinine clearance, ≤30 mL/min)

Assessments

- The US Food and Drug Administration (FDA) primary efficacy endpoint was early clinical response (ECR) at 96±24 hours after the first dose of study drug
- The European Medicines Agency primary efficacy endpoint (FDA secondary endpoint) was the investigator assessment of clinical response (IACR); this analysis reports IACR at test of cure (TOC; 5–10 days after the last dose of study drug)
- Treatment-emergent adverse events (TEAEs) were evaluated by category and preferred term (Medical Dictionary for Regulatory Activities, version 20.0)
- For this post hoc analysis, descriptive statistics were generated to characterize demographics, baseline characteristics, efficacy, and safety outcomes in the subpopulation of outpatients in LEAP 2

RESULTS

Demographics and Baseline Characteristics

- Overall, 42% (310/736) of patients started treatment as outpatients (LEF, 41% [151/368]; MOX, 43% [159/368])
- Demographics and baseline characteristics were generally similar in both treatment groups **(Table 1)** and broadly reflective of the patient population with CABP
 - 30% (94/310) of patients were ≥65 years old (LEF, 32% [49/151]; MOX, 28% [45/159]), and approximately 15% of patients in both groups were ≥75 years old
 - 28% (87/310) of patients had a history of hypertension (LEF, 26% [39/151]; MOX, 30% [48/159]), 16% (49/310) of patients had underlying chronic obstructive pulmonary disease or asthma (14% [21/151] and 18% [28/159], respectively), and 9% (29/310) of patients had underlying diabetes (7% [11/151] and 11% [18/159])
- 49% (153/310) of patients had ≥1 baseline pathogen (LEF, 55% [83/151]; MOX, 44% [70/159])
 - 28% (23/83) and 33% (23/70), respectively, had polymicrobial infections, which were most commonly caused by a combination of gram-positive and gram-negative pathogens, predominantly *Streptococcus pneumoniae* plus *Haemophilus influenzae* and/or *Moraxella catarrhalis* (11/23 LEF patients and 15/23 MOX patients)

Table 1. Demographics and Baseline Characteristics in Outpatients

Parameter	LEF (*n*=151)	MOX (*n*=159)
Age, y, mean (SD)	55.8 (16.8)	55.7 (15.3)
Female, *n* (%)	69 (45.7)	87 (54.7)
BMI, kg/m^2, mean (SD)	25.9 (6.3)	26.6 (6.3)
PORT risk class,* *n* (%)		
I	0	1 (0.6)
II	84 (55.6)	93 (58.5)
III	51 (33.8)	49 (30.8)
IV	15 (9.9)	15 (9.4)
V	1 (0.7)	1 (0.6)
CURB-65 score,† *n* (%)		
0	34 (22.5)	42 (26.4)
1	86 (57.0)	83 (52.2)
2	28 (18.5)	28 (17.6)
3	3 (2.0)	6 (3.8)
Comorbidities, *n* (%)		
Smoking history	65 (43.0)	54 (34.0)
Hypertension	39 (25.8)	48 (30.2)
COPD or asthma	21 (13.9)	28 (17.6)
Diabetes mellitus	11 (7.3)	18 (11.3)
Moderate to severe renal impairment (CrCl <60 mL/min)	22 (14.6)	28 (17.6)
Met SIRS criteria,‡ *n* (%)	138 (91.4)	147 (92.5)
Region, *n* (%)		
European Union	31 (20.5)	34 (21.4)
Non-European Union Europe	40 (26.5)	39 (24.5)
North America	9 (6.0)	11 (6.9)
Latin America	25 (16.6)	17 (10.7)
Rest of World§	46 (30.5)	58 (36.5)

BMI=body mass index; COPD=chronic obstructive pulmonary disease; CrCl=creatinine clearance; LEAP=Lefamulin Evaluation Against Pneumonia; LEF=lefamulin; MOX=moxifloxacin; PORT=Pneumonia Outcomes Research Team; SIRS=systemic inflammatory response syndrome; WBC=white blood cell (count).
*PORT risk class was calculated programmatically using data obtained at the site and reported in the electronic case report form and was not always consistent with the site-reported PORT risk class used for enrollment/stratification; as a result, 3 patients with PORT risk class I (LEF, *n*=1; MOX, *n*=2), including 1 outpatient in the MOX group, and 3 patients with PORT risk class V (LEF, *n*=1; MOX, *n*=2), including 1 outpatient in each treatment group, were enrolled in the LEAP 2 study.
†Defined as confusion of new onset, blood urea nitrogen >19 mg/dL, respiratory rate ≥30 breaths/min, systolic blood pressure <90 mm Hg or diastolic blood pressure ≤60 mm Hg, and age ≥65 years.
‡Defined as having ≥2 of the following 4 criteria at baseline: temperature <36°C or >38°C; heart rate >90 bpm; respiratory rate >20 breaths/min; and WBC <4000 cells/mm^3, WBC >12,000 cells/mm^3, or immature polymorphonuclear neutrophils >10%.
§Philippines, South Africa, and South Korea.

Efficacy

- ECR rates and IACR success rates at TOC were high and similar in both treatment groups among all outpatients, including among those with PORT risk class III or IV and CURB-65 score 2 or 3 **(Figure 2)**
 - The ECR rate was 88% (45/51) in the LEF group and 94% (46/49) in the MOX group for patients with PORT risk class III and 86% (24/28) in both treatment groups for patients with CURB-65 score 2
 - The IACR success rate was 92% (47/51) and 94% (46/49), respectively, for patients with PORT risk class III and 89% (25/28) and 86% (24/28) for patients with CURB-65 score 2
- LEF demonstrated high response rates among outpatients infected by common CABP pathogens **(Figure 3)**, including among patients with polymicrobial infections (ECR: LEF, 78% [18/23]; MOX, 91% [21/23]. IACR at TOC: 87% [20/23] and 83% [19/23], respectively)
- LEF demonstrated high response rates among outpatients with comorbidities such as advanced age, smoking history, history of hypertension, underlying COPD or asthma, and underlying diabetes **(Figure 4)**

Figure 2. ECR and IACR at TOC in Outpatients

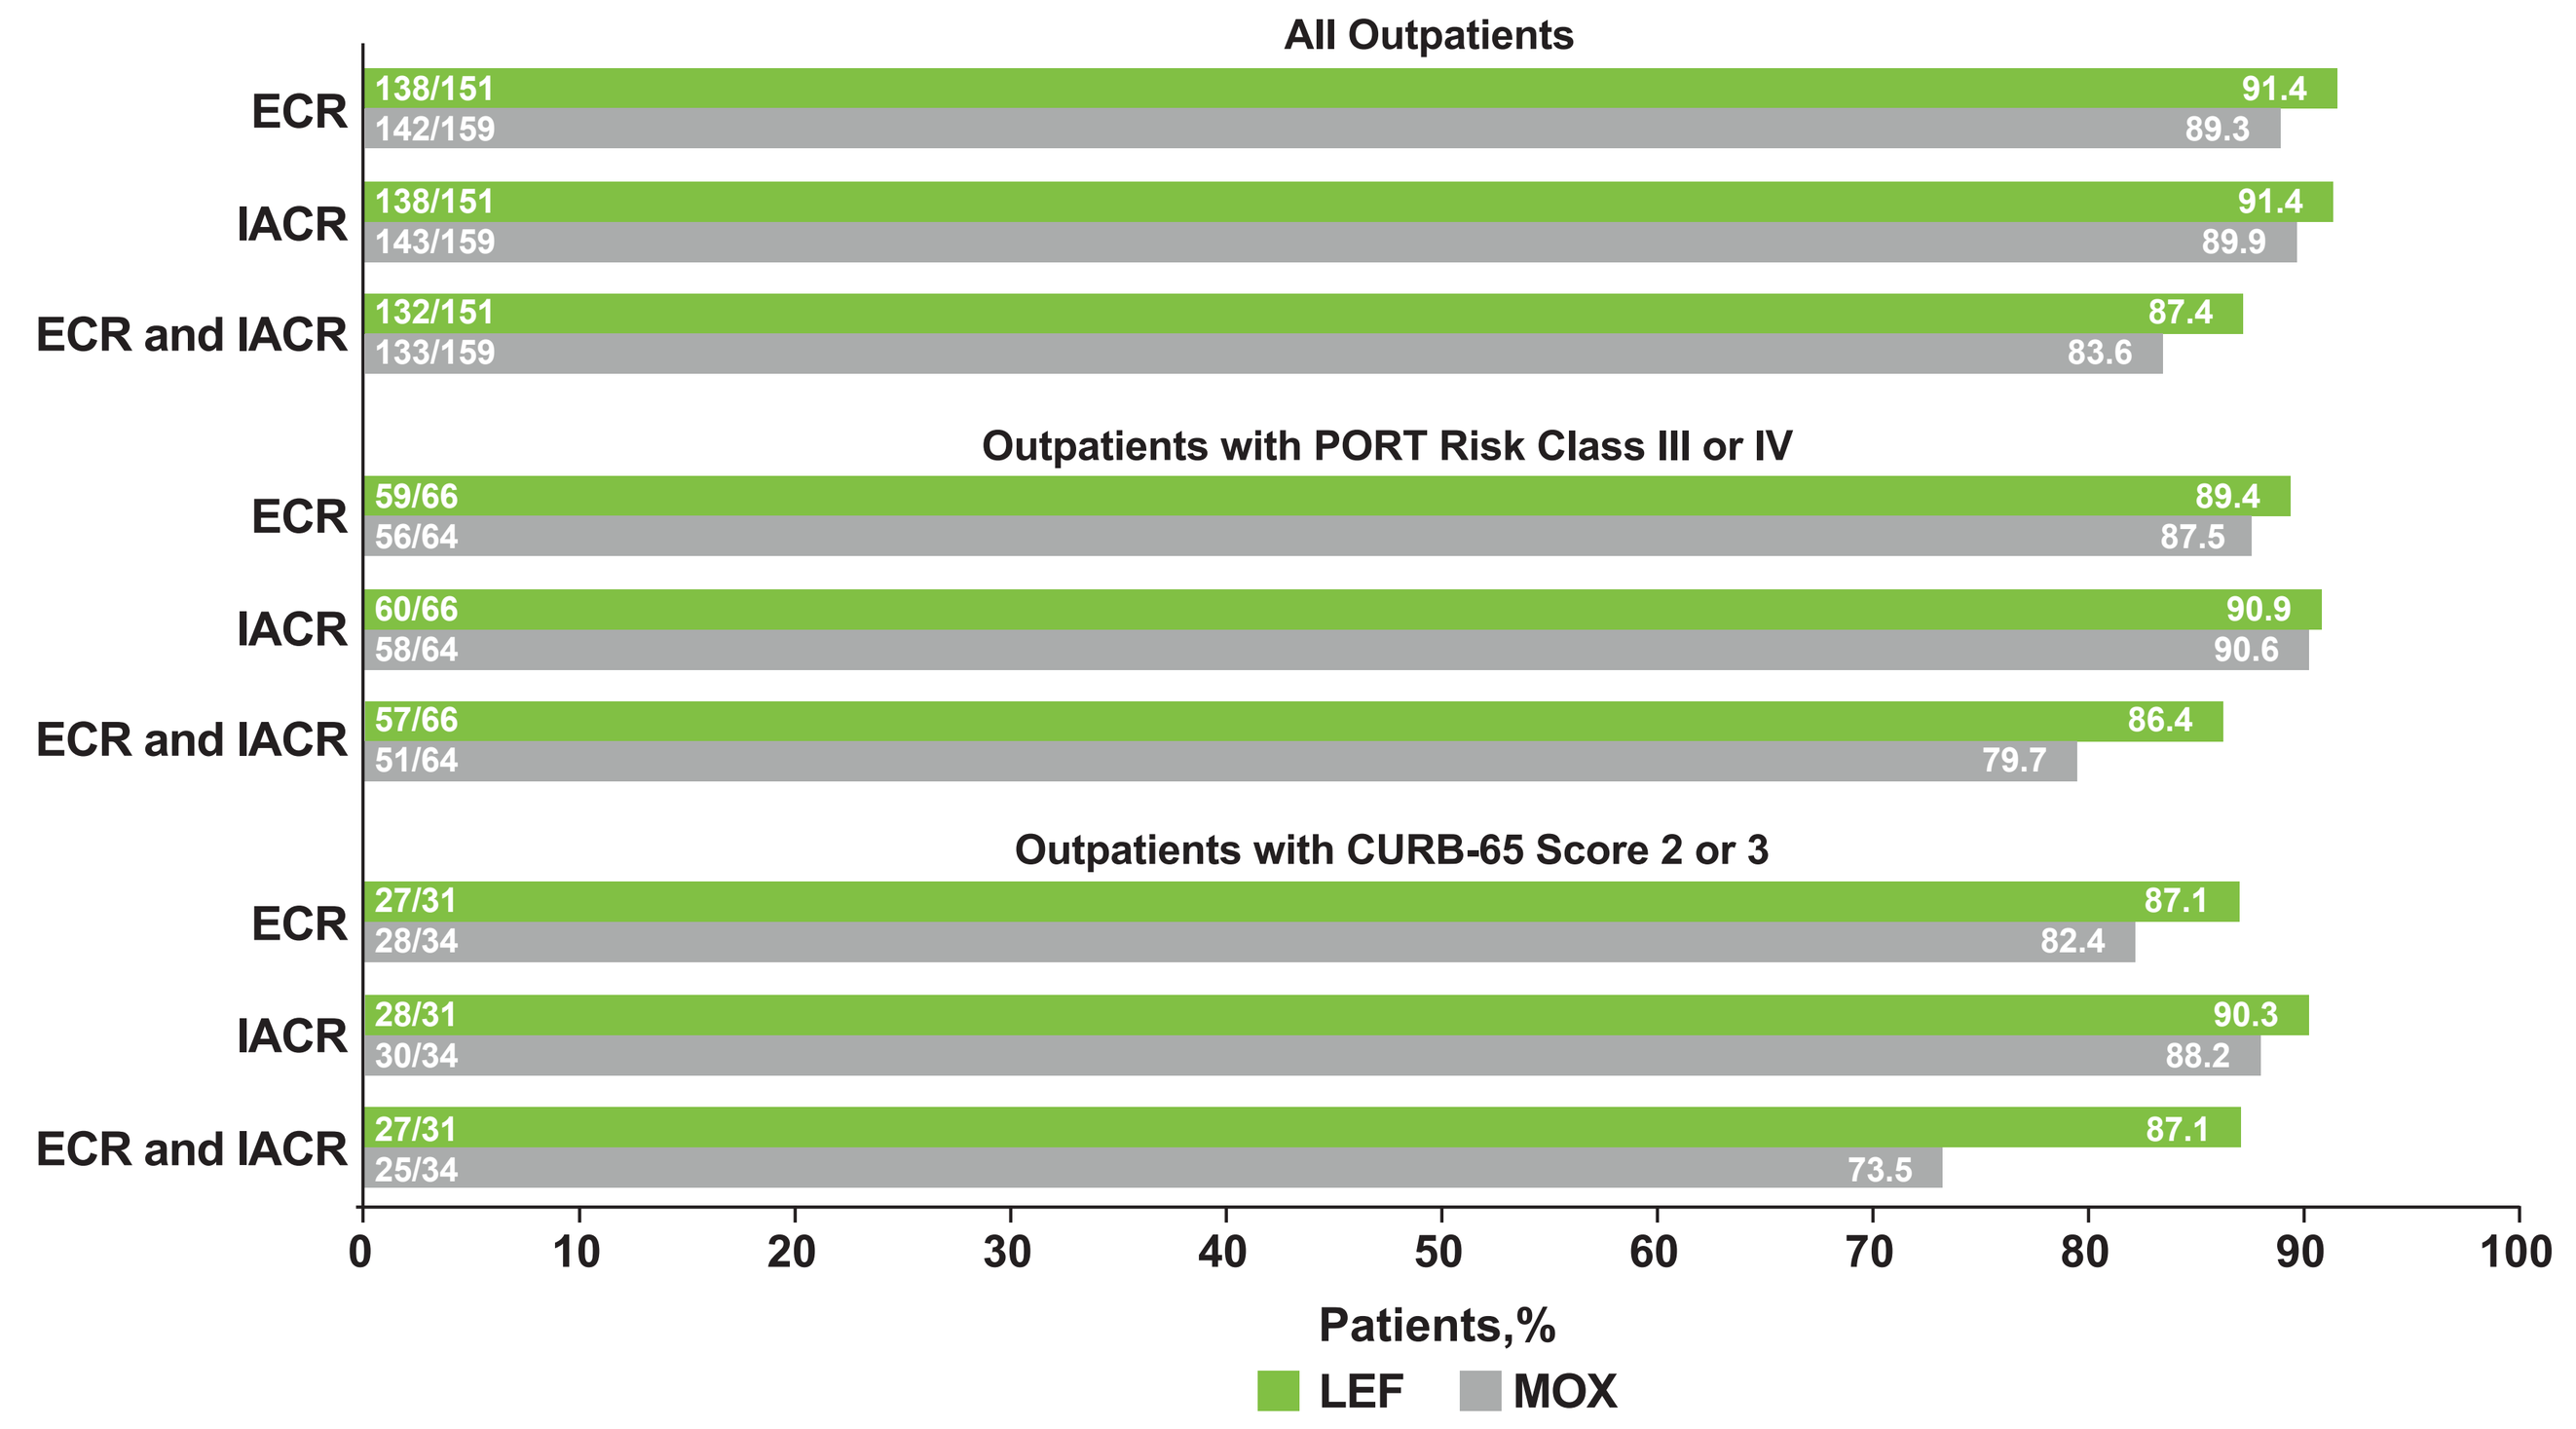
All Outpatients
ECR 138/151 91.4 142/159 89.3
IACR 138/151 91.4 143/159 89.9
ECR and IACR 132/151 87.4 133/159 83.6
Outpatients with PORT Risk Class III or IV
ECR 59/66 89.4 56/64 87.5
IACR 60/66 90.9 58/64 90.6
ECR and IACR 57/66 86.4 51/64 79.7
Outpatients with CURB-65 Score 2 or 3
ECR 27/31 87.1 28/34 82.4
IACR 28/31 90.3 30/34 88.2
ECR and IACR 27/31 87.1 25/34 73.5
0 10 20 30 40 50 60 70 80 90 100
Patients,%
LEF MOX

CURB-65=confusion of new onset, blood urea nitrogen >19 mg/dL, respiratory rate ≥30 breaths/min, systolic blood pressure <90 mm Hg or diastolic blood pressure ≤60 mm Hg, and age ≥65 years; ECR=early clinical response; IACR=investigator assessment of clinical response; LEF=lefamulin; MOX=moxifloxacin; PORT=Pneumonia Outcomes Research Team; TOC=test of cure.

Figure 3. ECR (A) and IACR at TOC (B) by Baseline Pathogen in Outpatients

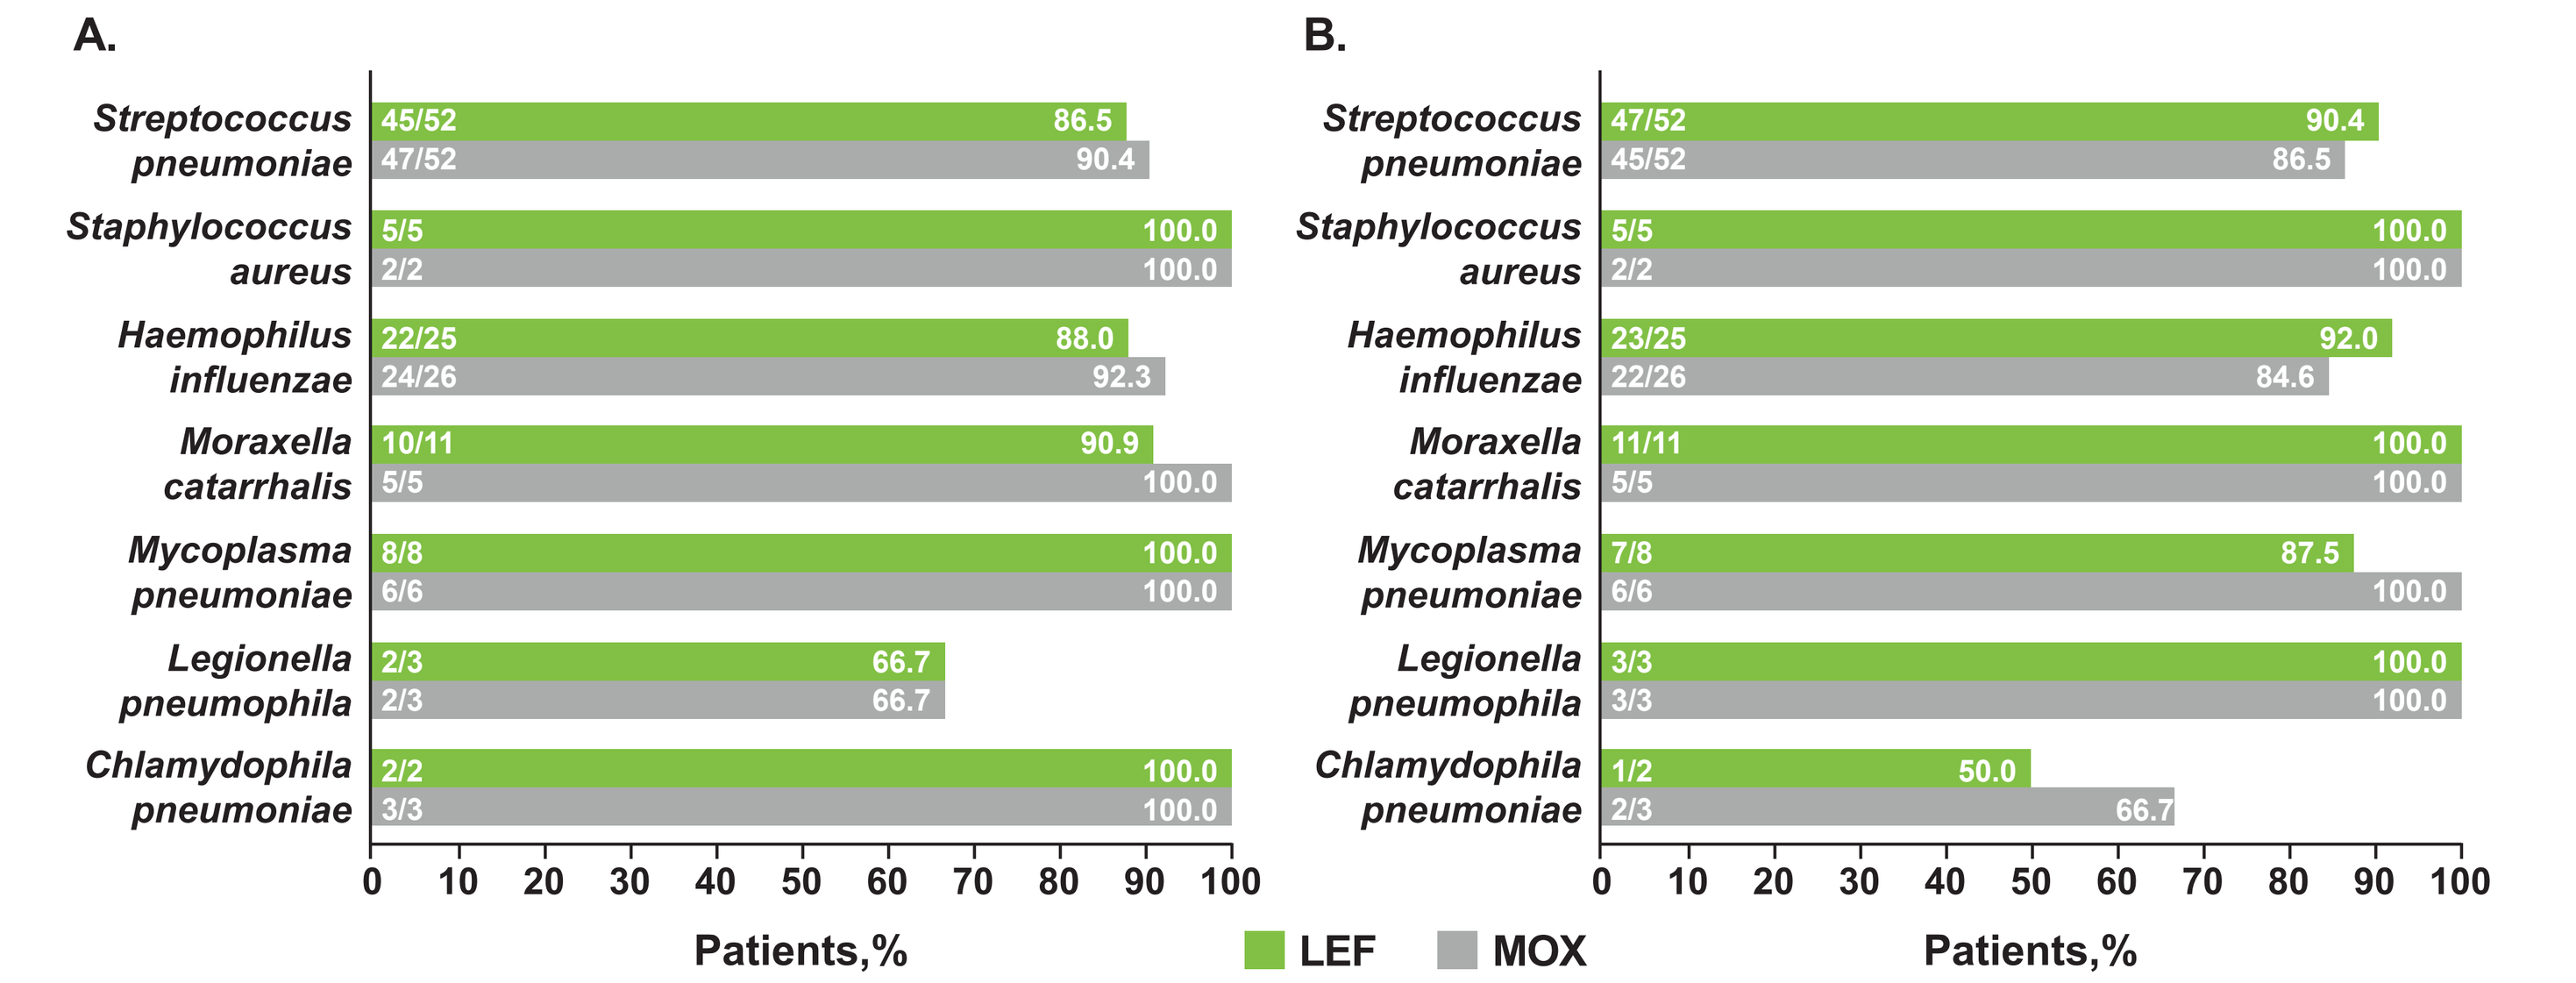
A.
Streptococcus pneumoniae 45/52 86.5 47/52 90.4
Staphylococcus aureus 5/5 100.0 2/2 100.0
Haemophilus influenzae 22/25 88.0 24/26 92.3
Moraxella catarrhalis 10/11 90.9 5/5 100.0
Mycoplasma pneumoniae 8/8 100.0 6/6 100.0
Legionella pneumophila 2/3 66.7 2/3 66.7
Chlamydophila pneumoniae 2/2 100.0 3/3 100.0
B.
Streptococcus pneumoniae 47/52 90.4 45/52 86.5
Staphylococcus aureus 5/5 100.0 2/2 100.0
Haemophilus influenzae 23/25 92.0 22/26 84.6
Moraxella catarrhalis 11/11 100.0 5/5 100.0
Mycoplasma pneumoniae 7/8 87.5 6/6 100.0
Legionella pneumophila 3/3 100.0 3/3 100.0
Chlamydophila pneumoniae 1/2 50.0 2/3 66.7
0 10 20 30 40 50 60 70 80 90 100
Patients,%
LEF MOX

ECR=early clinical response; IACR=investigator assessment of clinical response; LEF=lefamulin; MOX=moxifloxacin; TOC=test of cure.

Figure 4. ECR (A) and IACR at TOC (B) by Comorbidity* in Outpatients

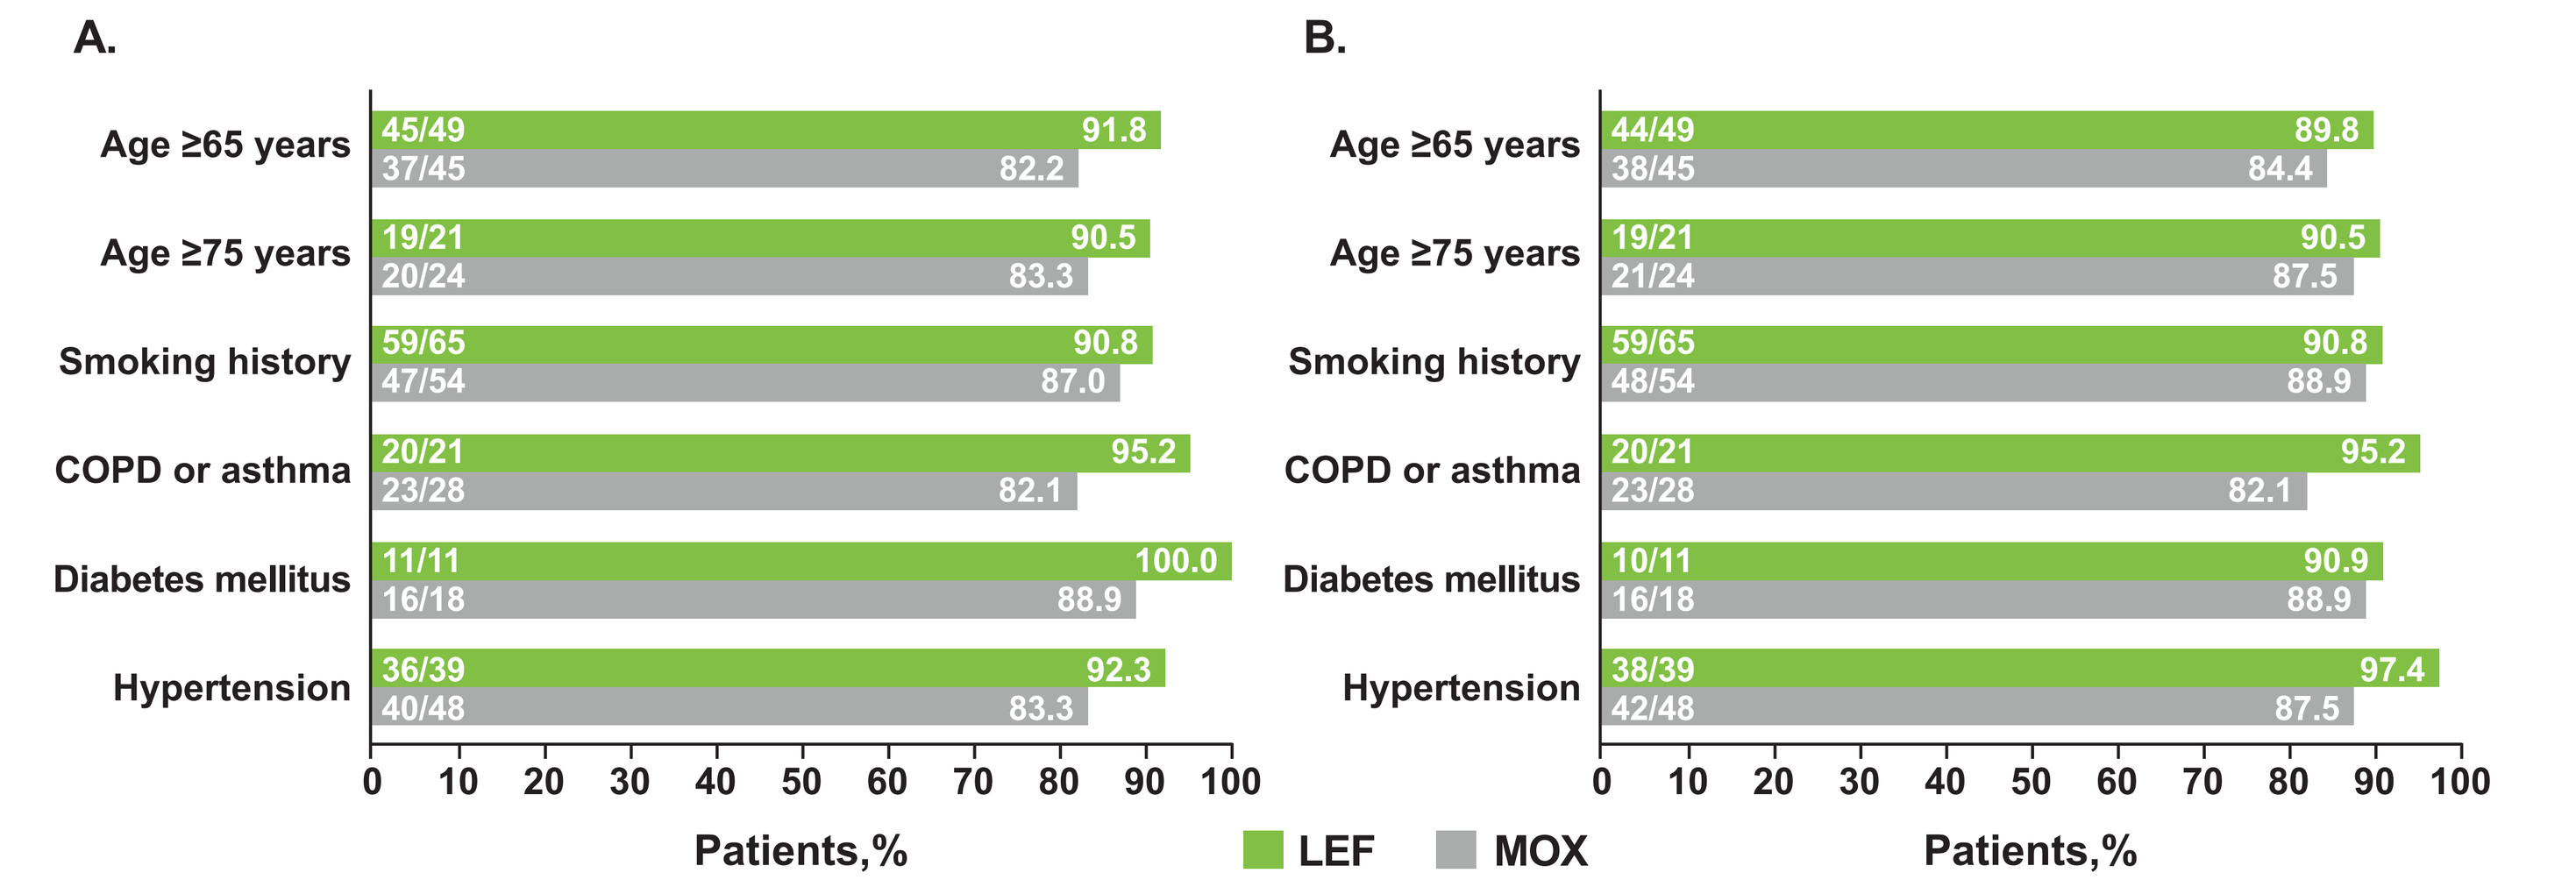
A.
Age ≥65 years 45/49 91.8 37/45 82.2
Age ≥75 years 19/21 90.5 20/24 83.3
Smoking history 59/65 90.8 47/54 87.0
COPD or asthma 20/21 95.2 23/28 82.1
Diabetes mellitus 11/11 100.0 16/18 88.9
Hypertension 36/39 92.3 40/48 83.3
B.
Age ≥65 years 44/49 89.8 38/45 84.4
Age ≥75 years 19/21 90.5 21/24 87.5
Smoking history 59/65 90.8 48/54 88.9
COPD or asthma 20/21 95.2 23/28 82.1
Diabetes mellitus 10/11 90.9 16/18 88.9
Hypertension 38/39 97.4 42/48 87.5
0 10 20 30 40 50 60 70 80 90 100
Patients,%
LEF MOX

COPD=chronic obstructive pulmonary disease; ECR=early clinical response; IACR=investigator assessment of clinical response; LEF=lefamulin; MOX=moxifloxacin; TOC=test of cure.
*A patient could have more than 1 comorbidity.

Safety

- TEAE rates were similar in both treatment groups, with low rates of TEAEs leading to study drug discontinuation **(Table 2)**
 - No one in the LEF outpatient group had a serious TEAE or was admitted for hospitalization during the study, compared with 5 (3%) serious TEAEs, including 2 deaths, in the MOX group
- The most common TEAEs in the LEF treatment group were gastrointestinal events **(Table 3)**, which were all mild to moderate in severity; 3 patients discontinued study drug due to a gastrointestinal TEAE (vomiting [LEF, *n*=1; MOX, *n*=1], abdominal pain upper [LEF, *n*=1])
 - Related TEAEs were generally reflective of overall TEAEs

Table 2. Overview of TEAEs in Outpatients

Event, *n* (%)	LEF (*n*=151)	MOX (*n*=159)
TEAE	52 (34.4)	48 (30.2)
Related TEAE	34 (22.5)	18 (11.3)
Serious TEAE	0	5 (3.1)*
Related serious TEAE	0	0
TEAE leading to DC of study drug	4 (2.6)	4 (2.5)
Related TEAE leading to DC of study drug	2 (1.3)	2 (1.3)
TEAE leading to death	0	2 (1.3)*

DC=discontinuation; LEF=lefamulin; MOX=moxifloxacin; TEAE=treatment-emergent adverse event.
*Serious TEAEs in the MOX group included 1 patient with worsening pneumonia on Day 4 leading to DC of study drug and hospitalization on Days 6–14 and 4 patients who completed study drug treatment (*n*=1 with acute cholecystitis on Day 18 leading to hospitalization on Days 18–21; *n*=1 with "death from natural causes" on Day 12; *n*=1 with angioedema on Day 3; and *n*=1 with cerebral infarction on Day 17 leading to hospitalization that day and then death on Day 18).

Table 3. TEAEs Reported in >1 Outpatient in Either Treatment Group

Preferred Term, *n* (%)	LEF (*n*=151)	MOX (*n*=159)
Diarrhea	29 (19.2)	3 (1.9)
Nausea	9 (6.0)	5 (3.1)
Vomiting	8 (5.3)	2 (1.3)
Headache	2 (1.3)	4 (2.5)
Dizziness	2 (1.3)	3 (1.9)
Hypertension	2 (1.3)	3 (1.9)
Abdominal pain	2 (1.3)	2 (1.3)
Blood creatine phosphokinase increased	2 (1.3)	1 (0.6)
Gastritis	2 (1.3)	1 (0.6)
Urinary tract infection	1 (0.7)	4 (2.5)
Alanine aminotransferase increased	1 (0.7)	2 (1.3)
Aspartate aminotransferase increased	1 (0.7)	2 (1.3)
Insomnia	0	2 (1.3)

LEF=lefamulin; MOX=moxifloxacin; TEAE=treatment-emergent adverse event.

CONCLUSIONS

- These data from LEAP 2 suggest that patients with PORT risk class III or IV, including those with polymicrobial infections or difficult-to-treat pathogens such as *Legionella pneumophila*, can be effectively managed as outpatients with 5 days of oral LEF as an alternative to fluoroquinolones for the treatment of CABP
- Oral LEF 5-day therapy was generally well tolerated, with low rates of study drug discontinuation due to TEAEs; no one in the LEF outpatient group had a serious TEAE or was admitted for hospitalization during the study
- LEF is a promising new monotherapy option for the outpatient treatment of CABP

REFERENCES

(1) Mandell LA, et al. *Clin Infect Dis.* 2007;44(suppl 2):S27-S72.
(2) Fine MJ, et al. *N Engl J Med.* 1997;336(4):243-250.
(3) Lim WS, et al. *Thorax.* 2003;58(5):377-382.
(4) Society for Healthcare Epidemiology of America, et al. *Infect Control Hosp Epidemiol.* 2012;33(4):322-327.
(5) Barlam TF, et al. *Clin Infect Dis.* 2016;62(10):e51-e77.
(6) Dellit TH, et al. *Clin Infect Dis.* 2007;44(2):159-177.
(7) Xenleta™ (lefamulin). Full Prescribing Information, Nabriva Therapeutics US, Inc., King of Prussia, PA, 2019.
(8) File TM Jr, et al. *Clin Infect Dis.* 2019; doi: 10.1093/cid/ciz090:[Epub ahead of print].
(9) Alexander E, et al. *JAMA.* 2019; doi: 10.1001/jama.2019.15468:[Epub ahead of print].

Acknowledgments

Funding for development of this poster was provided by Nabriva Therapeutics to C4 MedSolutions, LLC (Yardley, PA), a CHC Group company.

Disclosures

Jennifer Schranz, Elizabeth Alexander, David Fitts, David Mariano, Andrew Meads, and Steven P. Gelone are employees of/stockholders in Nabriva Therapeutics plc. Christian Sandrock has served as a consultant for Allergan and Nabriva Therapeutics, has received grants from the National Institutes of Health and the Health Resources & Services Administration, and has received nonfinancial support from the State of California. Gregory J. Moran has received grants from ContraFect and Nabriva Therapeutics.

Scan this QR code with your electronic device to receive a PDF file of the poster or visit posters.chcinc.com/IDWeek2019_LEAP2_Outpatients

IDWeek 2019: October 2–6, Washington, DC, USA